Meta-Analysis and Cross-species Validation Defines A Universal Cardiac Ischemia Signature

Zhenyu Ji[1], Anpuchchelvi Rajadurai[1], Njauw, Ching-Ni Jenny[1], Kishwor Poudel[1] and Hensin Tsao[1]*

[1]Wellman Center for Photomedicine, Massachusetts General Hospital, Boston, MA

MHSRS-26-4092

Objectives:

➔To derive a Universal Cardiac Ischemia Signature (UCIS-110) by Restricted Maximum Likelihood (REML)-based random-effects meta-analysis across three human ICM cohorts and one murine myocardial infarction dataset requiring concordance across all four.

➔To profile AC16 and HL-1 cardiomyocytes across acute ischemia, hypoxic adaptation, and chronic hypoxia by bulk RNA-seq, DESeq2, and fGSEA to map stage-specific transcriptional programs.

➔To determine whether the in vivo ICM transcriptome maps to acute or chronic hypoxic biology by Fisher's exact enrichment testing of UCIS-110 at each oxygen stage in both cell lines.

Background: Cardiac ischemia from hemorrhagic shock, blast injury, and service-related coronary disease remains a leading cause of disability in military personnel, yet no consensus cross-species transcriptional ischemic signature exists to guide therapeutic development.

Methods :We set out to define such a signature by applying Restricted Maximum Likelihood (REML) across three human ICM cohorts (GSE116250, GSE55296, GSE120852; 29 ICM vs. 29 non-failing left ventricles) and one murine 28-day LAD-ligation dataset (GSE236374). We next mapped the resulting signature to a specific oxygen state by profiling AC16 and HL-1 cardiomyocytes (n=3/group) across Stage I (acute ischemia, 20%->1% O2, 24h), Stage II (hypoxic adaptation, chronic vs. acute 1% O2), and Stage III (chronic hypoxia, 1% vs. 20% O2, both >30d) using bulk RNA-seq, DESeq2, pre-ranked fGSEA (143 gene sets; 1,000 permutations), cross-species overlap analysis, and Fisher's exact enrichment testing.

Results: A Universal Cardiac Ischemia Signature of 110 concordant genes was derived (UCIS-110; |LFC| >= log2(1.5), padj <0.05): 91 upregulated (UCIS-91), enriched in ECM/matrisome remodeling (NABA Core Matrisome, LogP=-11.8) with an immune cluster (CCL5, ZAP70, NKG7, CD8A), and 19 downregulated (UCIS-19), encoding contractile apparatus loss (TTN, KLHL41, SMYD1; muscle development LogP=-4.0). Stage I was characterized by HIF-1a/glycolysis activation (HYPOXIA NES=+2.72, GLYCOLYSIS NES=+2.55) with interferon suppression (INTERFERON_ALPHA NES=-2.53). Stage II showed HIF resolution with MYC/E2F and ribosomal biogenesis reactivation. Stage III showed HIF re-emergence with myogenesis induction (MYOGENESIS NES=+2.53) and oxidative phosphorylation suppression (NES=-2.58). Cross-species overlap confirmed Acute Ischemia (165 shared upregulated, 97 downregulated) and Chronic Hypoxia conservation. UCIS-110 enrichment was Chronic Hypoxia-dominant in both lines (AC16: OR=4.8; HL-1: OR=5.2; both p<0.001), exceeding Acute Ischemia (OR~2.0).

Conclusion: Taken together, these findings indicate that end-stage ICM reflects a stable chronic hypoxic transcriptional identity, not a perpetuation of acute ischemic injury, characterized by ECM remodeling, contractile loss, and sustained HIF activity; UCIS-110 provides a cross-species validated target set for countermeasure development against chronic cardiac ischemia.

This work is supported by the Air Force Office of Scientific Research under award number FA9550-23-1-0656.

