Transcranial Electric Stimulation (tES) in Warfighters with Moderate-to-Severe Insomnia

Amanda Studnicki[1], Ethan Cheraghpour[1], Angeliki Pollatou[1], Heather Decot[1], Grant Tays[2], Elizabeth Metzger[1], Stephen B. Simons[3], J. Kent Werner, Jr.[1]
1: Uniformed Services University, 2: Walter Reed Army Institute of Research, 3: Teledyne Scientific & Imaging

Sleep. Neuroscience. Technology.

Key Takeaway #1:

tES worn 30-min before bed elicited a therapeutic effect on insomnia symptoms compared to baseline, however the effects were not frequency-specific.

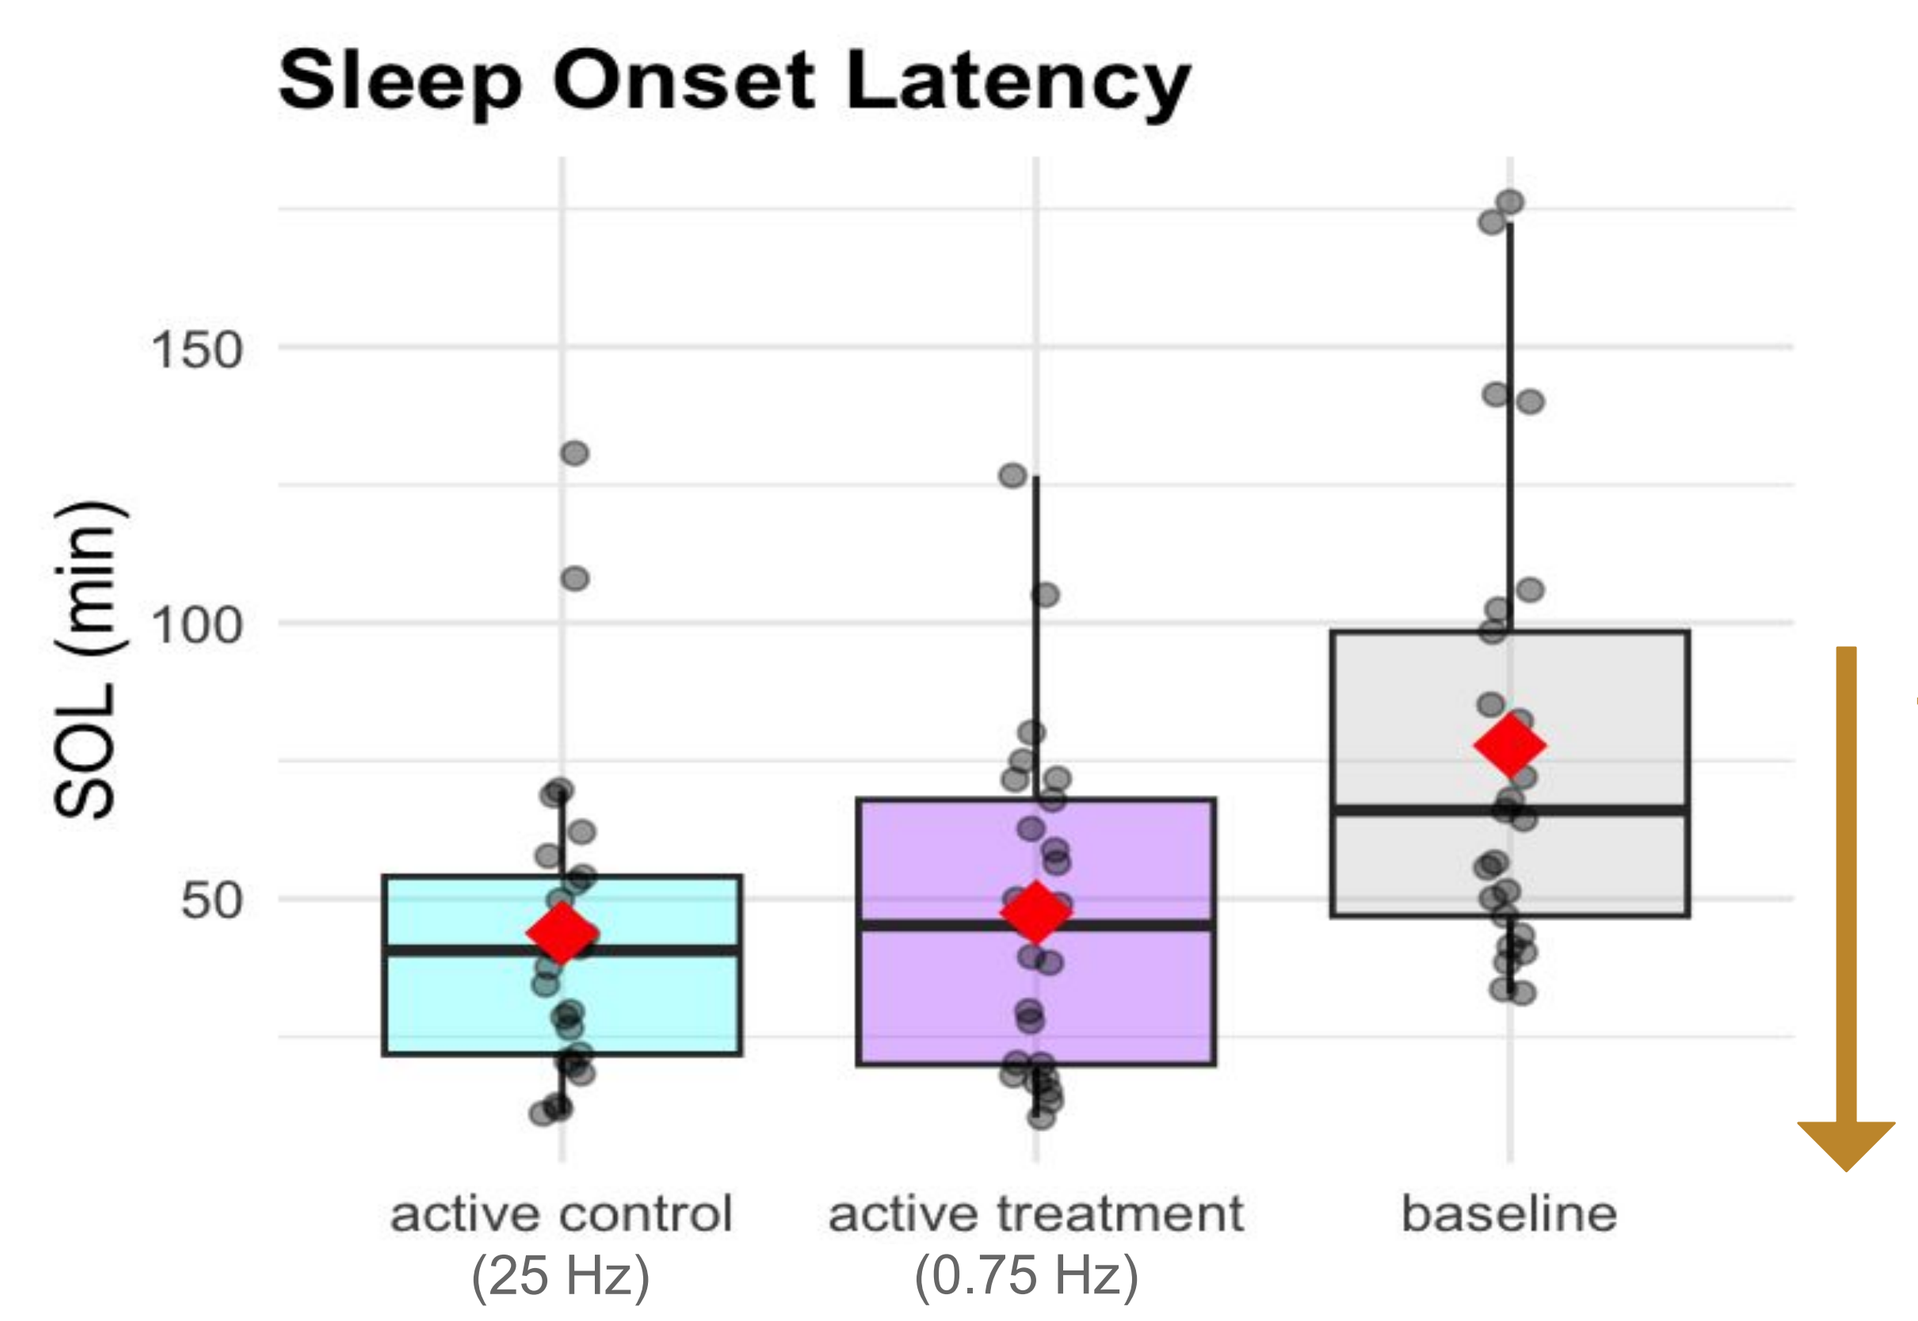
Sleep Onset Latency
SOL (min)
150
100
50
active control (25 Hz)
active treatment (0.75 Hz)
baseline

