Circulate
Collaborative Blood Intelligence

Real Time Civilian Blood Supply Intelligence as a Strategic Readiness Asset for Military Mobilization and Global Health Engagement

About Circulate

Ensuring a resilient and responsive blood supply is fundamental to military medical readiness. Understanding the nuances of seasonal availability and operational capacity of the civilian blood supply will contribute to quantifiable visibility into civilian backstop capacity, enabling more accurate surge modeling and risk assessment of civilian military response strategies. By integrating civilian operational intelligence into military planning frameworks, DoW can enhance joint preparedness, strengthen medical sustainment strategies, and ensure reliable access to lifesaving blood components.

Circulate is an independent, nonprofit entity holding the largest direct representation of U.S. blood supply data. **More than 60% of the blood supply is directly represented in Circulate's data.**

Data contributors:

American Red Cross · Gulf Coast Blood · oneblood Share your power. · vitalant

National data in a fragmented landscape:
- Always available, real-time data
- Highly granular, extensible dataset
- Inventory data at an ISBT 128 product code level
- Donor presentation → Phlebotomy → Inventory and transformations → Final disposition

Circulate has a suite of data-oriented product offerings including:
- Online portal with 15+ interactive dashboards
- Annual State of the U.S. Blood Supply report

2026 RBC and Platelet Daily National Civilian Inventory

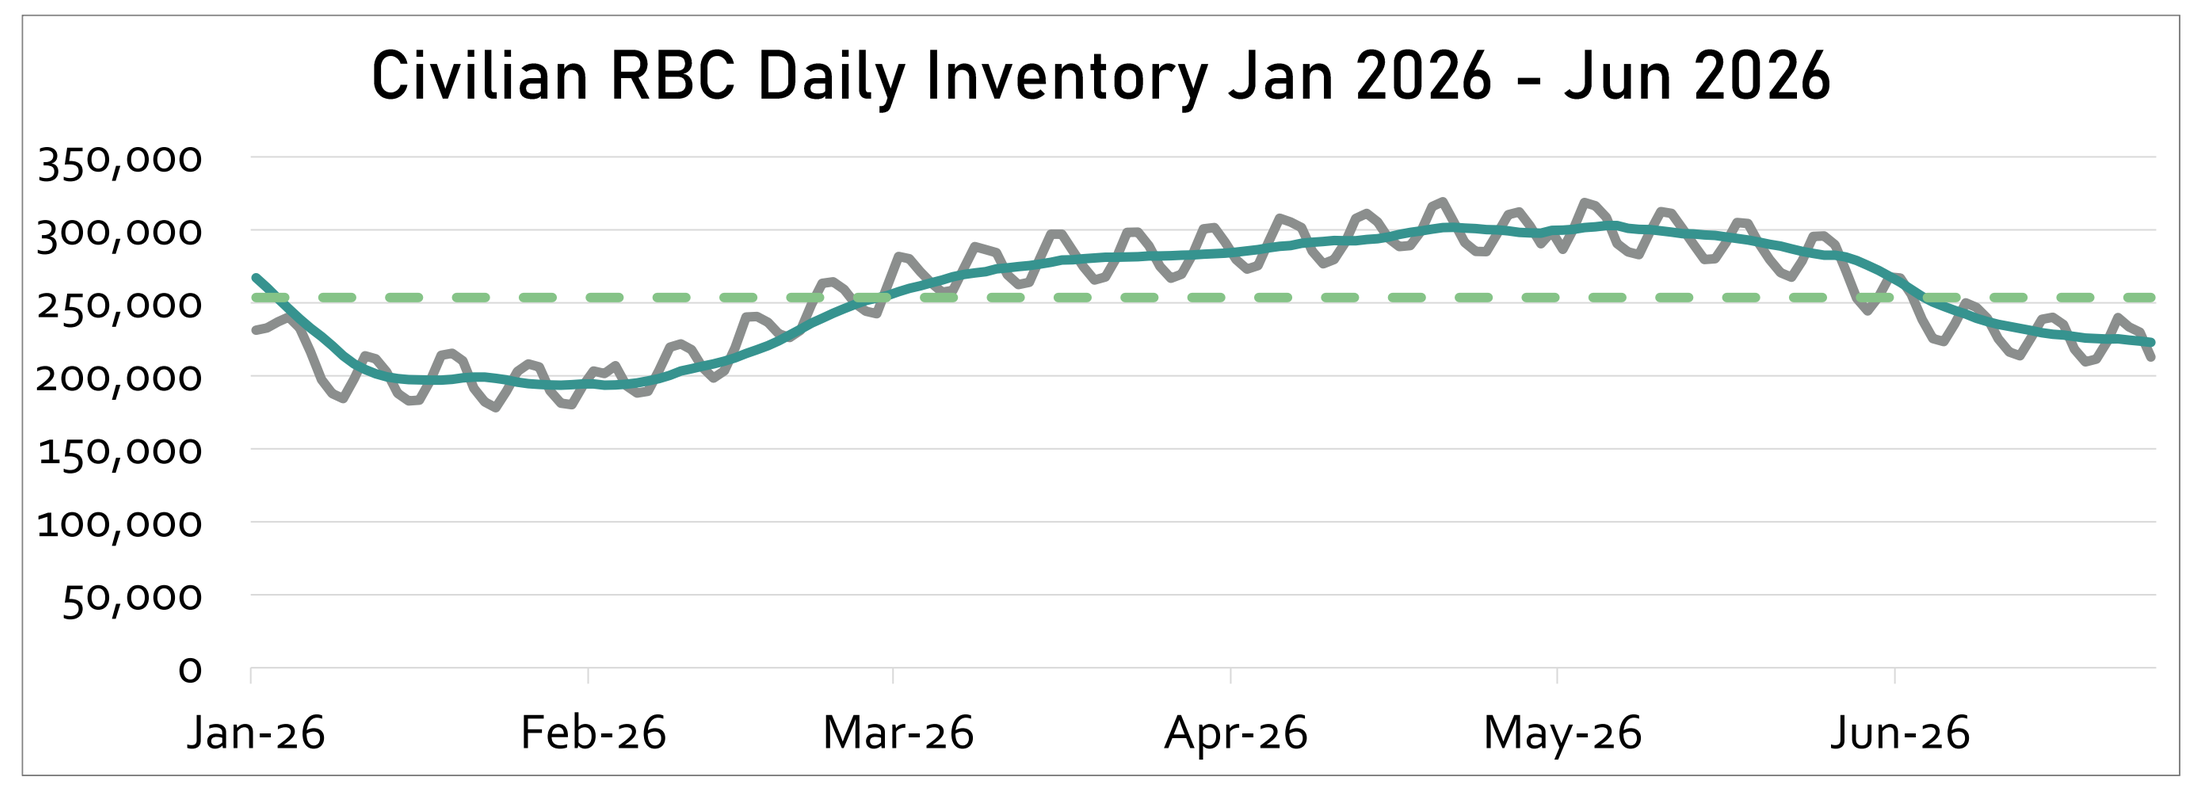
Civilian RBC Daily Inventory Jan 2026 - Jun 2026