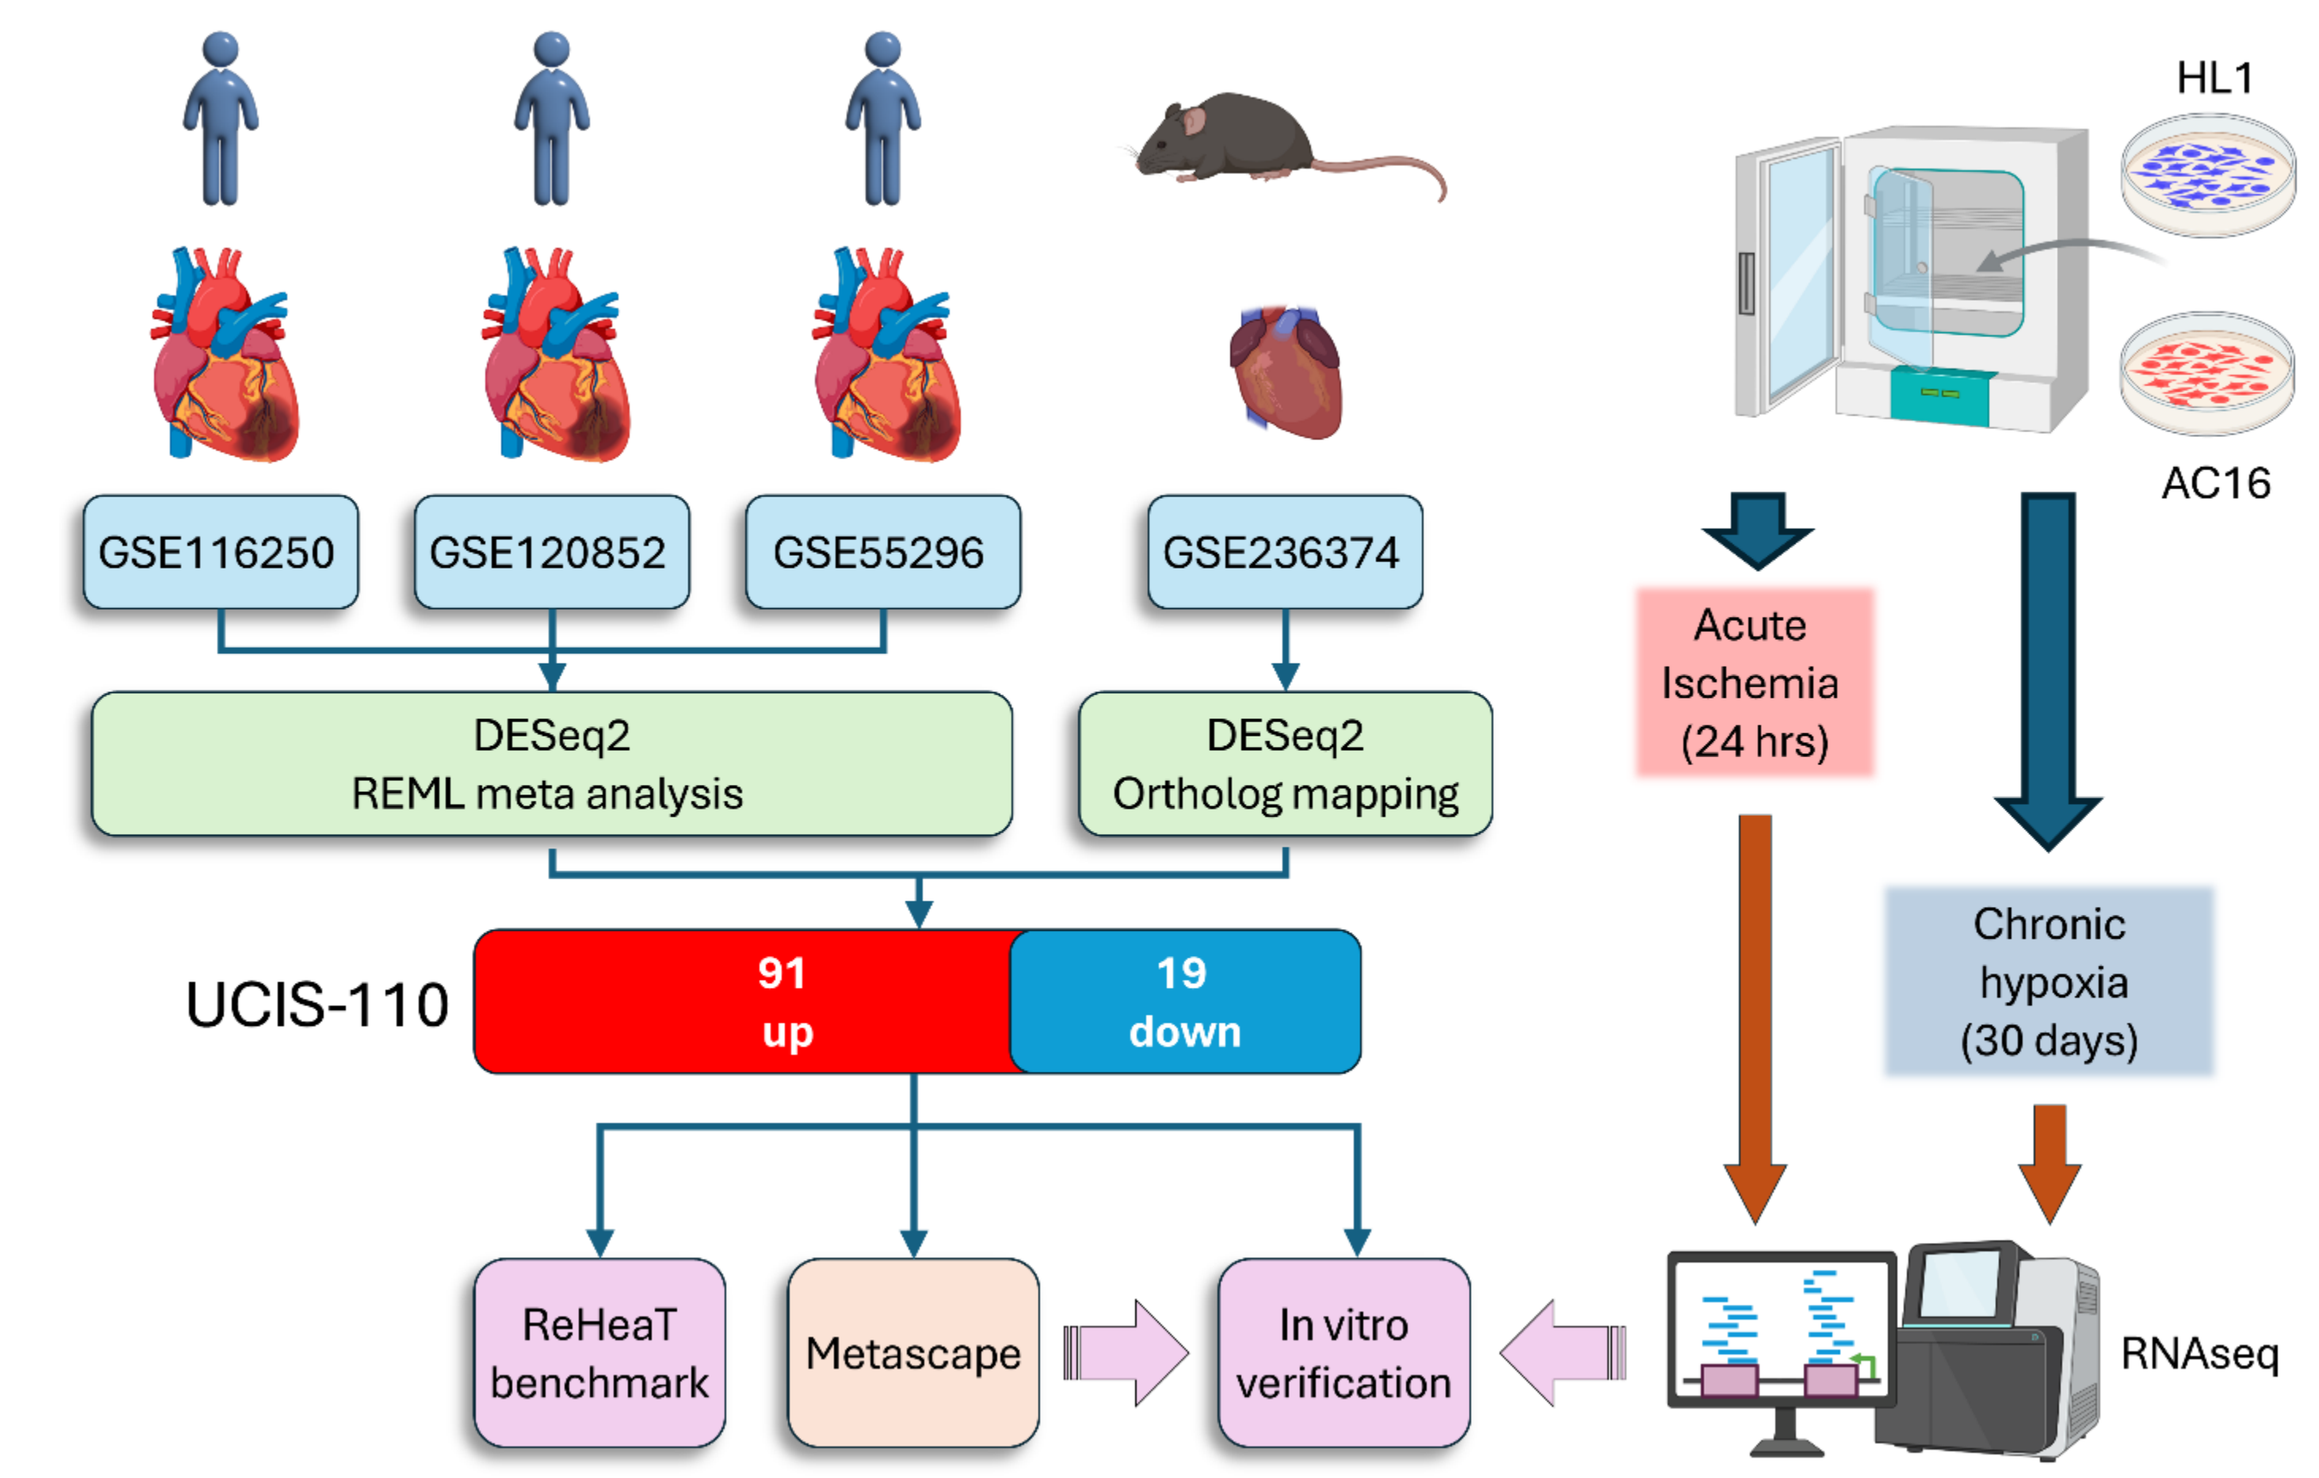
HL1
AC16
GSE116250
GSE120852
GSE55296
GSE236374
DESeq2
REML meta analysis
DESeq2
Ortholog mapping
Acute Ischemia (24 hrs)
Chronic hypoxia (30 days)
UCIS-110
91 up
19 down
ReHeaT benchmark
Metascape
In vitro verification
RNAseq

Figure 1. Overall schema of study. (a) Three public human left ventricular transcriptomic datasets (GSE116250, GSE120852, and GSE55296) were analyzed to identify ischemic cardiomyopathy (ICM)-associated genes. Differential expression was performed using DESeq2 (RNA-seq datasets) or Welch's t-test (GSE55296), followed by REML random-effects meta-analysis. Genes showing concordant regulation in a murine myocardial infarction model (GSE236374) were retained, yielding the UCIS-110 signature (91 upregulated, 19 downregulated). UCIS-110 was then benchmarked against the ReHeaTΔ14/18 heart failure consensus, functionally characterized by Metascape enrichment analysis, and experimentally validated by RNA-seq in human AC16 and mouse HL-1 cardiomyocytes subjected to acute ischemia (1% O_2, 24 h) or chronic hypoxia (1% O_2, 30 days).

