DAISY™ Self-administrable Therapeutic Applicator

DISC

DAISY™ Benefits

DAISY™ is the perfect platform for mass therapeutics administration, from vaccines and chemical/biological countermeasures to pain management and more.

Self-administrable

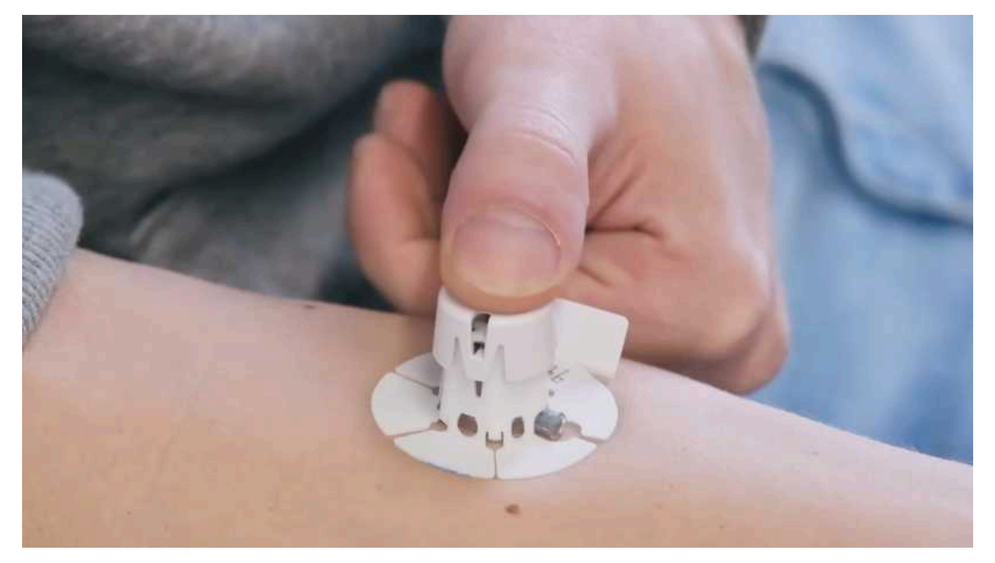

Eliminates healthcare provider administration

Dose Sparing Capabilities

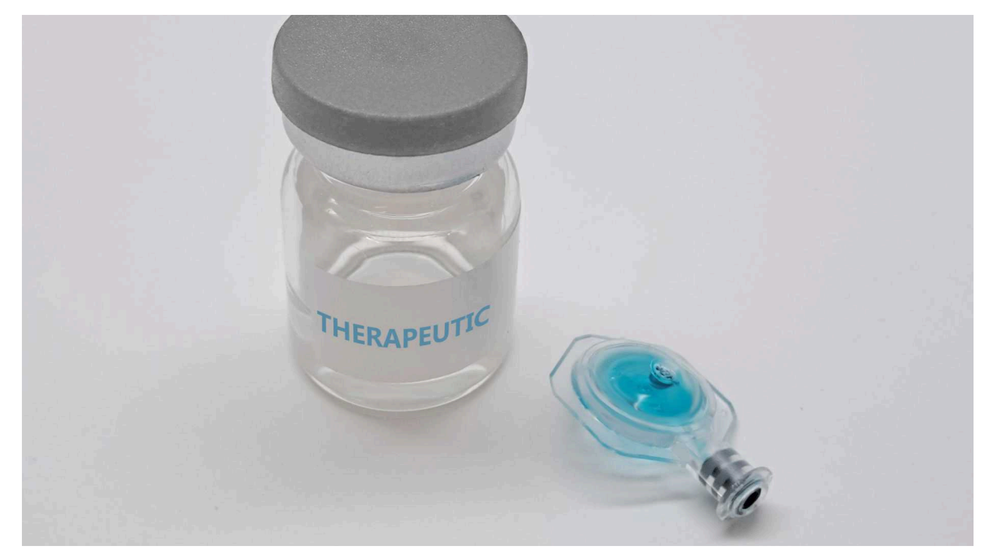

THERAPEUTIC

1/5th - 1/10th volume of intramuscular vaccine dosage

Point-of-need Manufacturing

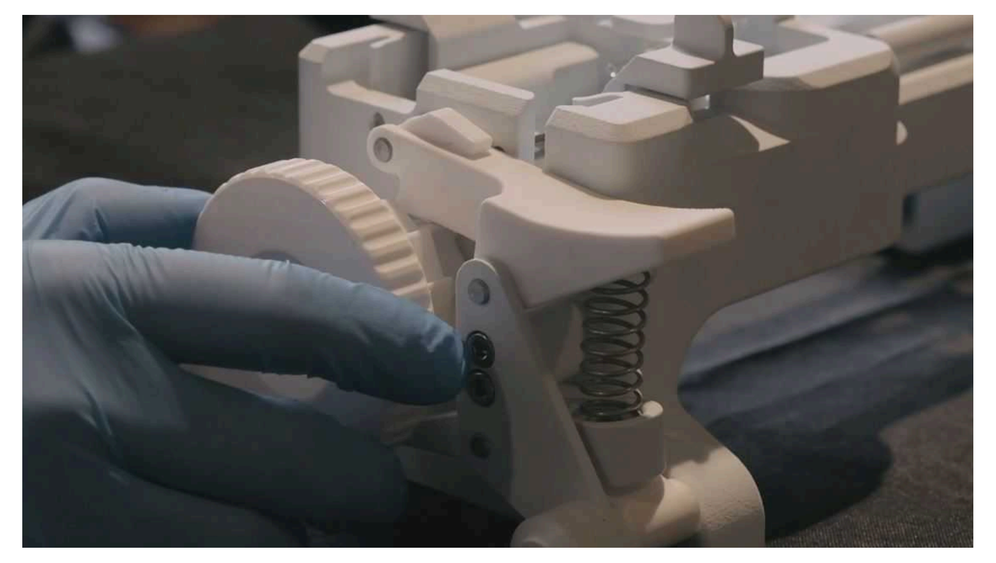

24 doses per 3mL vial

Mitigate Patient Fear

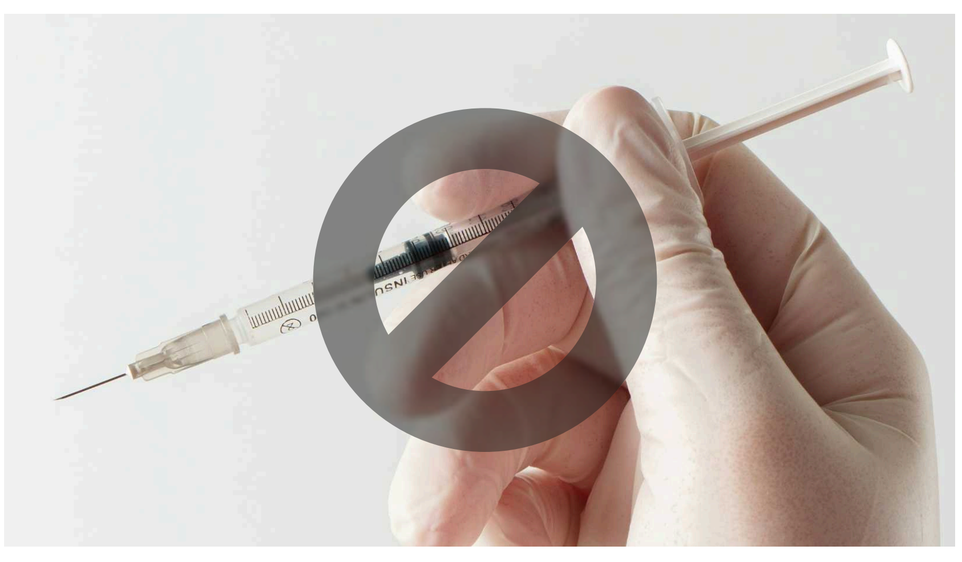

Pain-free delivery with microneedles

Logistical Efficiency

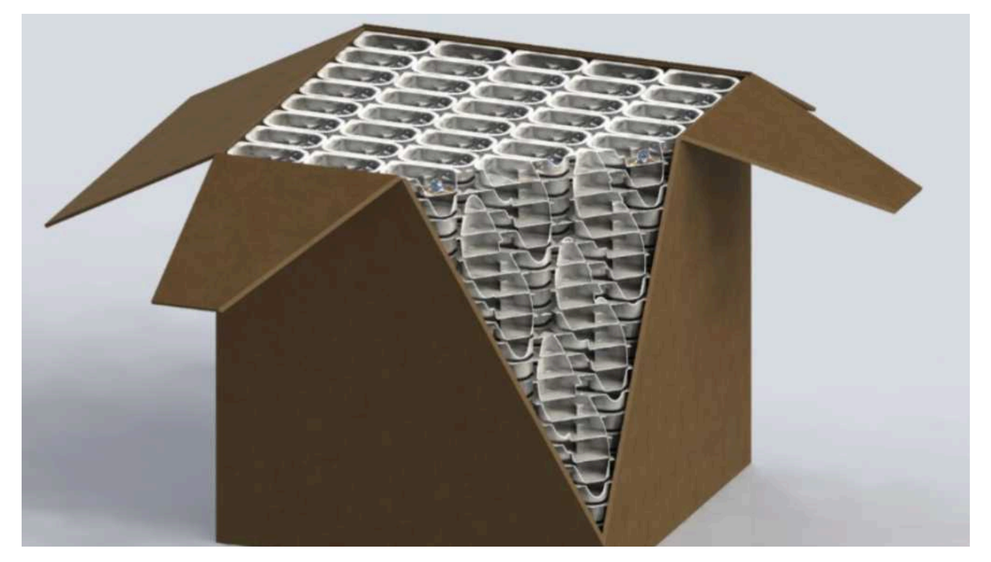

Standard logistics, no cold chain requirements

Less Overall Waste

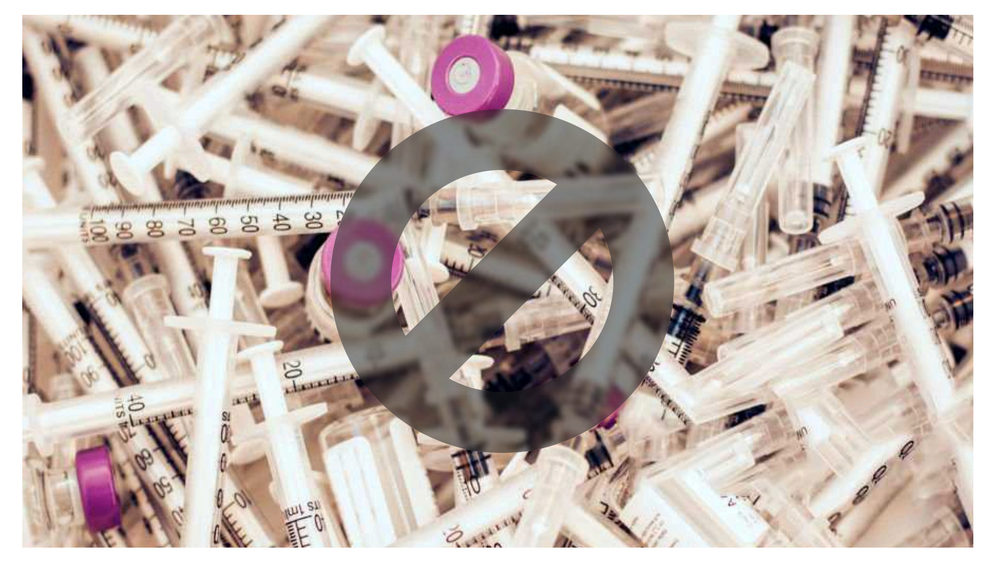

Fewer vials and syringes

Delivers Existing Formulations

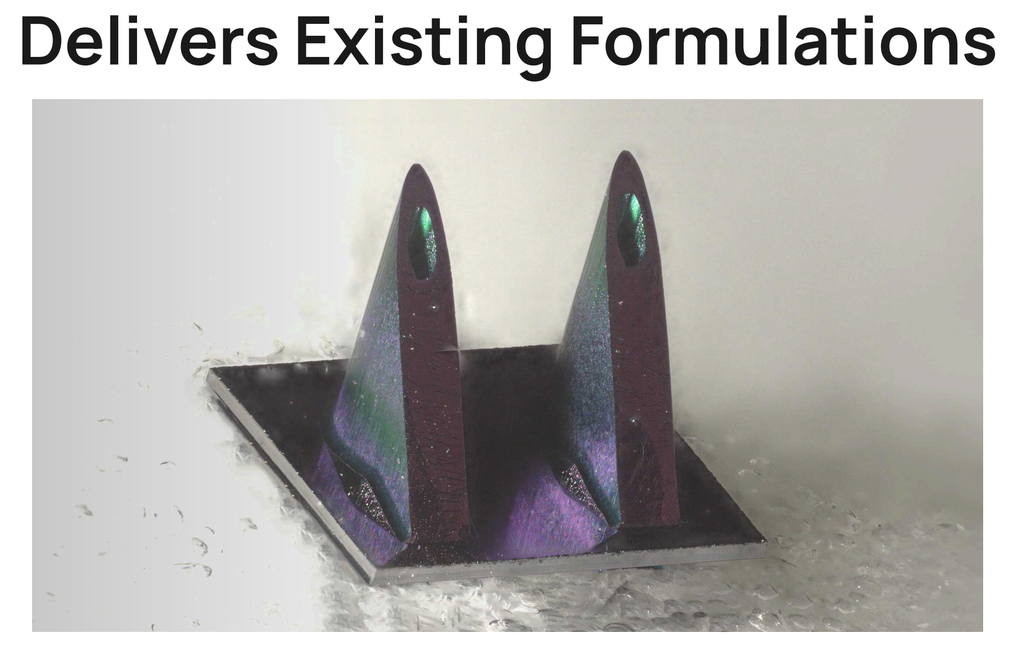

FDA cleared hollow silicone microneedle technology for intradermal delivery

Provide Traceability

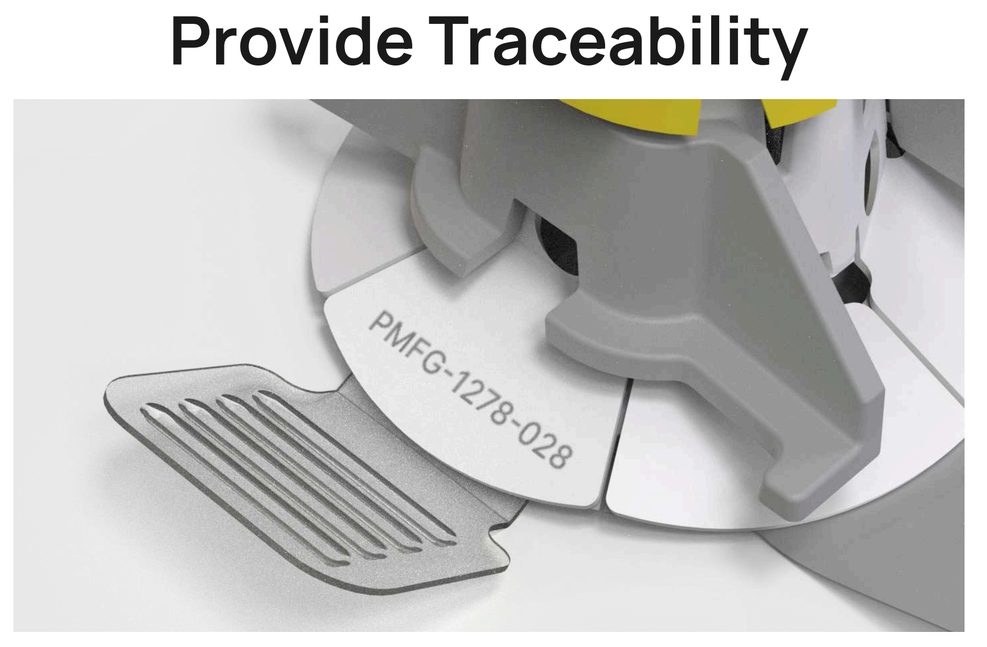

PMFG-1278-028

Each device receives its own unique identifier for tracking purposes

Simple, Reliable, Reproduceable

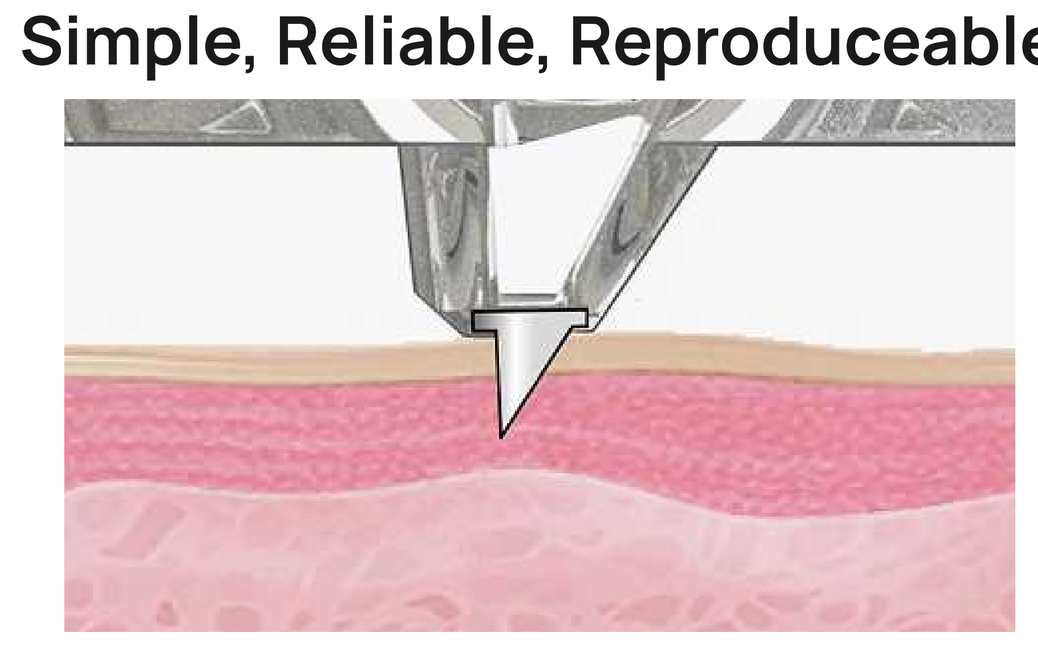

Consistent delivery into the dermal layer

DAISY™ Device

With DoW CARES Act funding, we built an intradermal therapeutics delivery device that can be self-administered, is virtually painless and can dispense existing liquid therapies.