253,500
Average Daily RBC Inventory,
Jan – Jun 2026

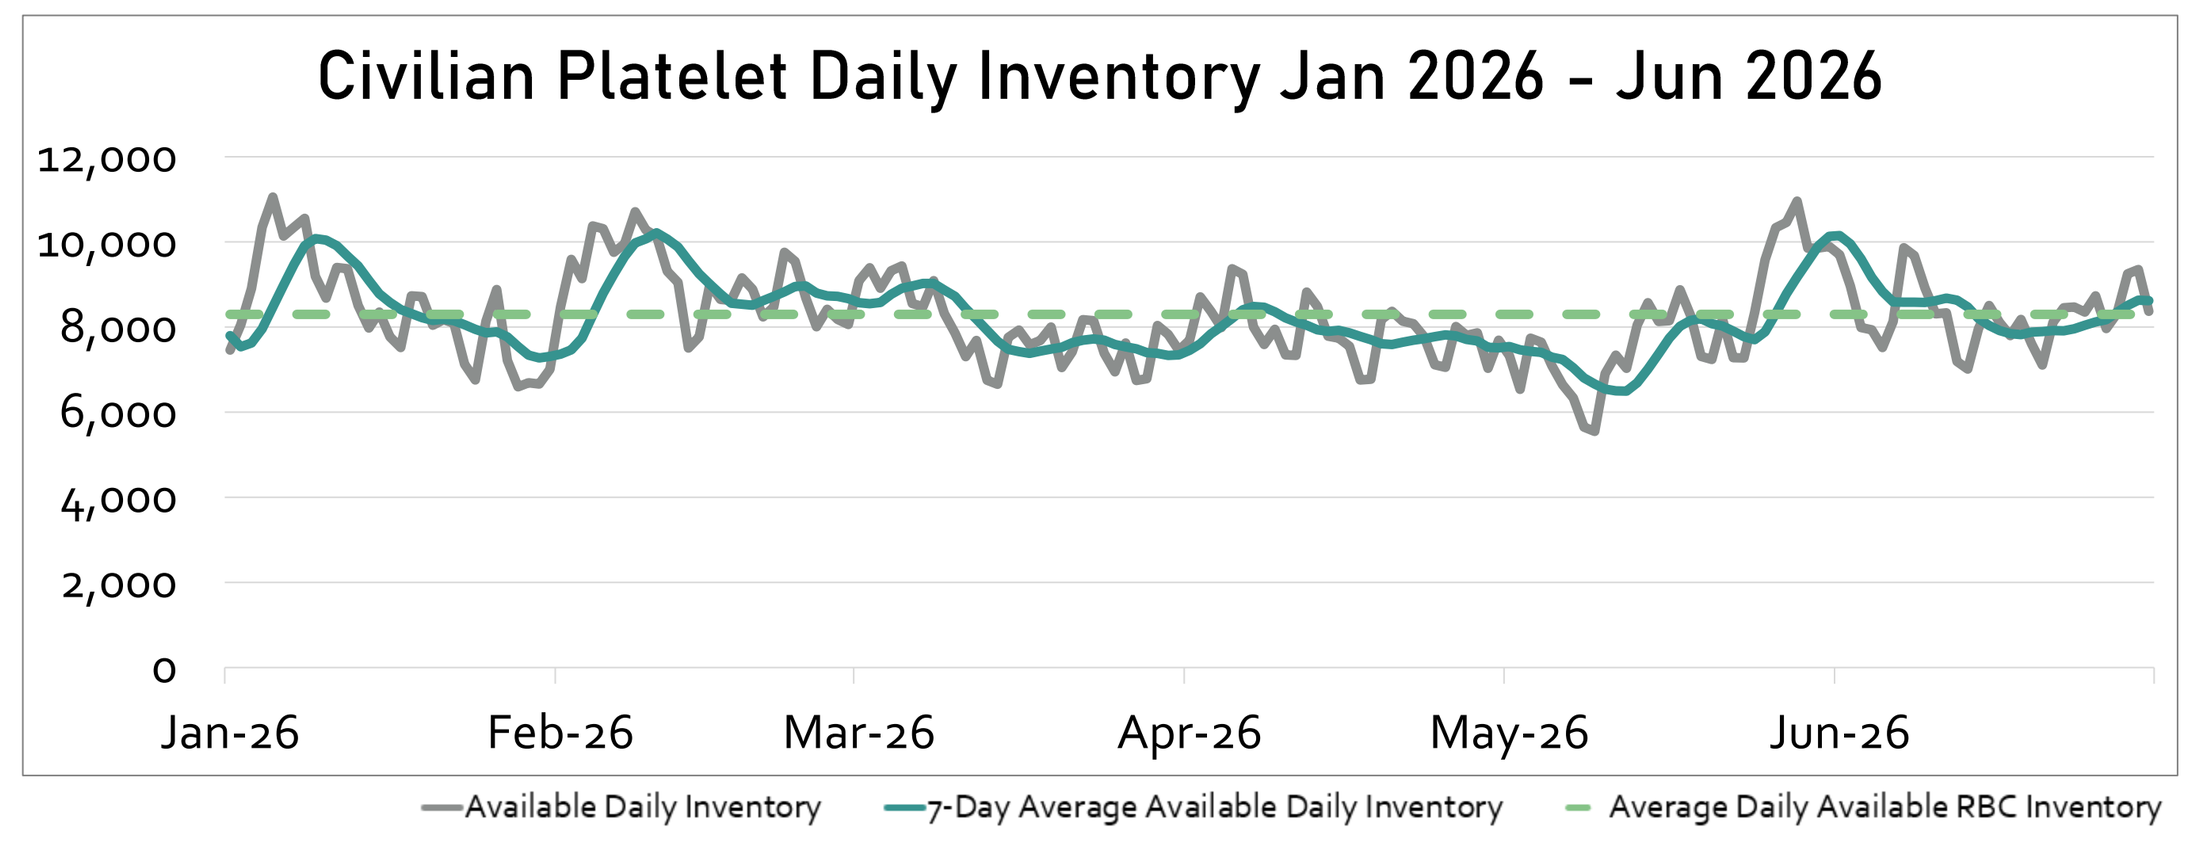
Civilian Platelet Daily Inventory Jan 2026 - Jun 2026
Available Daily Inventory
7-Day Average Available Daily Inventory
Average Daily Available RBC Inventory