Figure 1. Median + IQR (box plot) and mean (red diamond) sleep onset latency for N=25 participants.

therapeutic effect (participants fell asleep faster while wearing the headset)

Introduction

- Insomnia is prevalent in the military, especially when returning home from deployment [1]
- Sleep medications can be limited by side effects and recurring costs [2]
- TES shows promise as a safe, non-invasive alternative to improve sleep quality, possibly by enhancing frontal slow wave activity before bed [3]

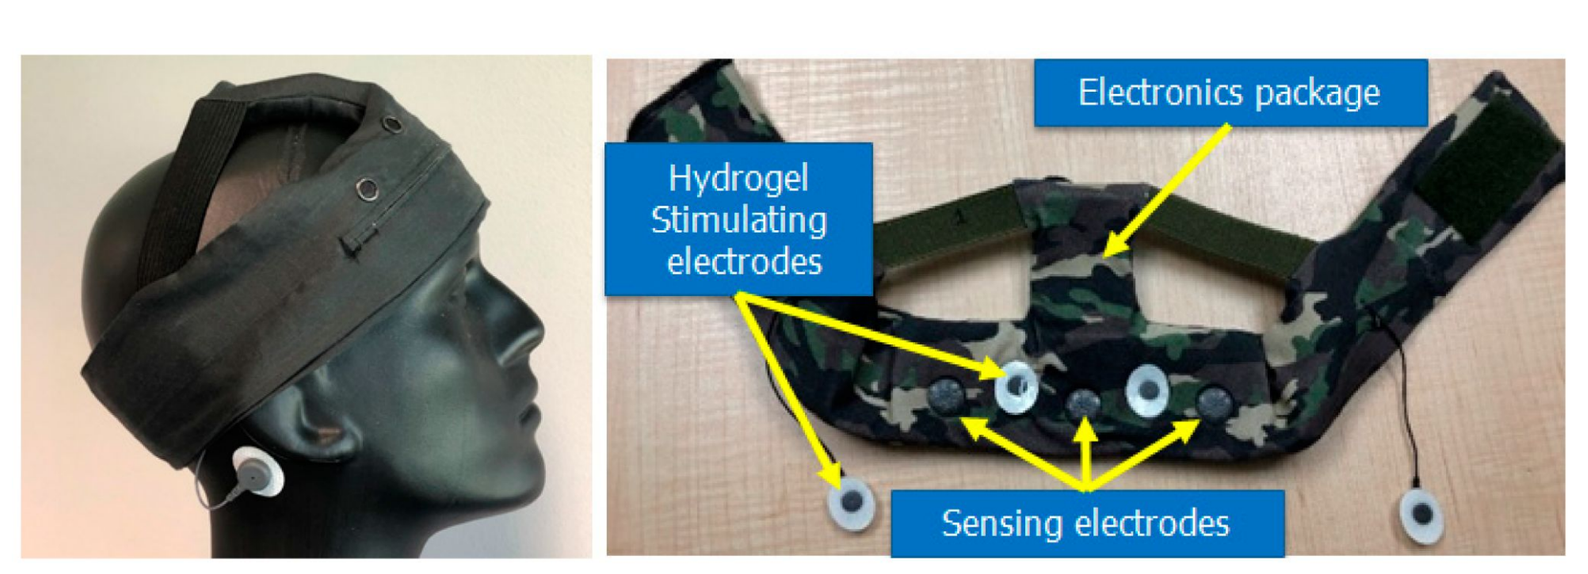
Electronics package
Hydrogel Stimulating electrodes
Sensing electrodes

Figure 2. Teledyne's PeakSleep tES headset has 3 frontal EEG electrodes and 2 stimulating electrodes [3]

We aimed to validate the efficacy of Teledyne Scientific's PeakSleep active stimulation headset to reduce insomnia in a military cohort.

Methods

Table 1. Metrics of Interest

Metric:	Calculation:
Sleep Onset Latency (SOL)	sleep start - latest bedtime
Insomnia Severity Index (ISI)	aggregate score (self report)
Wake After Sleep Onset (WASO)	wake between sleep start & stop
Frontal Slow Wave Activity	Δ Fpz delta power

