Ultra-Thin, Helmet-Compatible Cerebrovascular Sensing: Advancing Operator State Monitoring for Army Future Vertical Lift

Thibault Roumengous*, Aiden J. Chauntry, R. Casey Boutwell, James Dieffenderfer, Jon Walter, Christopher Flippen, Brett Goldbach
NIRSense Inc., 1717 E Cary St, Richmond, VA 23223 *Thibault@NIRSense.com

Wearable Somatic and Cerebral Oximetry

Introduction:

The increased agility, speed, and altitude of the Army's future vertical lift (FVL) aircraft are expected to dramatically increase physiological stress on aircrews during flight. These stressors pose a severe risk to mission success, necessitating real-time operator state monitoring (OSM) to continuously optimize performance. Following a successful Phase I effort and expert workshop at Fort Rucker, NIRSense gathered critical FVL OSM requirements to guide this Phase II redesign.

Approach:

NIRSense develops rugged, wearable physiological monitoring systems featuring simultaneous photoplethysmography (PPG) and near-infrared spectroscopy (NIRS) for cerebral oximetry. This Phase II effort implements the FVL OSM requirements into a revised cerebrovascular sensing system, the "Apollo" prototype, developed through iterative human-factors evaluations with the U.S. Army Aeromedical Research Laboratory (USAARL).

