FROM DRONE BLAST TO STABILIZATION:
Evaluation of SEAL Aerosolized Chitosan Platform for Rapid Burn Control on the Battlefield

BC3 Technologies, Baltimore, MD, USA

THE PROBLEM:
Drone blast is now the dominant casualty mechanism in modern conflict.

A single blast event produces simultaneous penetrating hemorrhage, fragmentation wounds with complex geometry, and thermal injury. No single currently fielded hemostatic product addresses all three simultaneously. Extended evacuation timelines can push tourniquet dwell times beyond the traditional two-hour safe ischemic limit, increasing limb ischemia, nerve damage, and amputation risk.

The battlefield requires one product that controls hemorrhage, arrests burn progression, reaches blast wound geometry gauze cannot access, and enables safe tourniquet conversion with no training.

BLASTS AND BURN PROGRESSION

The classical burn injury model comprises three concentric zones of tissue damage: a central zone of necrosis, a surrounding zone of stasis, and an outer zone of hyperemia. The zone of stasis may progress to full-thickness necrosis within 24–48 h post-burn.

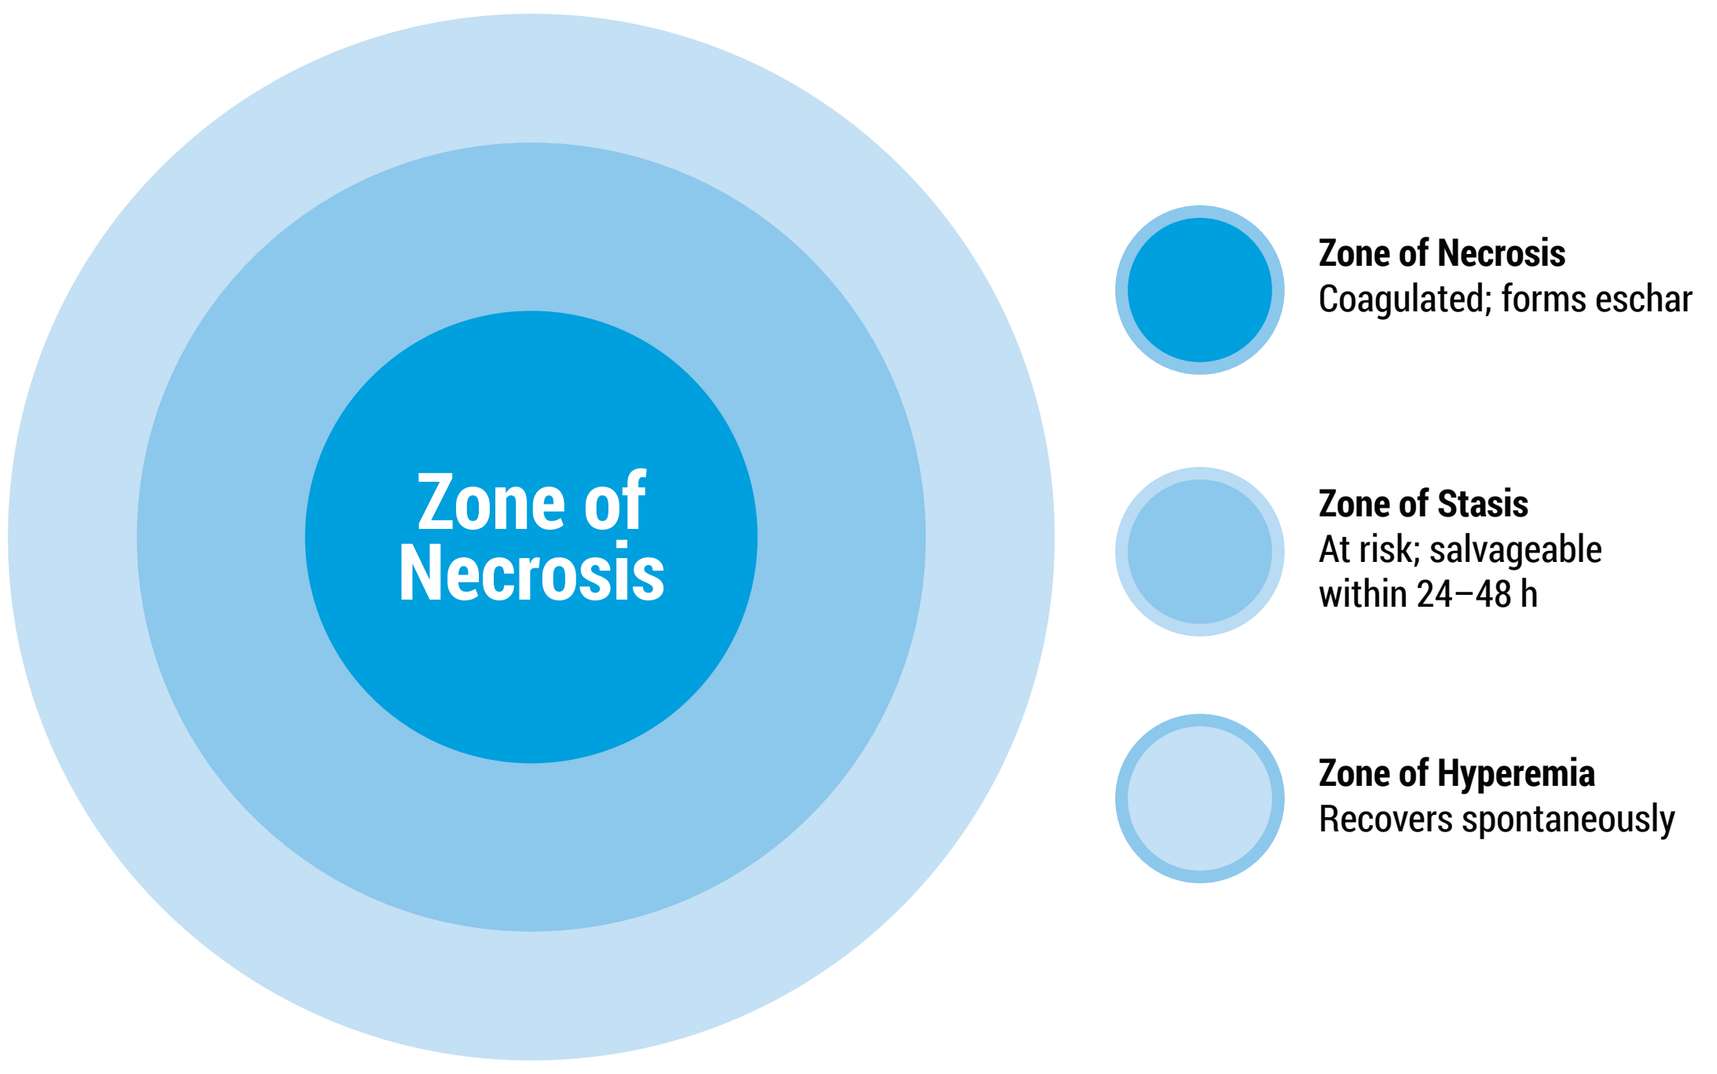
Zone of Necrosis
Zone of Necrosis
Coagulated; forms eschar
Zone of Stasis
At risk; salvageable within 24–48 h
Zone of Hyperemia
Recovers spontaneously

Figure 1: Classical three-zone model of burn injury. The zone of stasis is the primary target for early intervention.

Early cooling and wound-bed stabilization are therefore central to preserving viable tissue.

WHY SEAL?

SEAL is a chitosan-based aerosol that immediately cools the wound, adheres as a protective hemostatic and antimicrobial layer, and provides synergistic benefit in blast-associated burn wounds with traumatic hemorrhage by addressing cooling, infection control, and bleeding in a single application.

