Standardized Readiness Assessment for Wearable Physiological Monitoring Systems to Inform Warfighter Readiness

Andy Lam, Ph.D.
Daniel Kingsley, Ph.D., P.E.
Kevin O'Brien, Ph.D., P.E.
Elizabeth T. Lu, P.E.

INTRODUCTION

Wearable physiological monitoring systems are increasingly being used to assess health, performance, and readiness of the Human Weapon System. These systems often combine independently developed hardware, sensors, software, analytics, communications pathways, and decision-support applications. This complexity creates challenges for consistently evaluating technology maturity and readiness.

Current readiness assessments frequently focus on individual components rather than the complete wearable ecosystem. As a result, developers and acquisition programs can encounter uncertainty regarding what testing is appropriate, what evidence is required, and how readiness should be characterized across technologies and operational contexts.

This work proposes a standardized methodology that guides application of Technology Readiness Levels (TRL), Integration Readiness Levels (IRL), and Human Readiness Levels (HRL) to wearable physiological monitoring systems and their Critical Technology Elements (CTE).

PROBLEM STATEMENT

Wearable physiological monitoring systems include multiple interdependent components:

- Sensors and wearable devices
- Edge computing devices and mobile platforms
- Communication pathways
- Algorithms
- Software functions
- Applications
- Enterprise/cloud infrastructures
- Human users and decision makers

Different components mature at different rates and require different evaluation criteria. A single readiness assessment approach does not adequately characterize all elements of a wearable ecosystem.

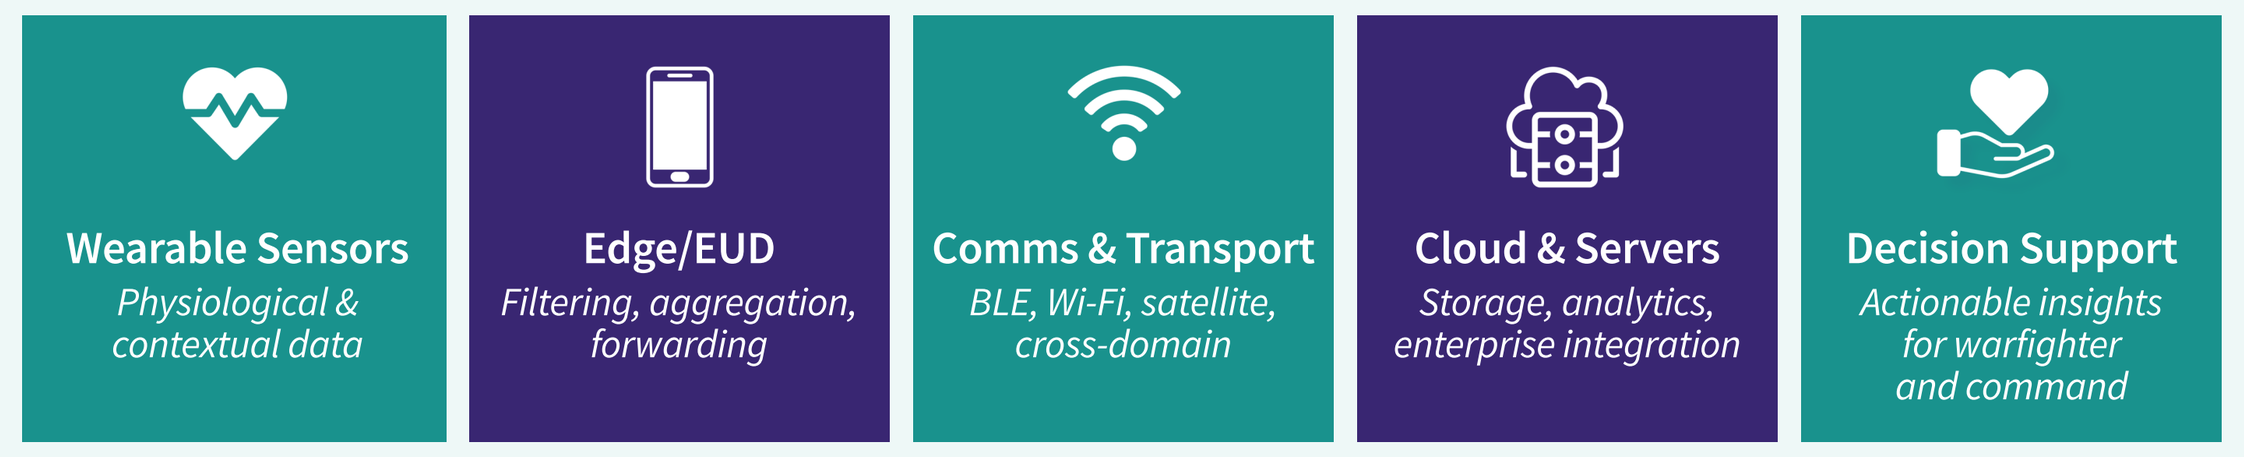
Wearable Sensors
Physiological & contextual data
Edge/EUD
Filtering, aggregation, forwarding
Comms & Transport
BLE, Wi-Fi, satellite, cross-domain
Cloud & Servers
Storage, analytics, enterprise integration
Decision Support
Actionable insights for warfighter and command