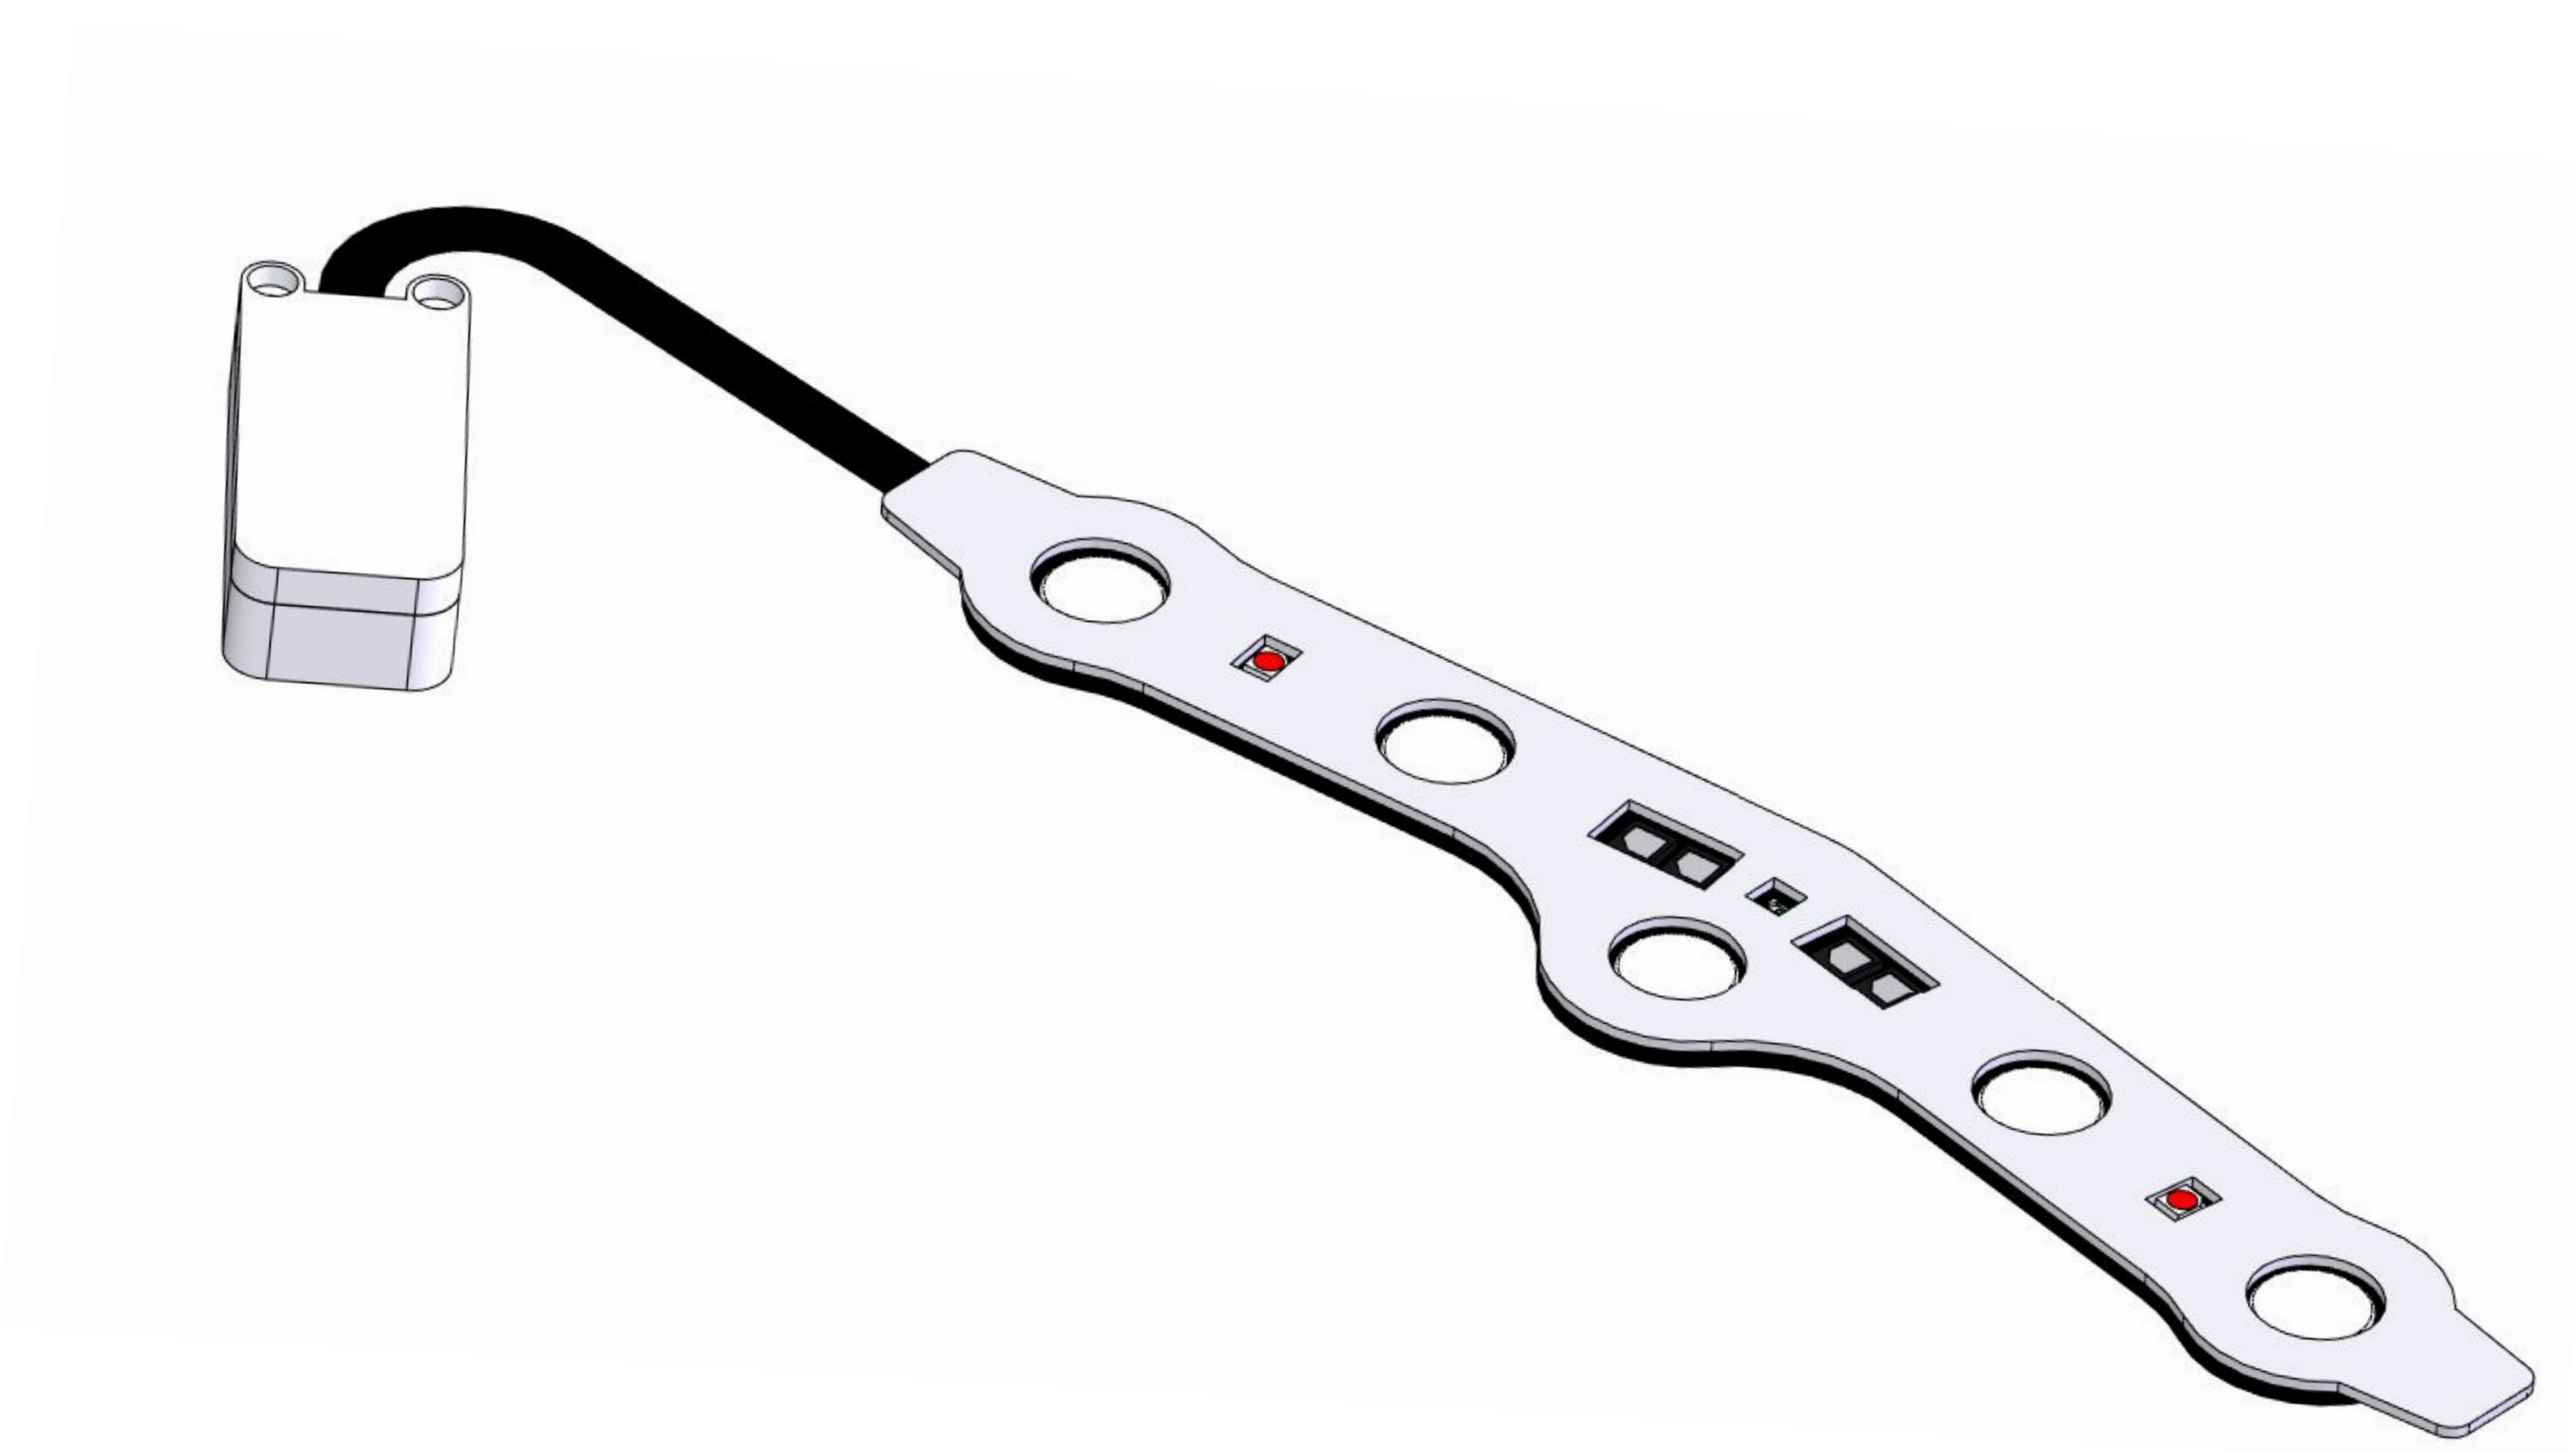

Apollo Patch Design:

The Apollo system uses a "Side Tail" configuration that reduces the flexible patch footprint by over 50%. The flexible multimodal patch (NIRS, PPG, and EEG) maintains a sub-3 mm thickness at the forehead,or comfort without modifying the HGU-56/P helmet liner, while the rigid electronics are relocated to a secure module outside primary pressure zones.