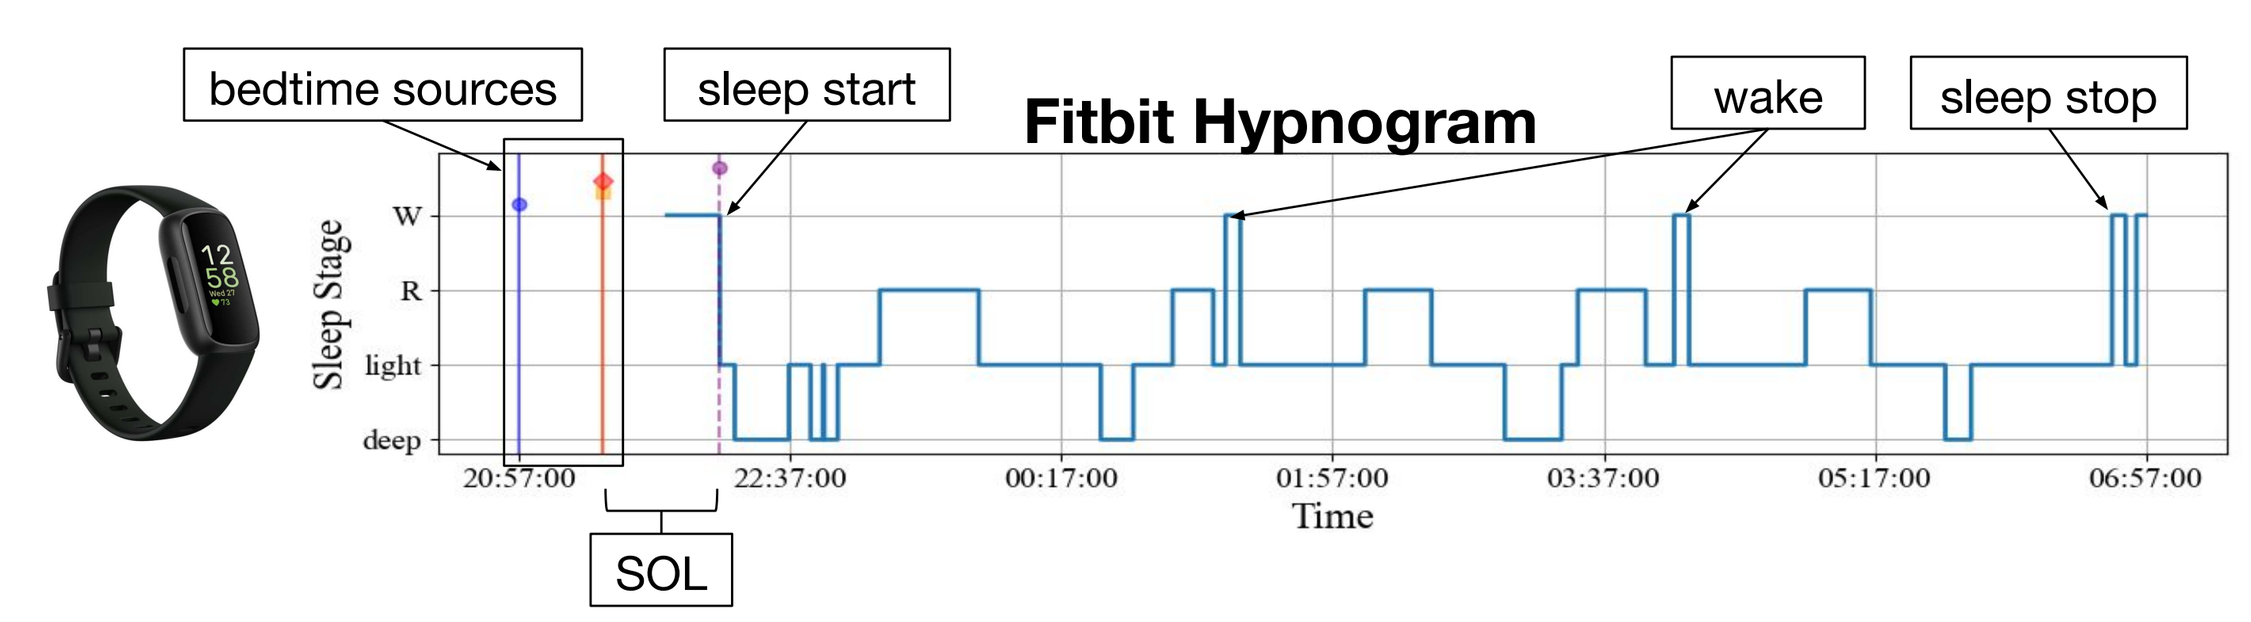
bedtime sources
sleep start
Fitbit Hypnogram
wake
sleep stop
Sleep Stage
W
R
light
deep
20:57:00
22:37:00
00:17:00
01:57:00
03:37:00
05:17:00
06:57:00
Time
SOL

Figure 3. Hypnogram in the Fitbit Inspire 3 used to calculate SOL and WASO.

Study design:

- Double-blind, randomized, crossover, phase II clinical trial in military personnel with moderate-to-severe insomnia (ISI ≥ 15; SOL ≥ 30 min.)

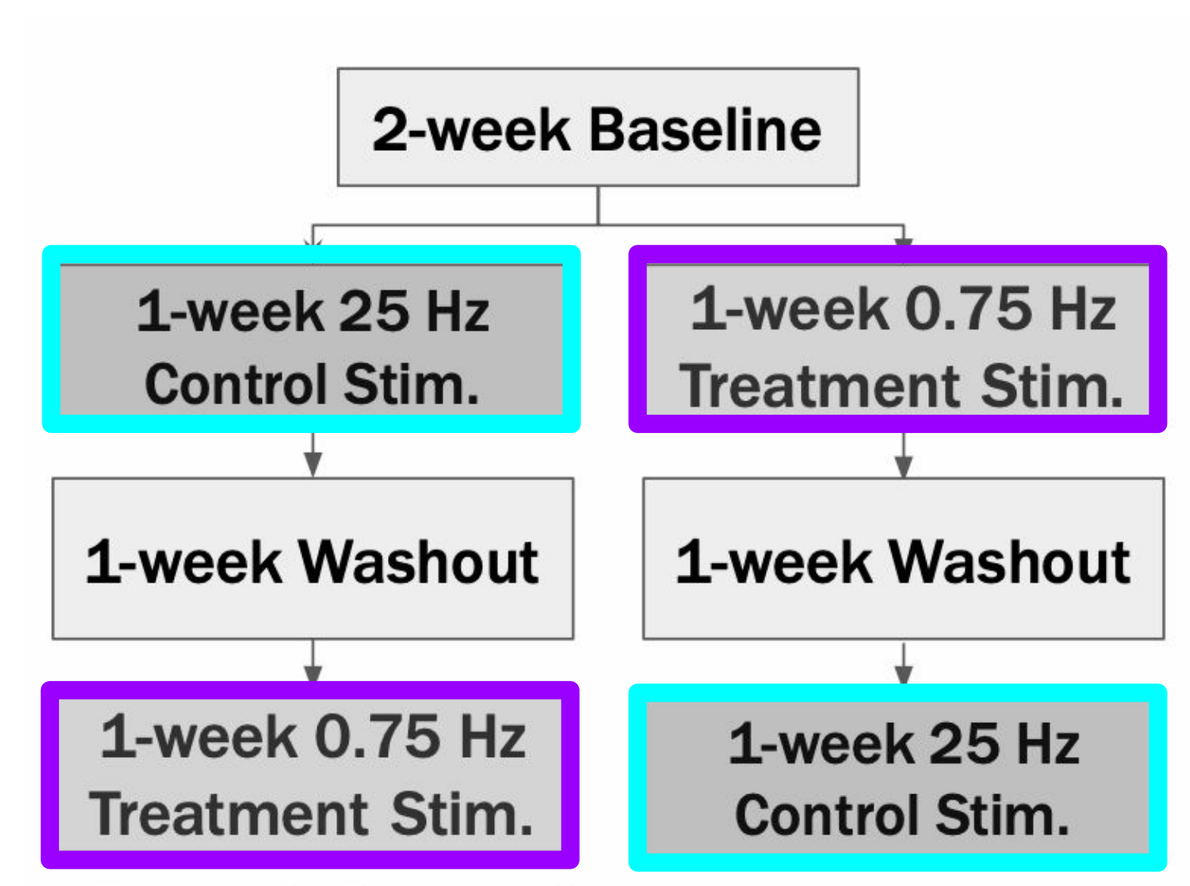
2-week Baseline
1-week 25 Hz Control Stim.
1-week 0.75 Hz Treatment Stim.
1-week Washout
1-week Washout
1-week 0.75 Hz Treatment Stim.
1-week 25 Hz Control Stim.