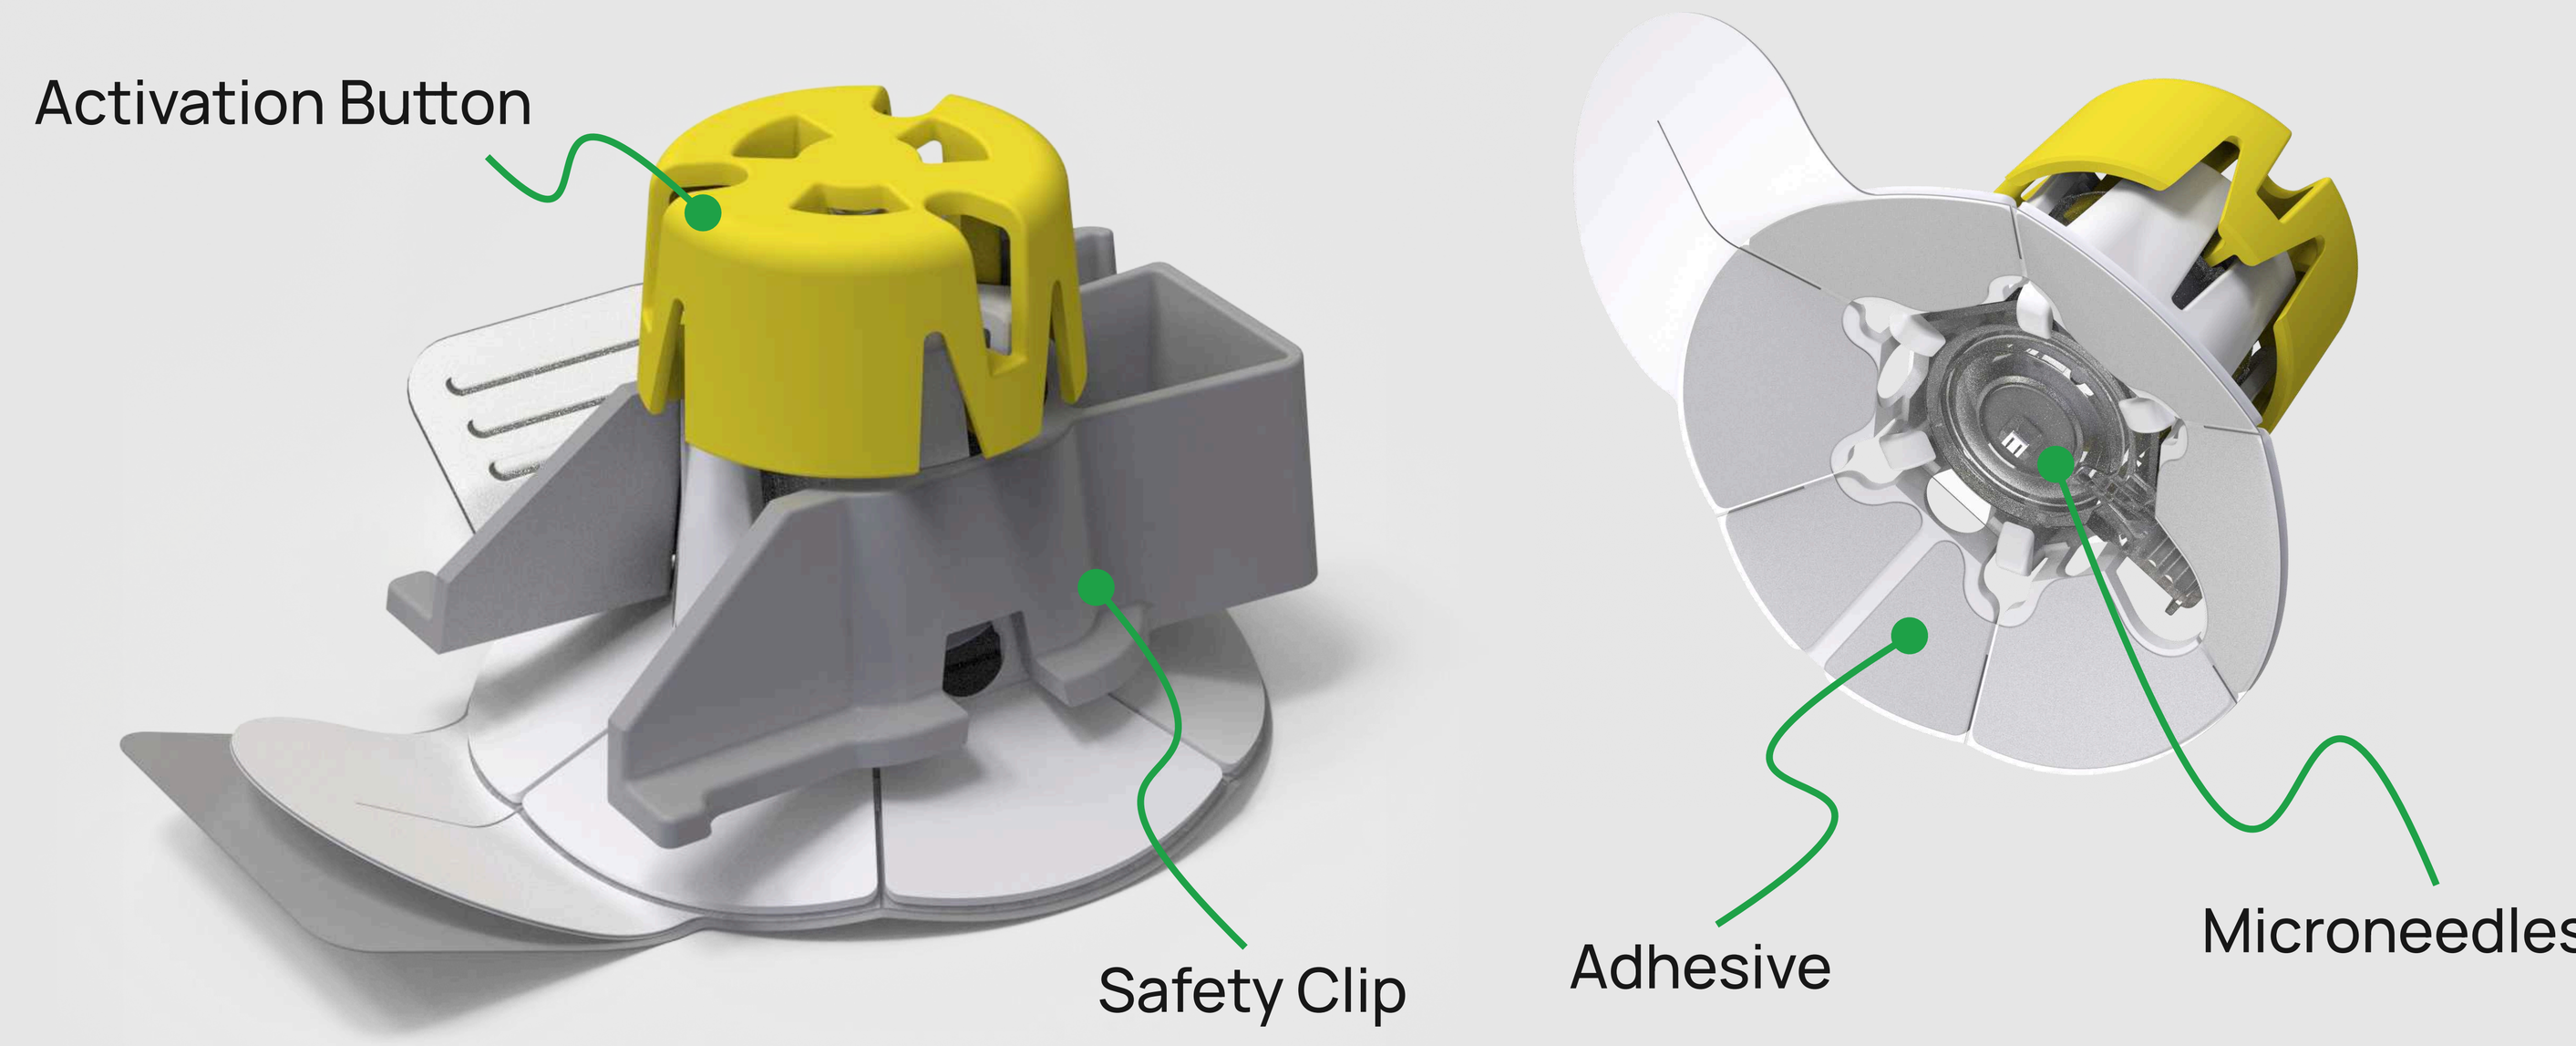

Activation Button
Safety Clip
Adhesive
Microneedles

Dimensions	1.75” D × 1.25” H (4.45 cm × 3.175 cm)
Weight	9g
Internal Reservoir Capacity	120µl
Delivery	100µl
Injection Depth	Intradermal
Delivery Time	<2minute
Shipping and Storage Temperature	-40°C to 60°C per ISTA 3A
Usage Temperature	5°C to 40°C
Delivery Mechanism	Manual press of the activation button
TRL/MRL	7/7

*Specification subject to change

DAISY™ Scalability

We built a prototype machine to demonstrate the scalability of the manufacturing process.