- **Immediate surface cooling** to dissipate residual heat and preserve the zone of stasis.
- **Durable chitosan adherence** forming a hemostatic and antimicrobial barrier.
- **Aerosol delivery** reaching deep, irregular, and junctional wounds unreachable by gauze.
- **Enhanced burn wound healing** through support of tissue regeneration and repair.

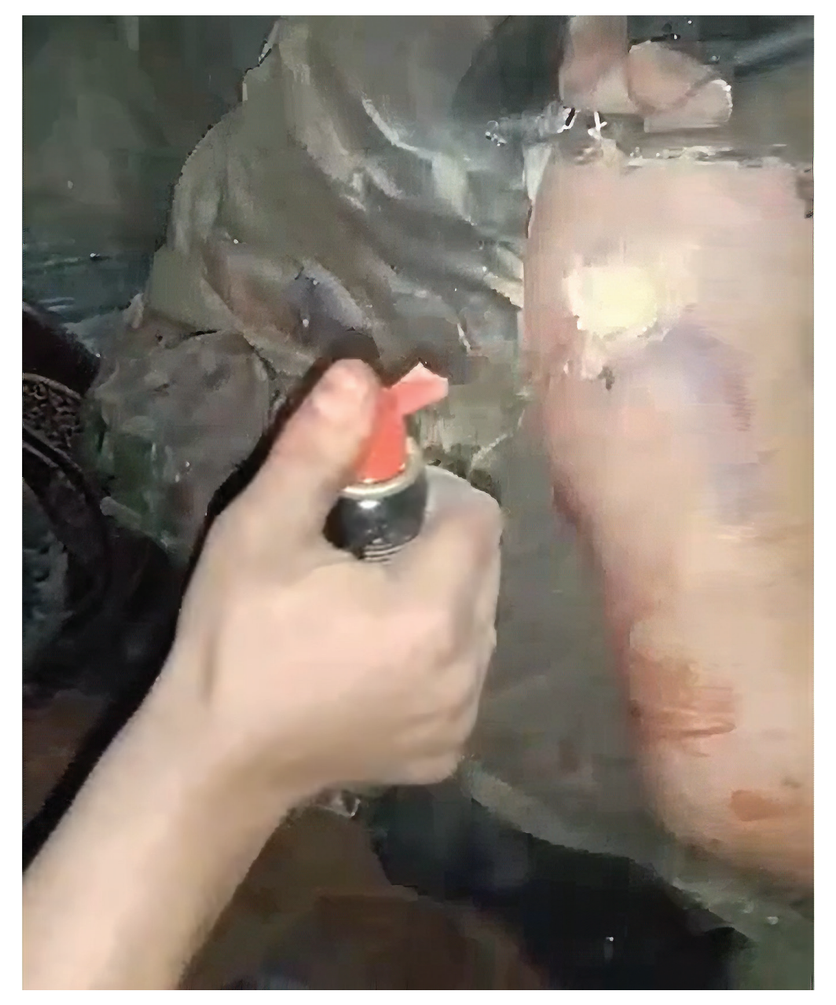

Figure 2. SEAL in Ukraine battlefield application.

MATERIALS AND METHODS

Full-thickness burns were created with a 3 cm stainless-steel device heated to 200 ± 10 °C and applied for 40 seconds. Wounds were randomized to SEAL or Triple Helix Collagen Powder control, with biopsies collected at days 14, 28, and 60.

RESULTS

MACROSCOPIC WOUND HEALING

Both SEAL and control wounds showed progressive improvement at all assessment time-points. In comparison to control, key outcomes of SEAL included:

- **More organized healing:** SEAL wounds looked more uniform with cleaner margins and fewer/ smaller necrotic crusts from day 14 onward.
- **Earlier surface recovery:** First signs of re-epithelialization appeared in SEAL by day 28 (none in controls at that point).