METHODOLOGY

Step 1 – Characterize the Wearables Ecosystem

The wearable ecosystem can consist of interconnected layers supporting end-to-end data collection, processing, transmission, analysis, and decision support. Components may include wearable sensors, end-user devices, edge computing resources, applications, cloud/server infrastructure, and secure communication paths (Figure 1).

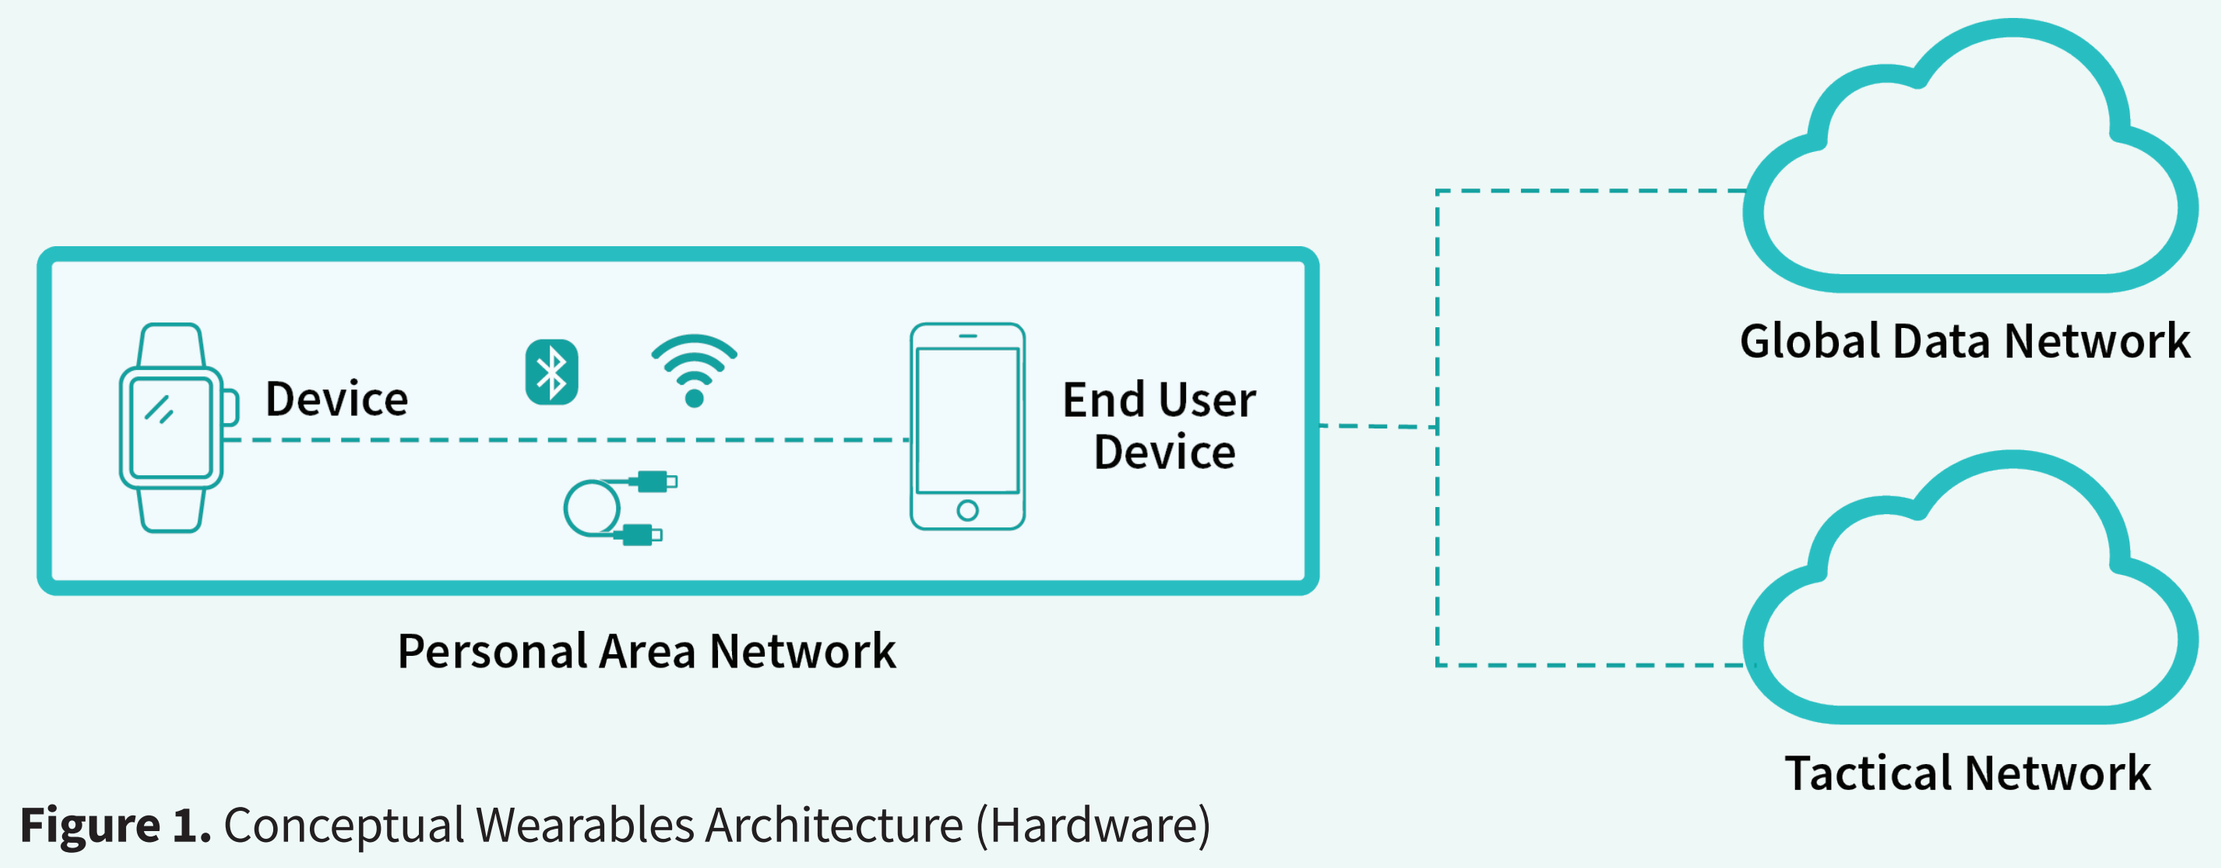
Device
End User Device
Personal Area Network
Global Data Network
Tactical Network

Figure 1. Conceptual Wearables Architecture (Hardware)

Software elements can include algorithms, software functions, and applications (Figure 2), each with maturity and integration requirements and therefore warrant different readiness assessments.