8,300
Average Daily Platelet Inventory,
Jan – Jun 2026

Resiliency of the U.S. Blood Supply

At its fastest rate, the U.S. civilian red blood cell supply has increased at a rate of +2% or 4,000+ components per day. Daily available RBC inventory increased 37% over a three-week period.

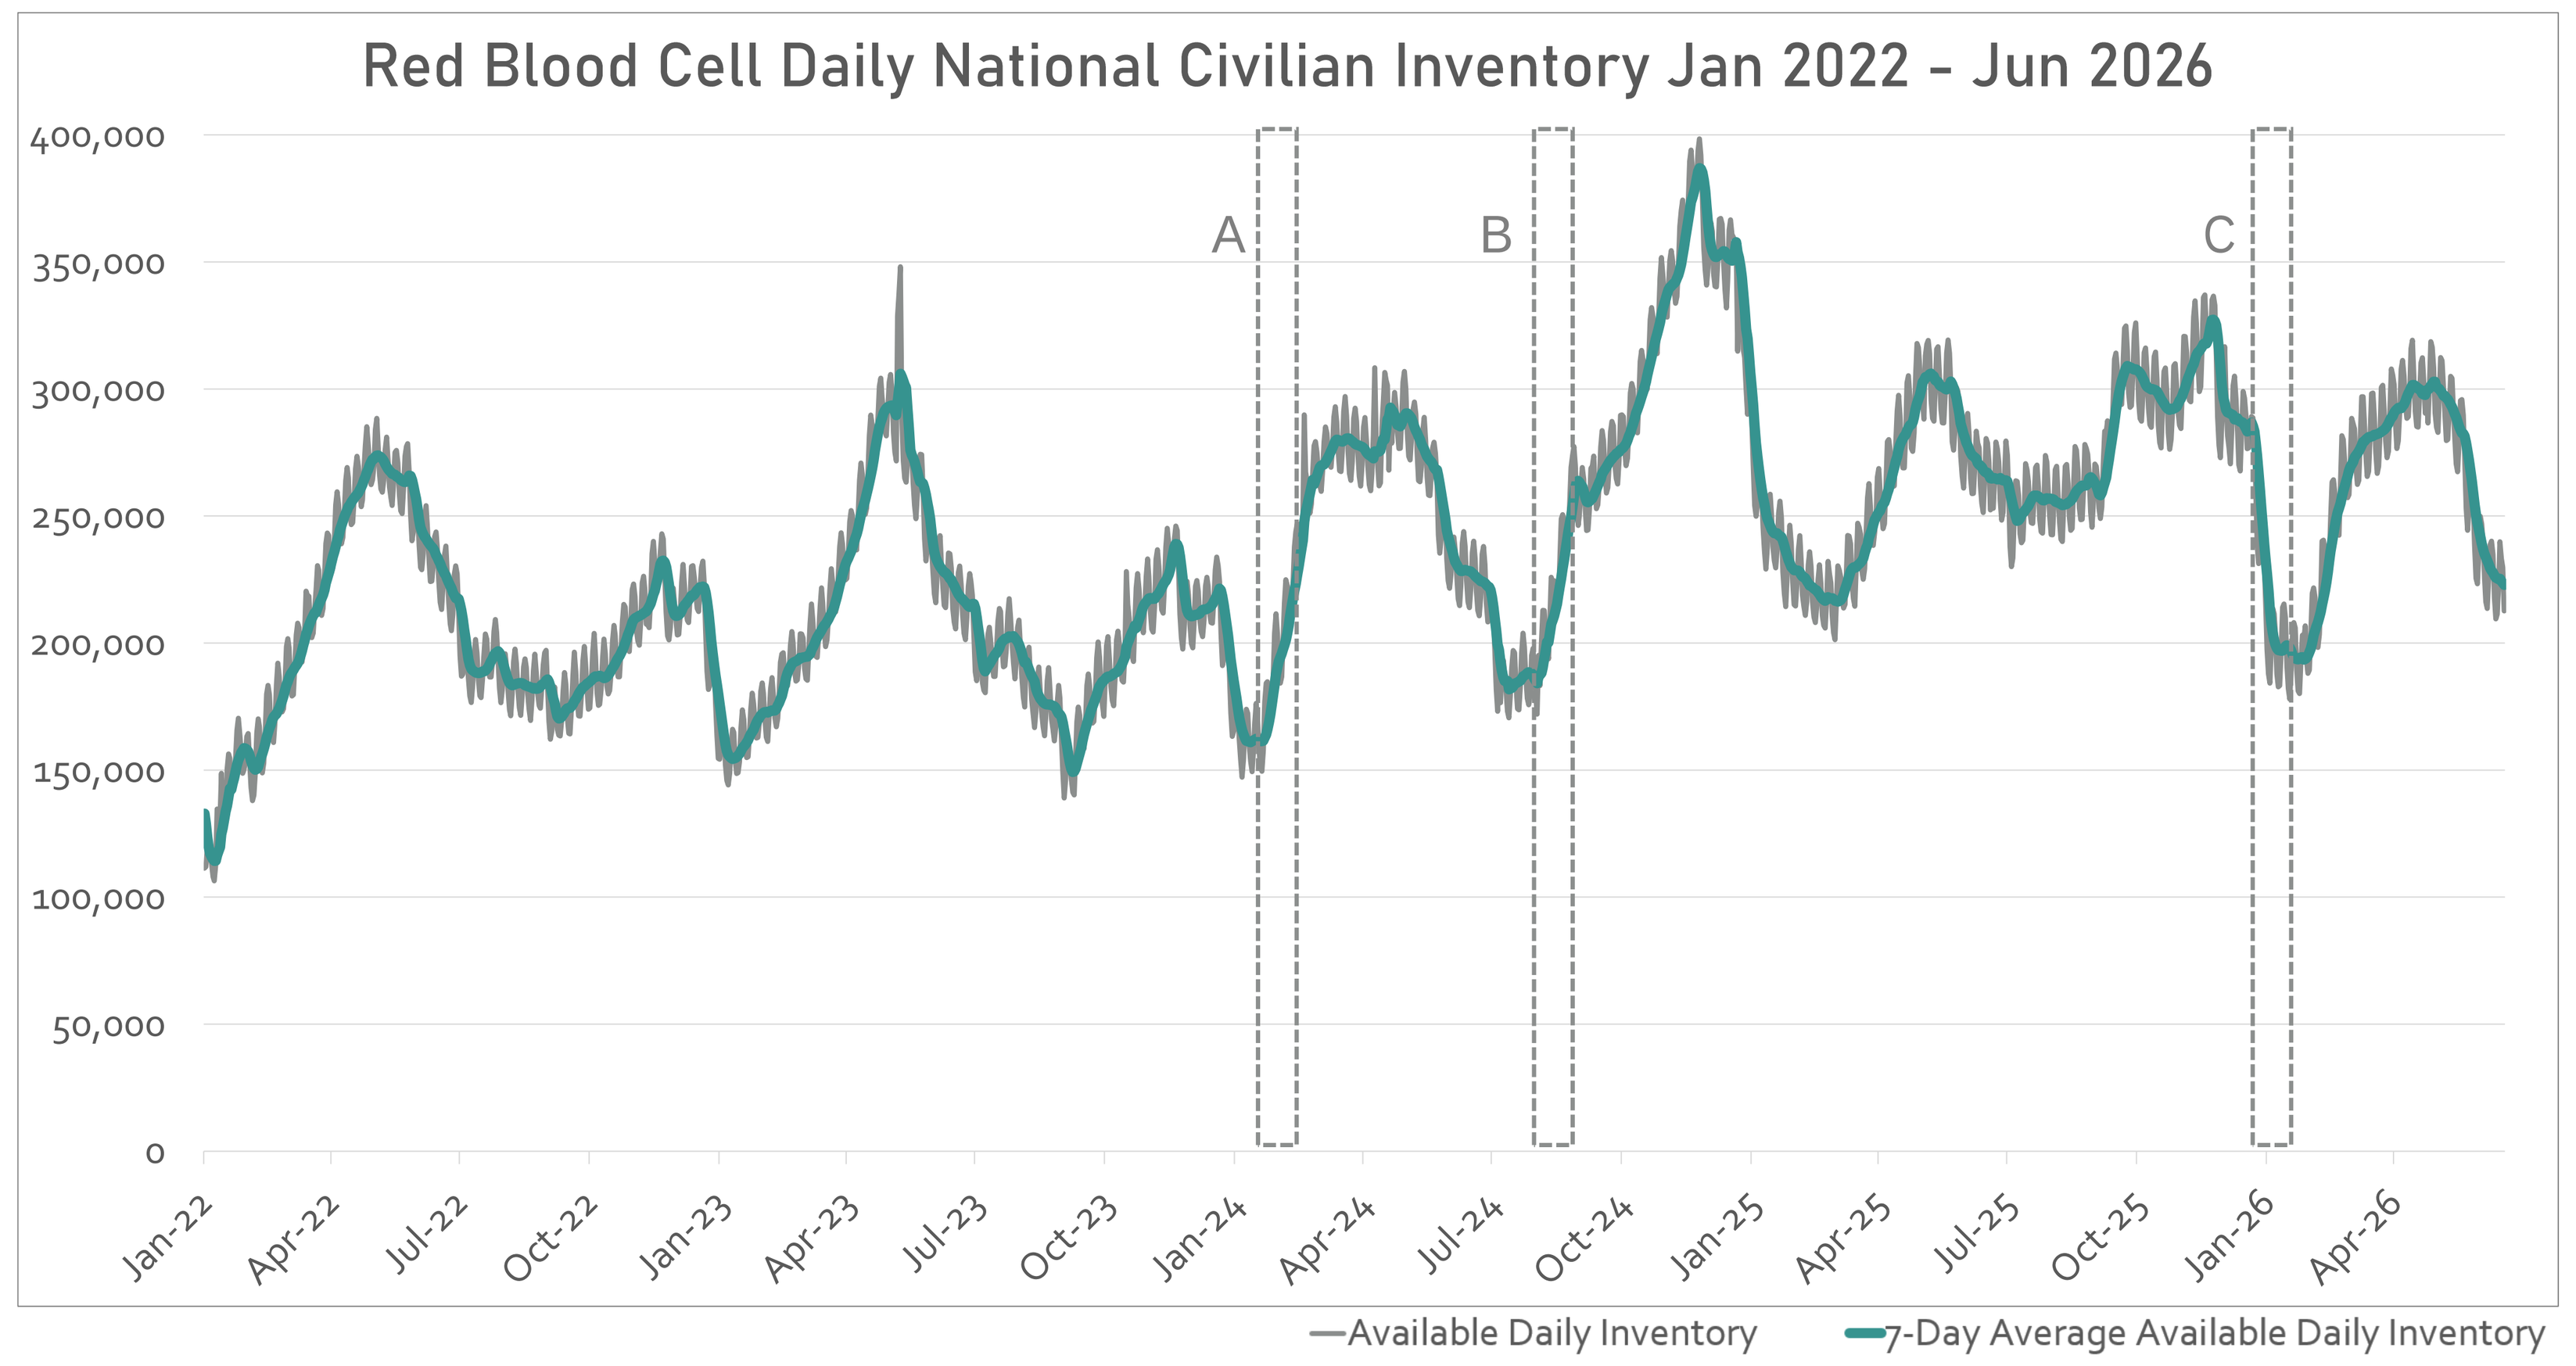
Red Blood Cell Daily National Civilian Inventory Jan 2022 - Jun 2026
A
B
C
Available Daily Inventory
7-Day Average Available Daily Inventory