Figure 4. Data collection flow diagram of the ~5-week paradigm, participants counter-balanced by stimulation order.

Stimulation:

- Headset worn immediately before trying to fall asleep
- 30 min: 60s EEG baseline → 100 trials (8s ON + 10s OFF)
- Amplitude scaled by real-time measure of skin impedance

Results

- N = 25 participants completed the full 5-week paradigm and had at least 3 nights of viable data in baseline and stimulation sessions (age 43 +/- 12 years, 13 females, 13 reported using sleep medication prior to participation)

Table 2. Summary of Outcomes

Characteristic	active control N = 25[1]	active treatment N = 25[1]	baseline N = 25[1]	p-value[2]
SOL (min)	44 (±29)	47 (±30)	78 (±42)	0.002
ISI	16 (±6)	16 (±5)	20 (±3)	0.014
WASO (min)	37 (±13)	35 (±12)	37 (±10)	0.9
TST (min)	405 (±73)	394 (±61)	397 (±53)	0.8
Awakenings	19.9 (±6.1)	19.4 (±5.1)	20.4 (±5.2)	0.9
Light Sleep %	0.57 (±0.06)	0.57 (±0.06)	0.58 (±0.05)	0.9
Deep Sleep %	0.15 (±0.04)	0.16 (±0.03)	0.15 (±0.03)	>0.9
REM Sleep %	0.19 (±0.05)	0.18 (±0.06)	0.18 (±0.04)	0.7
Resting HR (bpm)	67 (±10)	70 (±10)	68 (±7)	0.4
PHQ-9	8 (±5)	8 (±4)	9 (±5)	0.8

[1] Mean (±SD)
[2] Kruskal-Wallis rank sum test

Table 2. SOL and ISI dropped in both active treatment and active control conditions, as compared to baseline

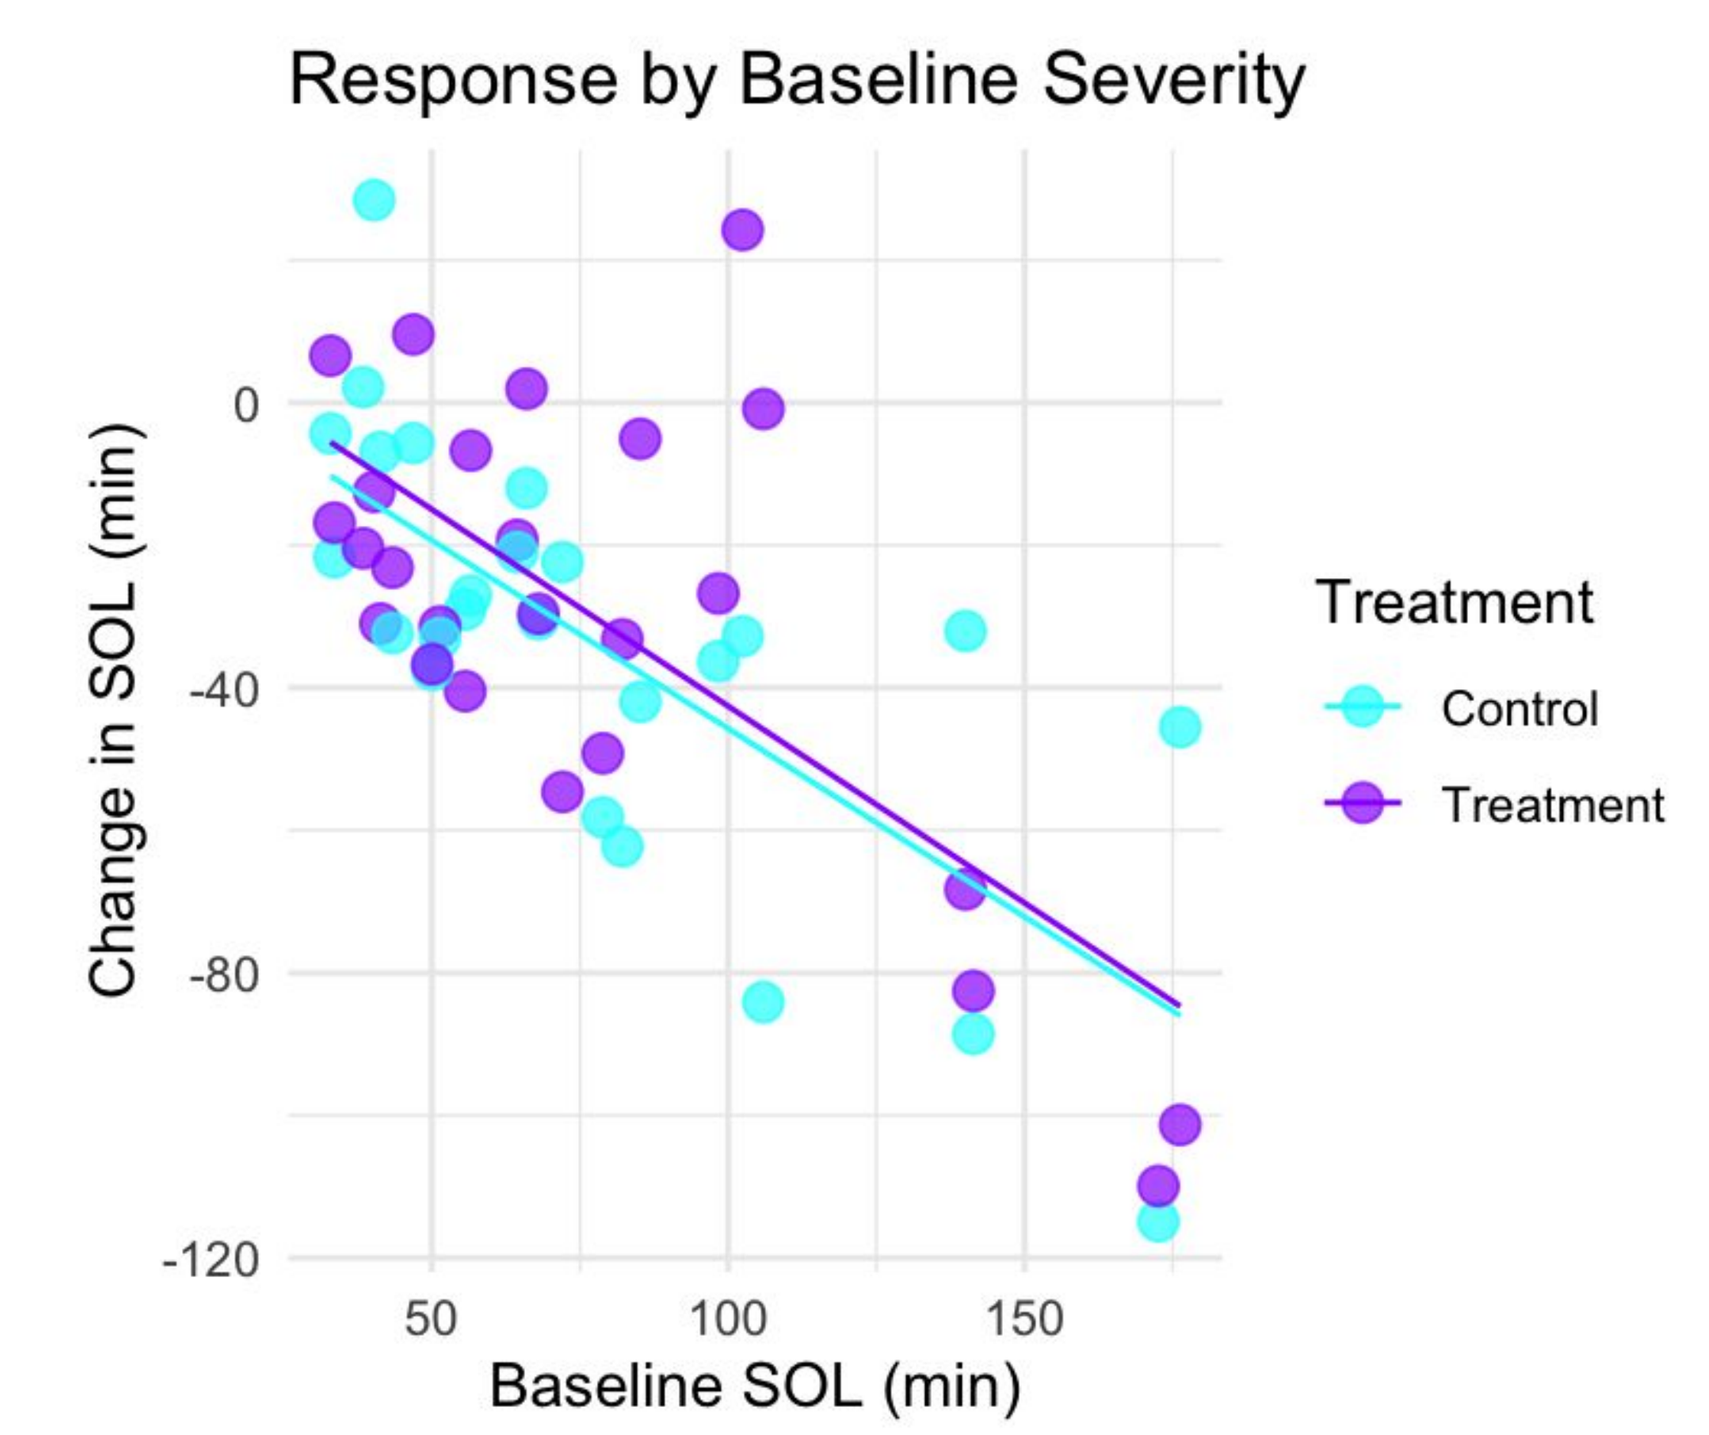
Response by Baseline Severity
Change in SOL (min)
0
-40
-80
-120
50
100
150
Baseline SOL (min)
Treatment
Control
Treatment