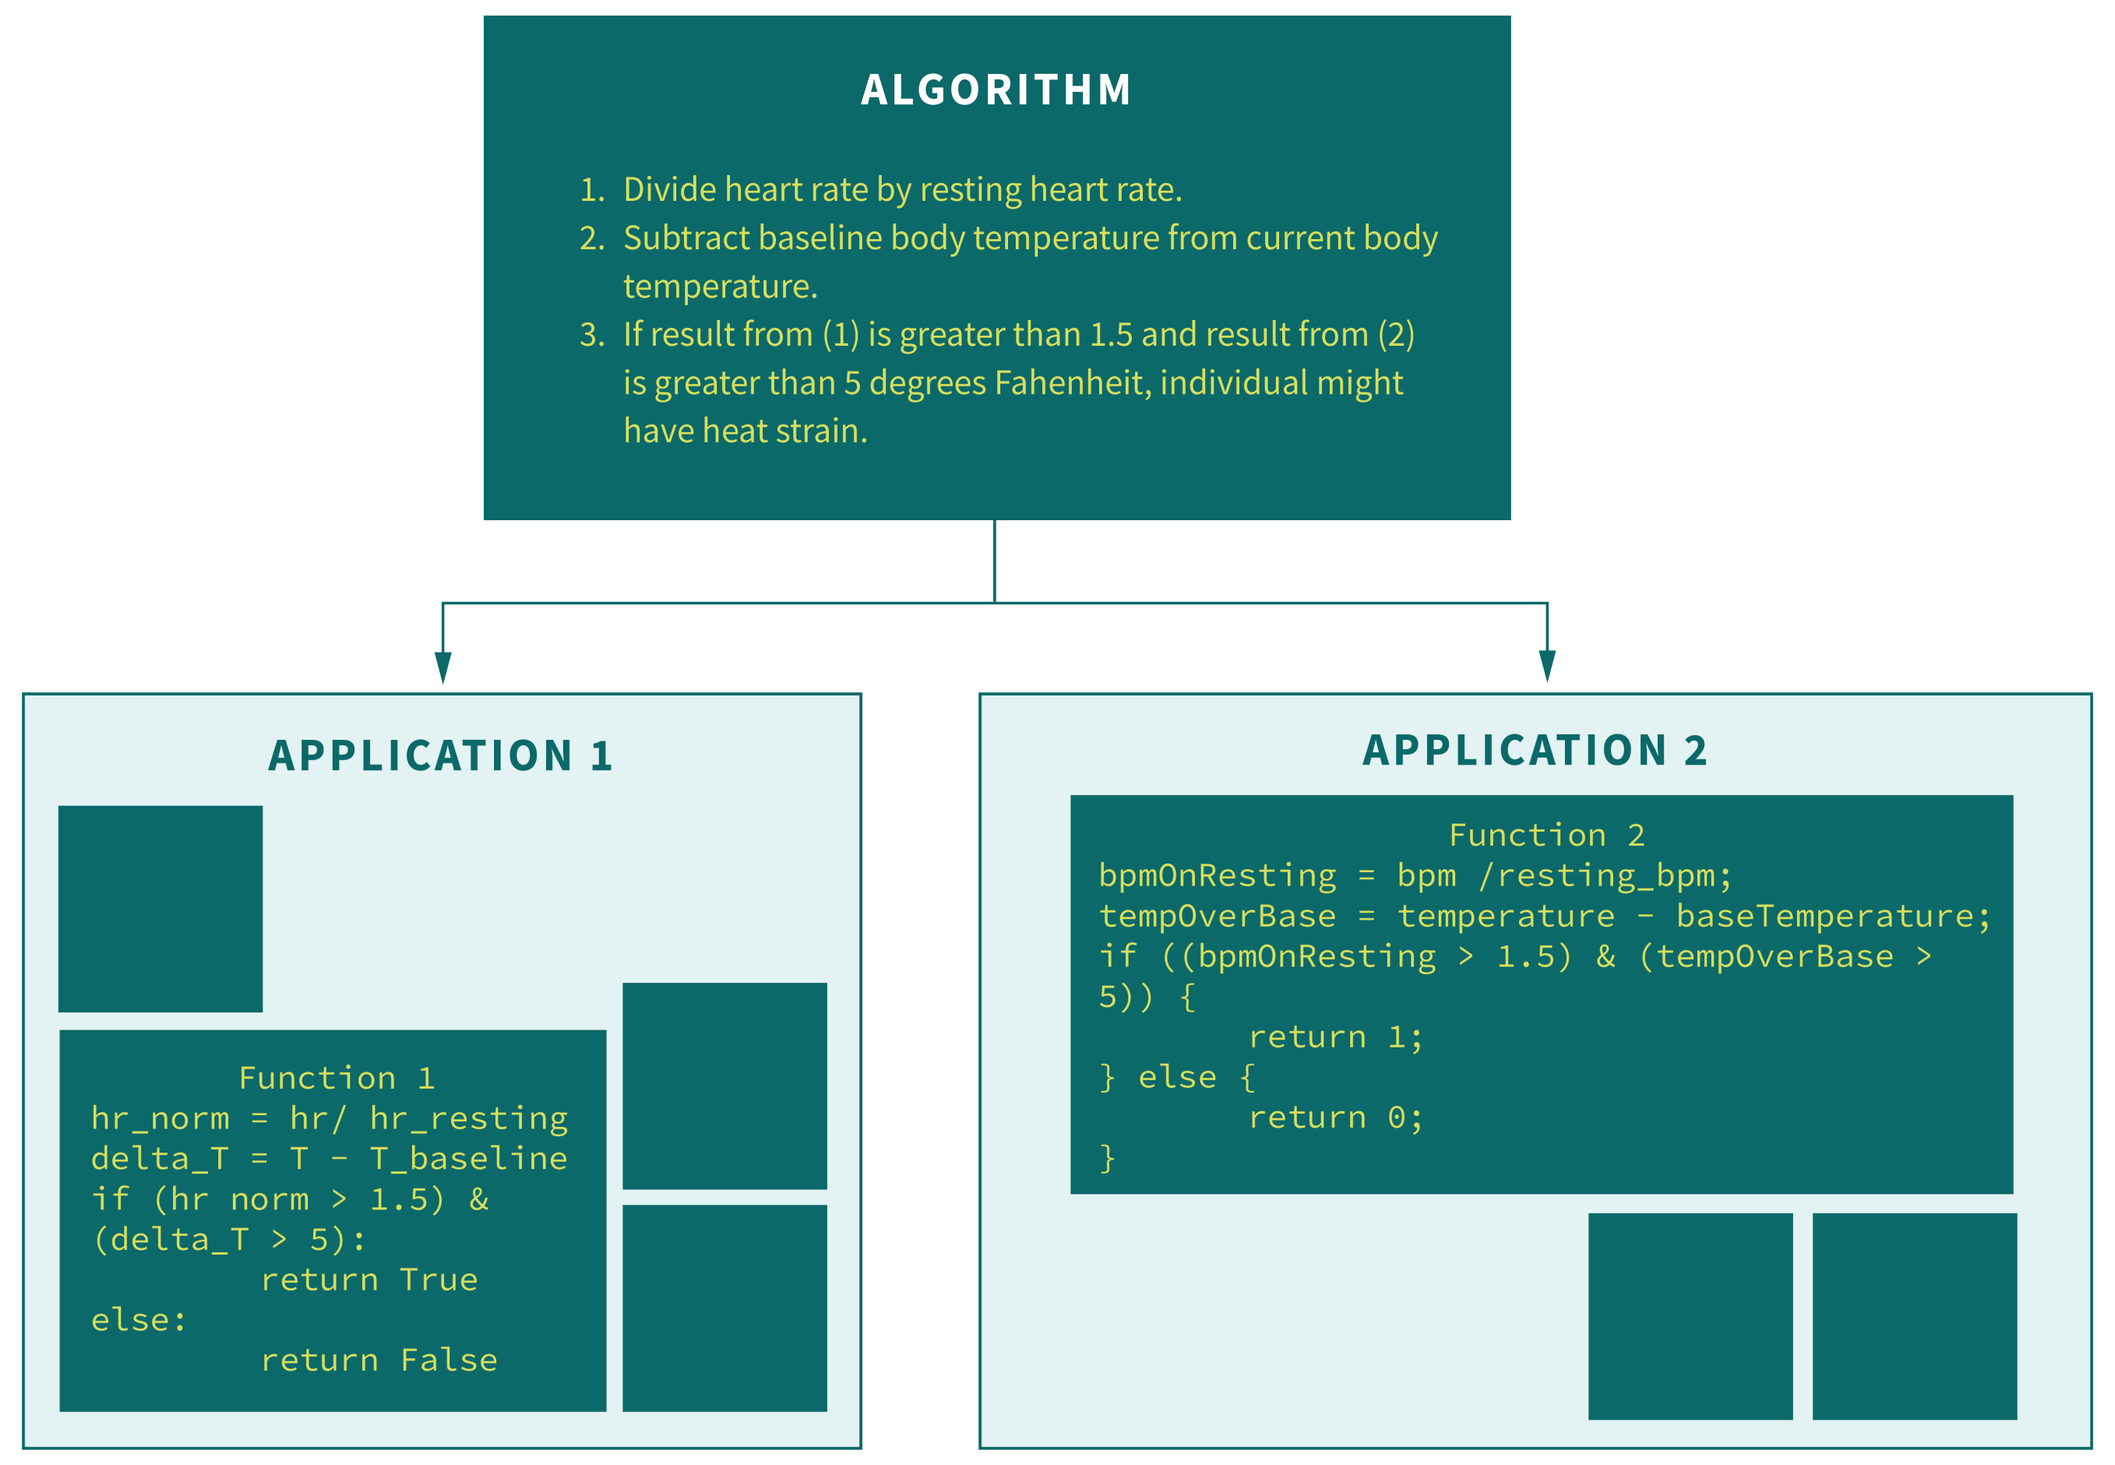
ALGORITHM
1. Divide heart rate by resting heart rate.
2. Subtract baseline body temperature from current body temperature.
3. If result from (1) is greater than 1.5 and result from (2) is greater than 5 degrees Fahenheit, individual might have heat strain.
APPLICATION 1
Function 1
hr_norm = hr/ hr_resting
delta_T = T - T_baseline
if (hr norm > 1.5) &
(delta_T > 5):
return True
else:
return False
APPLICATION 2
Function 2
bpmOnResting = bpm /resting_bpm;
tempOverBase = temperature - baseTemperature;
if ((bpmOnResting > 1.5) & (tempOverBase > 5)) {
return 1;
} else {
return 0;
}

