OxiWear: FDA-Cleared Ear-Worn Pulse Oximeter for Military Clinical Decision Support

OxiWear
Jamie Wood, PhD
OxiWear Inc, Arlington VA
jamie@oxiwear.com

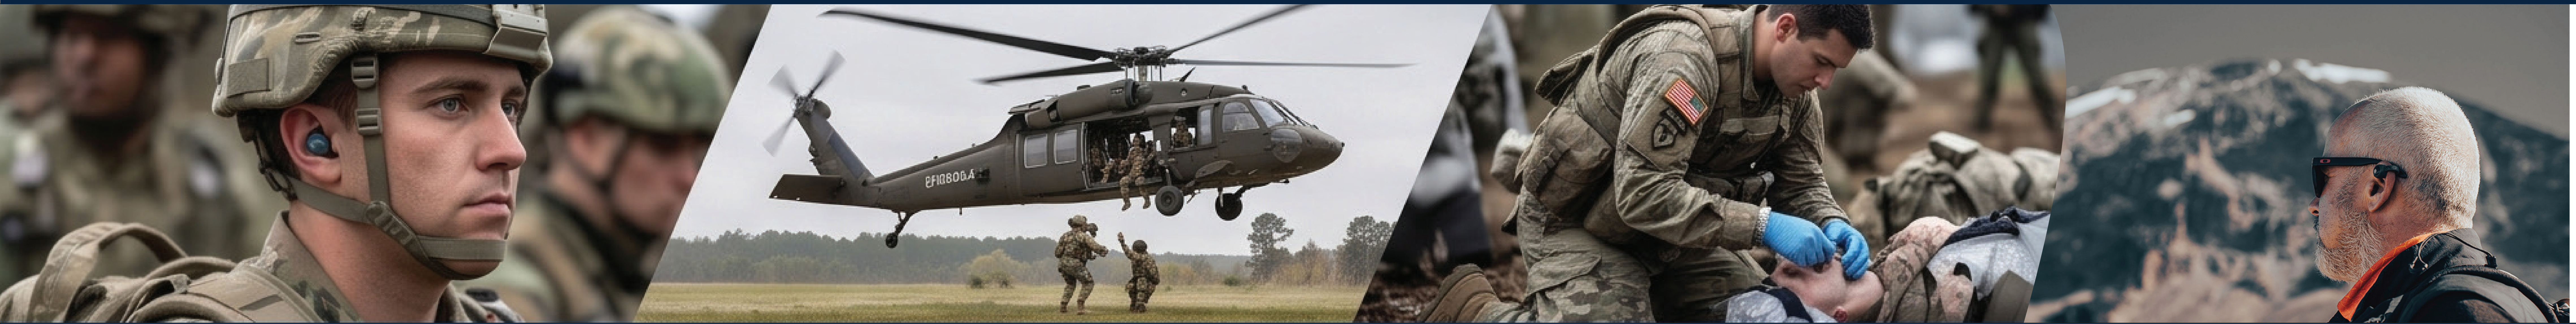

FDA-cleared ear-worn continous SpO_2 and pulse rate measurements during motion for military operational and clinical environments.

BACKGROUND

THE CHALLENGE

- Military personnel frequently experience hypoxia and physiological deterioration during:
 - High-altitude operations
 - CASEVAC
 - En Route Care
 - Prolonged Field Care
 - Combat casualty care
- Current fingertip pulse oximeters are often unreliable because they are affected by:
 - » Motion
 - » Peripheral vasoconstriction
 - » Cold exposure
 - » Extremity injury
 - » Limited hands-free operation

Why the Ear?