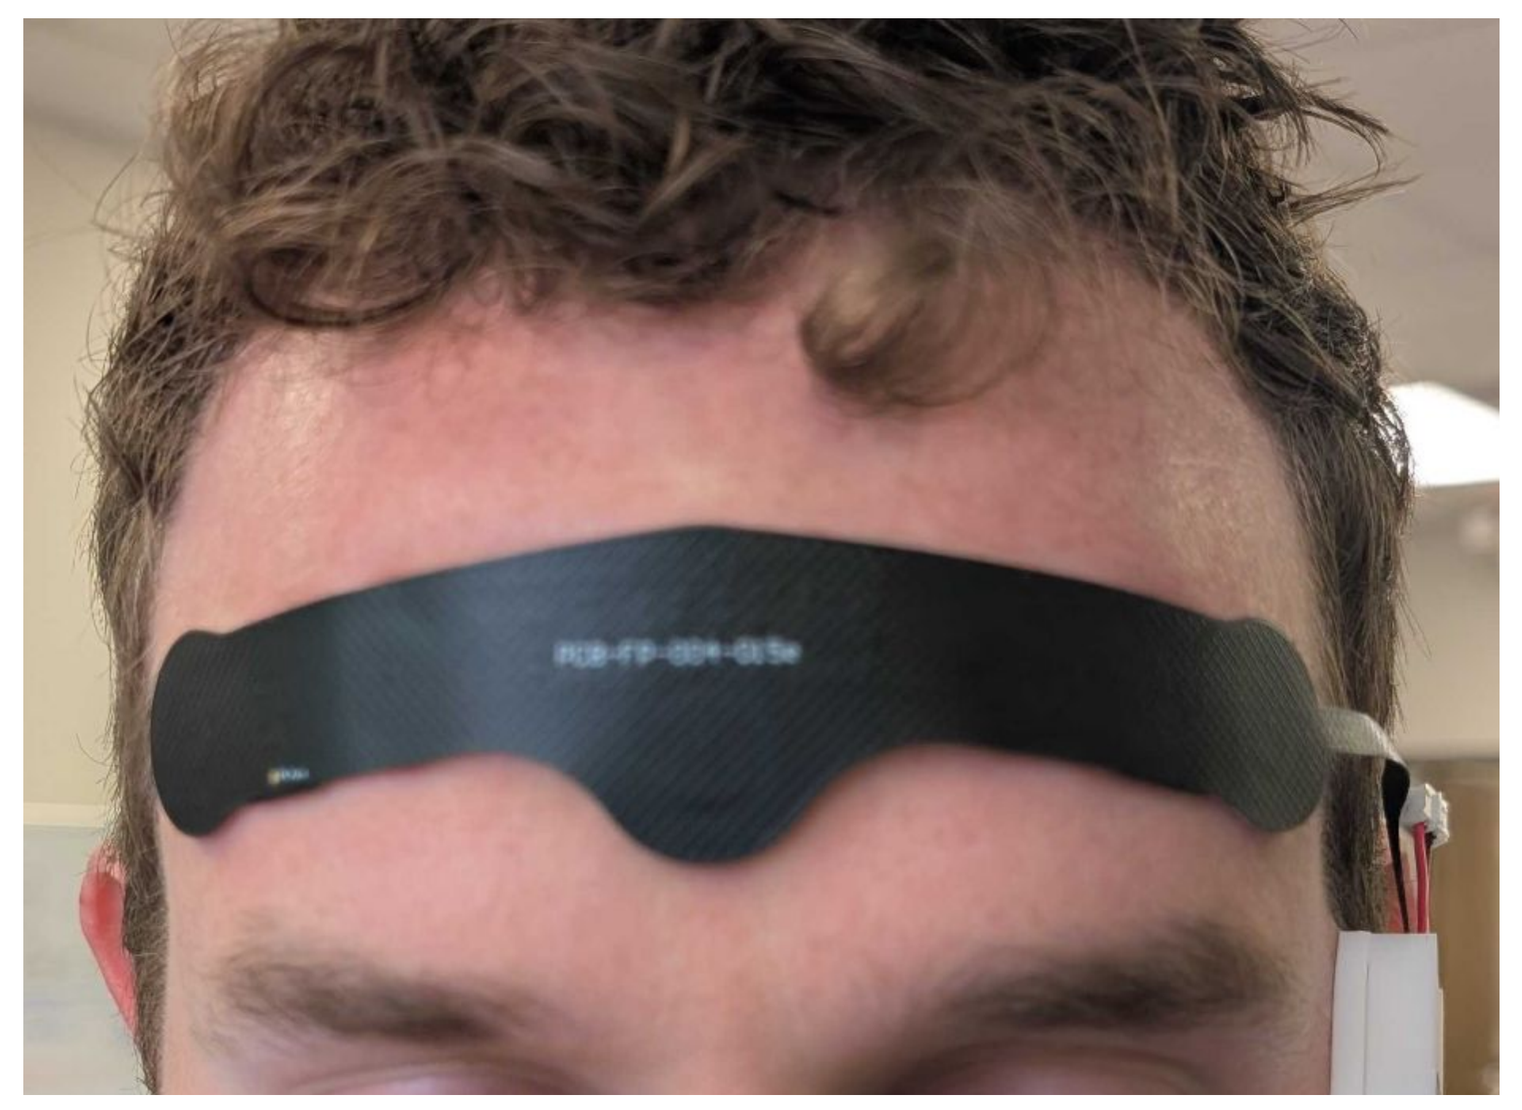

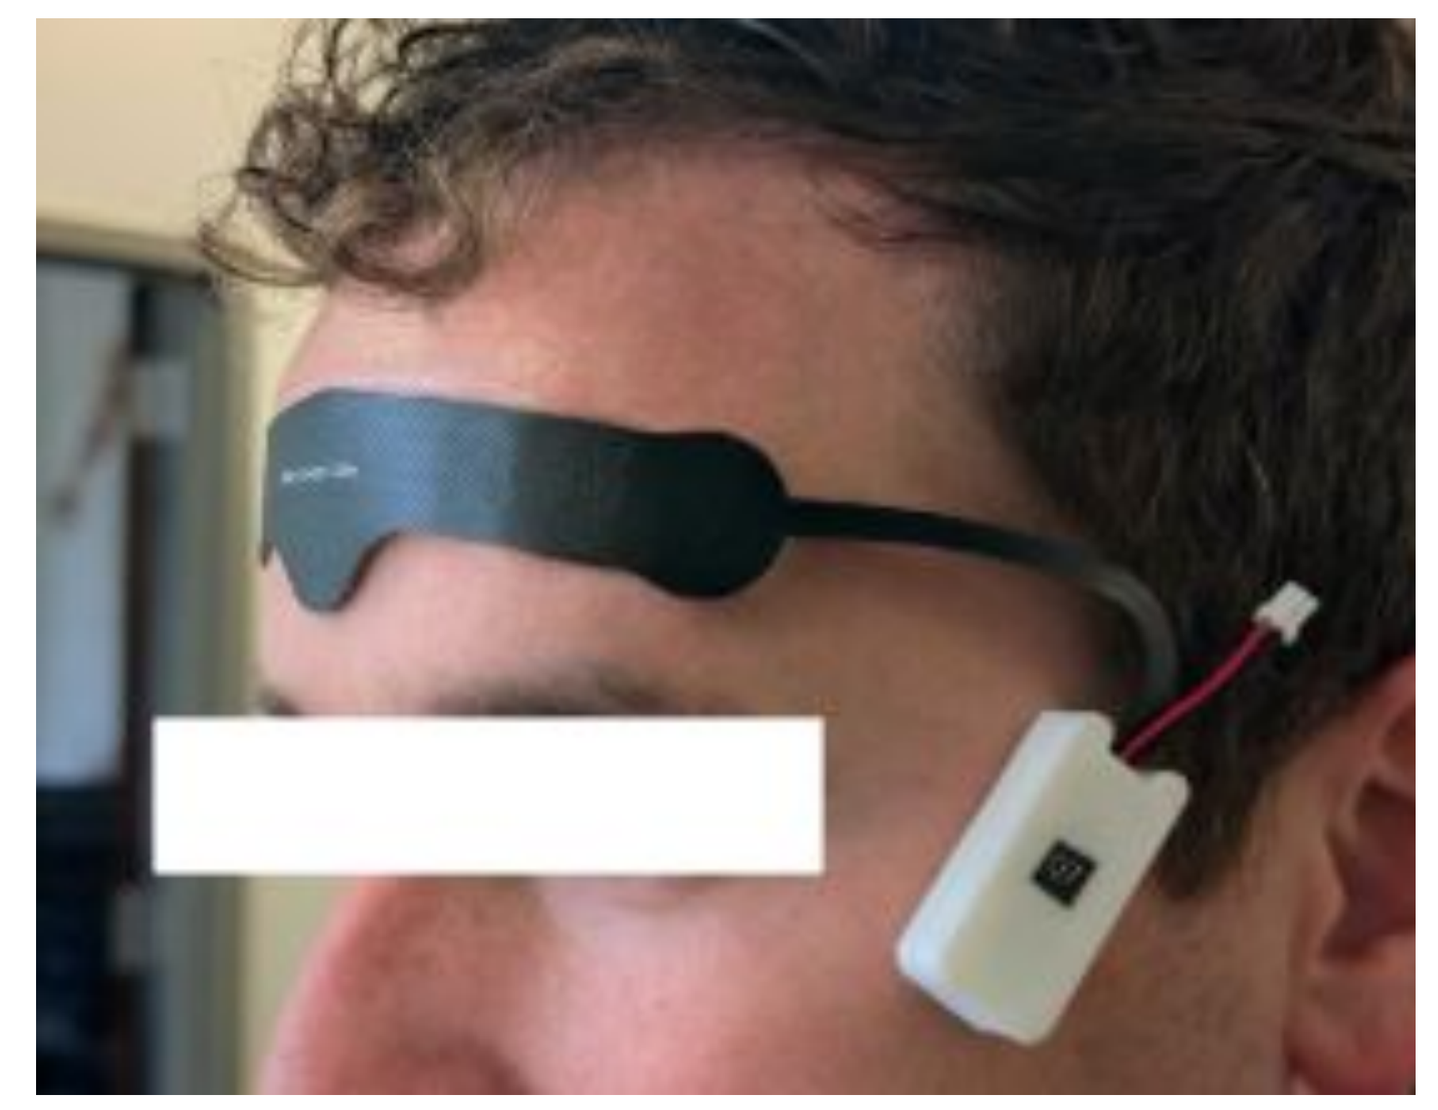

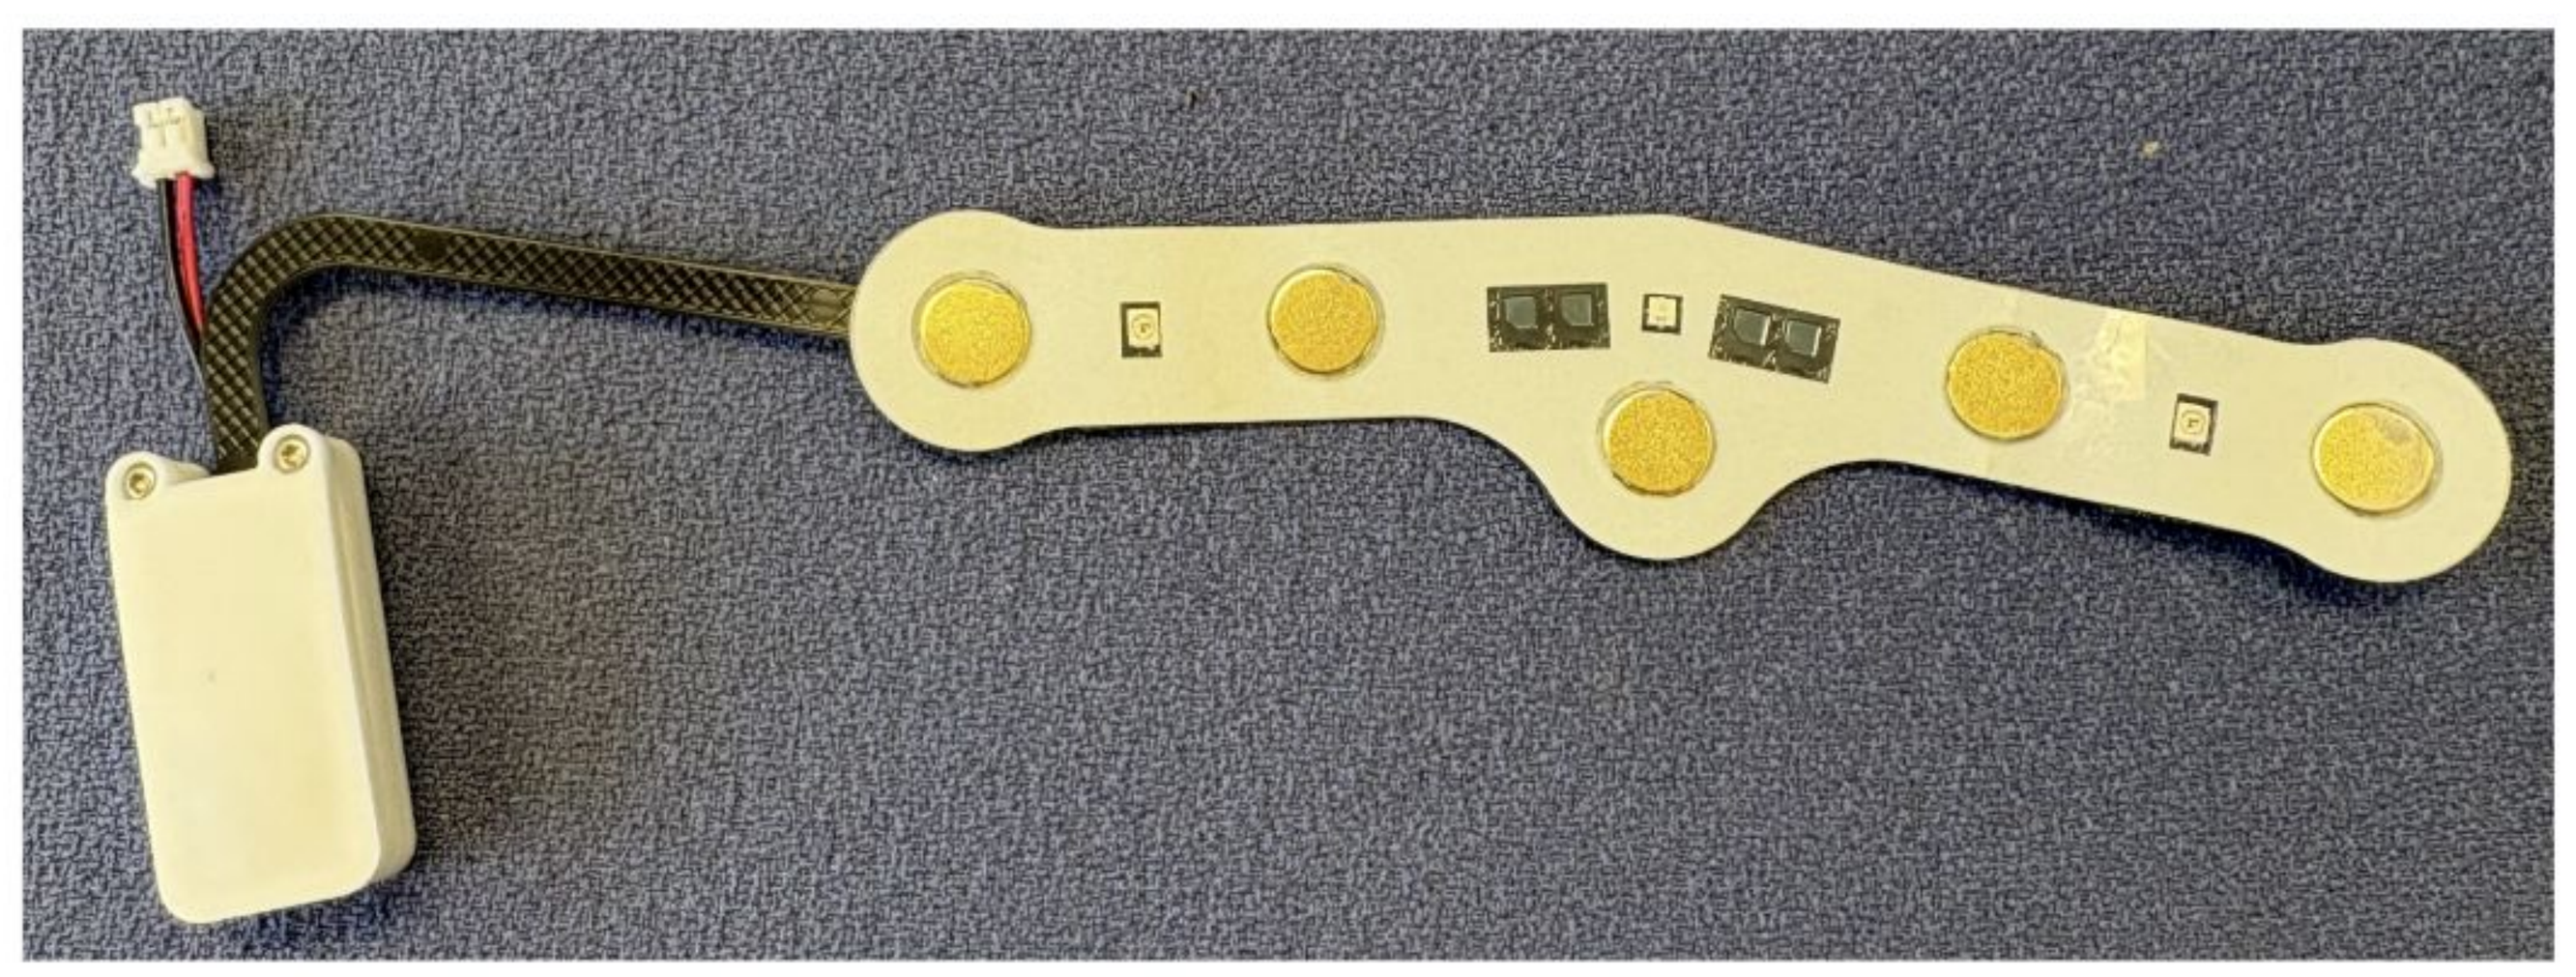

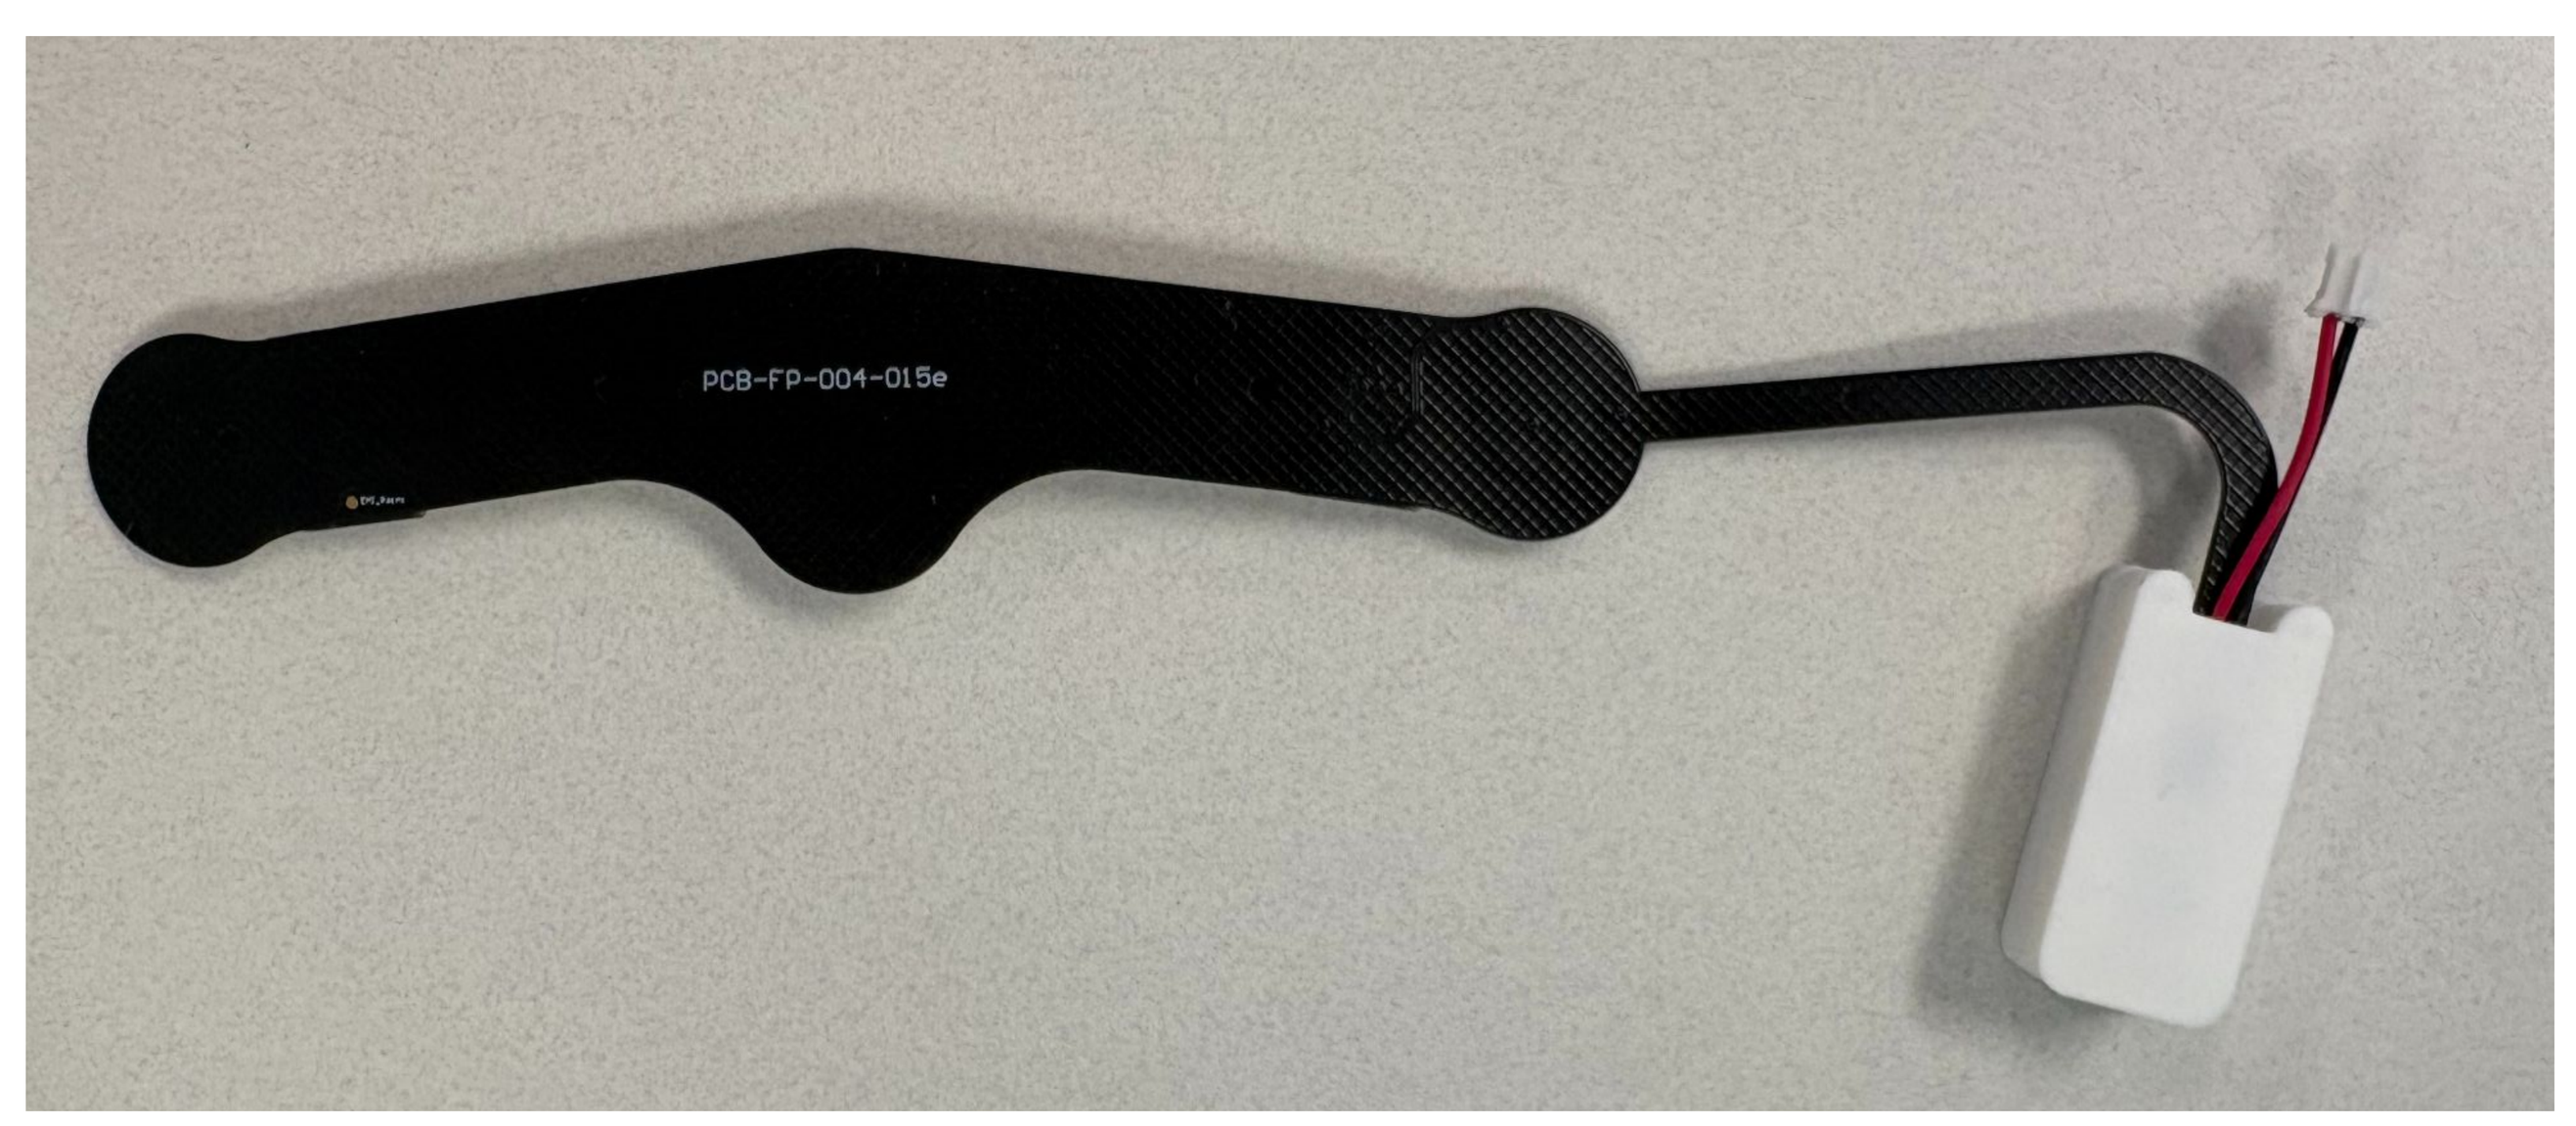
PCB-FP-004-015e

Under-Helmet Integration:

On-head and under-helmet fit checks confirmed the redesigned geometry orients the rigid control module more vertically. Worn on the forehead beneath a seated HGU-56/P flight helmet with the visor down, the module tucks neatly around the earmuff without conflicting with the helmet shell, no liner modification required.