Figure 2. Example of Relationship between Algorithms, Software Functions, and Application

Readiness must be evaluated across technology maturity, system integration maturity, and human interaction maturity, rather than through component performance alone.

Step 2 – Identify the Critical Technology Element

CTE development associated with wearable systems typically falls into one of a limited number of categories.

- New/updated hardware component
- New/updated wearable device
- Novel use of existing hardware
- New operating system
- New algorithm
- New software function
- New application
- Novel use of an existing algorithm or device

Each category follows a different readiness evaluation pathway (Table 1).

Table 1: Scope of Testing & Evaluation (T&E) for New/Updated/Novel Use Wearable System CTEs

Wearables CTE	Type[1]	CTE TRL T&E Scope	CTE IRL T&E	System IRL T&E
New/Updated Component	HW	Sensor	Sensor → Device	Sensor → Function(s) → System
Updated Device (legacy SW)	HW	Device	Device → OS	Device → Function(s) → System
New Device (new SW)	HW/SW	Device; OS	Device → OS	Device → Function(s) → System
New OS (legacy HW)	SW	Function(s)	Function(s) →Device	Function(s) → Device → System
Novel Usage (legacy HW/SW)	HW/SW	Device; OS	Device → OS	Function(s) → Device → System
New Algorithm or Novel Use of Existing Algorithm	SW	Algorithm	Algorithm → Function	Algorithm → Device → System
New Function/Capability	SW	Function	Function → Application	Function → Device → System
New Application	SW	Application	Function(s) → Application	Function(s) → Device → System

[1] HW = hardware, SW = software, OS = operating system

Given this, the opportunity exists to describe and clarify how to apply the respective definitions for TRL and IRLs to the categories of CTEs typically associated with wearable systems.

Step 3 – Apply Appropriate Readiness Constructs

Table 2: Readiness Constructs

Description	Scope
Technology Readiness Level Component maturity for hardware, algorithms, software functions & applications	• Device • Hardware • Algorithm • Function • Application
Integration Readiness Level Interoperability across sensors, devices, functions, systems	• Sensor → Device • Algorithm → Function • Function → Application • Device → Operating System • End-to-End Wearable Ecosystem
Human Readiness Level Safe, effective user interaction per ANSI/HFES 400-2021	• User trust • Cognitive workload • Decision support effectiveness • Human-system interaction • Operational usability

Step 4 – Evaluate Common Domains

The methodology identifies recurring assessment domains that should be considered across readiness evaluations.