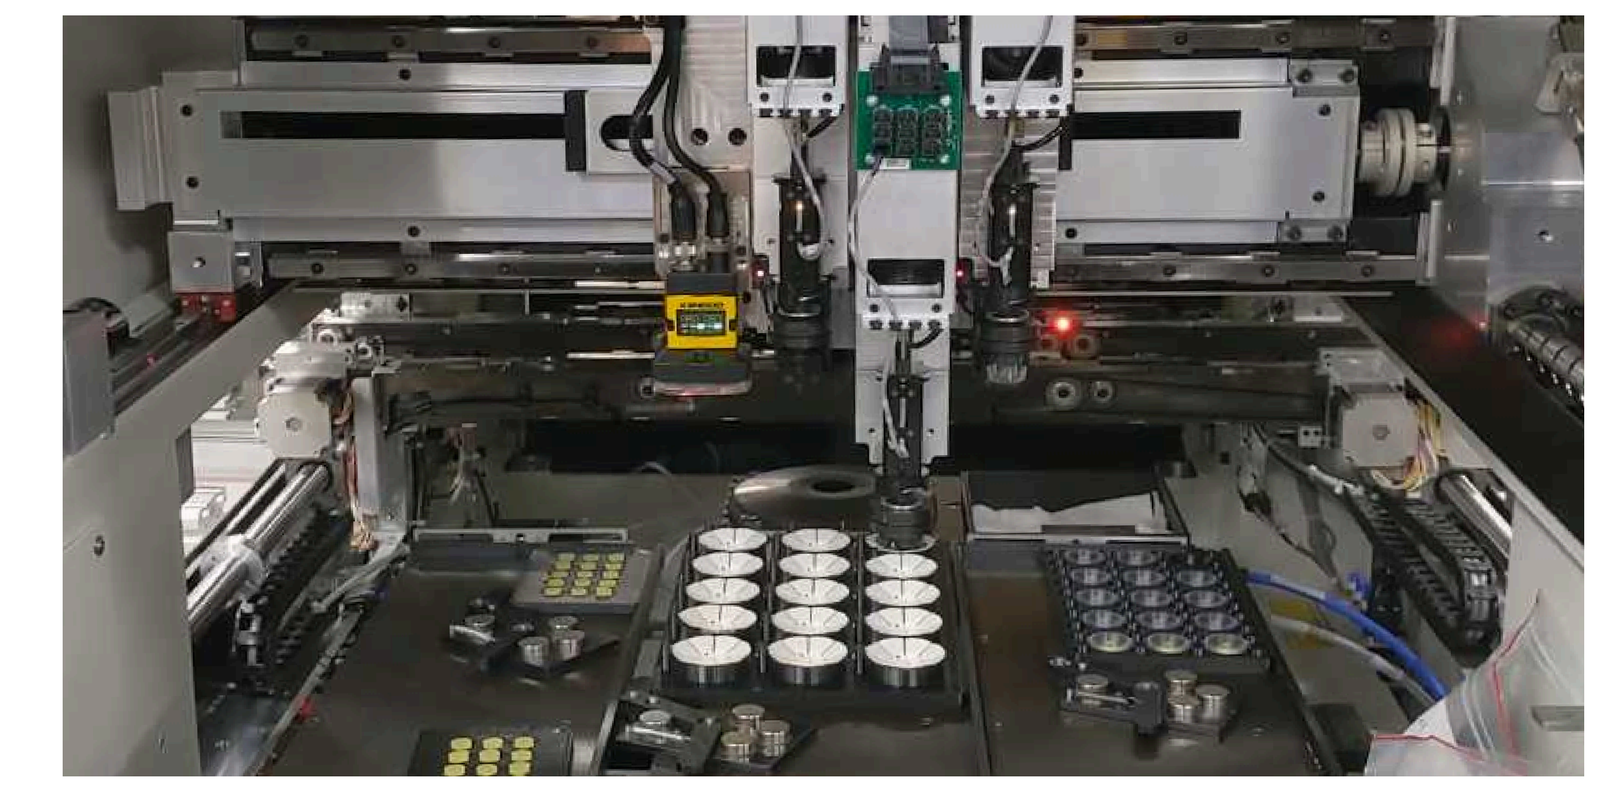

To produce the microneedles, we use a process similar to the semi-conductor industry’s process of making computer chips.

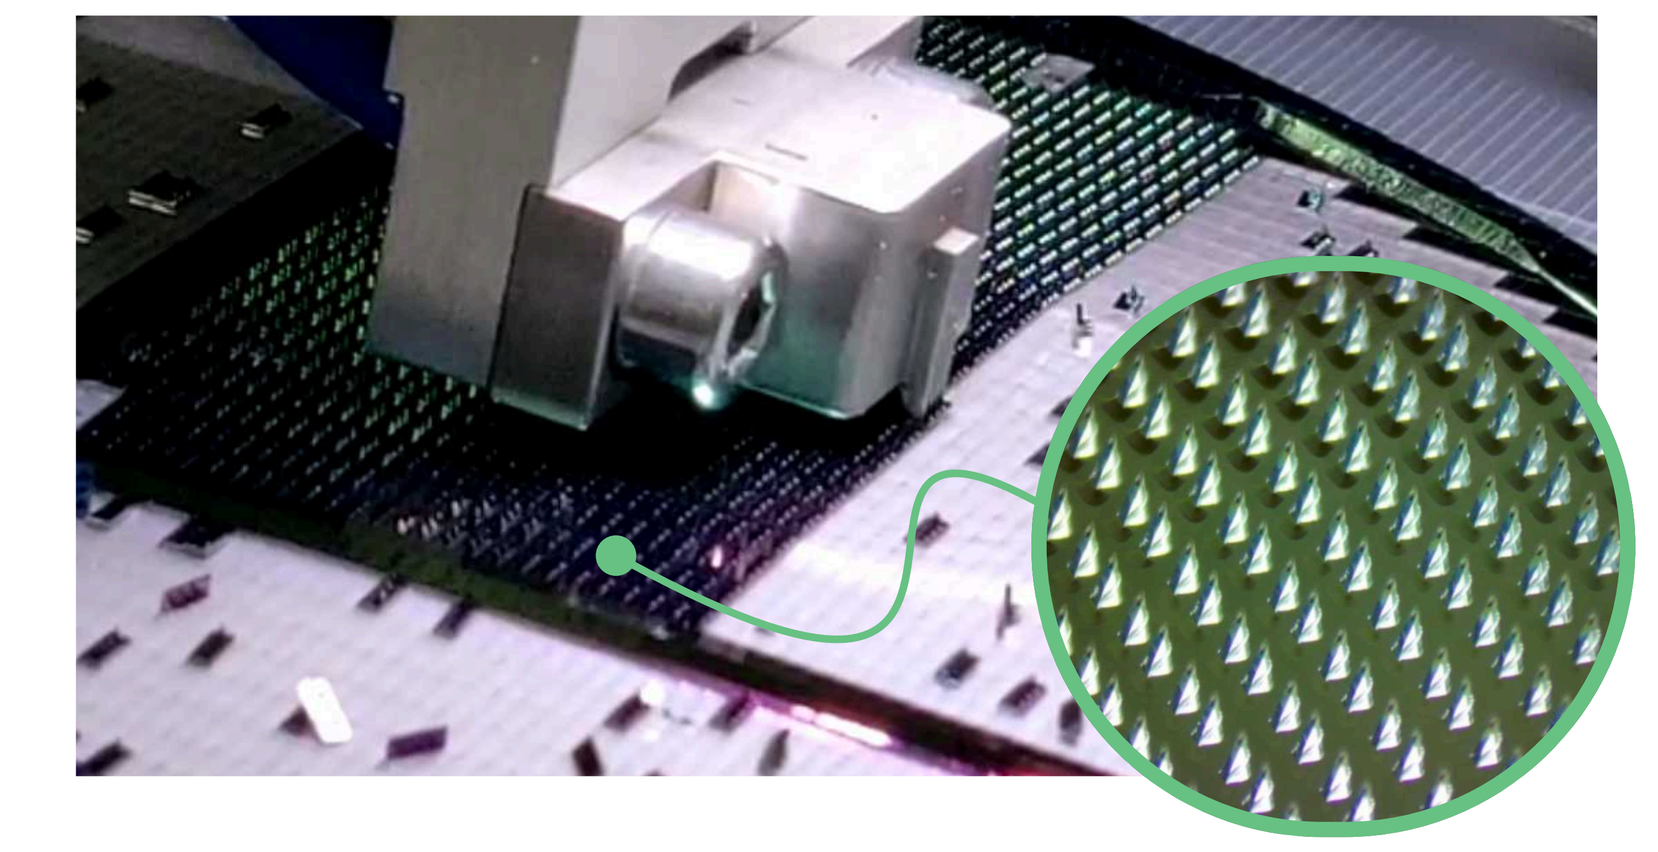

DAISY™ Uses Liquid Therapeutics

DAISY™ uses liquid therapeutics as opposed to coated or dissolving solid microneedle technology, eliminating the need for therapy reformulation.