Figure 5. Subjects with severe insomnia (high SOL) had greatest change in SOL, regardless of stim type (cyan: 25 Hz vs purple: 0.75 Hz). Active Control Pearson's R value = -0.723 ($p < 0.001$). Active Treatment Pearson's R value = -0.694 ($p = 0.001$).

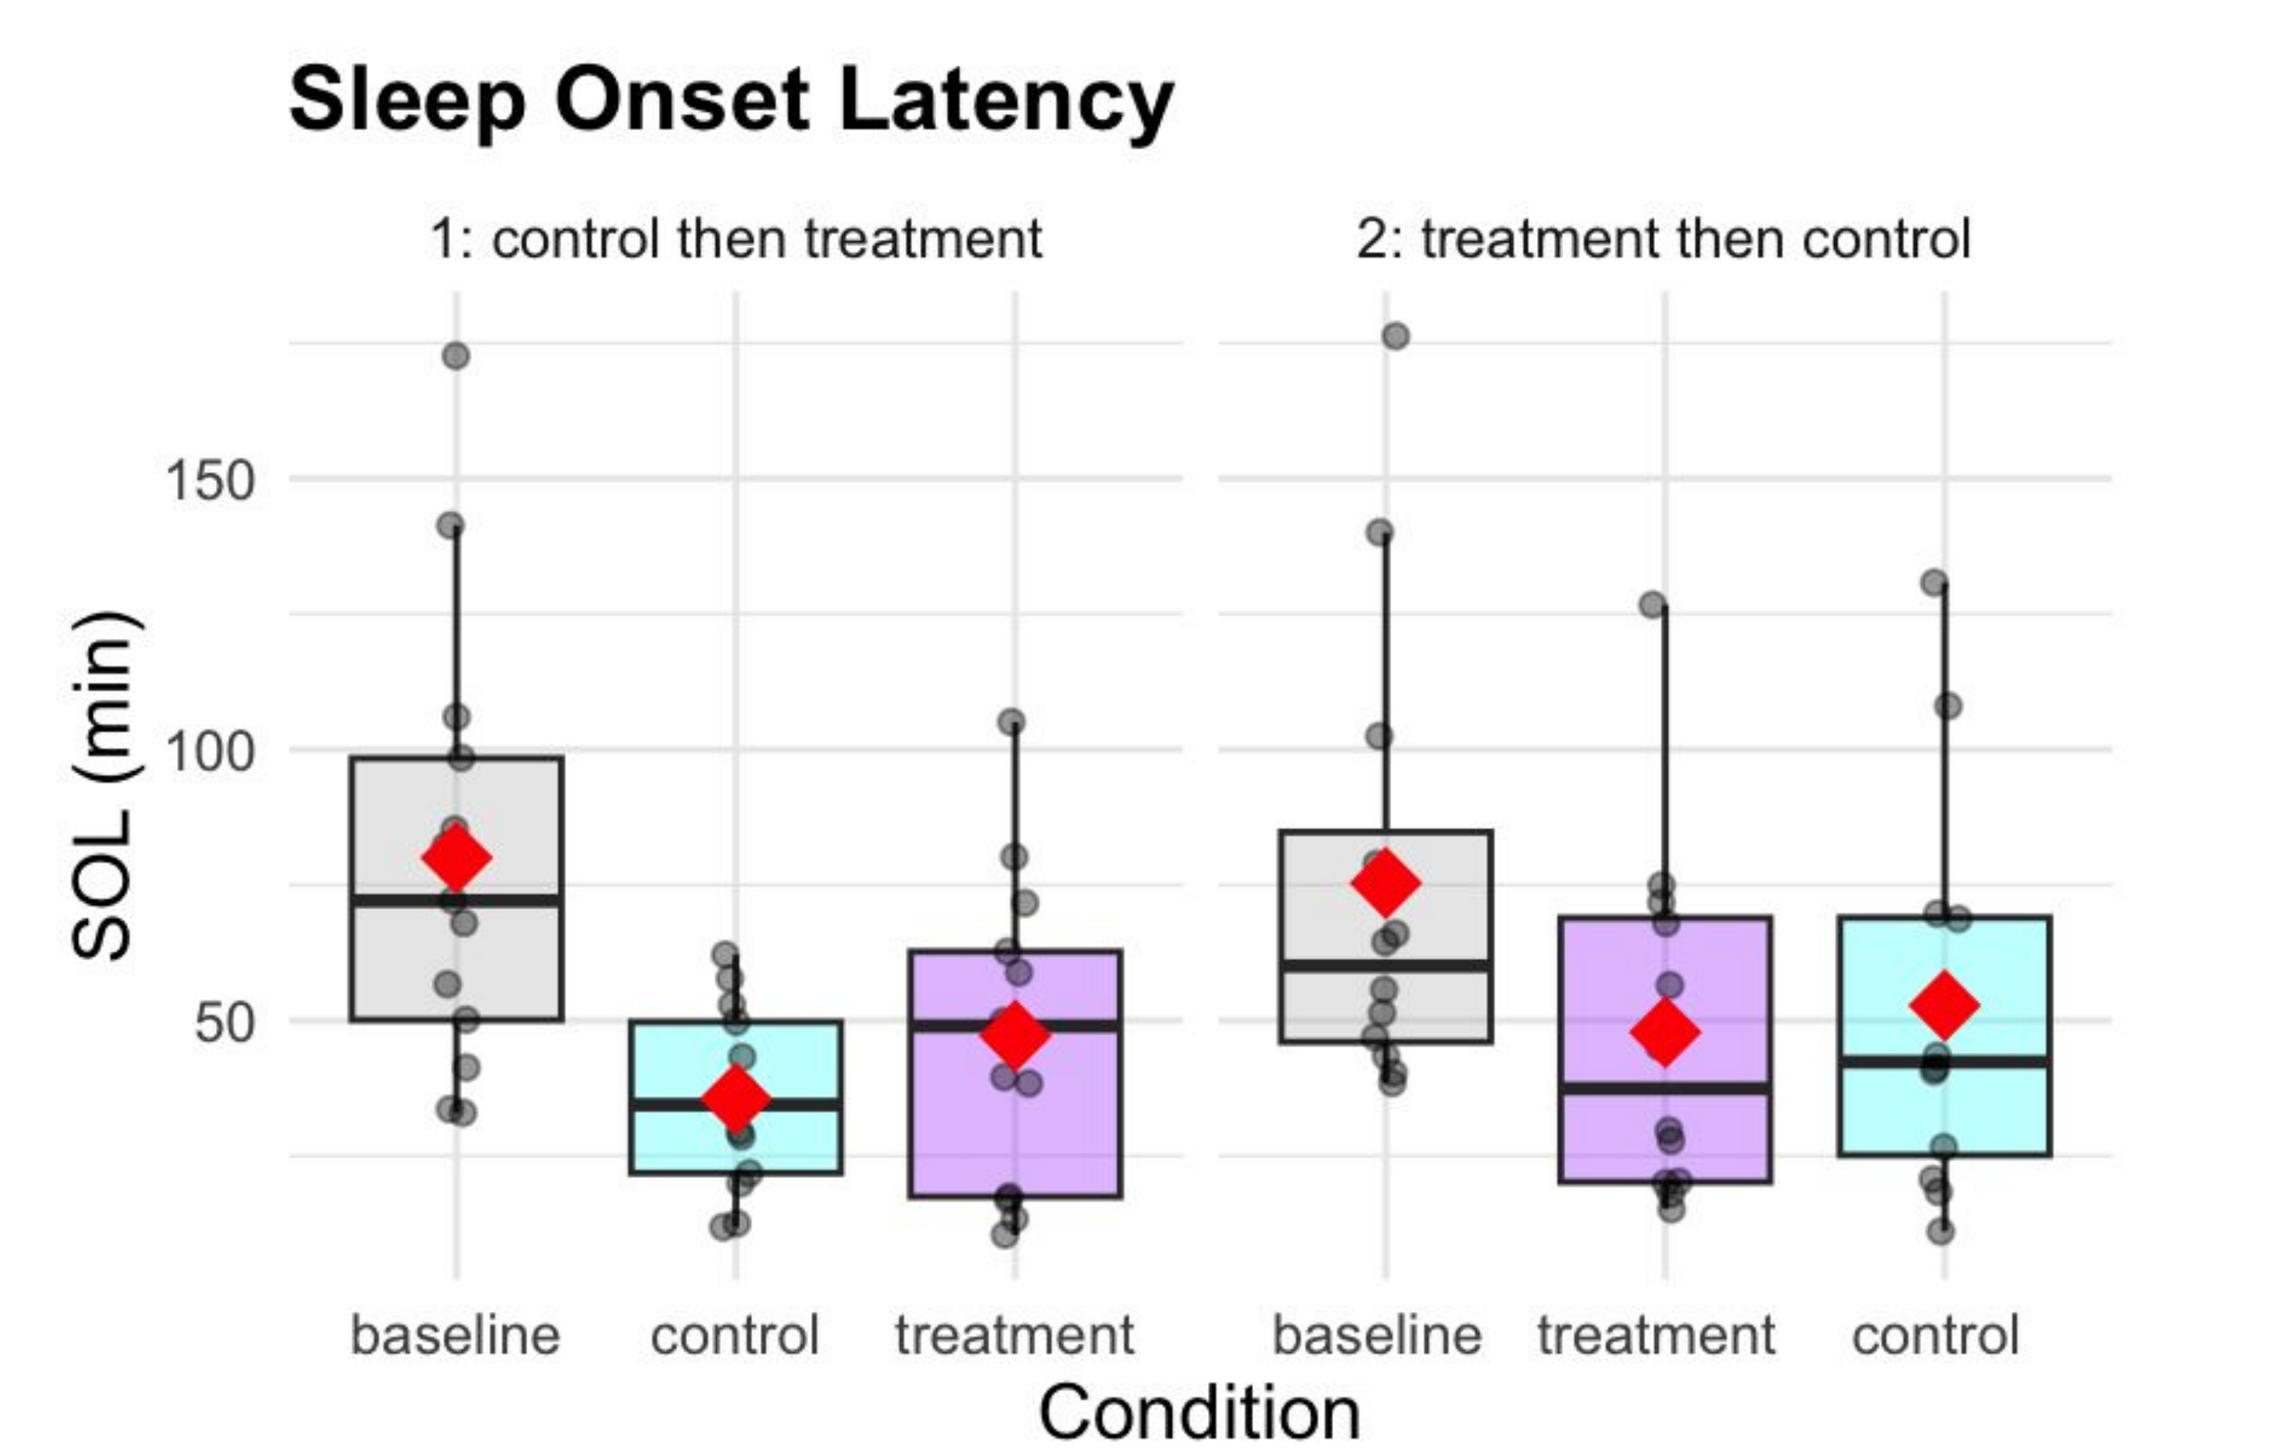
Sleep Onset Latency
1: control then treatment
2: treatment then control
SOL (min)
150
100
50
baseline
control
treatment
baseline
treatment
control
Condition