Table 3: Key Evaluation Topics

Topic	Typical Scope
Human System Interface	Assesses how new technologies are able to effectively interface with the human system and how prepared individuals or teams are to effectively engage with new technologies or environments.
Functional Validations	Confirms that each component (i.e., wearable sensor, firmware module, or analytics process) performs its intended tasks under expected conditions.
Performance and Reliability Assessments	Measures critical metrics such as data accuracy, communication stability, and resilience to environmental or input errors.
Integration and Interoperability Checks	Ensures that wearable devices, edge computing units, servers, and networks interconnect seamlessly while identifying potential interface failure points.
Cybersecurity and Data Protection	Probes vulnerabilities across components, verifies secure data transmission across BLE, Wi-Fi, satellite, and cross-domain links, and validates that updates uphold Zero Trust principles.
Data Architecture and Metadata Alignment	Confirms conformance with the wearables data model designated by the CPE CBRND JPM CBRN IEW, acting through its Chemical-Biological (CB) Wearables Project Office.
Device Wear Compliance and Usability Evaluations	Assesses real-world user interactions and confirms minimum usage thresholds are met to support use case requirements.
Quality Assessment	Compares current state of critical metrics (Performance & Reliability) to success metrics.
Optimization Strategy	Acknowledges technology-specific efforts and achievements that align with the overall purpose of system.
Policy and Regulations Compliance	Assesses real-world feasibility of usage and confirms minimum compliance thresholds are met to support authorizations to operate.

Step 5 – Advance via Exit Criteria Guidance

The Hardware and Software Technology Readiness Levels and Integration Readiness Levels guidance provided in the DoD Technology Readiness Assessment Guidance were supplemented with guidance specific to wearable physiological monitoring systems and their components. The same was done for the Human Readiness Level guidance from the American National Standards Institute (ANSI) and Human Factors and Ergonomics Society (HFES) 400-2021 standard. The following tables were generated.

- Applying Hardware TRLs to a New/Updated Wearable Device/Hardware Component(s) or Novel Use of an Existing Wearable Device
- Applying Software TRLs to a New/Updated/Novel Use of Existing Physiological Monitoring Algorithm
- Applying Software TRLs to a New/Modified Function
- Applying IRLs to a New/Updated Application Containing a New/Modified/Existing Feature (Algorithm or Function)
- Application of Integration Readiness Levels for Wearable Systems
- Application of Human Readiness Levels for Wearable Systems

RESULTS

Standardization
- Consistent approach to readiness characterization across varying wearable technologies
- Common terminology for developers, testers, and decision makers

Repeatability
- Repeatable readiness evaluation framework for diverse wearable technologies
- Reduced variability in assessment approaches

Improved Test & Evaluation Planning
- Clear identification of required evidence
- Better alignment of testing activities with maturity objectives

Reduced Program Risk
- Improved understanding of technical gaps
- Early identification of integration and usability risks

Increased Decision Confidence
- More reliable readiness determinations
- Better-informed technology transition decisions

OPERATIONAL IMPACT

The framework supports wearable systems intended to inform health, performance, and readiness across military operational environments and medical roles of care. A consistent readiness assessment methodology enables trusted use of wearable-derived physiological information by:

- Improving confidence in readiness metrics
- Supporting individualized monitoring
- Enhancing operational decision support
- Reducing technical and programmatic risk
- Increasing mission effectiveness

CONCLUSIONS

1. Wearable physiological monitoring systems require evaluation of hardware, software, integration, and human interaction as a unified ecosystem.
2. Different wearable CTE categories require different readiness assessment pathways.
3. Application of TRLs, IRLs, and HRLs provides a structured and evidence-based evaluation framework.
4. Standardized readiness assessments improve test planning, reduce risk, and support confident readiness decisions for the Human Weapon System.

DEVELOPMENTAL STATUS

The methodology is currently being applied and refined as part of CPE CBRND JPM CBRN IEW, acting through its CB Wearables Project Office to evaluate wearable physiological monitoring systems and associated technologies.

ACKNOWLEDGMENTS

This work was funded by CPE CBRND JPM CBRN IEW, acting through its CB Wearables Project Office to evaluate wearable physiological monitoring systems and associated technologies. The views expressed in the presentation are those of the authors and do not necessarily express the office policy and position of CPE CBRND, the US government, or the institutions affiliated with the authors.

REFERENCES

1. Exponent, *Standardized Methodology for Evaluating Wearable Systems* Report to JPEO-CBRND, 18 August 2025
2. United States Department of Defense, *Technology Readiness Assessment Guidebook*, p. 5, Feb 2025.
3. TRA-Guide-Feb2025-Cleared.pdf, accessed 28 July 2025.
4. Human Factors and Ergonomics Society News, June 19, 2025, accessed on July 16, 2025.
5. Massachusetts Institute of Technology Lincoln Laboratory and Exponent, *Wearables Systems Readiness Level Assessment Phase 1 Report*. February 2024.