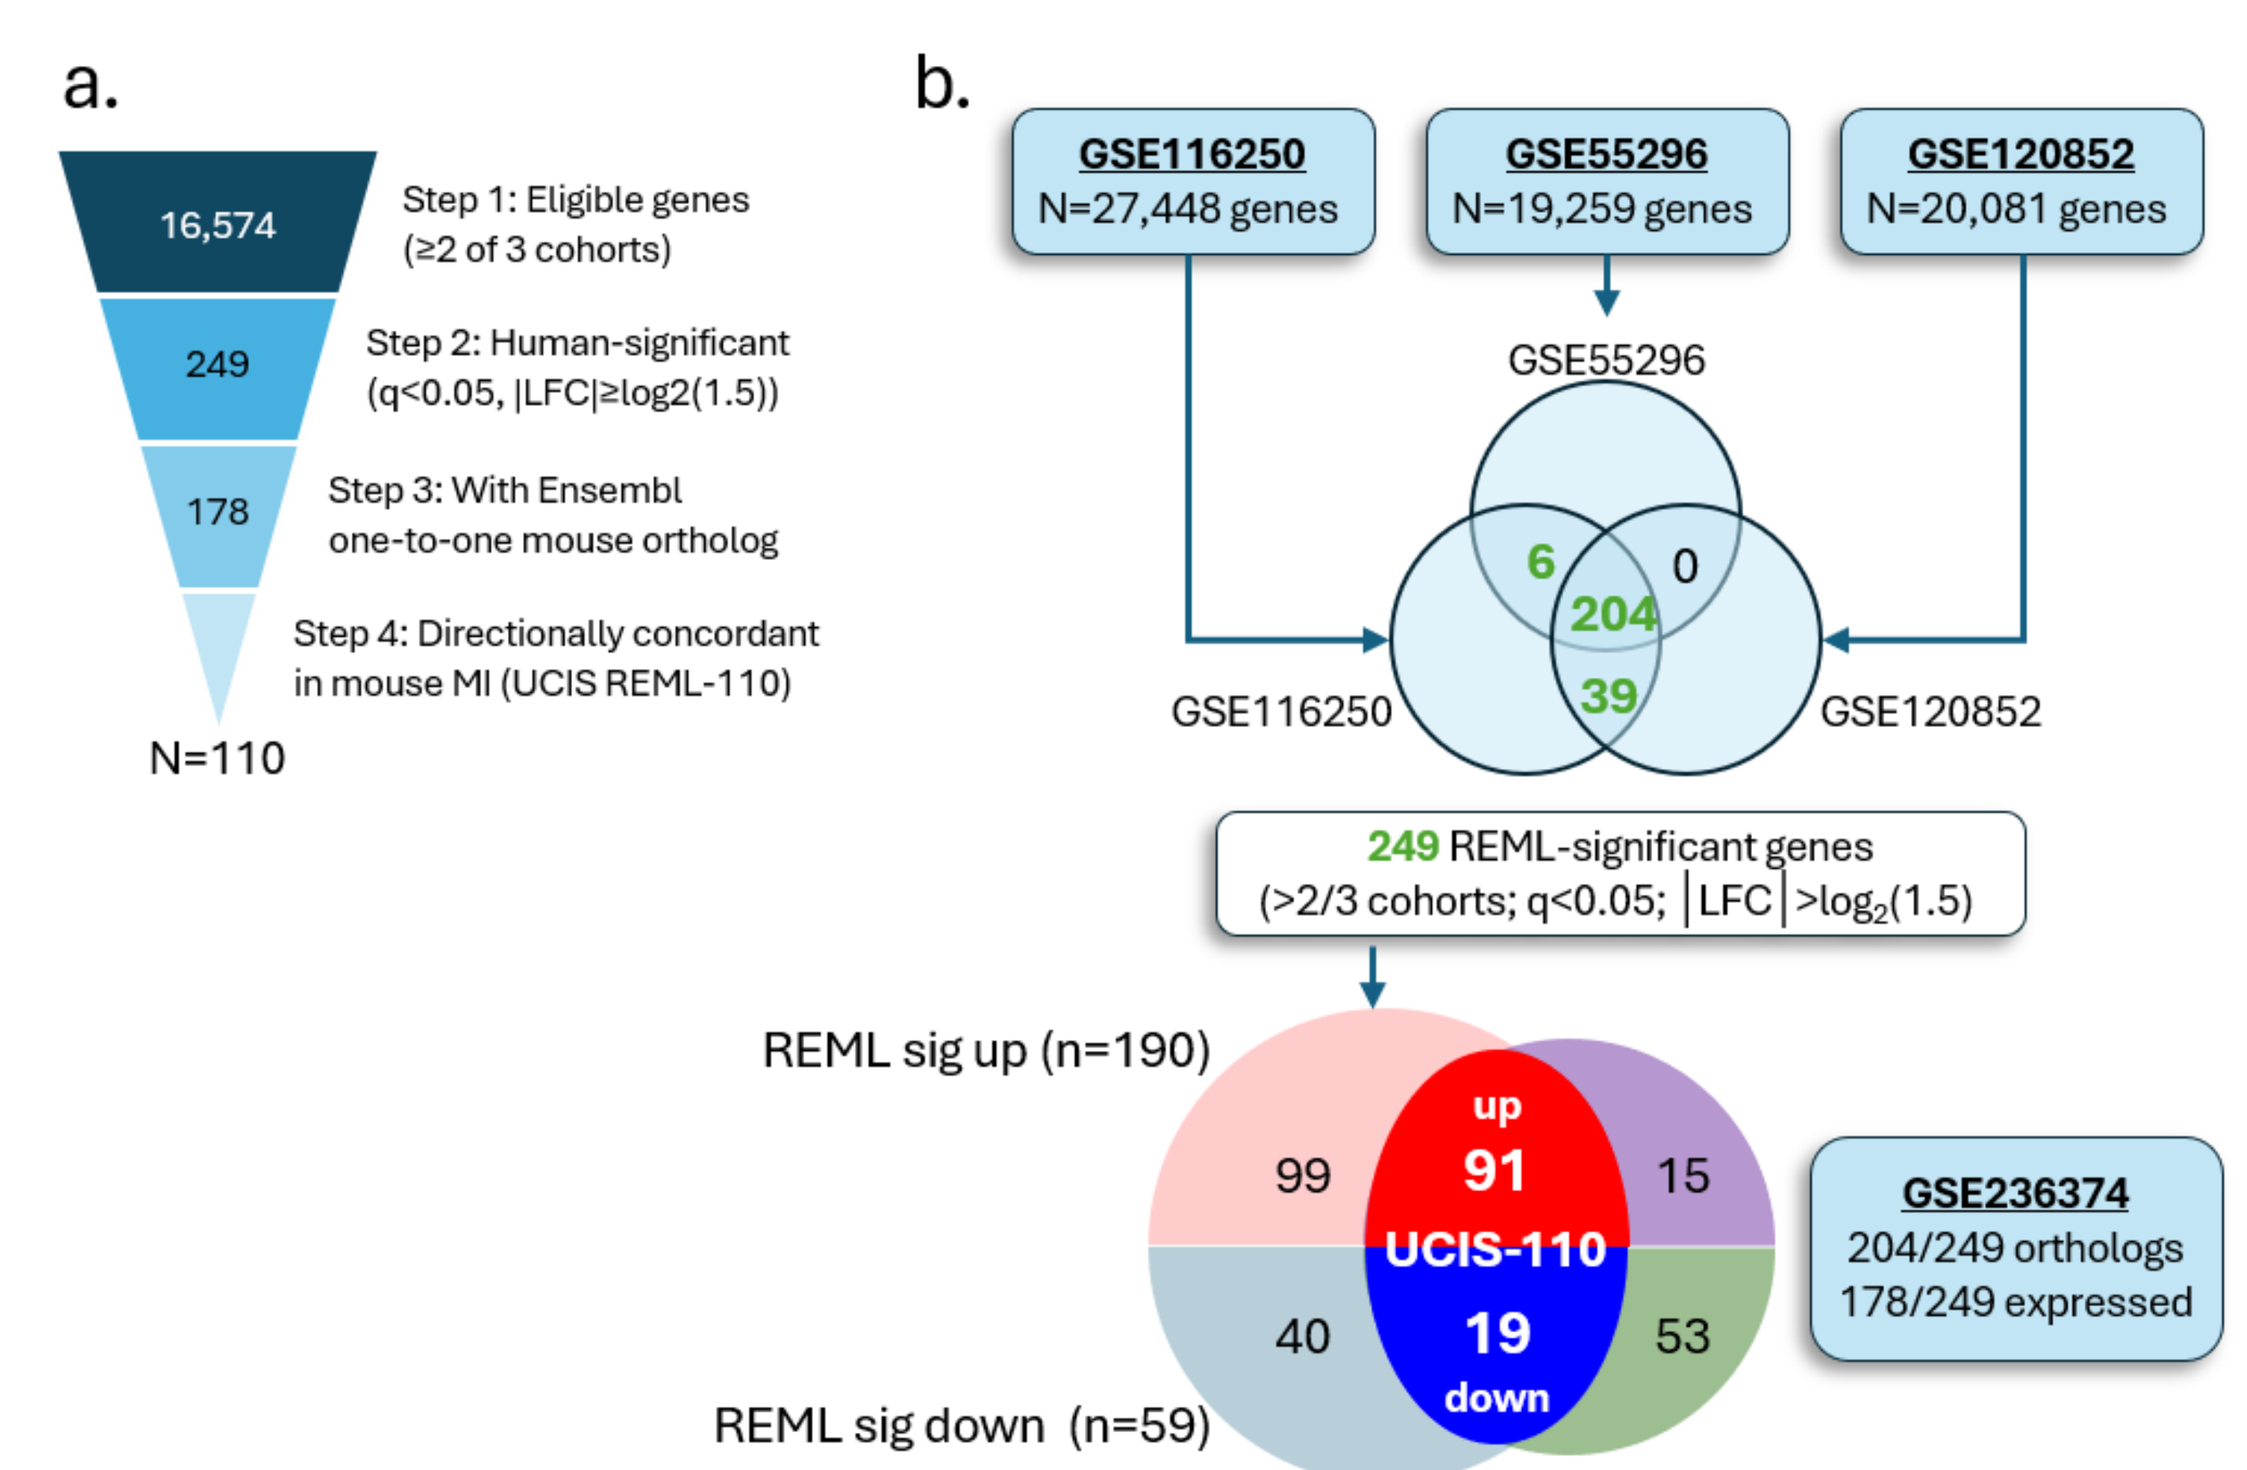
a.
16,574
Step 1: Eligible genes (≥2 of 3 cohorts)
249
Step 2: Human-significant (q<0.05, |LFC|≥log2(1.5))
178
Step 3: With Ensembl one-to-one mouse ortholog
Step 4: Directionally concordant in mouse MI (UCIS REML-110)
N=110
b.
GSE116250 N=27,448 genes
GSE55296 N=19,259 genes
GSE120852 N=20,081 genes
GSE55296
6
0
204
39
GSE116250
GSE120852
249 REML-significant genes (>2/3 cohorts; q<0.05; |LFC|>log2(1.5)
REML sig up (n=190)
99
up 91
15
UCIS-110
40
19 down
53
REML sig down (n=59)
GSE236374
204/249 orthologs
178/249 expressed