In the past four years, the largest increase in the civilian RBC inventory is best characterized as an intense iteration of a standard seasonal cycle. This growth occurred without extraordinary circumstances or exhortations to the donor base, indicating possible additional capacity that would more likely be constrained by the supply chain.

Despite shocks to the blood supply and operators, the largest decrease in the RBC inventory was a period of significant inventory shedding after months of inventory accumulation.

A: February 2024 recorded the **fastest rate of RBC inventory growth**:
- **4,000+ RBCs/day** from 2/2 – 2/6
- **+2% daily increase in RBC inventory** from 2/1 – 2/5
- **+37% increase in RBC inventory** across a 3-week period, from 1/30 – 2/21
 61,100 additional RBCs

B: RBC inventory also increased rapidly in August/September 2024:
- 4,000+ RBCs/day from 9/3 – 9/4
- +2% daily increase in RBC available inventory from 8/23 – 8/30
- +33% increase in RBC inventory across a 3-week period, from 8/14 – 9/4
 64,300 additional RBCs

C: At its fastest rate, the U.S. civilian red blood cell supply has depleted at a rate of -3% or 7,000+ components/day.

For the period Jan 2022 – Jun 2026, January 2026 recorded the **fastest rate of RBC inventory depletion**:
- **-7,000+ RBCs/day** from 1/1 – 1/9
- **-3% daily decrease in RBC inventory** from 1/2 - 1/10
- **-31% decrease in RBC inventory** across a 3-week period, from 12/27 – 1/19
 89,900 fewer RBCs

Within the same 3-week period, 33,800 RBCs were discarded and the daily discard rate peaked at 2% on 1/2.

Case Studies

Cyberattack

One U.S. blood operator was the victim of a cyberattack in 2024 and greatly reduced or fully stopped operations from 7/29 – 8/8 (weeks 31 and 32). It is possible to observe some of the impact of this event in the national estimates of the U.S. blood supply:

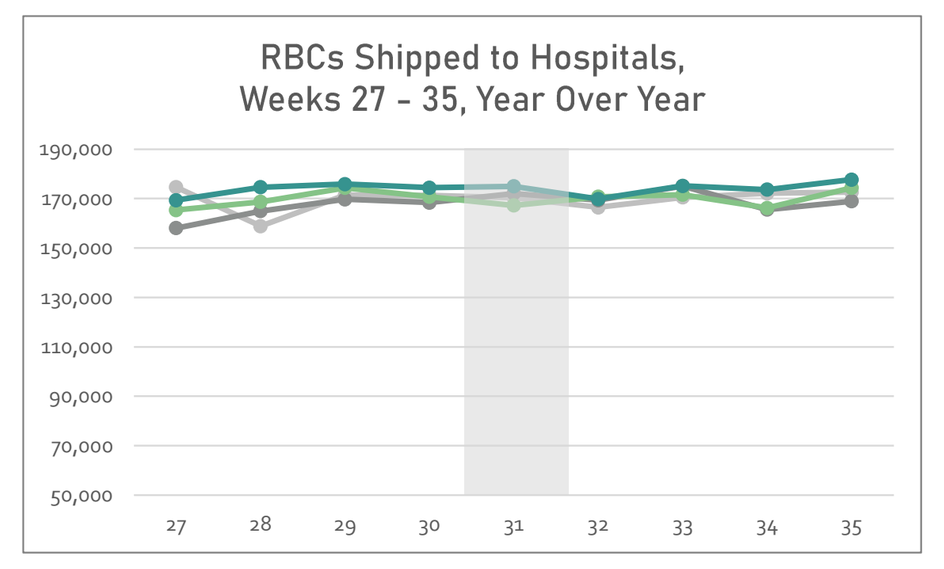
RBCs Shipped to Hospitals, Weeks 27 - 35, Year Over Year

