An Updated Mortality Study of Beryllium Workers, 1925-2020

Tiffani Fordyce, PhD[1]. Megan Leonhard, MPH[2]. Alesia M Jung, PhD[1]. Jessica Lin, MS[2]. Heather Watson, PhD[2]. Gregory Heldt, MS[3]. Paolo Boffetta, MD[4].

[1]Exponent, Inc., Menlo Park, CA. [2]Exponent, Inc., Bellevue, WA
[3]Materion Brush, Inc., Elmore, OH [4]Stony Brook University, Stony Brook, NY

INTRODUCTION

Beryllium is used for essential defense missions. Beryllium metal is lighter than aluminum, stiffer than steel and is invisible to X-ray. Applications include space-based missile tracking, key avionics platforms, high-altitude unmanned surveillance, coastline defense detectors, undersea fiberoptics, and fusion energy. Beryllium is essential to every nuclear weapon and its unique properties are critical to future defense innovations. The Department of War (DoW) identified beryllium as the very first material to be classified as both "critical" and "strategic" to the United States (US). This study, the largest ever study of beryllium workers, is designed to provide comprehensive evidence on the presence or absence of risk of mortality from lung cancer, all cancers, all causes, other non-malignant respiratory disease (ONMRD), and cancers with suggested associations to beryllium exposure (kidney, central nervous system (CNS) and bladder) among workers exposed to insoluble beryllium. The DoW only uses insoluble forms of beryllium.

MATERIALS AND METHODS

We conducted a retrospective cohort study of 17,149 workers employed in 15 US beryllium manufacturing facilities (1925-2020) with 95 years of potential follow-up from first employment. Facilities were classified as either insoluble-only or mixed soluble/insoluble based on the forms of beryllium processed; because individual quantitative exposure data and detailed job histories were unavailable, facility type served as a surrogate for potential exposure. Mortality and cause of death were ascertained through the National Death Index, supplemented with Social Security Administration data, pension death audit records, genealogy sources, and death certificates coded by a certified nosologist. Data were analyzed using standardized mortality ratios (SMRs) and 95% confidence intervals and Cox Proportional Hazard models.

RESULTS

Lung cancer mortality was not elevated at facilities with exposures only to insoluble forms of beryllium during any period, it was only elevated at facilities including soluble beryllium exposed workers and only for those workers hired pre-1955. Mortality from all cancers was borderline significantly elevated among workers with time since hire of 30+ years for the total cohort. ONMRD mortality was also significantly elevated in the total cohort, driven by elevated mortality among mixed soluble/insoluble use facilities and workers hired pre-1955, and among those with longer duration of employment or longer time since hire, but was not elevated in facilities with exposure to only insoluble beryllium. No elevated mortality due to kidney, bladder, or CNS cancer was observed in this cohort across facility type or exposure-duration categories, however in the individual facility analysis, bladder cancer was elevated at one facility with mixed exposures, based on 19 deaths (SMR 1.71; 95% CI 1.03–2.67). The Cox models and 10-year lag sensitivity analyses generally supported the main SMR findings.

This research was funded by DoW grant # TX220306P1. Conflicts of Interest: A co-investigator on the project (Dr. Constantine Dumas) and one of the authors (Gregory Heldt) are employed by Materion, a beryllium manufacturing company. Authors Dr. Boffetta and Dr. Fordyce both previously consulted with Materion, and Materion is providing additional financial support for the next phase of this study.