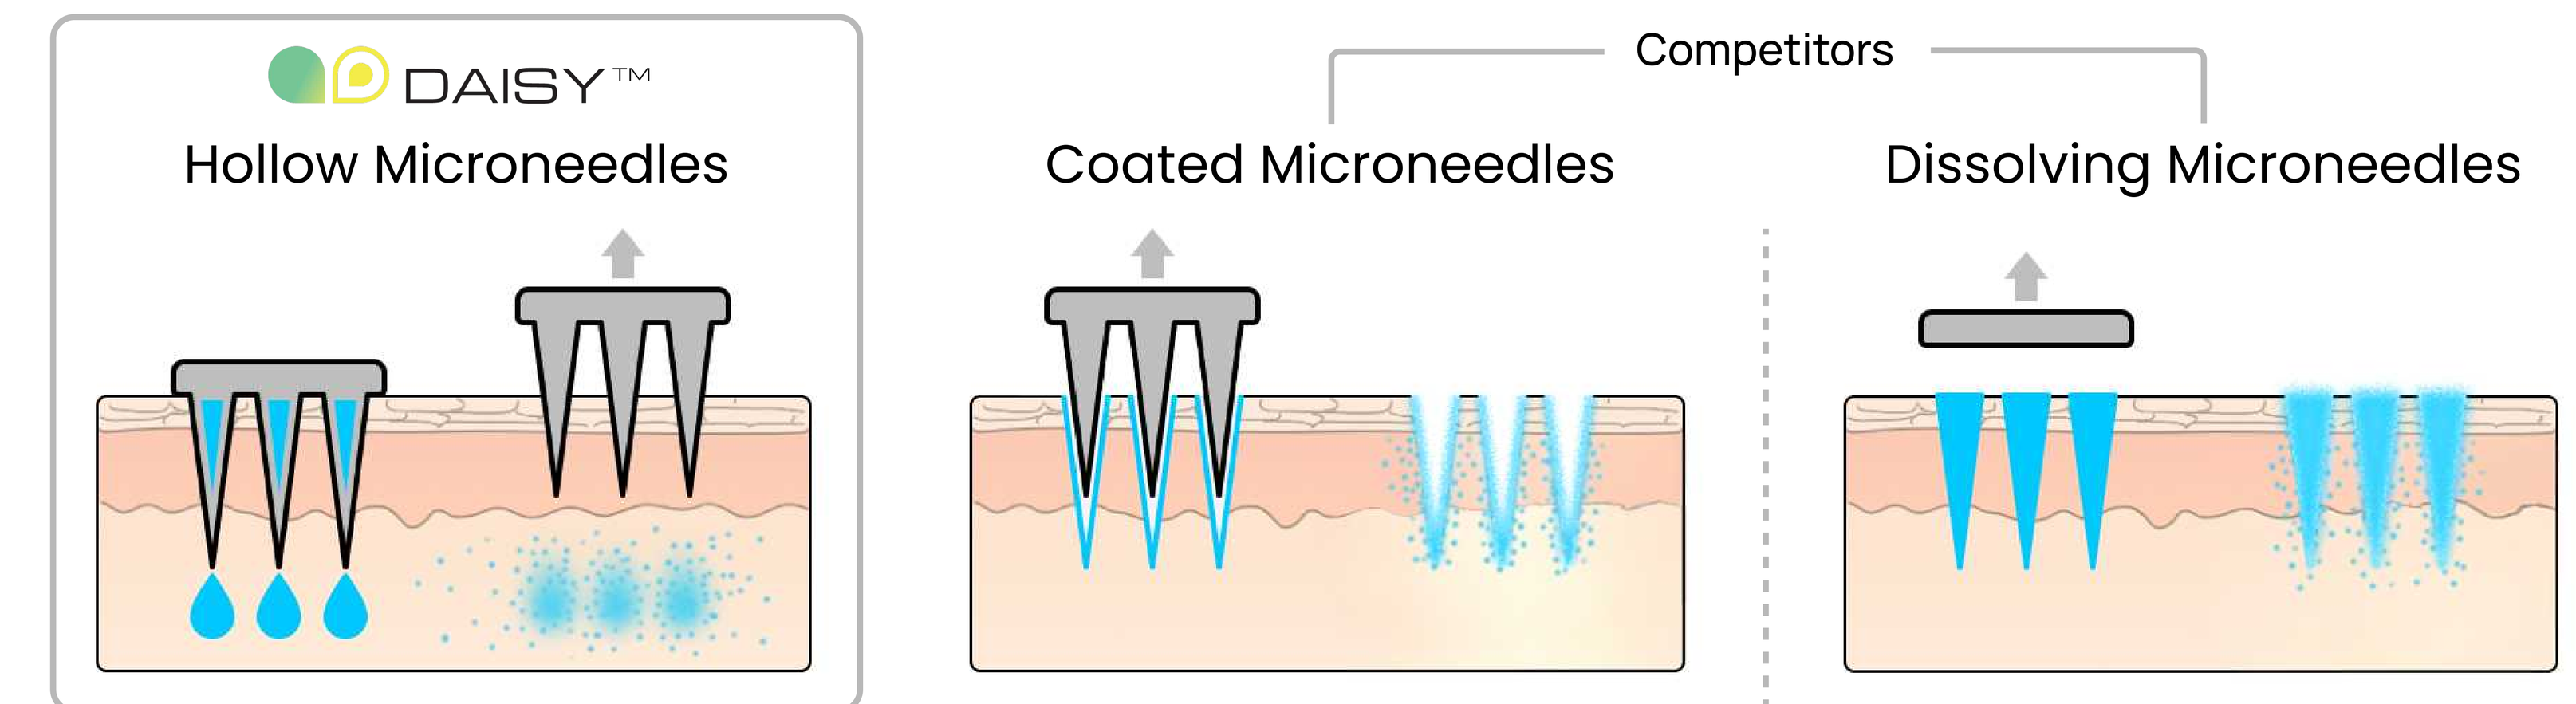

DAISY™
Hollow Microneedles
Competitors
Coated Microneedles
Dissolving Microneedles

DAISY™ utilizes FDA cleared hollow microneedle technology, allowing for precise dose control and better scalability.