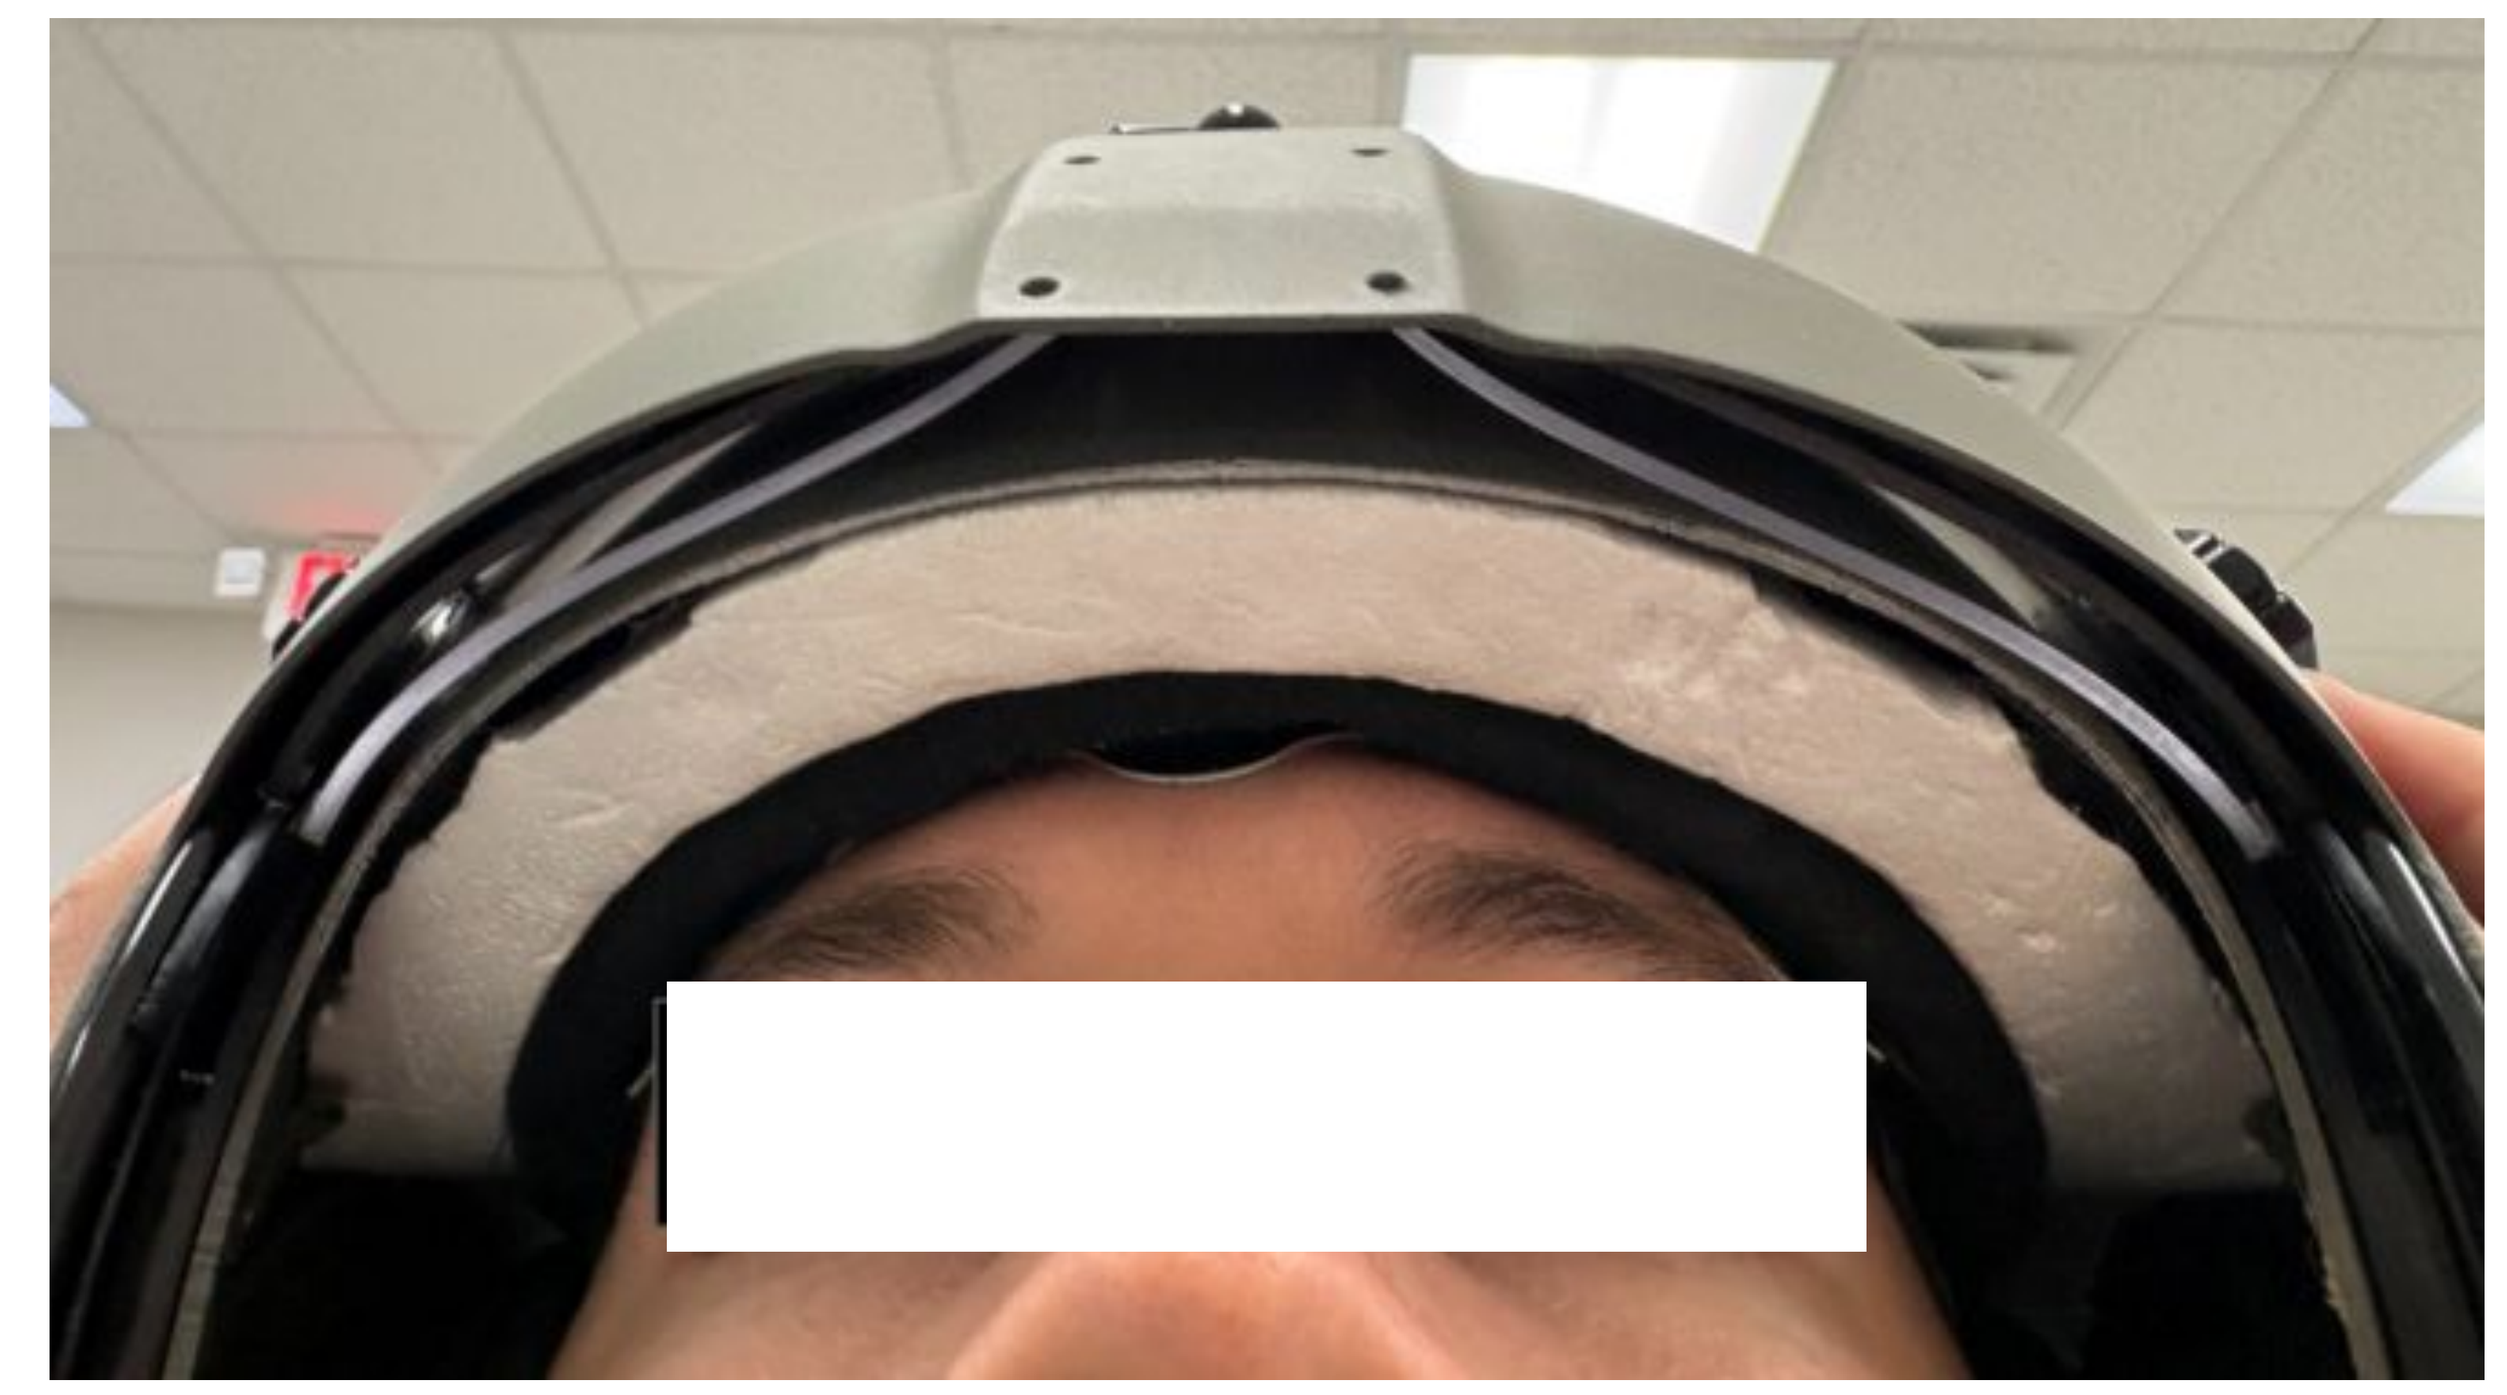

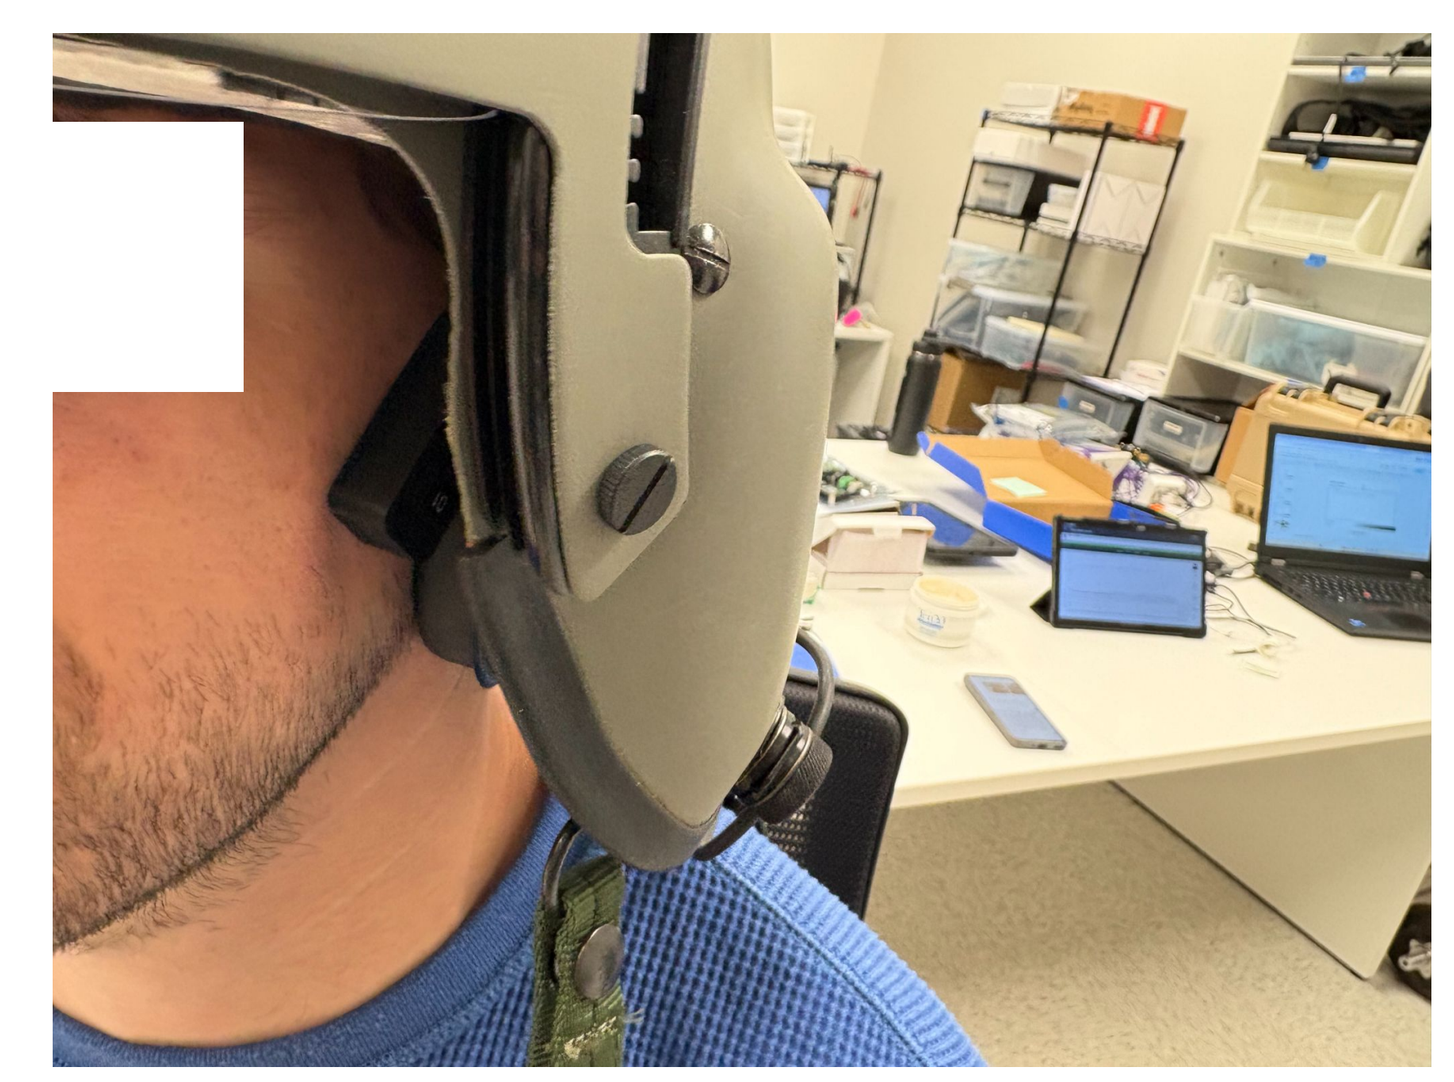

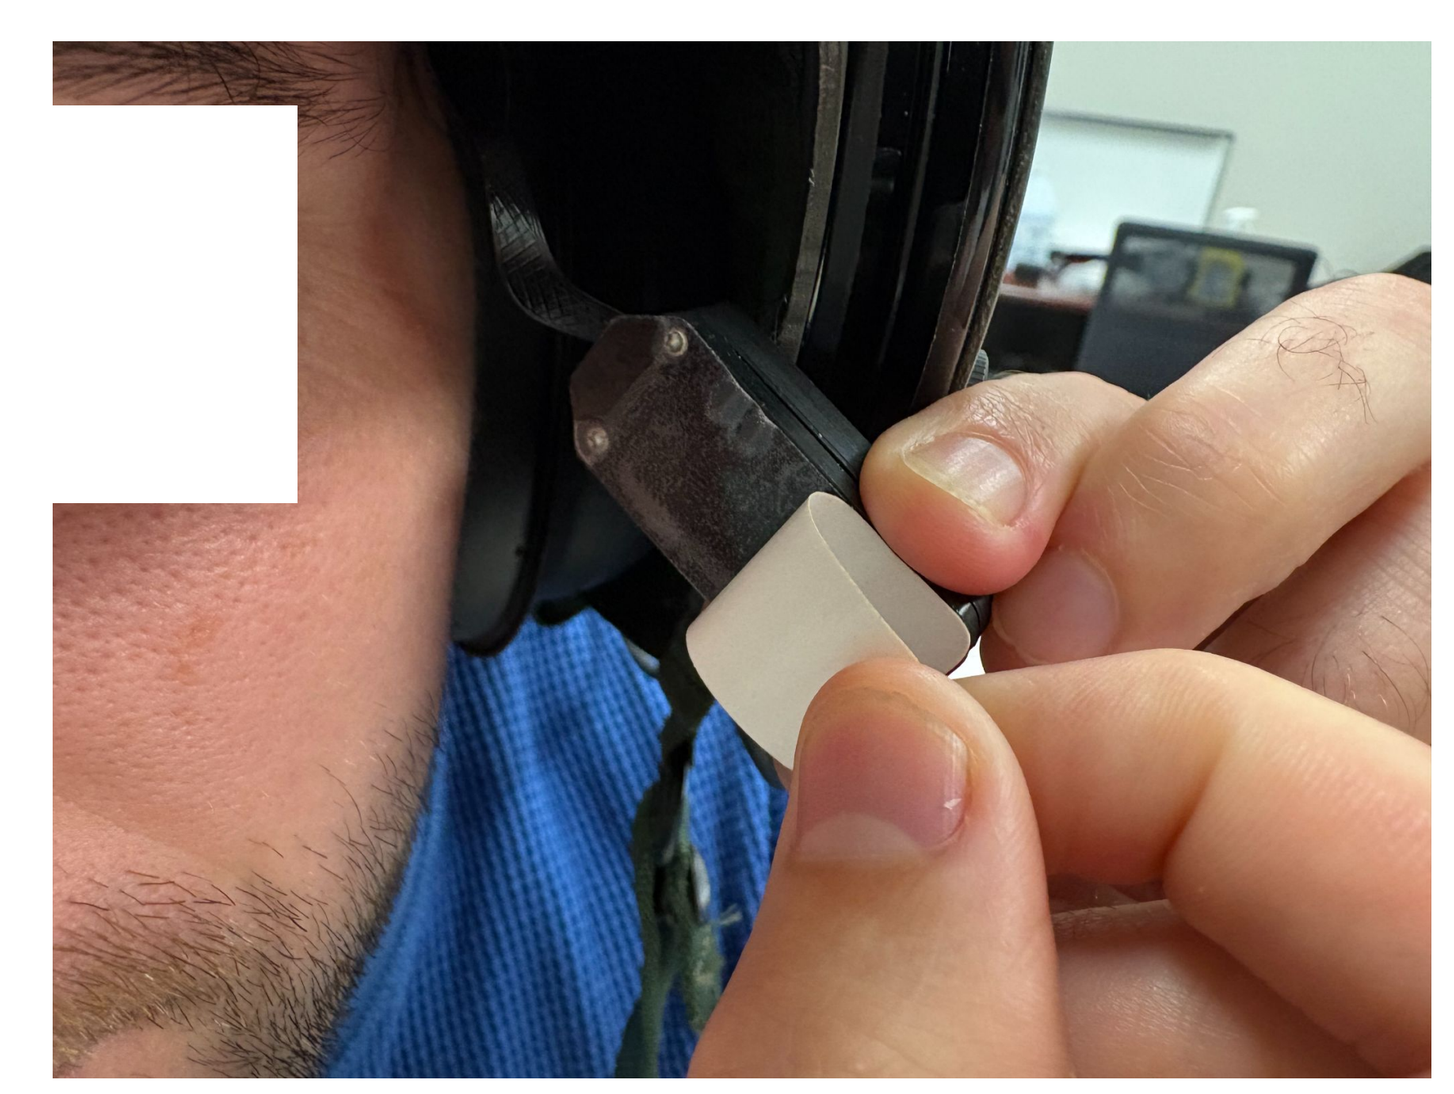

Multimodal Sensing & Data:

Apollo simultaneously streams NIRS, PPG, and EEG over Bluetooth to a mobile application with over 8h of continuous battery life. Software enhancements achieved sampling rates of 128 Hz (NIRS) and 256 Hz (EEG), with real-time display of tissue oxygen saturation (StO_2), SpO_2, pulse rate, and bilateral EEG.