Table 1. Standardized mortality ratios by year of hire, duration of employment and time since hire

	Mixed beryllium			Insoluble beryllium			Total cohort		
	Obs	SMR	95% CI	Obs	SMR	95% CI	Obs	SMR	95% CI
Lung Cancer									
Year of hire									
< 1955	393	1.27	1.15-1.40	50	0.93	0.69-1.23	443	1.22	1.11-1.34
1955+	189	0.88	0.76-1.02	151	0.90	0.76-1.05	340	0.89	0.80-0.99
p heterogeneity		<0.001			0.884			<0.001	
Duration of employment									
< 5 yrs	406	1.17	1.05-1.28	135	0.92	0.77-1.08	541	1.09	0.99-1.18
5-14 yrs	85	1.04	0.83-1.29	33	0.78	0.54-1.09	118	0.95	0.79-1.14
15-29 yrs	45	0.88	0.64-1.17	25	1.05	0.68-1.55	70	0.94	0.73-1.18
30+ yrs	46	1.11	0.81-1.48	8	1.01	0.44-1.99	54	1.09	0.82-1.42
p for linear trend*		0.276			0.006			0.380	
Time since hire									
< 10 yrs	3	0.19	0.04-0.56	10	0.91	0.44-1.68	13	0.49	0.26-0.83
10-19 yrs	32	0.81	0.55-1.14	16	0.60	0.34-0.97	48	0.72	0.53-0.95
20-29 yrs	77	0.95	0.75-1.19	45	0.96	0.70-1.28	122	0.95	0.79-1.14
30+ yrs	470	1.22	1.11-1.33	130	0.95	0.79-1.13	600	1.15	1.06-1.24
p for linear trend*		<0.001						<0.001	
ONMRD									
Year of hire									
< 1955	197	1.38	1.19-1.58	27	1.05	0.69-1.53	224	1.33	1.16-1.52
1955+	99	1.12	0.91-1.37	67	0.96	0.74-1.21	166	1.05	0.89-1.22
p heterogeneity		0.113			0.765			0.022	
Duration of employment									
< 5 yrs	180	1.15	0.99-1.33	56	0.88	0.66-1.14	236	1.07	0.94-1.22
5-14 yrs	46	1.33	0.97-1.77	15	0.82	0.46-1.35	61	1.15	0.88-1.48
15-29 yrs	37	1.68	1.18-2.32	19	1.89	1.14-2.95	56	1.74	1.31-2.26
30+ yrs	33	1.77	1.22-2.49	4	1.12	0.30-2.86	37	1.66	1.17-2.29
p for linear trend*		<0.001			<0.001			<0.001	
Time since hire									
< 10 yrs	1	0.47	0.01-2.60	1	0.43	0.01-2.37	2	0.44	0.05-1.60
10-19 yrs	8	0.99	0.43-1.96	3	0.42	0.09-1.22	11	0.72	0.36-1.29
20-29 yrs	29	1.29	0.87-1.86	19	1.24	0.75-1.93	48	1.27	0.94-1.68
30+ yrs	258	1.30	1.14-1.47	71	1.00	0.78-1.26	329	1.22	1.09-1.36
p for linear trend*		0.007			0.036			<0.001	
All Cancers									
Year of hire									
< 1955	104 1	1.03	0.97-1.09	183	1.05	0.91-1.22	1224	1.03	0.97-1.09
1955+	605	0.90	0.83-0.97	490	0.90	0.82-0.98	1095	0.90	0.85-0.95
p heterogeneity		0.008			0.077			0.001	
Duration of employment									
< 5 yrs	113 4	0.99	0.94-1.06	460	0.96	0.87-1.05	1594	0.98	0.93-1.03
5-14 yrs	231	0.88	0.77-1.00	122	0.90	0.74-1.07	353	0.88	0.79-0.98
15-29 yrs	149	0.93	0.79-1.09	70	0.93	0.73-1.18	219	0.94	0.82-1.07
30+ yrs	132	1.04	0.87-1.23	21	0.82	0.51-1.26	153	1.00	0.85-1.17
p for linear trend*		<0.001			0.007			0.012	
Time since hire									
< 10 yrs	25	0.35	0.23-0.52	34	0.83	0.58-1.16	59	0.53	0.40-0.68
10-19 yrs	92	0.65	0.52-0.79	64	0.75	0.58-0.95	156	0.68	0.58-0.80
20-29 yrs	211	0.86	0.75-0.98	128	0.91	0.76-1.08	339	0.87	0.78-0.97
30+ yrs	131 8	1.07	1.02-1.13	447	0.99	0.90-1.09	1765	1.05	1.00-1.10
p for linear trend*		<0.001			<0.001			<0.001	