DAISY™ Helps to Make Troop Deployment More Efficient

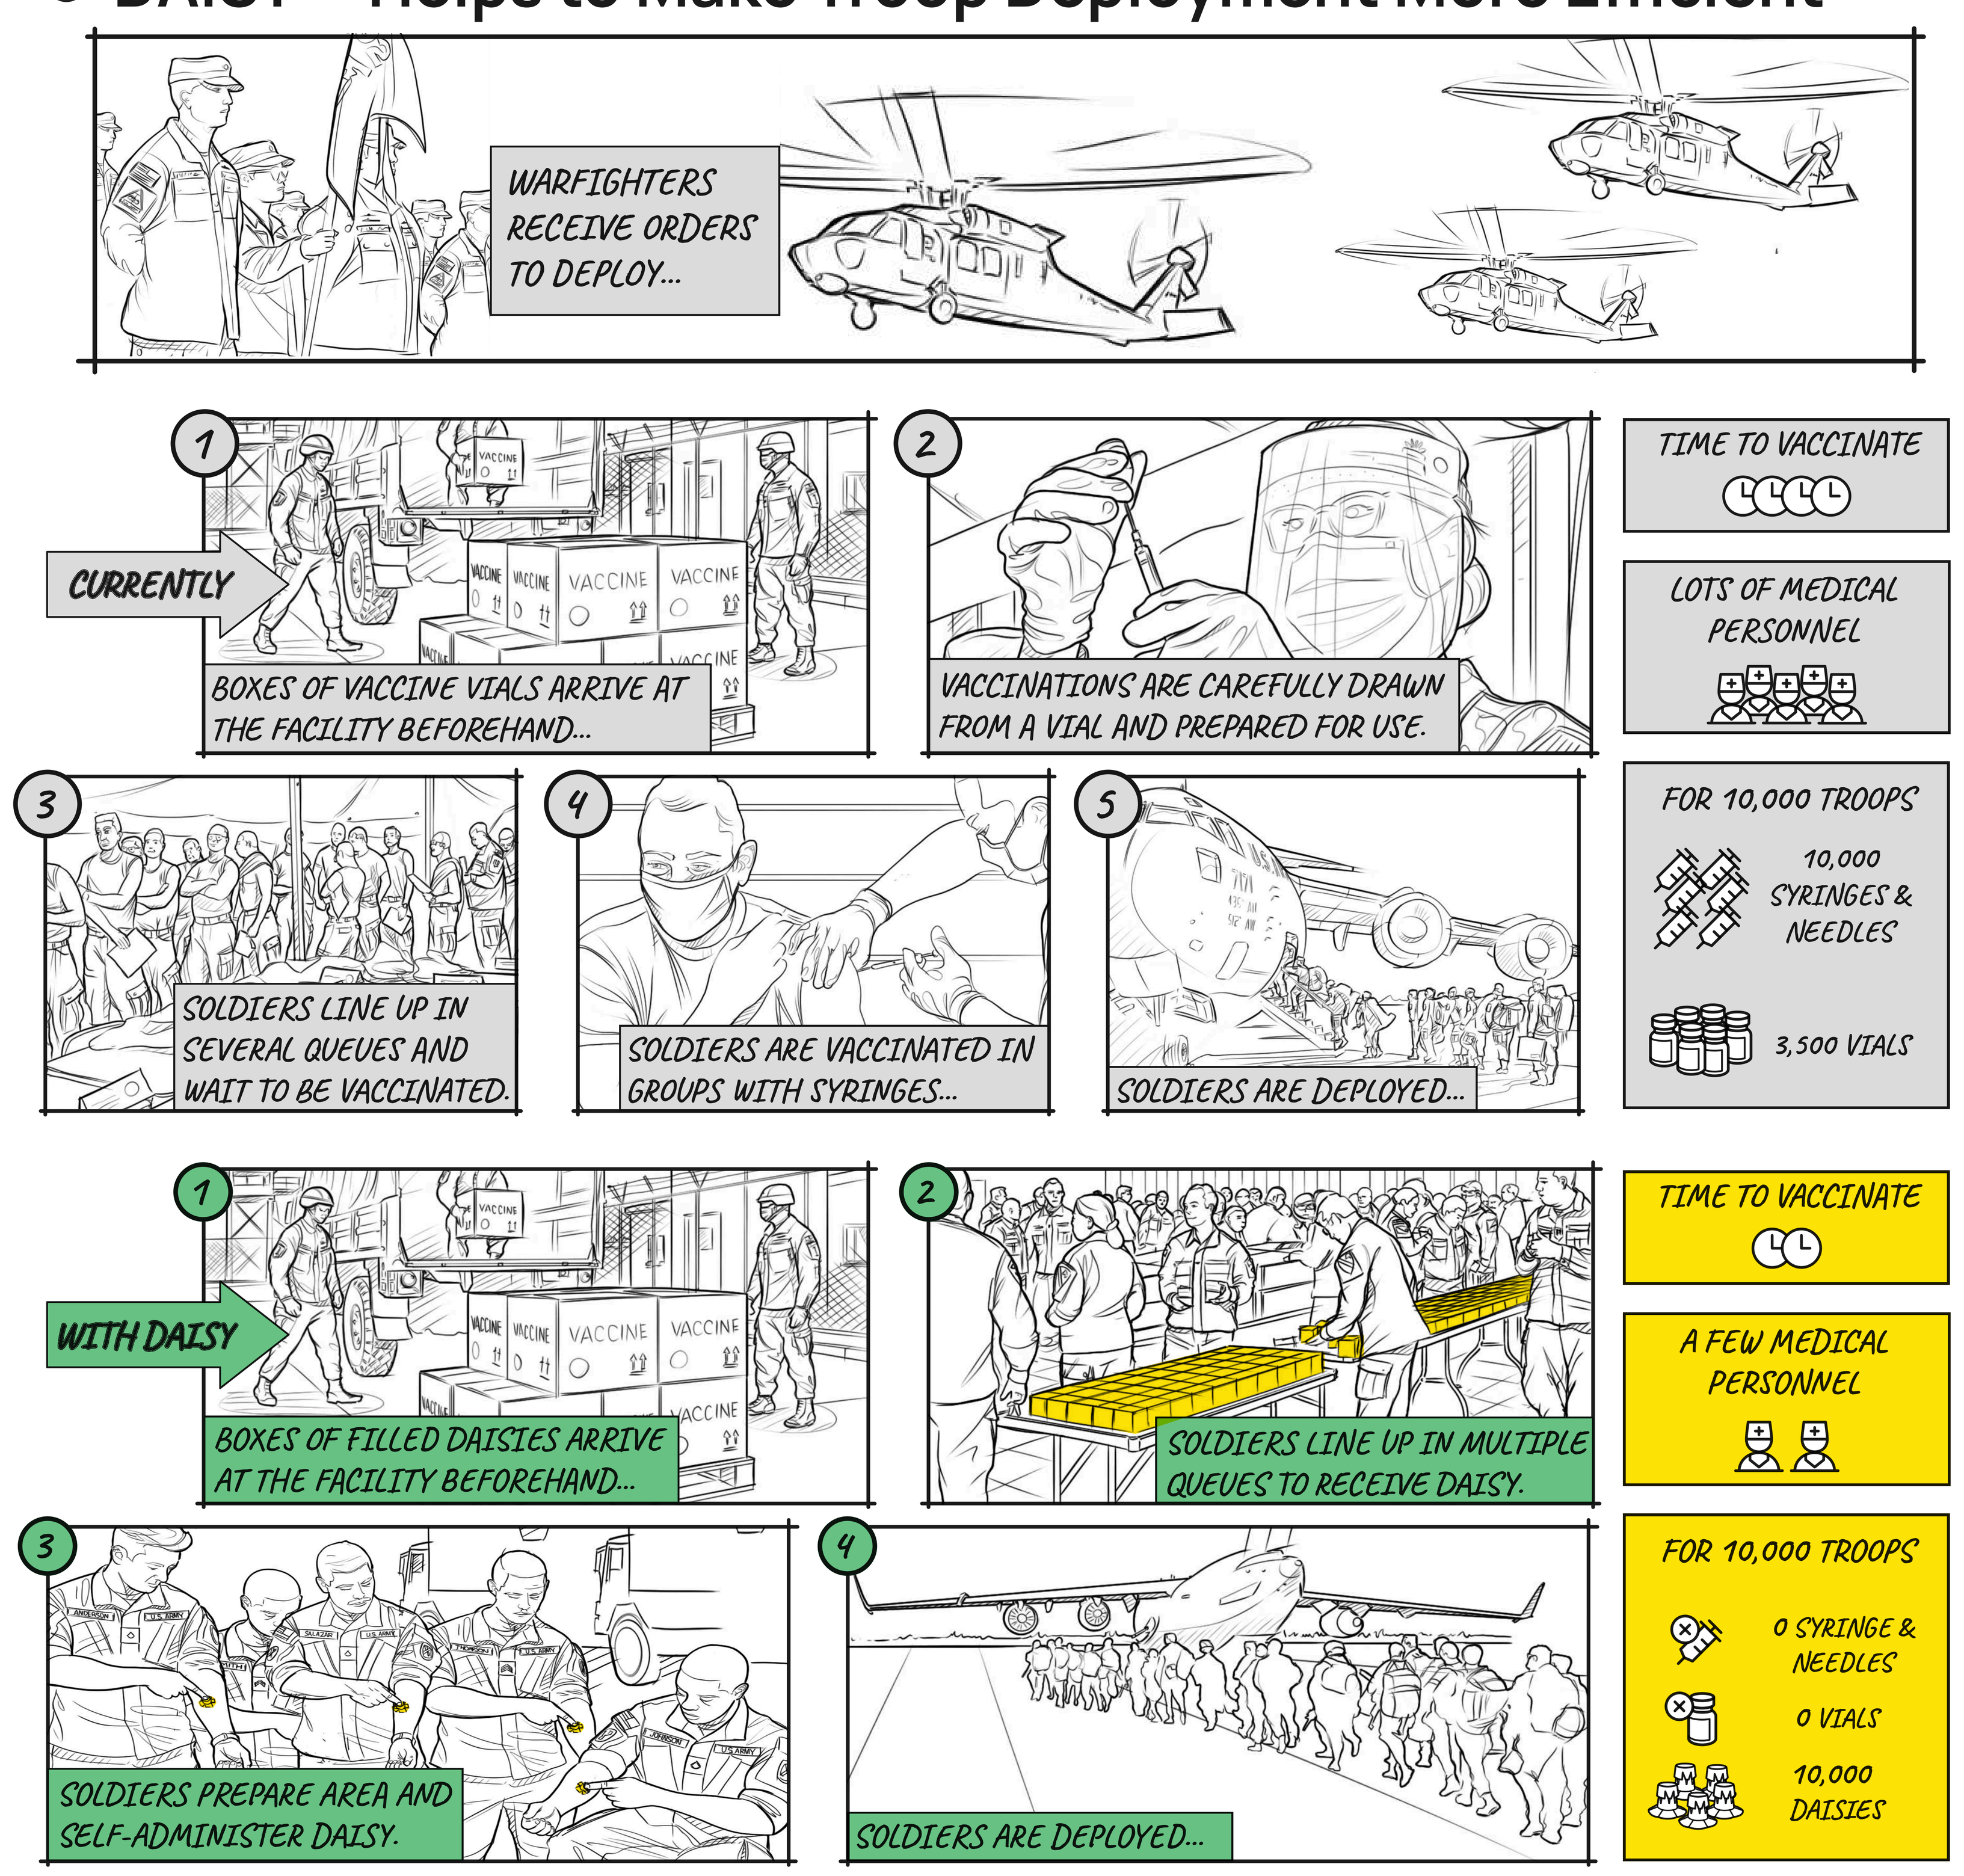

WARFIGHTERS RECEIVE ORDERS TO DEPLOY...
CURRENTLY
1
BOXES OF VACCINE VIALS ARRIVE AT THE FACILITY BEFOREHAND...
2
VACCINATIONS ARE CAREFULLY DRAWN FROM A VIAL AND PREPARED FOR USE.
3
SOLDIERS LINE UP IN SEVERAL QUEUES AND WAIT TO BE VACCINATED.
4
SOLDIERS ARE VACCINATED IN GROUPS WITH SYRINGES...
5
SOLDIERS ARE DEPLOYED...
TIME TO VACCINATE
LOTS OF MEDICAL PERSONNEL
FOR 10,000 TROOPS
10,000 SYRINGES & NEEDLES
3,500 VIALS
WITH DAISY
1
BOXES OF FILLED DAISIES ARRIVE AT THE FACILITY BEFOREHAND...
2
SOLDIERS LINE UP IN MULTIPLE QUEUES TO RECEIVE DAISY.