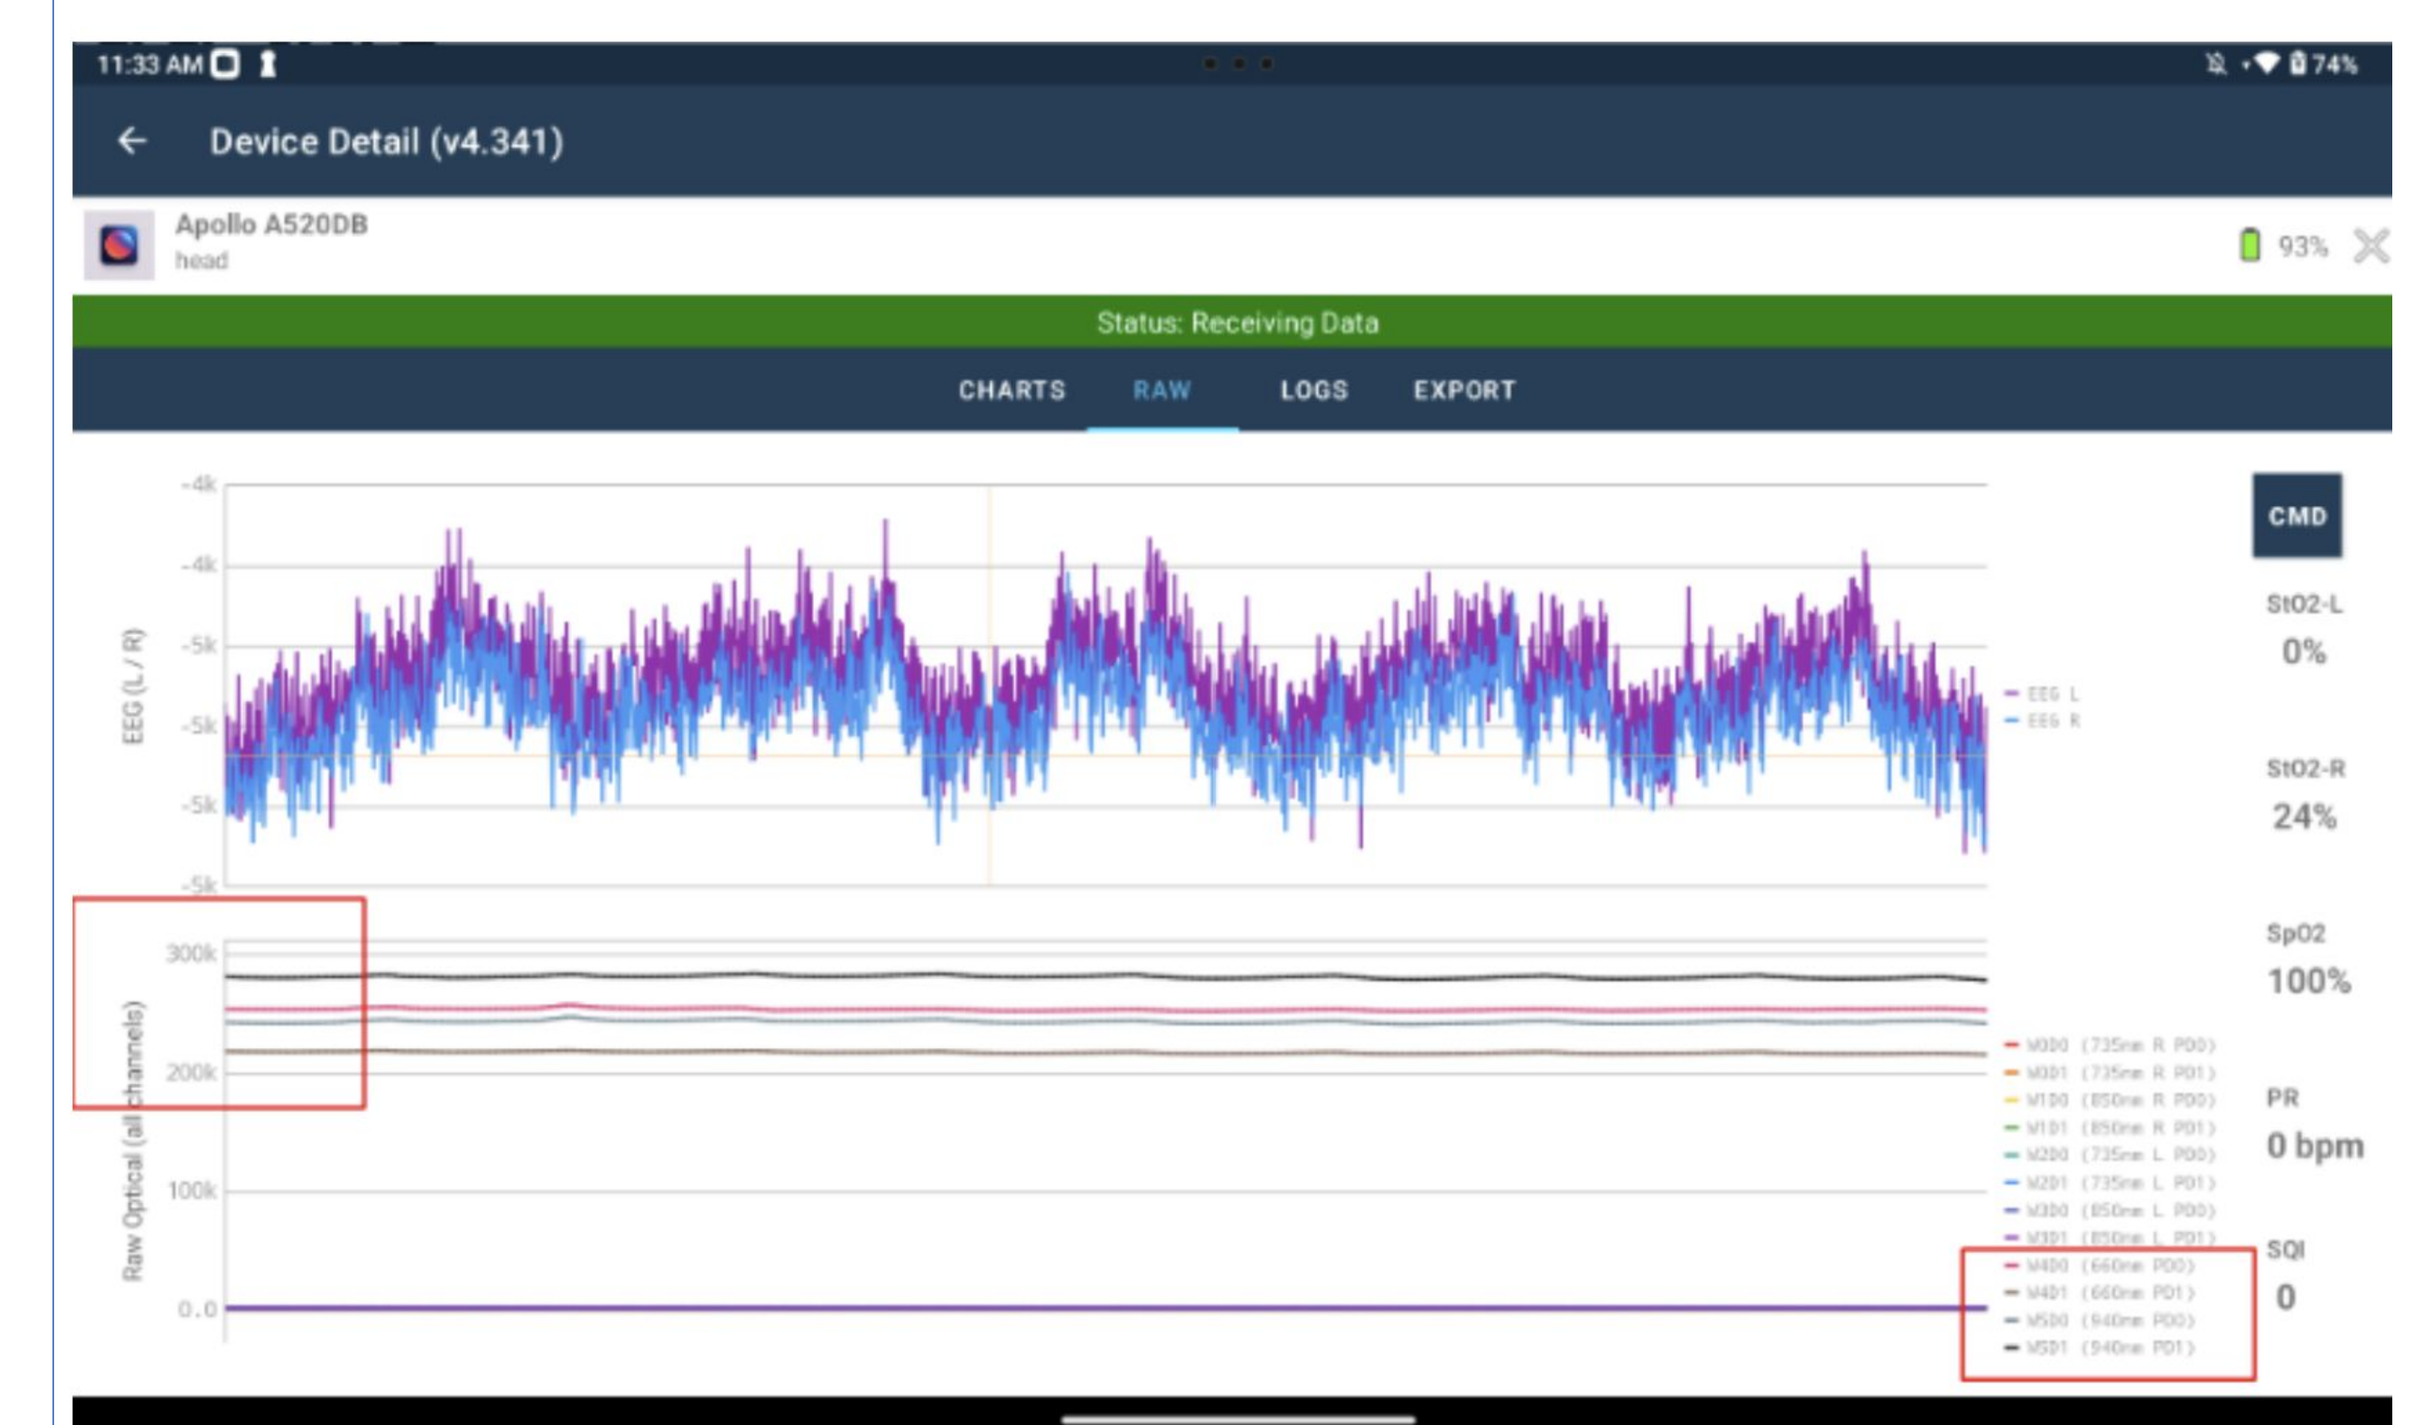
11:33 AM
74%
Device Detail (v4.341)
Apollo A520DB
head
93%
Status: Receiving Data
CHARTS
RAW
LOGS
EXPORT
EEG (L / R)
Raw Optical (all channels)
CMD
StO2-L
0%
StO2-R
24%
SpO2
100%
PR
0 bpm
SQI
0