Obs, observed deaths
SMR, standardized mortality ratio
CI, confidence interval
*** weights are mid-points of categories of duration and time since hire**

Figure 1. Standardized Mortality Ratios and 95% Confidence Intervals by Cause of Death and Facility Type

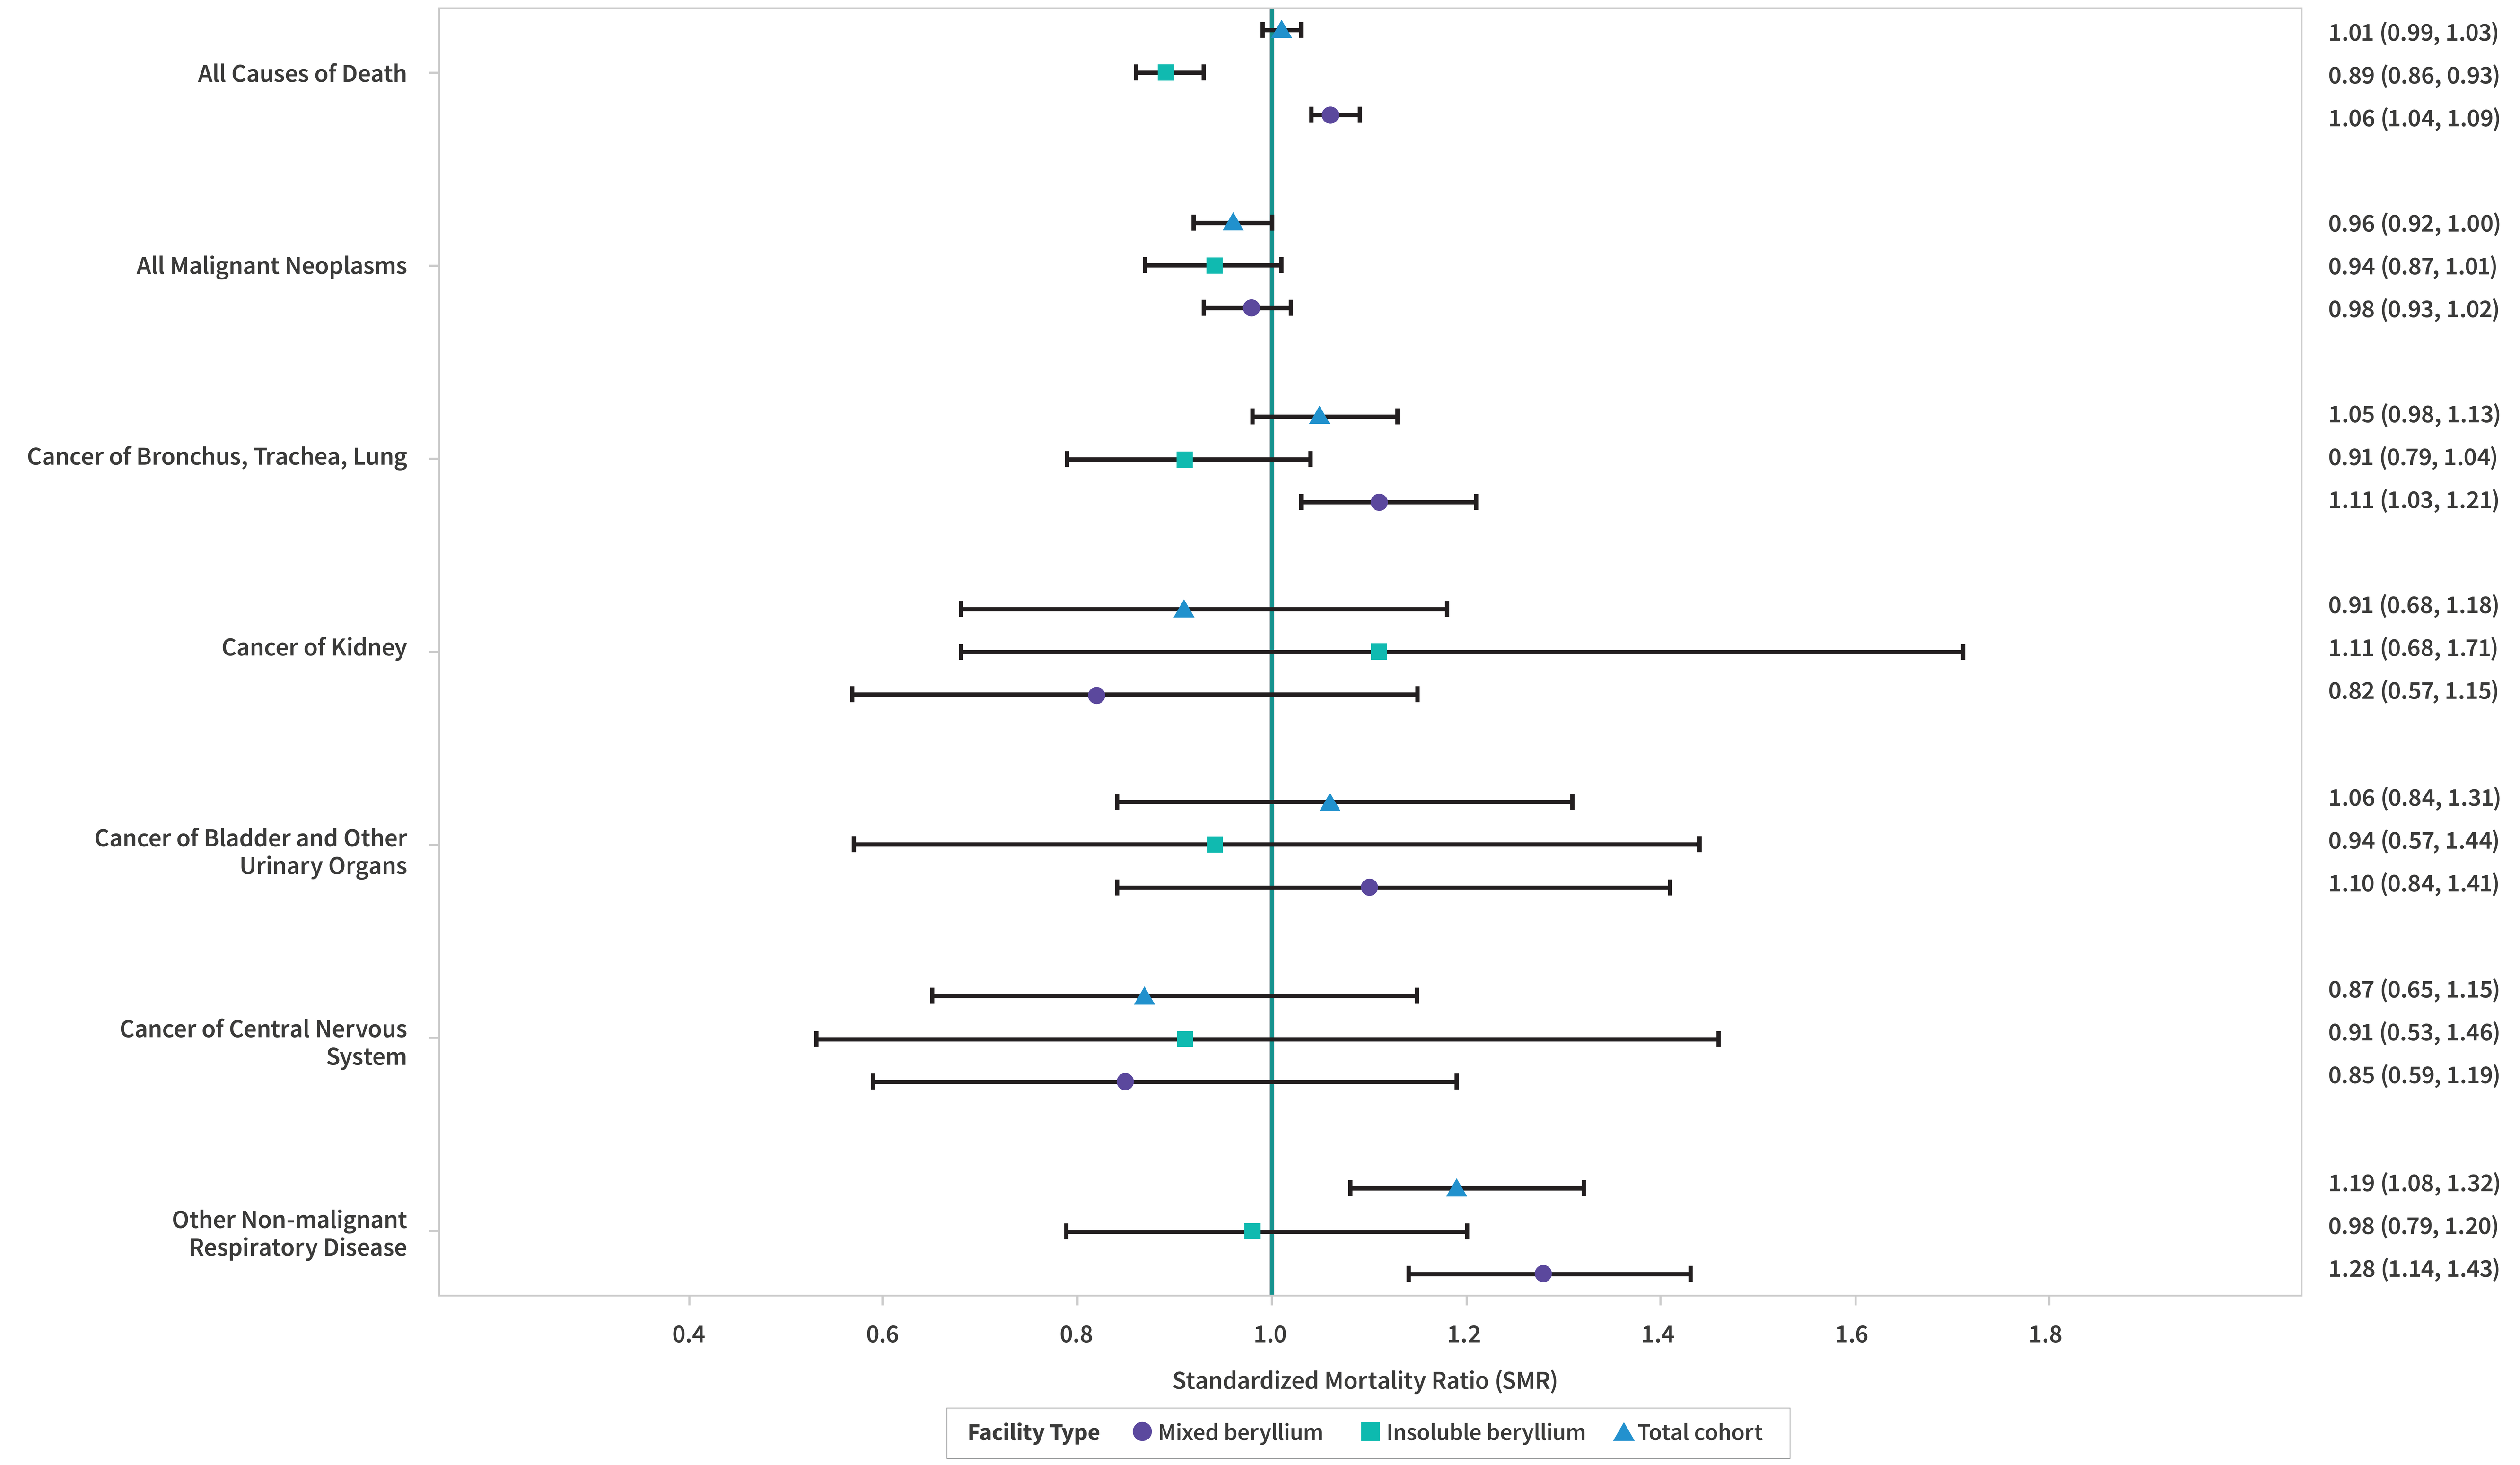
All Causes of Death
1.01 (0.99, 1.03)
0.89 (0.86, 0.93)
1.06 (1.04, 1.09)
All Malignant Neoplasms
0.96 (0.92, 1.00)
0.94 (0.87, 1.01)
0.98 (0.93, 1.02)
Cancer of Bronchus, Trachea, Lung
1.05 (0.98, 1.13)
0.91 (0.79, 1.04)
1.11 (1.03, 1.21)
Cancer of Kidney
0.91 (0.68, 1.18)
1.11 (0.68, 1.71)
0.82 (0.57, 1.15)
Cancer of Bladder and Other Urinary Organs
1.06 (0.84, 1.31)
0.94 (0.57, 1.44)
1.10 (0.84, 1.41)
Cancer of Central Nervous System
0.87 (0.65, 1.15)
0.91 (0.53, 1.46)
0.85 (0.59, 1.19)
Other Non-malignant Respiratory Disease
1.19 (1.08, 1.32)
0.98 (0.79, 1.20)
1.28 (1.14, 1.43)
0.4
0.6
0.8
1.0
1.2
1.4
1.6
1.8
Standardized Mortality Ratio (SMR)
Facility Type
Mixed beryllium
Insoluble beryllium
Total cohort