FINGER VS EAR

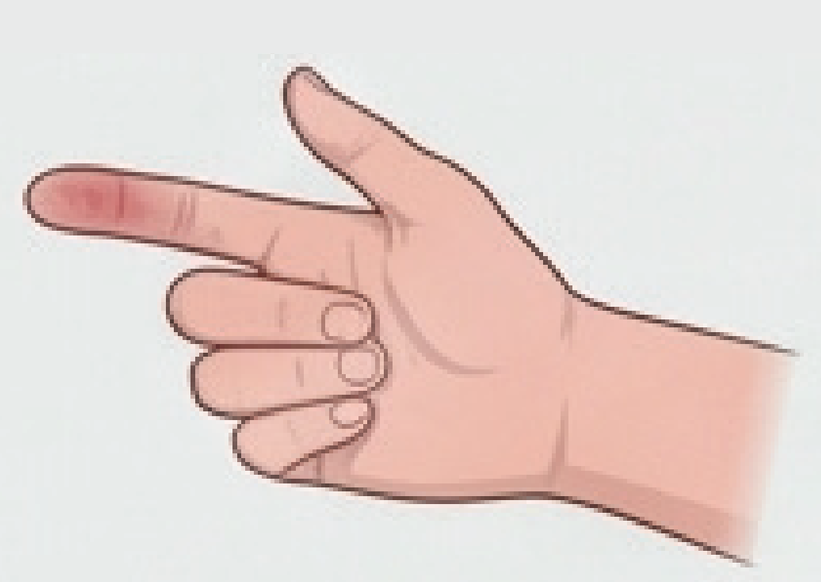

Finger

High motion artifact
Vasoconstriction
Hands occupied
Extremity injuries

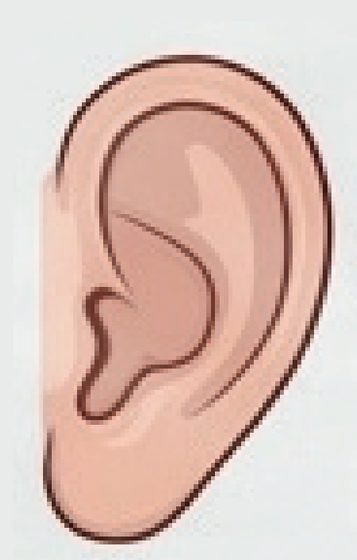

Ear

Stable blood flow
Less affected
Hands-free
Preserved measurement

OXIWEAR SOLUTION

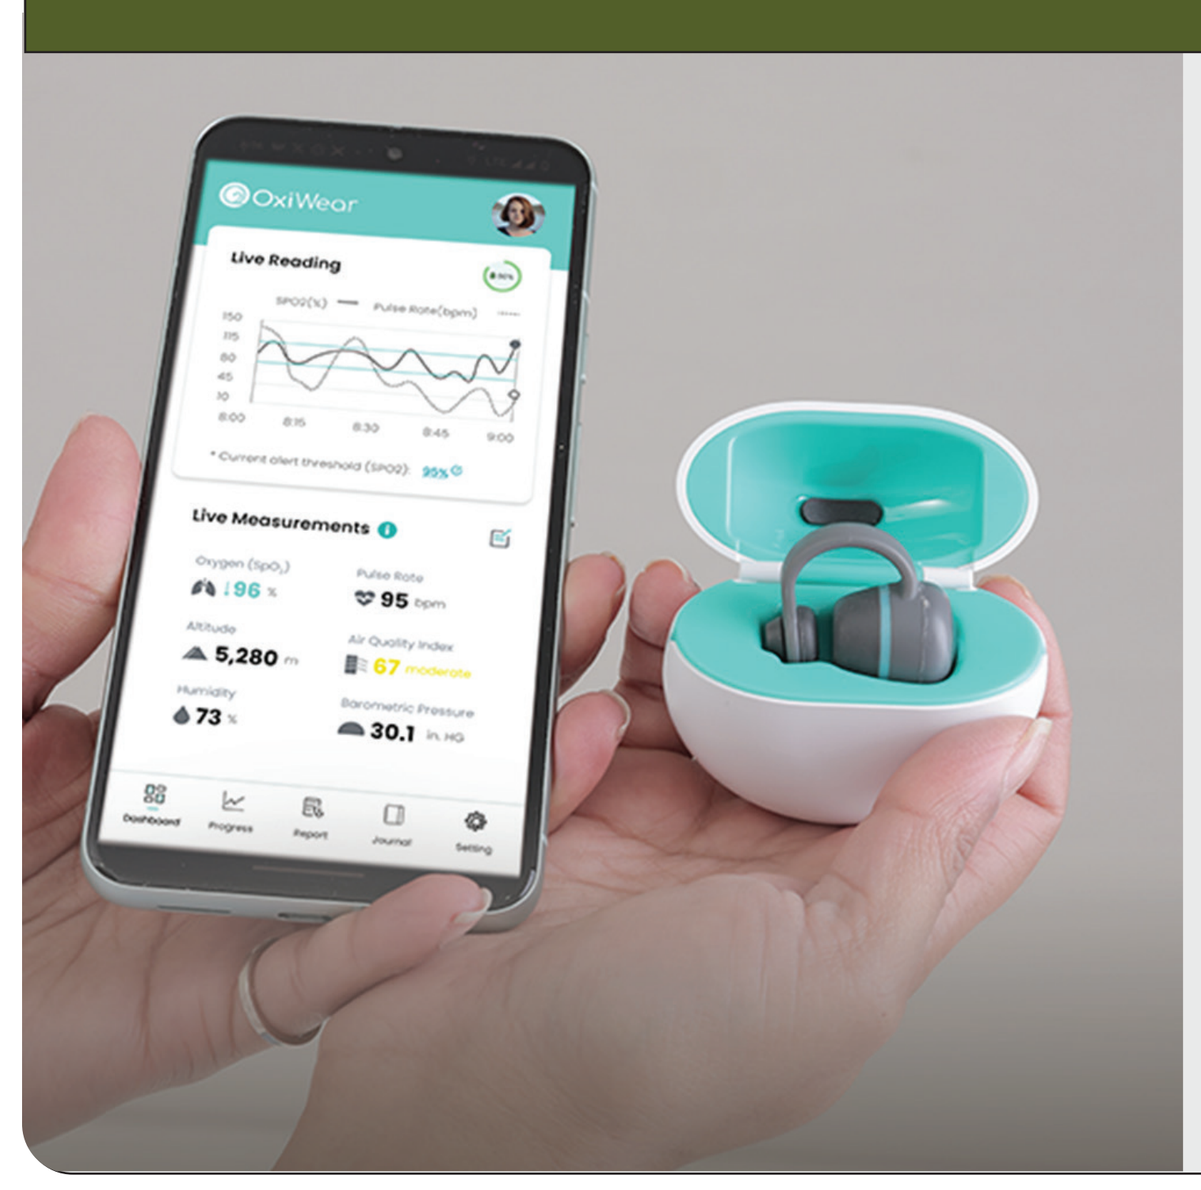

- **FDA-cleared Class II medical device**
 - Continuous SpO_2 measurements
 - Continuous Pulse Rate measurements
 - Ergonomic ear-worn form factor
 - Ultra-lightweight (5.3 g) wearable
 - Real-time physiological alerts
 - Tactical platform interoperability
 - Validated across all skin tones

VALIDATION

Clinically Validated Performance

ISO 80601-2-61 VALIDATION

Workflow → → → → Role Study

Desaturation Study

UCSF Hypoxia Laboratory

- 24 healthy participants
- Arterial Blood Gas (ABG) reference
- SpO_2 Range: 70–100%

Motion Study

- 30 participants
- Rigorous Walk / Jog protocol
- Compared directly against Nellcor PM10N

Results

SpO_2 Accuracy	
No Motion A_{RMS}	Motion A_{RMS}
3.42%	1.74%

Pulse Rate Accuracy	
No Motion A_{RMS}	Motion A_{RMS}
1.14 BPM	3.62 BPM

VALIDATED ACROSS ALL FITZPATRICK SKIN TYPES

MILITARY APPLICATIONS

- **Point of Injury :** Continuous physiological assessment
- **CASEVAC :** Uninterrupted monitoring during transport.
- **En Route Care :** Supports critical medical decision-making
- **High Altitude Operations :** Earlier detection of hypoxia
- **Diverse Force :** Validated across all skin tones.
- **Low-Light Operations :** Objective SpO_2 measurement when cyanosis cannot be visually assessed.

FUTURE INTEGRATION

OxiWear → BATDOK → CDP → JOMIS → Medical Command

DEVELOPMENT STATUS

FDA Cleared → Commercial Deployment → TRL 6 → MIL-STD-810 Hardening → IP65 → Military Integration

KEY TAKEAWAYS

CONTINUOUS MEASUREMENTS
Unlike spot-check pulse oximeters

MOTION VALIDATED
Reliable and resilient during movement and transport.

EAR-WORN DESIGN
Measurement integrity is preserved when hands or extremities are compromised.

OPERATIONAL READINESS
Seamlessly supports monitoring from point of Injury through en route care.

CLINICALLY VALIDATED
ISO 80601-2-61 compliant, FDA Cleared, and commercially available.