Figure 6. Both Order 1 (25 Hz control → 0.75 Hz treatment) and Order 2 (0.75 Hz treatment → 25 Hz control) showed reductions in SOL compared to baseline.

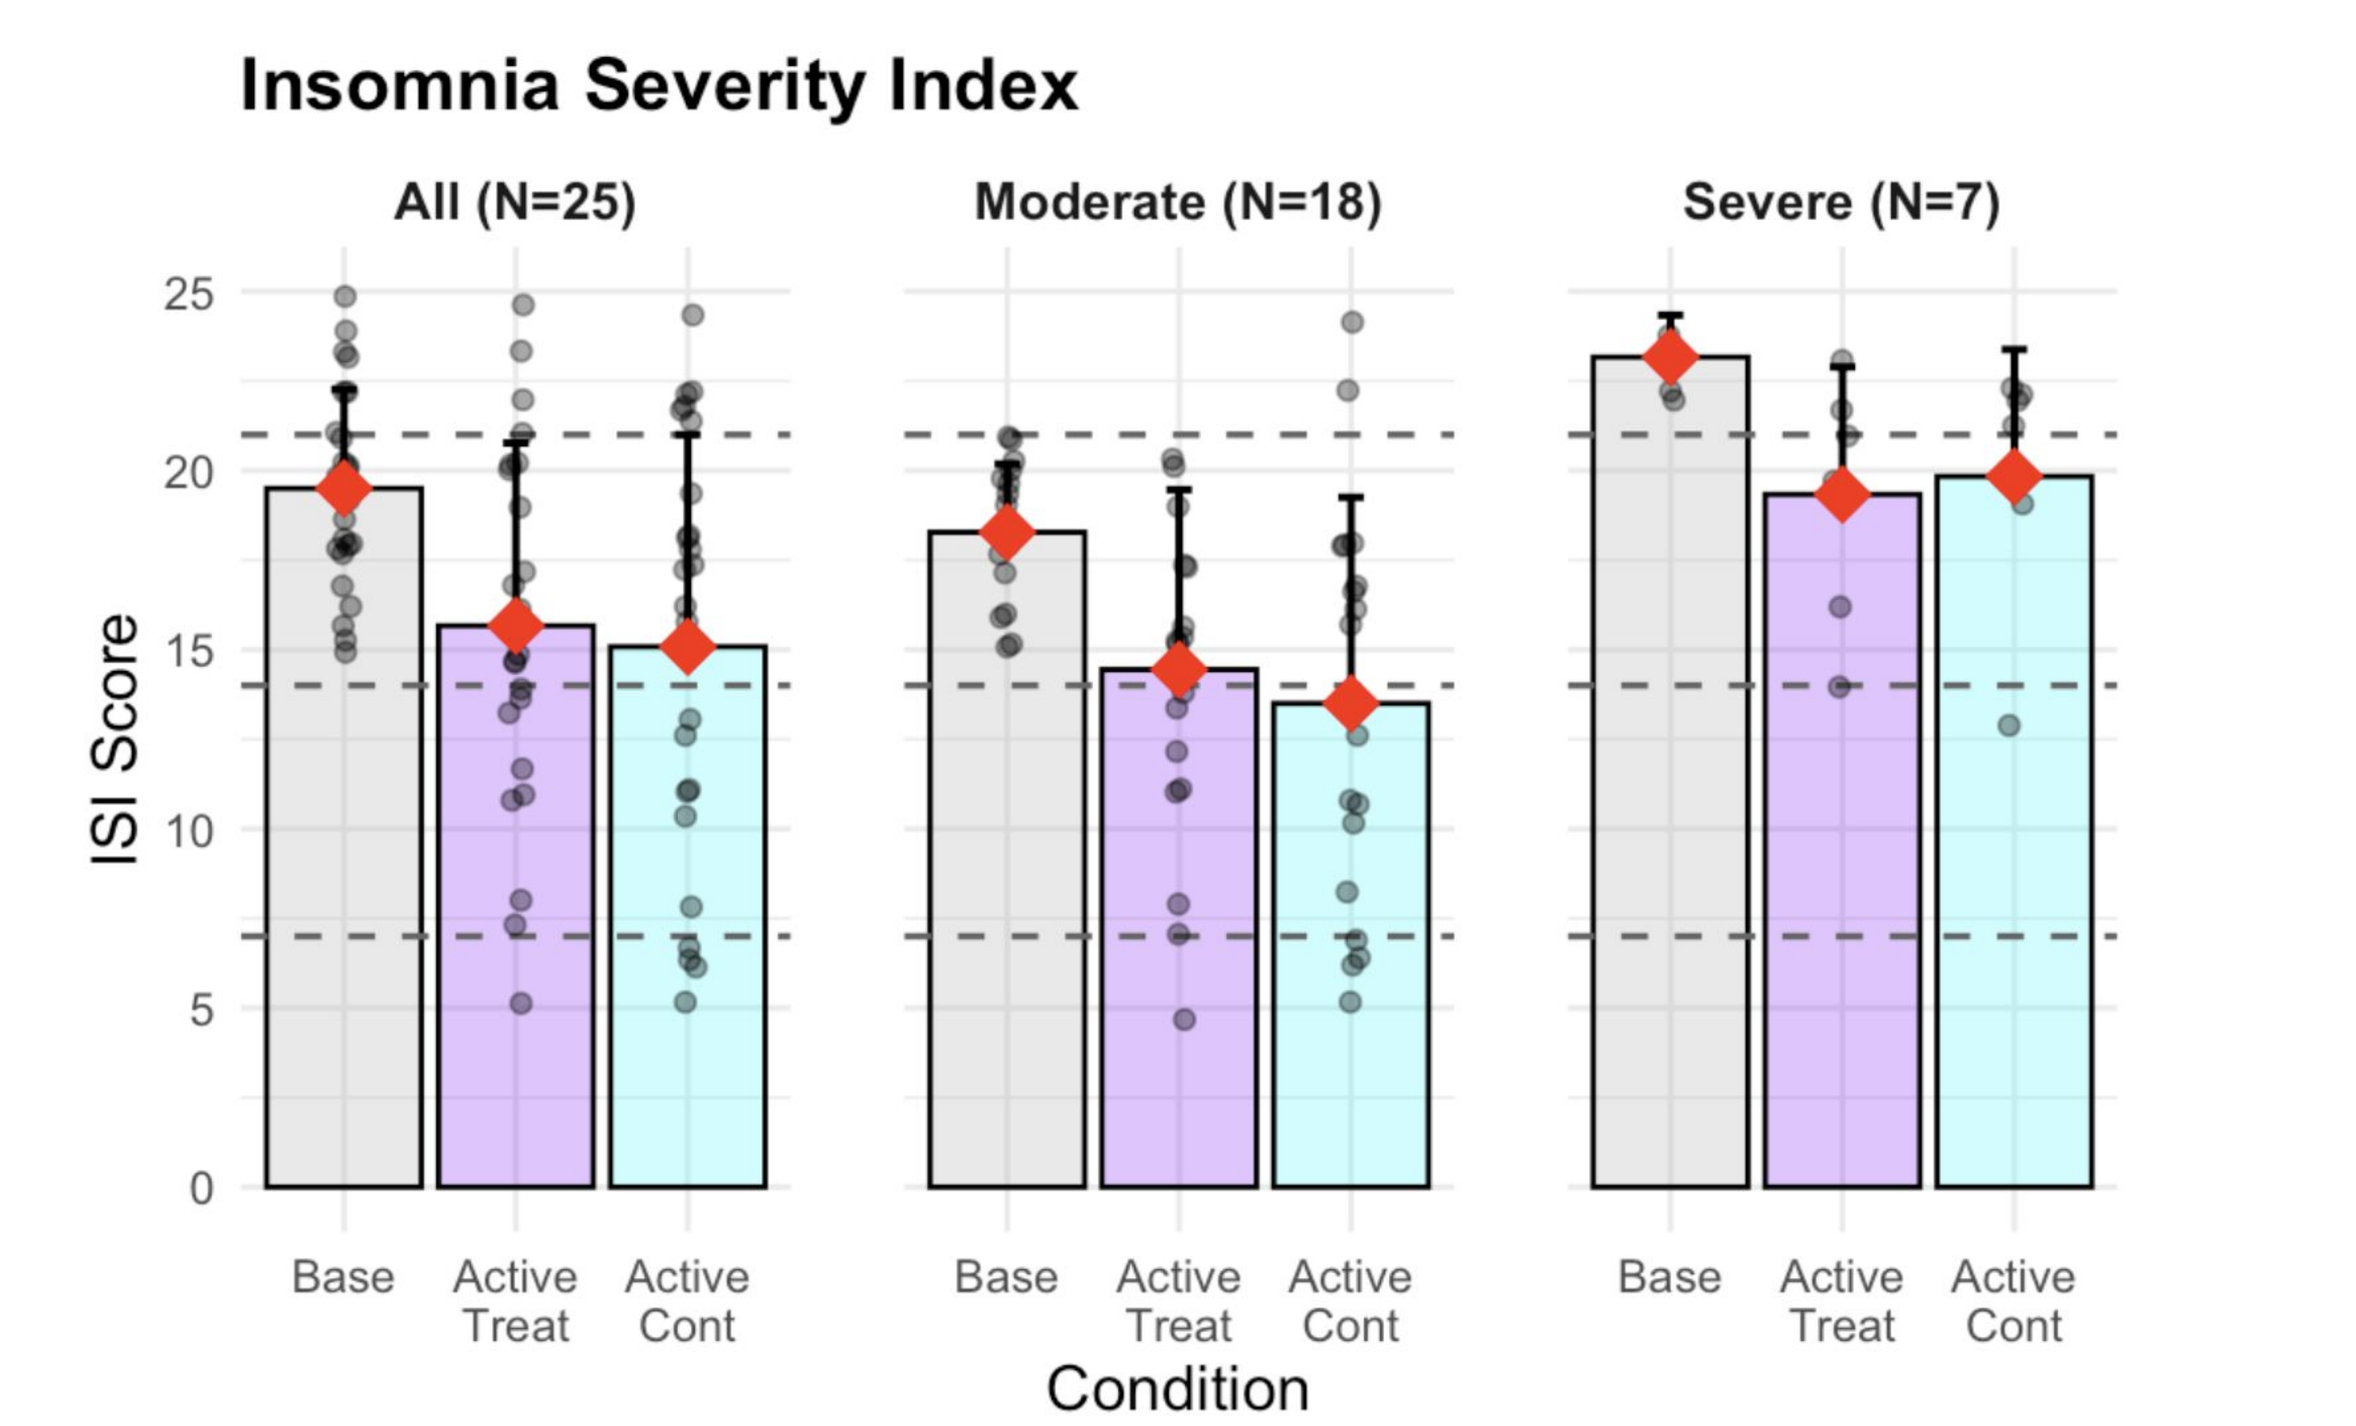
Insomnia Severity Index
All (N=25)
Moderate (N=18)
Severe (N=7)
ISI Score
25
20
15
10
5
0
Base
Active Treat
Active Cont
Condition