CONCLUSIONS

Elevated risks were concentrated in specific historical and exposure contexts. Lung cancer mortality was elevated among workers employed in facilities that processed both soluble and insoluble forms of beryllium, particularly among those hired before 1955, when exposures were very high. In contrast, workers employed in facilities with exposure to only insoluble beryllium forms showed no elevated lung cancer mortality in any time period evaluated, and workers hired in 1955 or later did not show increased lung cancer mortality regardless of facility type.

The most consistent excess mortality finding was for ONMRD, which was elevated in the total cohort but was driven primarily by workers in mixed-exposure facilities, especially those hired before 1955. However, ONMRD is a broad mortality category that includes chronic beryllium disease as well as other respiratory diseases, and beryllium-related deaths may have been misclassified within this grouping. As a result, ONMRD provides an important signal of respiratory disease mortality, but it does not allow precise estimation of mortality risk from chronic beryllium disease specifically.

No increase in lung cancer or other cancers for any timeframe for workers exposed to only insoluble beryllium (the only form utilized in DoW defense systems) was observed in this study. The study's statistical power provided for a very robust analysis of the largest subcohort of workers exposed only to insoluble beryllium (which comprises over 98% of beryllium use) and provides strong support for the absence of elevated lung cancer or other mortality risks from exposure to insoluble forms of beryllium.

A key limitation of this study is that facility type was used as a surrogate for exposure. While this approach allowed for broad comparisons between facilities with insoluble-only beryllium exposure and facilities with both soluble and insoluble beryllium exposure, it may also introduce exposure misclassification. Completion of a planned job exposure matrix for each facility to assign quantitative exposure values, and nested case-control analyses to control for confounding will be critical for determining whether the observed patterns are truly attributable to soluble versus insoluble forms of beryllium.