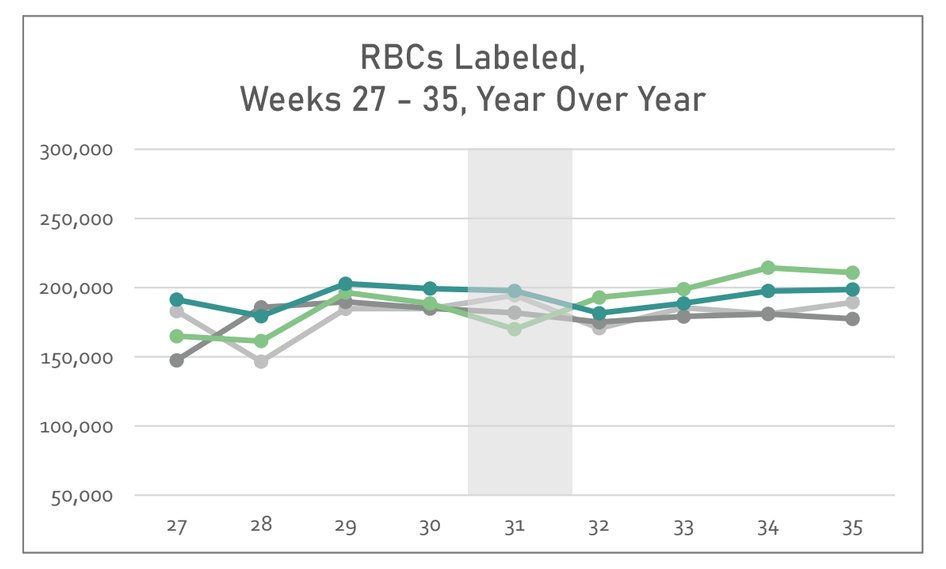
RBCs Labeled, Weeks 27 - 35, Year Over Year

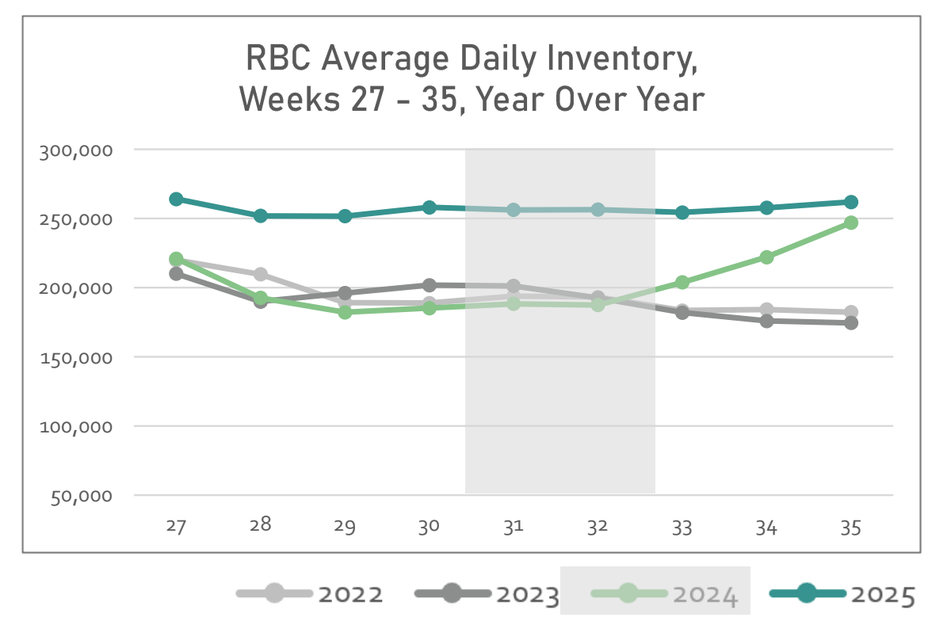
RBC Average Daily Inventory, Weeks 27 - 35, Year Over Year
2022
2023
2024
2025

Week 31 recorded the lowest number of labeled RBCs and lowest number of RBCs shipped to hospital customers in four years. A rebound in the number of labeled RBCs in the following weeks likely indicates WIP held for later labeling.

Weeks 31 and 32 recorded the lowest average available daily RBC inventory in four years.

Weather Events

In early January 2025 (weeks 2 – 5) there were severe fires in southern California, and significant winter weather storms across the Midwest, northeast, and southeastern United States. There were observable shocks to the blood collection system, but they were minimized by high RBC inventory from the end of 2024.

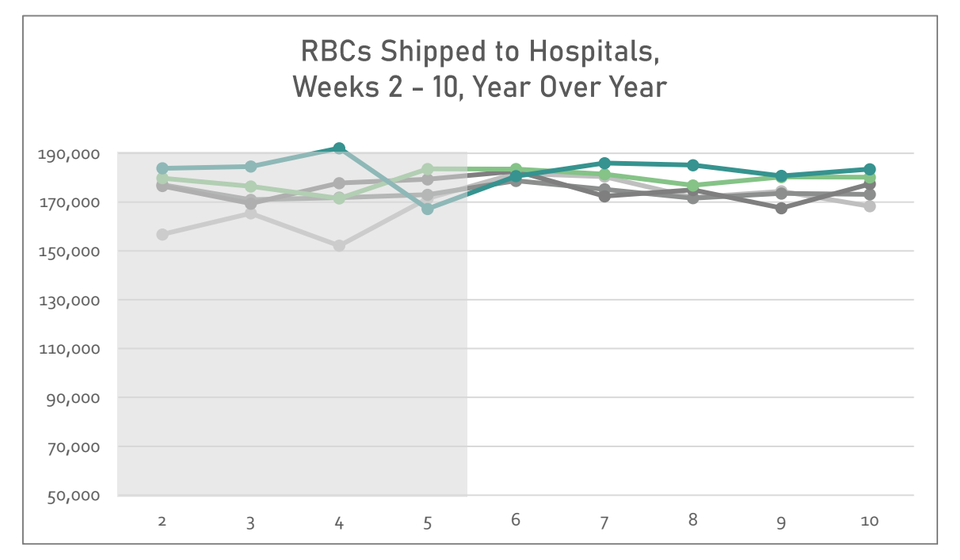
RBCs Shipped to Hospitals, Weeks 2 - 10, Year Over Year