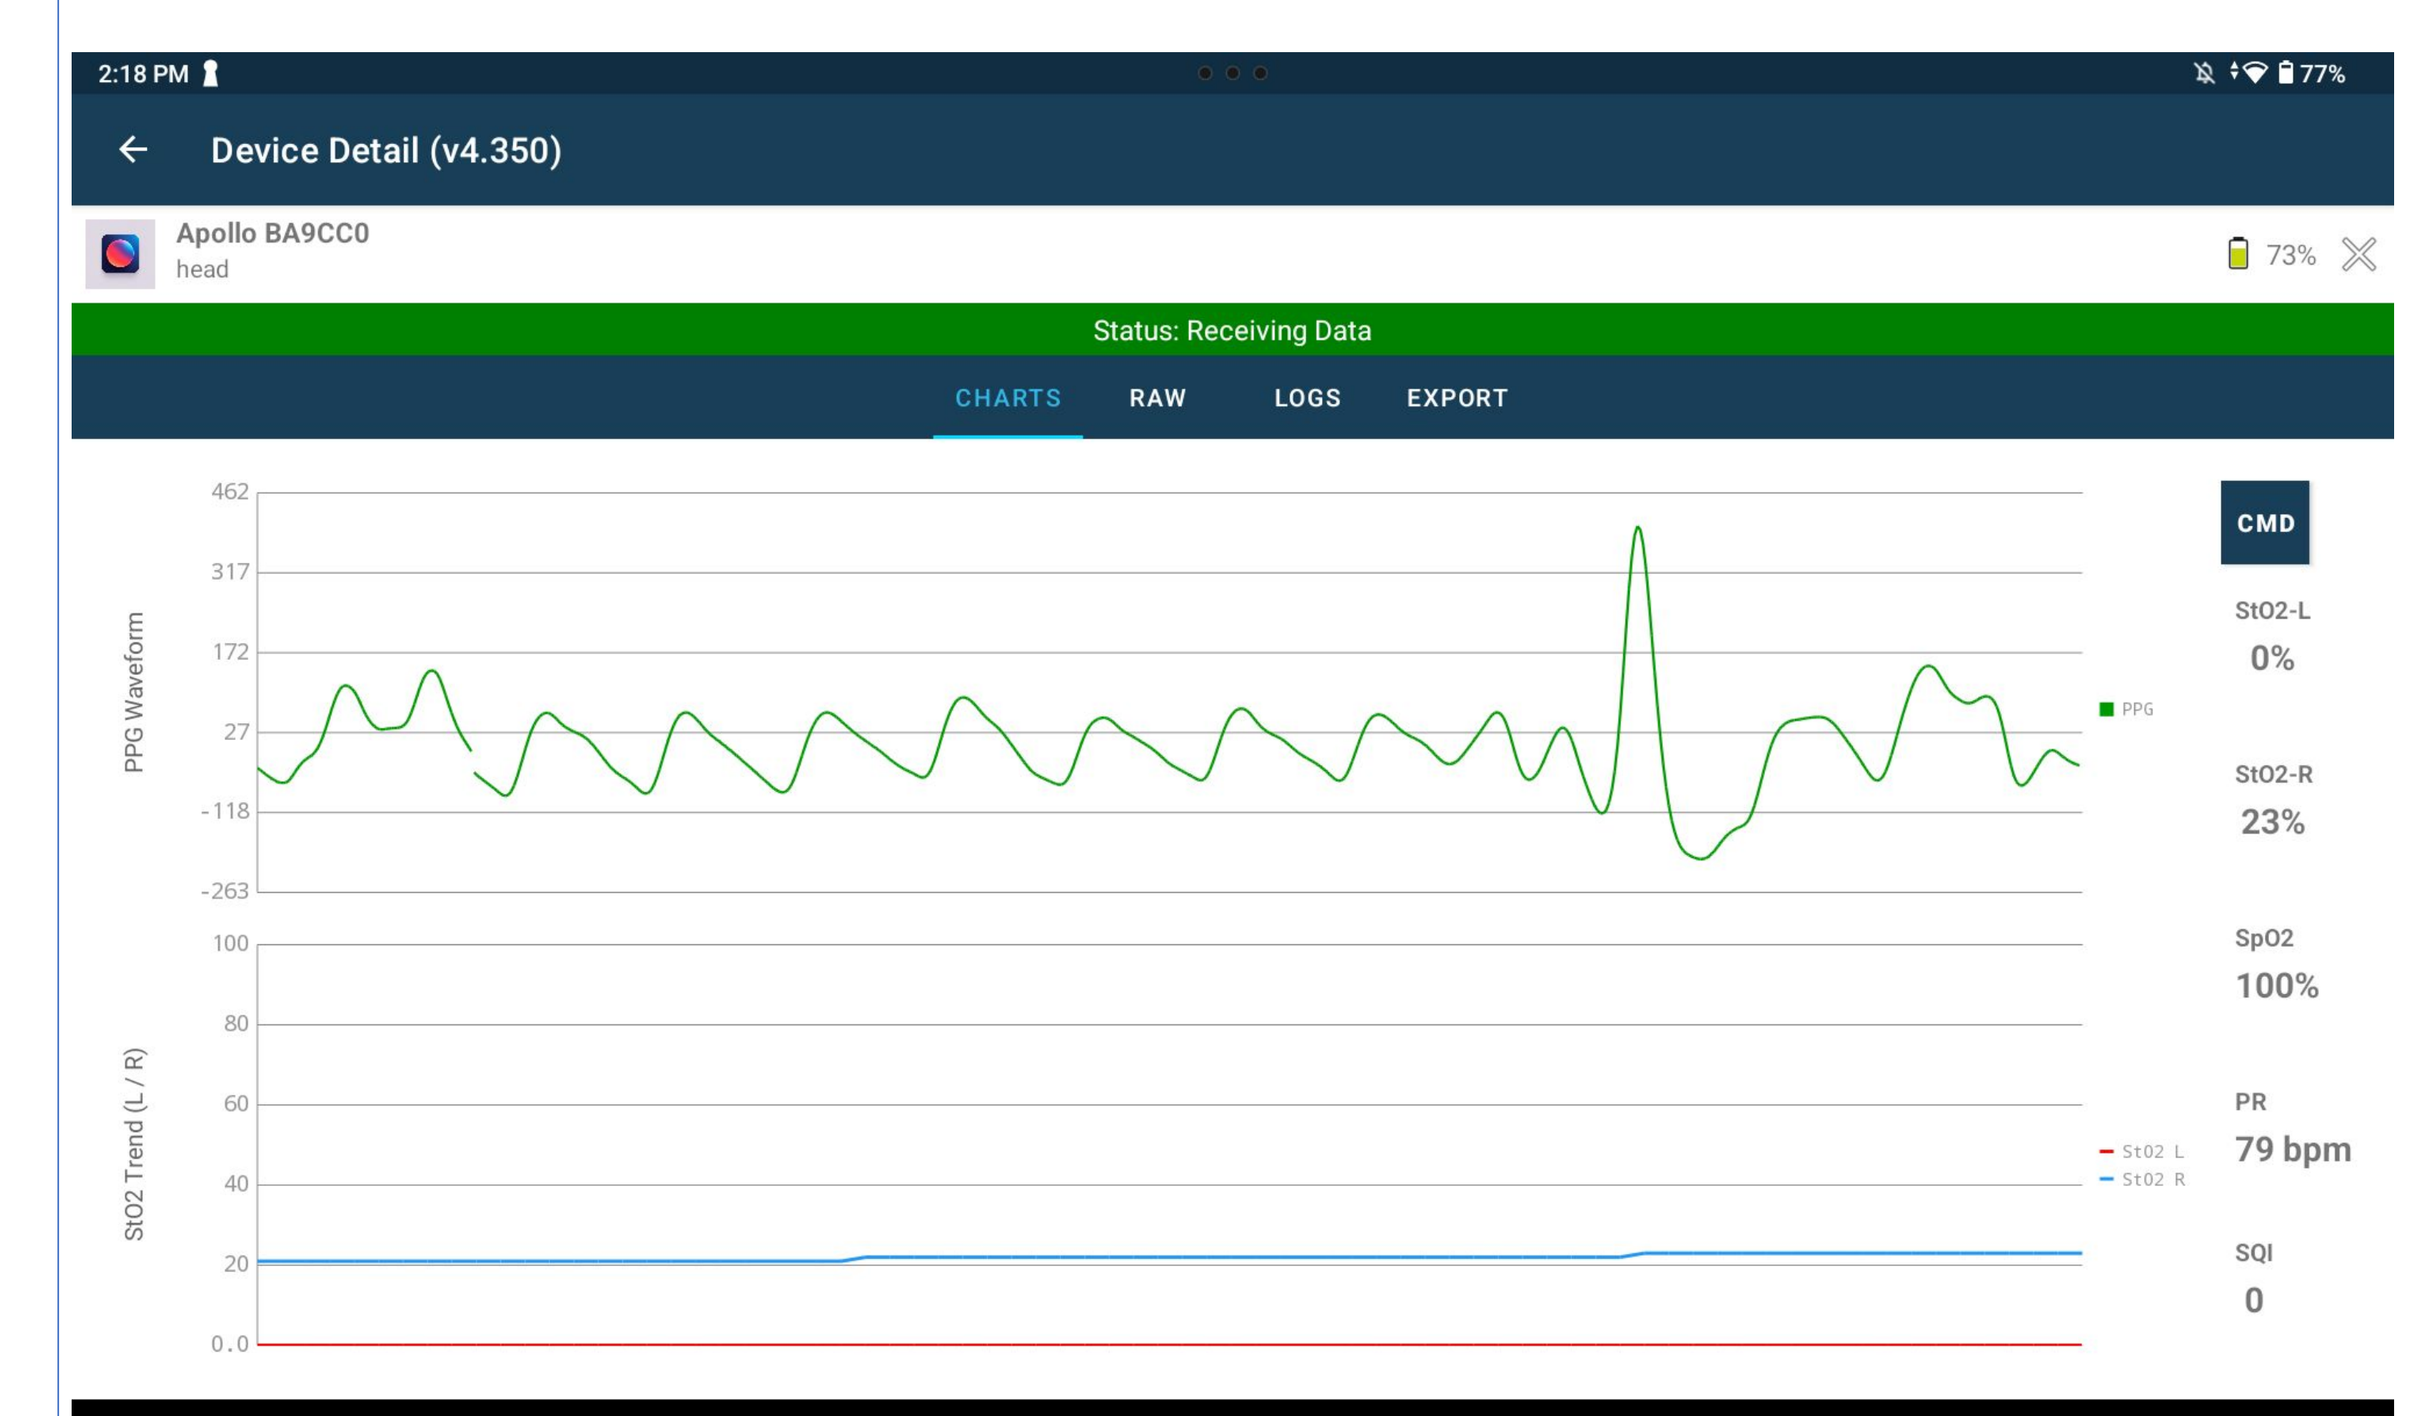
2:18 PM
77%
Device Detail (v4.350)
Apollo BA9CC0
head
73%
Status: Receiving Data
CHARTS
RAW
LOGS
EXPORT
PPG Waveform
StO2 Trend (L / R)
462
317
172
27
-118
-263
100
80
60
40
20
0.0
PPG
StO2 L
StO2 R
CMD
StO2-L
0%
StO2-R
23%
SpO2
100%
PR
79 bpm
SQI
0

Path to Flight:

- Passed Verification & Validation for mechanical durability, adhesion, and electrical integrity
- Several units delivered to USAARL for under-helmet usability evaluation
- Preparing H-60 Airworthiness Release (AWR) with JECETS-reinforced routing for rotary-wing flight testing

Acknowledgements: The views, opinions and/or findings contained in this report are those of the author(s) and should not be construed as an official Department of the Army position, policy or decision unless so designated by other documentation. This material is based upon work supported by the U.S. Army Medical Research and Development Command (USAMRDC) / U.S. Army Medical Research Acquisition Activity (USAMRAA) under Contract No. HT9425-25-C-0006. The U.S. Government is authorized to reproduce and distribute reprints for Governmental Purposes notwithstanding any copyright notation thereon.