Figure 2. Derivation of the UCIS-110 signature. (a) Filtering workflow. Among 16,540 genes present in at least two of three human cohorts, 249 were significant by REML meta-analysis (BH-FDR q < 0.05; |pooled LFC| ≥ $\log_2[1.5]$). Of these, 178 had a detectable one-to-one mouse ortholog expressed in the murine myocardial infarction dataset (GSE236374) and were evaluated for cross-species directional concordance. (b) Venn diagram showing cohort membership of the 249 REML-significant genes: 204 were present in all three cohorts, 39 in GSE116250 and GSE120852 only, 6 in GSE116250 and GSE55296 only, and none in GSE120852 and GSE55296 only. Gene counts indicate dataset membership rather than per-cohort significance. (Bottom right) Cross-species filtering. Of the 249 significant genes, 204 had one-to-one mouse orthologs and 178 were expressed in GSE236374. Directionally concordant genes (91 upregulated, 19 downregulated) were retained as the UCIS-110 signature, whereas genes lacking detectable orthologs or showing discordant regulation were excluded.

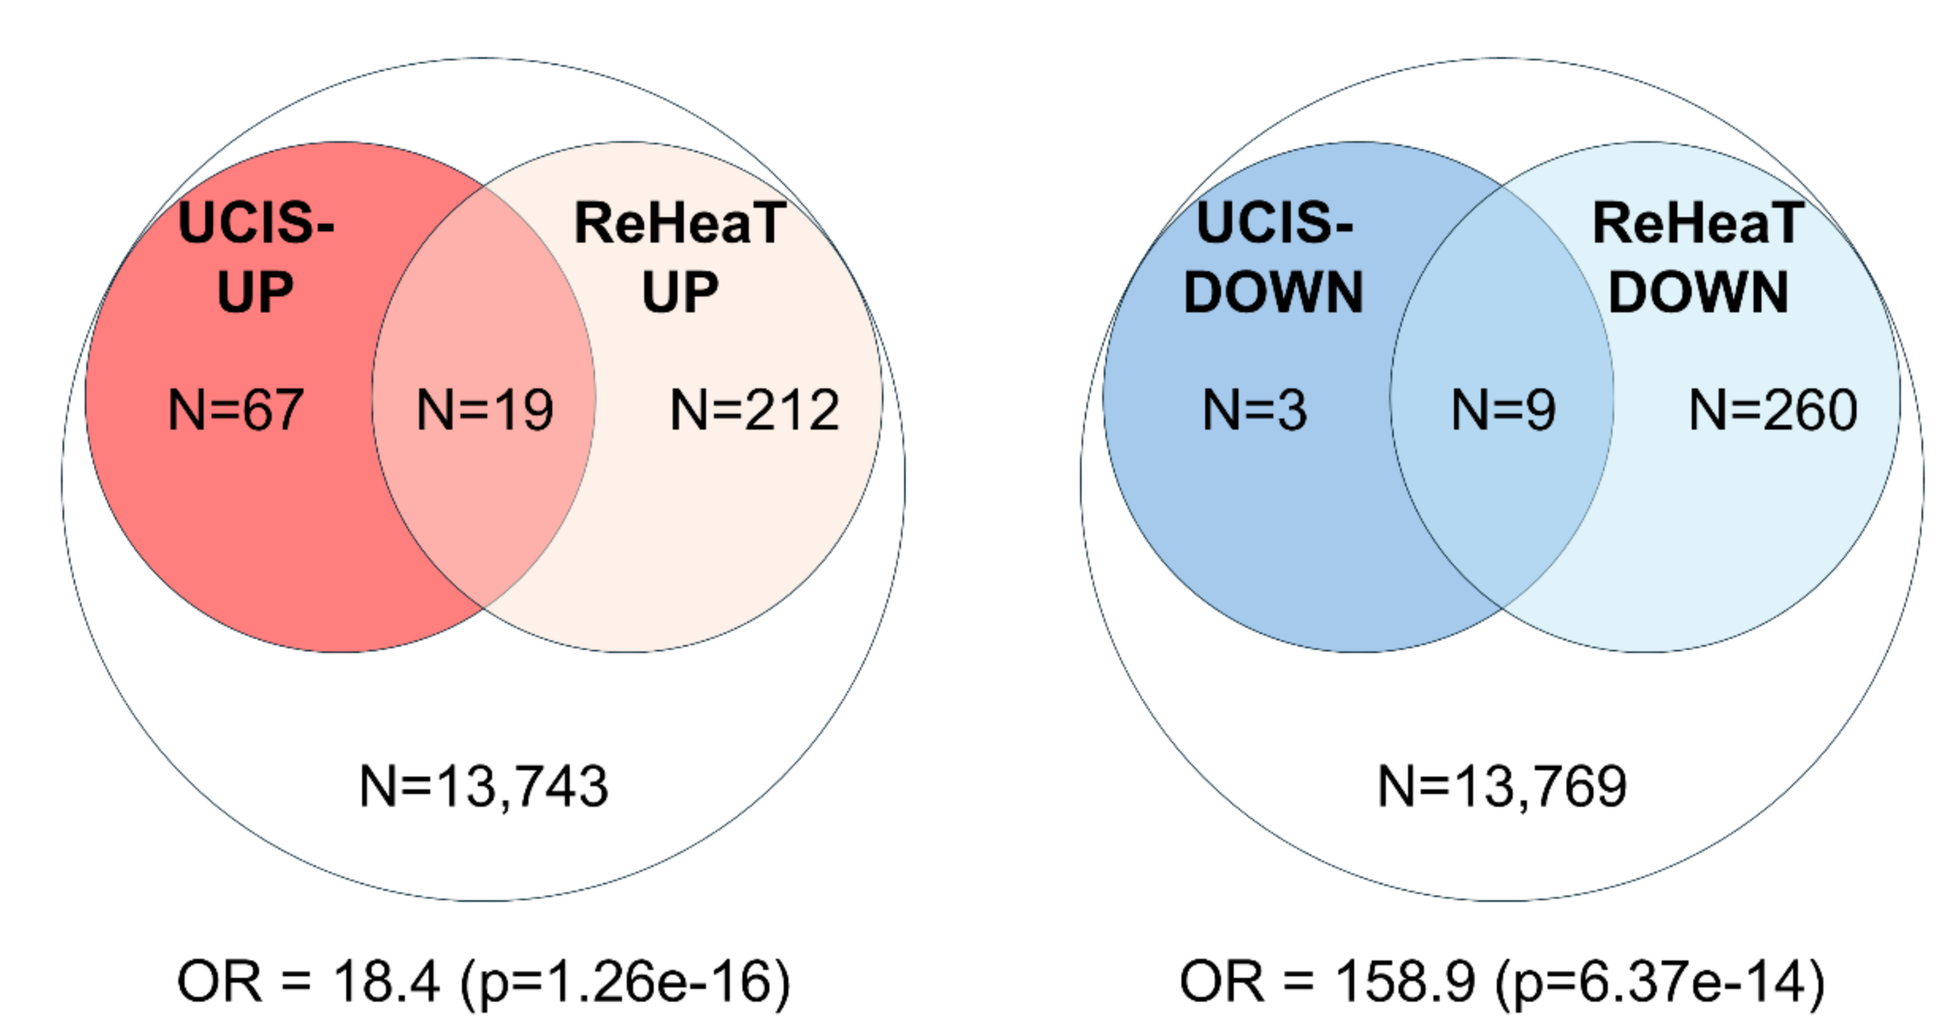
UCIS-UP
ReHeaT UP
N=67
N=19
N=212
N=13,743
OR = 18.4 (p=1.26e-16)
UCIS-DOWN
ReHeaT DOWN
N=3
N=9
N=260
N=13,769
OR = 158.9 (p=6.37e-14)