Figure 7. Mean +/- SD ISI aggregate score dropped from 20 +/- 3 at baseline to 16 +/- 6 in active control and 16 +/-5 in active treatment (i.e., right on the border of "subthreshold" insomnia levels).

Discussion

- This work is impactful where sleep interventions could enhance operational readiness
- Participants showed improved SOL and ISI while wearing the PeakSleep headset, regardless of stimulation frequency, compared to pre-treatment baseline
- Wearables are limited in their ability to accurately determine sleep start
- This study is limited by not having a true sham to control for the placebo effect

Disclaimer: The opinions and assertions expressed herein are those of the author(s) and do not reflect the official policy or position of the Uniformed Services University of the Health Sciences or the Department of War. Any opinions, findings and conclusions or recommendations expressed in this material are those of the author(s) and do not necessarily reflect the views of the USAMRAA. Neither I nor my family members have a financial interest in any commercial product, service, or organization providing financial support for this research. This abstract is Approved for Public Release - Teledyne Approved. **Funding Sources**: This material is based upon work supported by the United States Army Medical Research Acquisition Activity (USAMRAA) under Contract No. W81XWH22C0051.

References: [1] CH Good et al., "Sleep in the United States Military", *Neuropsychopharm. Reviews,* 2019. [2]Lie J., Tu K., Shen D., Wong B., "Pharmacological Treatment of Insomnia", *P&T,* 2015. [3] Simons S. et al., "A study on the effect of a wearable device using 0.75 Hz tES on sleep onset insomnia", *Front. Neurosci.* 2024.