3
SOLDIERS PREPARE AREA AND SELF-ADMINISTER DAISY.
4
SOLDIERS ARE DEPLOYED...
TIME TO VACCINATE
A FEW MEDICAL PERSONNEL
FOR 10,000 TROOPS
0 SYRINGE & NEEDLES
0 VIALS
10,000 DAISIES

Fill Station

The portable filling station provides point-of-need final manufacturing of self-administrable intradermal therapeutics devices.

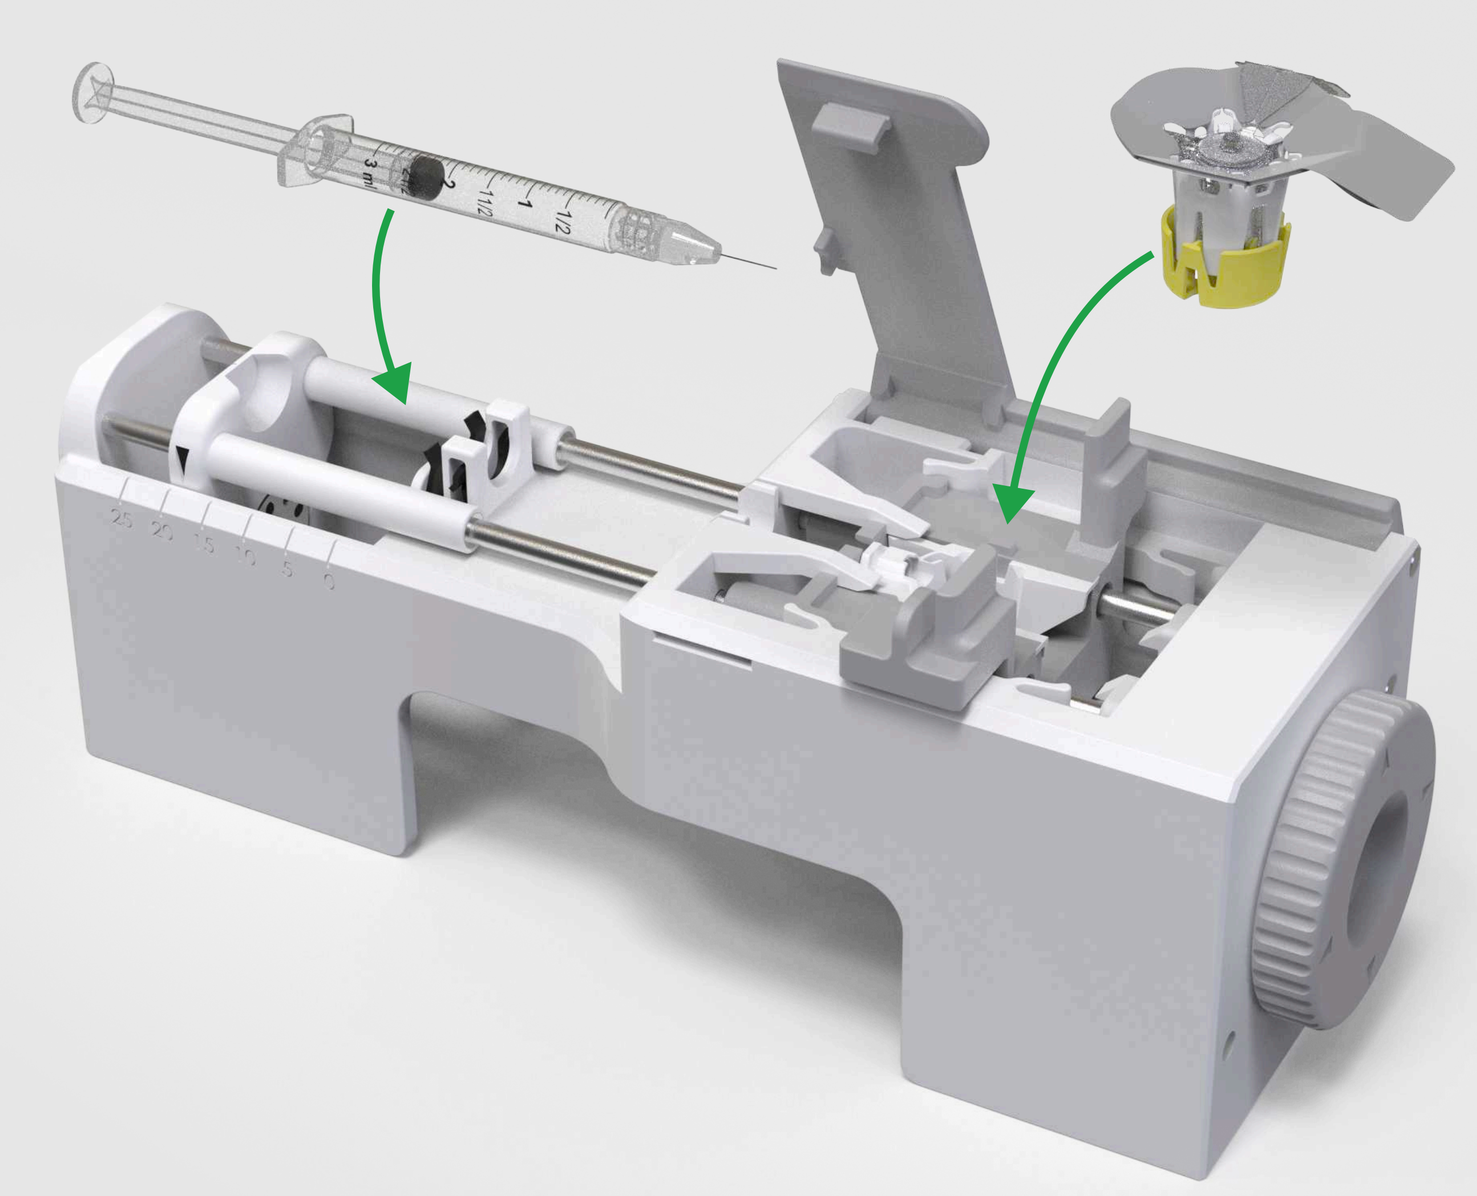

Dimensions	10” L × 4” W × 3” H (254cm x 101cm x 76cm)
Weight	~1 lb
Fill accuracy	±1 µL (meets ±5% requirement)
Fill rate	Up to 24 devices per minute with a 3mL syringe
Usage Temperature	-40°C to 40°C
Power requirements	None (manual operation)
TRL/MRL	6/4