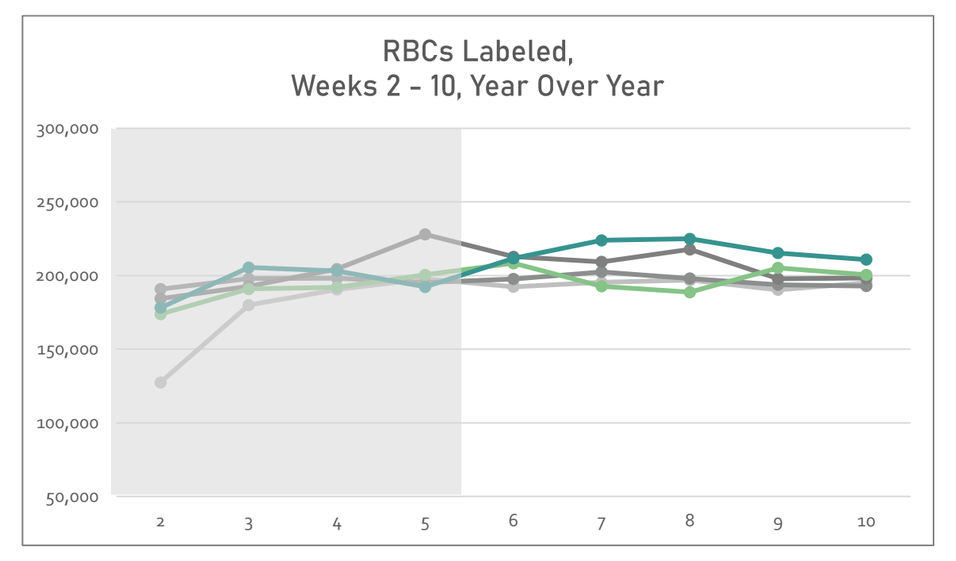
RBCs Labeled, Weeks 2 - 10, Year Over Year

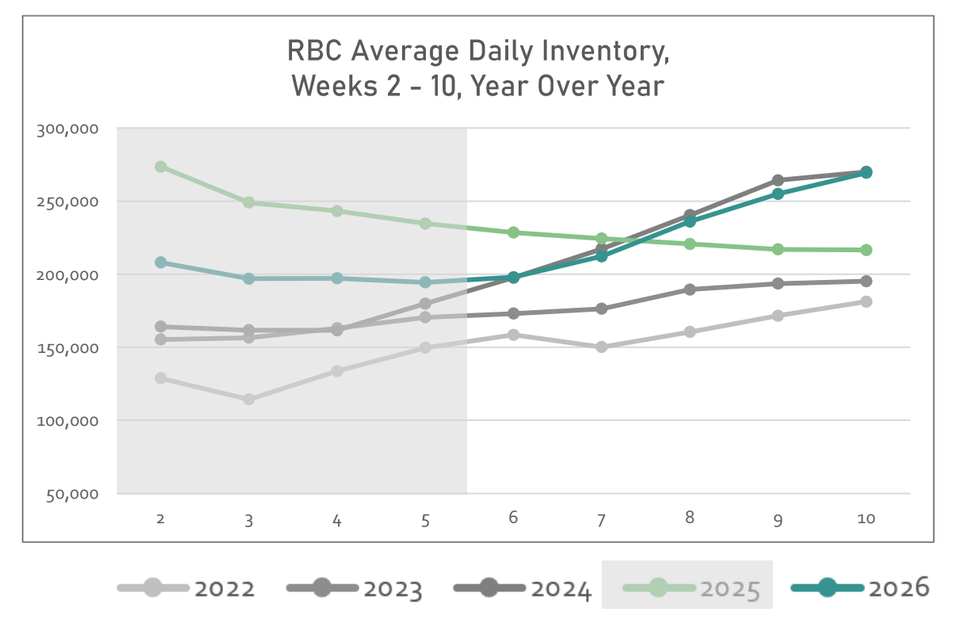
RBC Average Daily Inventory, Weeks 2 - 10, Year Over Year
2022
2023
2024
2025
2026

Weeks 2 – 5 recorded the second highest total number of RBCs shipped in five years.

Weeks 2 – 5 in 2025 recorded the second lowest total number of RBCs labeled in five years, reflecting the stress on the blood collection system.

Weeks 2 – 5 in 2025 recorded the lowest average daily available inventory per week in five years.