Figure 3. Benchmarking of UCIS-110 against the ReHeaTΔ14/18 heart failure consensus. UCIS-110 was compared with a non-overlapping ReHeaT reference generated after excluding the two discovery-overlapping cohorts, Sweet18 (GSE116250) and Tarazon14 (GSE55296). The resulting ReHeaTΔ14/18 consensus comprises the top 500 genes ranked by Fisher's combined P value across the remaining 14 cohorts (14,041-gene background), including 231 upregulated and 269 downregulated genes based on the mean $\log_2$ fold change.

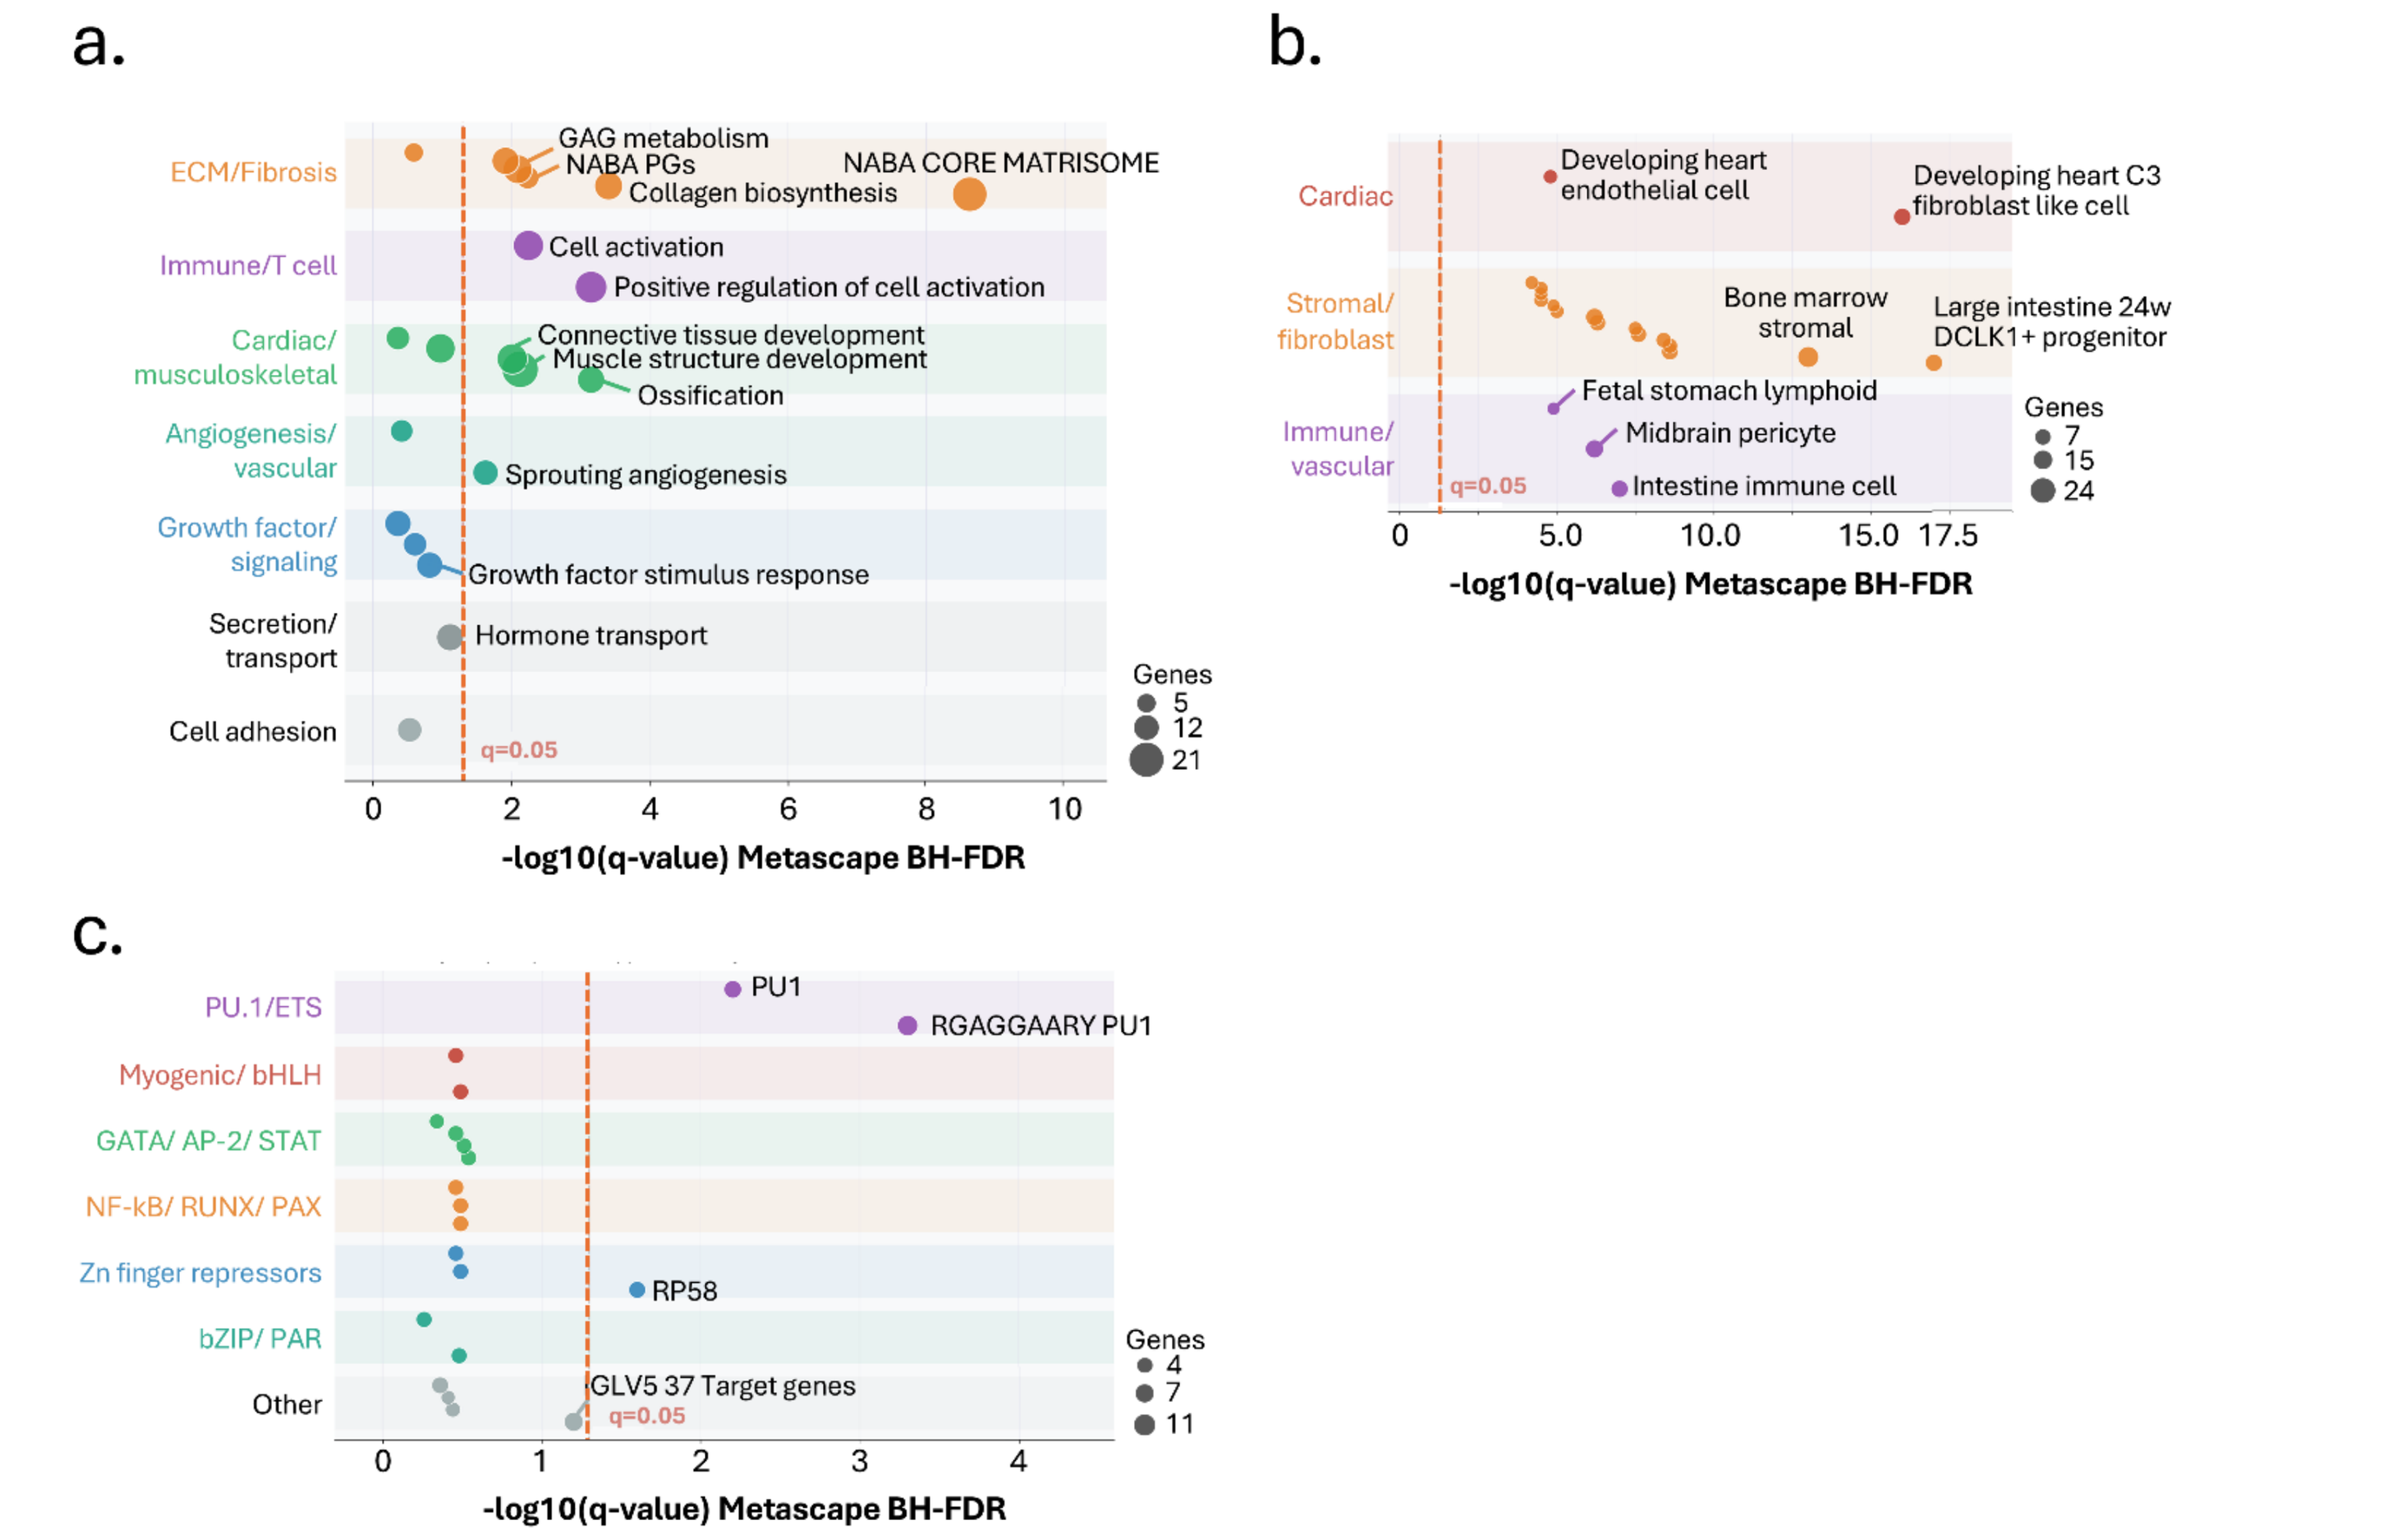
a.
ECM/Fibrosis
Immune/T cell
Cardiac/musculoskeletal
Angiogenesis/vascular
Growth factor/signaling
Secretion/transport
Cell adhesion
GAG metabolism
NABA PGs
Collagen biosynthesis
NABA CORE MATRISOME
Cell activation
Positive regulation of cell activation
Connective tissue development
Muscle structure development
Ossification
Sprouting angiogenesis
Growth factor stimulus response
Hormone transport
q=0.05
Genes
-log10(q-value) Metascape BH-FDR
b.
Cardiac
Stromal/fibroblast
Immune/vascular
Developing heart endothelial cell
Developing heart C3 fibroblast like cell
Bone marrow stromal
Large intestine 24w DCLK1+ progenitor
Fetal stomach lymphoid
Midbrain pericyte
Intestine immune cell
c.
PU.1/ETS
Myogenic/ bHLH
GATA/ AP-2/ STAT
NF-kB/ RUNX/ PAX
Zn finger repressors
bZIP/ PAR
Other
PU1
RGAGGAARY PU1
RP58
GLV5 37 Target genes