*Specification subject to change

Filling Process

1

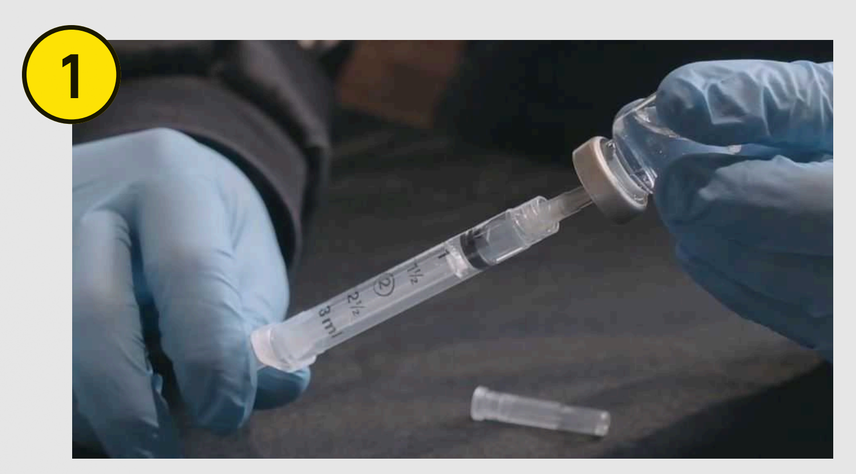

Draw therapeutic from the vial into a 3mL syringe

2

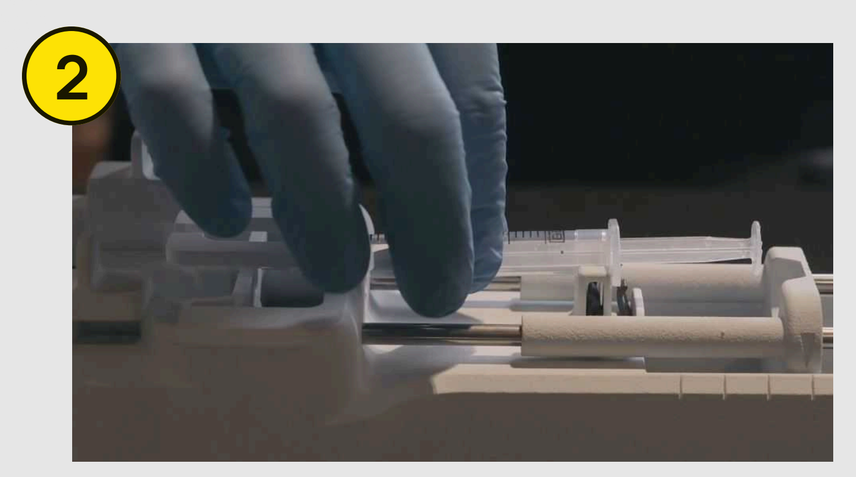

Insert filled syringe into carriage

3

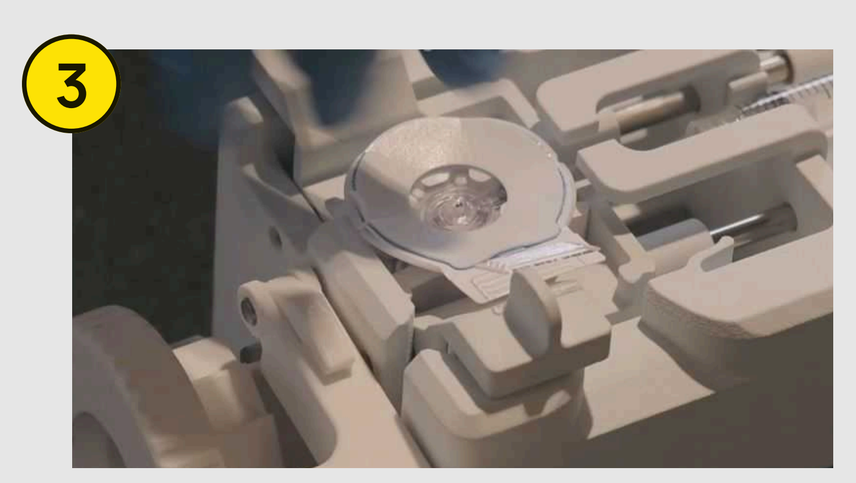

Load a Daisy™ into carriage and lock into place

4

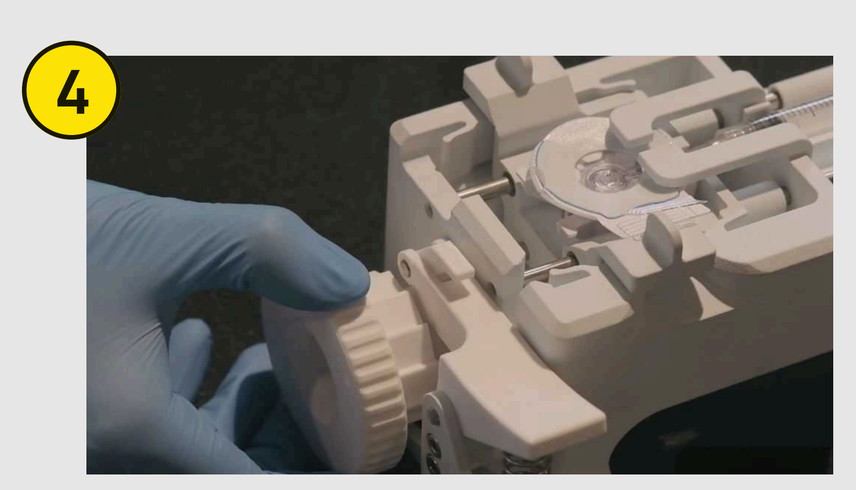

Rotate the drive handle to fill and remove filled Daisy™ from carriage

DAISY™ Enables a Faster Emergency Response

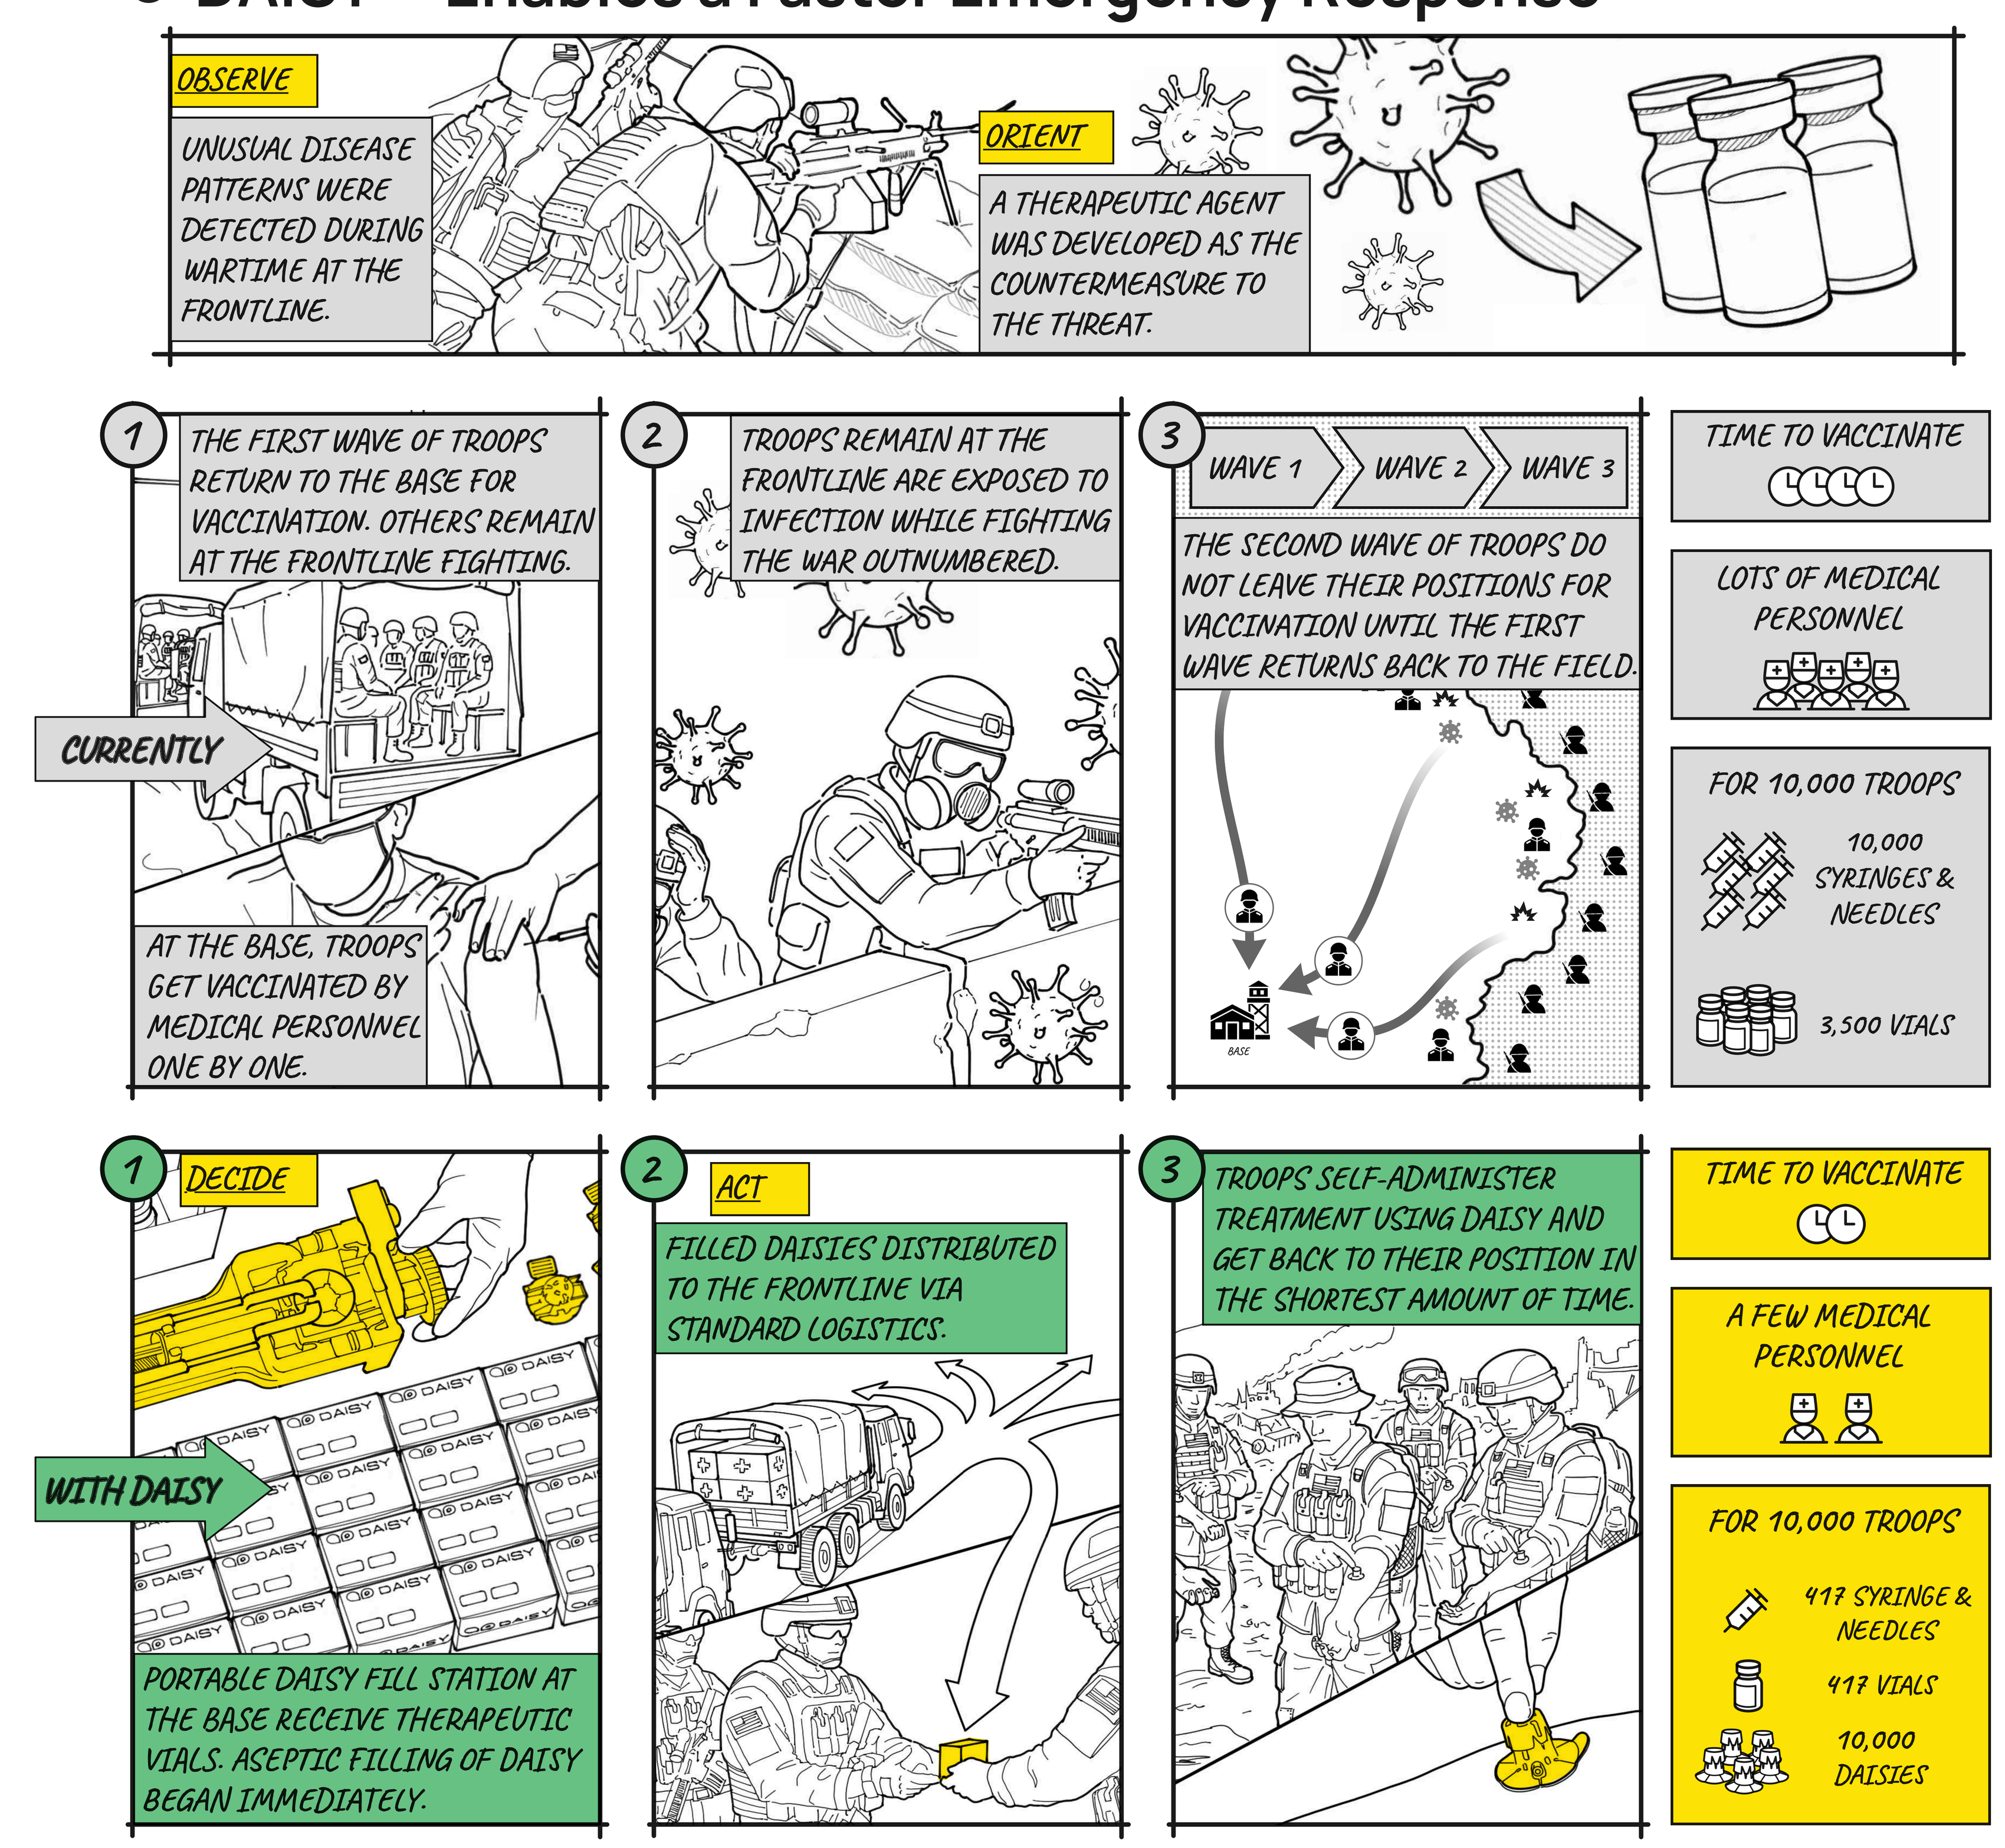

OBSERVE
UNUSUAL DISEASE PATTERNS WERE DETECTED DURING WARTIME AT THE FRONTLINE.
ORIENT
A THERAPEUTIC AGENT WAS DEVELOPED AS THE COUNTERMEASURE TO THE THREAT.
CURRENTLY
1
THE FIRST WAVE OF TROOPS RETURN TO THE BASE FOR VACCINATION. OTHERS REMAIN AT THE FRONTLINE FIGHTING.