Emerging Products

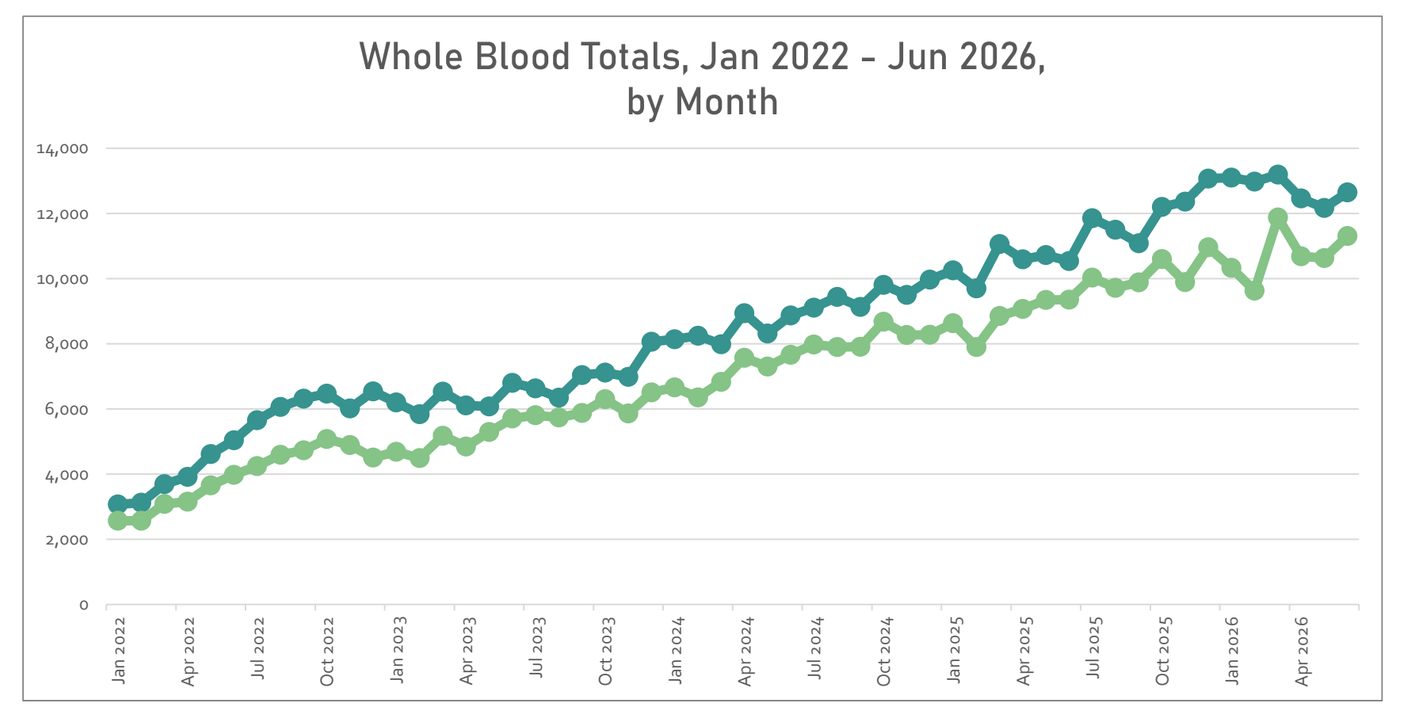
Whole Blood Totals, Jan 2022 - Jun 2026, by Month

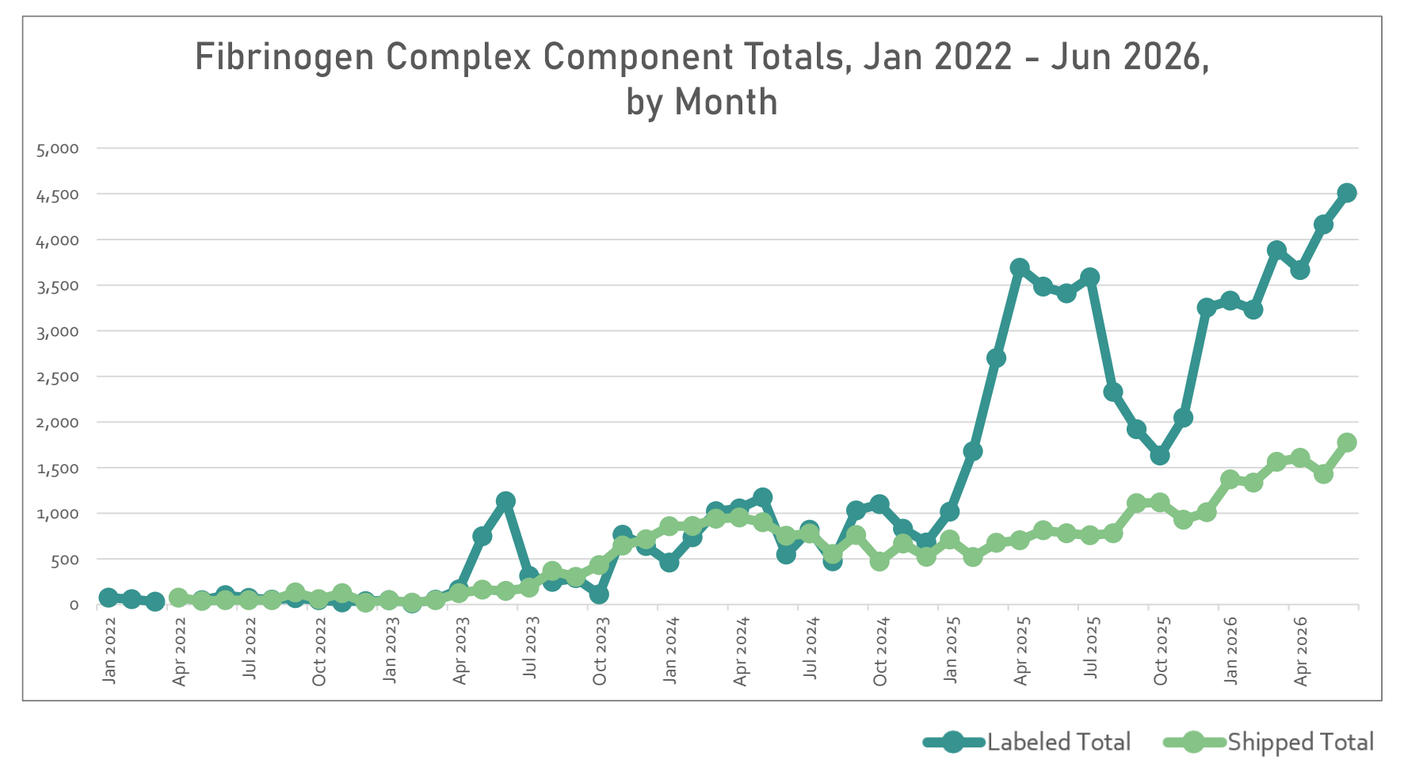
Fibrinogen Complex Component Totals, Jan 2022 - Jun 2026, by Month
Labeled Total
Shipped Total

- Whole blood manufacturing has increased 26% – 35% each year since 2022.
- Whole blood shipments have increased 25% – 41% each year since 2022.

- Fibrinogen complex component manufacturing has at least doubled every year since 2022.
- Fibrinogen complex component shipments at least doubled each year from 2022 – 2024, but dropped off to only +10% from 2024 – 2025.

For more information or to download Circulate's 2025 State of the U.S. Blood Supply Report, visit circulateblood.org

Poster authored by Katie Risvold, Executive Director, Circulate