Figure 4. Pathway, cell-type, and transcription factor enrichment analysis of the UCIS-110 upregulated arm. (a) Metascape overrepresentation analysis (ORA) of the 91 upregulated UCIS-110 genes showing the top 20 non-redundant pathway clusters, grouped into seven biological themes: extracellular matrix/fibrosis, immune and T-cell activation, cardiac and musculoskeletal remodeling, angiogenesis and vascular remodeling, growth factor and morphogen signaling, secretion and transport, and cell-cell adhesion. Bubble size indicates the number of genes per cluster and color represents $-\log_{10}(q)$. The dashed red line marks the significance threshold (q = 0.05). Three additional clusters below the plotting range are indicated by stacked dots. The 19 downregulated genes showed no significant enrichment and are not shown. (b) Metascape cell-type enrichment analysis of the 91 upregulated genes. The top 20 enriched cell types are grouped into cardiac, stromal/fibroblast-like, and immune/vascular classes. Stromal and fibroblast-like cell types were most strongly enriched, indicating that the UCIS-110 upregulated signature predominantly reflects activated fibroblast and stromal programs. Bubble size and color are as in (a). (c) MSigDB transcription factor motif enrichment of the 91 upregulated genes. The top 20 enriched motifs are grouped into seven transcription factor families. PU.1/ETS motifs were the strongest enrichment, with additional enrichment of RP58 and other transcription factor families. Bubble size and color are as in (a), and the dashed red line denotes q = 0.05.