Animal/Wound/Product	Beginning Day 0	Day 10	Day 25	Day 40	End Day 60
1/1/SEAL					
1/3/Control					

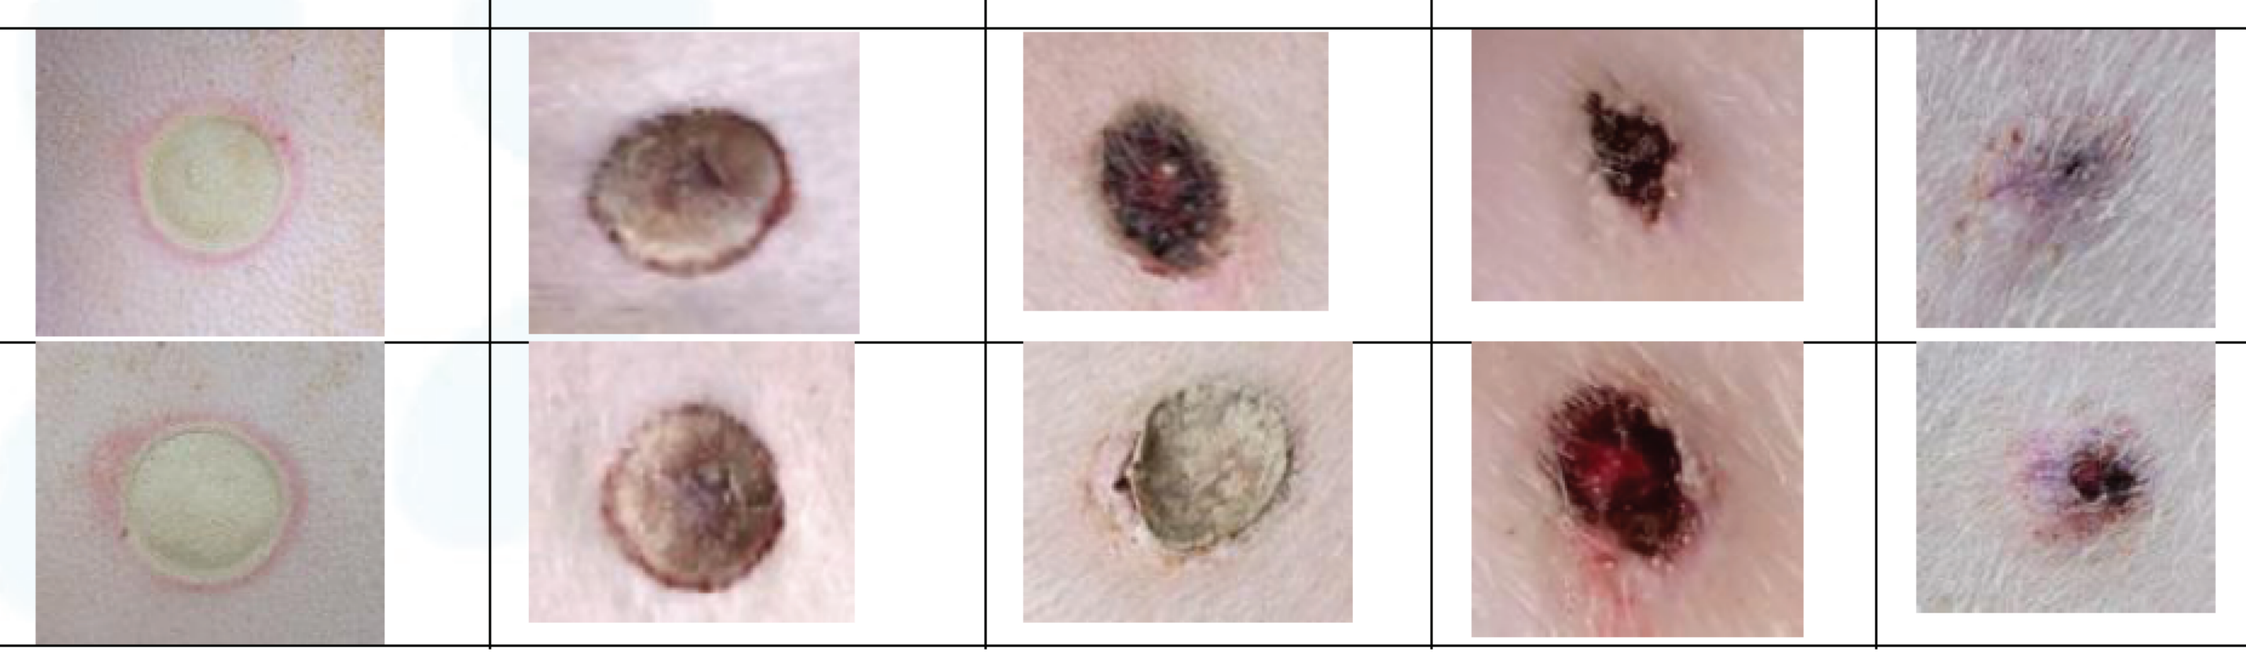

Figure 3: Representative photographs of the burn wound healing process. *Upper row:* SEAL-treated wound. *Lower row:* Collagen-treated control wound. Images show progressive healing over 60 days.

HISTOPATHOLOGY

SEAL was non-irritant and biocompatible, with no evidence of necrosis, abscess, abnormal fibrosis, or local toxicity. Compared to control, key outcomes of SEAL included:

- **Faster calming of inflammation:** By day 60, SEAL showed lower residual polymorphonuclear neutrophils and macrophages, indicating quicker resolution of acute inflammation.
- **Tissue quality trends:** Slightly higher angiogenesis at day 60 and less fatty infiltrate at day 28 with SEAL, while fibrosis/scar maturation matched controls.

INTERPRETATION

SEAL supports rapid, orderly tissue regeneration and efficient closure of severe full-thickness burns. Immediate cooling on deployment, durable chitosan adhesion, and a favorable inflammatory profile suggest potential preservation of the burn zone of stasis.

OPERATIONAL RELEVANCE

Blast injuries frequently combine burns, hemorrhage, contamination, and irregular wound geometry. SEAL is intended to support bleeding control, burn stabilization, and access to wound shapes that standard gauze may not easily reach.

The SEAL aerosolized chitosan platform is presented as usable from point of injury through prolonged field care with minimal user training:

CAPABILITY	MECHANISM	EVIDENCE
Hemorrhage Control	Coagulation cascade independent chitosan seal	**Swine femoral artery bleeding model:** 100% survival vs 75% Celox, 50% less blood loss