AT THE BASE, TROOPS GET VACCINATED BY MEDICAL PERSONNEL ONE BY ONE.
2
TROOPS REMAIN AT THE FRONTLINE ARE EXPOSED TO INFECTION WHILE FIGHTING THE WAR OUTNUMBERED.
3
WAVE 1
WAVE 2
WAVE 3
THE SECOND WAVE OF TROOPS DO NOT LEAVE THEIR POSITIONS FOR VACCINATION UNTIL THE FIRST WAVE RETURNS BACK TO THE FIELD.
BASE
TIME TO VACCINATE
LOTS OF MEDICAL PERSONNEL
FOR 10,000 TROOPS
10,000 SYRINGES & NEEDLES
3,500 VIALS
WITH DAISY
1
DECIDE
PORTABLE DAISY FILL STATION AT THE BASE RECEIVE THERAPEUTIC VIALS. ASEPTIC FILLING OF DAISY BEGAN IMMEDIATELY.
2
ACT
FILLED DAISIES DISTRIBUTED TO THE FRONTLINE VIA STANDARD LOGISTICS.
3
TROOPS SELF-ADMINISTER TREATMENT USING DAISY AND GET BACK TO THEIR POSITION IN THE SHORTEST AMOUNT OF TIME.
TIME TO VACCINATE
A FEW MEDICAL PERSONNEL
FOR 10,000 TROOPS
417 SYRINGE & NEEDLES
417 VIALS
10,000 DAISIES