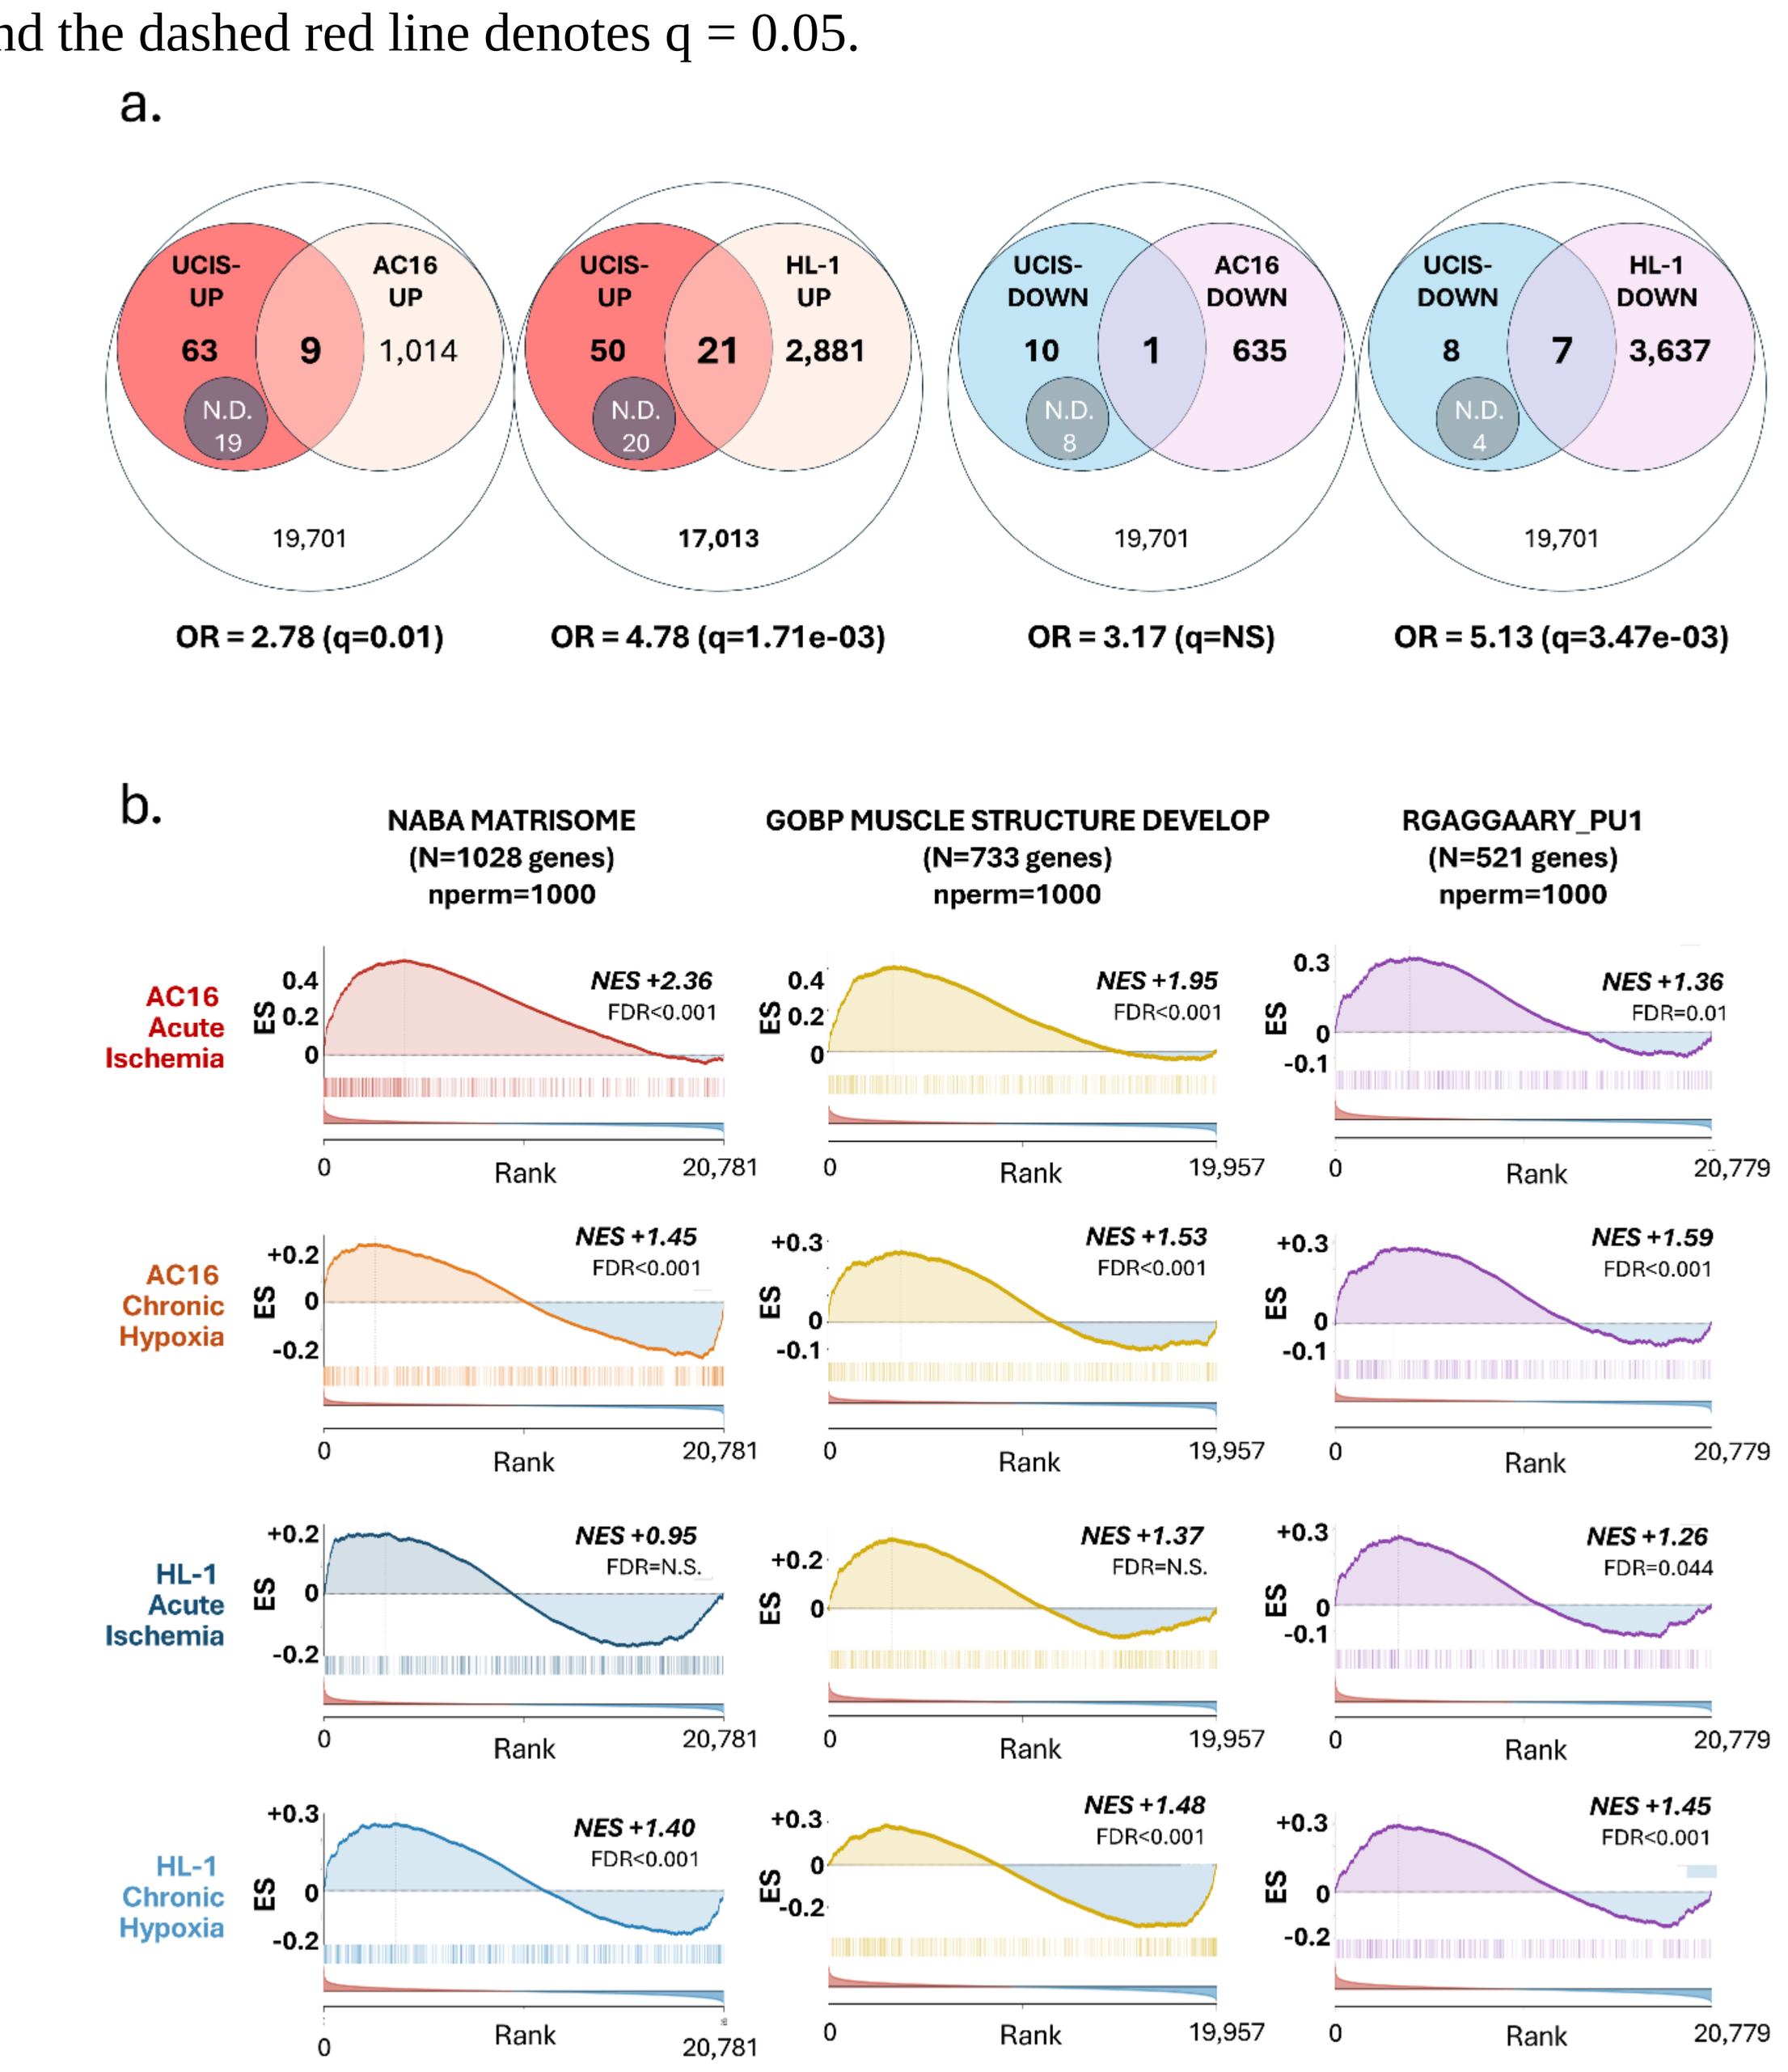
a.
UCIS-UP
AC16 UP
63
9
1,014
N.D. 19
19,701
OR = 2.78 (q=0.01)
UCIS-UP
HL-1 UP
50
21
2,881
N.D. 20
17,013
OR = 4.78 (q=1.71e-03)
UCIS-DOWN
AC16 DOWN
10
1
635
N.D. 8
19,701
OR = 3.17 (q=NS)
UCIS-DOWN
HL-1 DOWN
8
7
3,637
N.D. 4
19,701
OR = 5.13 (q=3.47e-03)
b.
NABA MATRISOME (N=1028 genes) nperm=1000
GOBP MUSCLE STRUCTURE DEVELOP (N=733 genes) nperm=1000
RGAGGAARY_PU1 (N=521 genes) nperm=1000
AC16 Acute Ischemia
AC16 Chronic Hypoxia
HL-1 Acute Ischemia
HL-1 Chronic Hypoxia
NES +2.36 FDR<0.001
NES +1.95 FDR<0.001
NES +1.36 FDR=0.01
NES +1.45 FDR<0.001
NES +1.53 FDR<0.001
NES +1.59 FDR<0.001
NES +0.95 FDR=N.S.
NES +1.37 FDR=N.S.
NES +1.26 FDR=0.044
NES +1.40 FDR<0.001
NES +1.48 FDR<0.001
NES +1.45 FDR<0.001
Rank
ES

Figure 5. Gene-level and pathway-level verification of UCIS-110 in AC16 and HL-1 cardiomyocyte ischemia models. (a) Gene-level validation. Venn diagrams show the overlap between UCIS-110 genes and concordantly differentially expressed genes in AC16 and HL-1 cardiomyocytes, separated into upregulated (91 genes) and downregulated (19 genes) arms. The dashed N.D. region denotes UCIS genes not detected in the corresponding cell line and therefore excluded from concordance testing. Overlapping regions indicate UCIS genes that were significantly and directionally concordant (DESeq2 BH-FDR q < 0.25) in at least one ischemic condition (acute or chronic). Enrichment was assessed by one-sided Fisher's exact test with BH-FDR correction. (b) Pathway-level validation by pre-ranked GSEA. Enrichment curves for three external gene sets—NABA Matrisome, GO Muscle Structure Development, and PU.1/SPI1 target genes—across four conditions: AC16 acute ischemia (1% O_2, 24 h), AC16 chronic hypoxia (1% O_2, 30 days), HL-1 acute ischemia (1% O_2, 24 h), and HL-1 chronic hypoxia (1% O_2, 30 days). Genes were ranked by the DESeq2 Wald statistic. Normalized enrichment score (NES) and FDR are shown in each panel.