Burn Progression Arrest and Healing	Propellant evaporative cooling in recommended temperature range	**Porcine full-thickness burn study:** rapid wound closure, superior regeneration vs FDA-cleared collagen control
Blast Wound Management	Aerosol reaches deep, irregular, junctional wounds	MIL STD 810H certified, LTT confirmed in heparinized coagulopathic models
Tourniquet Conversion	Hemostatic seal applied as tourniquet pressure reduces	Actively used by Ukrainian combat medics. LTT confirmed skill level independence

VALIDATION AND FIELD USE

Ukrainian Air Assault Forces Command: formal military approval; more than 1,800 cans reportedly deployed in active combat operations.

Ukraine SOF live tissue training, November to December 2025: 24 personnel across three skill levels; findings supported skill-level independence and promising time-to-hemostasis performance.

UAE military live tissue training, Q1 2026: SEAL reportedly controlled bleeding across all test cases, while Combat Gauze rebled on removal.

CONCLUSION:

SEAL combines aerosolized chitosan hemostasis with immediate cooling in a single platform. Preclinical burn data and operational use reports support continued clinical, regulatory, and defense evaluation. The product is positioned as a single-agent option for hemorrhage control, burn progression arrest, blast wound management, and tourniquet conversion.

Contact BC3 Technologies
Wayne Grube, CEO: waynegrube@bc3tech.com
Arthur Rosen, CFO: art@bc3tech.com
Rivelino Montenegro, Head Regulatory Affairs: rivelino@bc3tech.com
bc3tech.com | sealwoundcare.com

SEAL
HEMOSTATIC WOUND SPRAY