CHEMICAL OXYGEN GENERATION TO SUPPORT PROLONGED FIELD CARE AND DELAYED EVACUATION

Andrew Maier | Molecular Products Inc., Louisville, CO

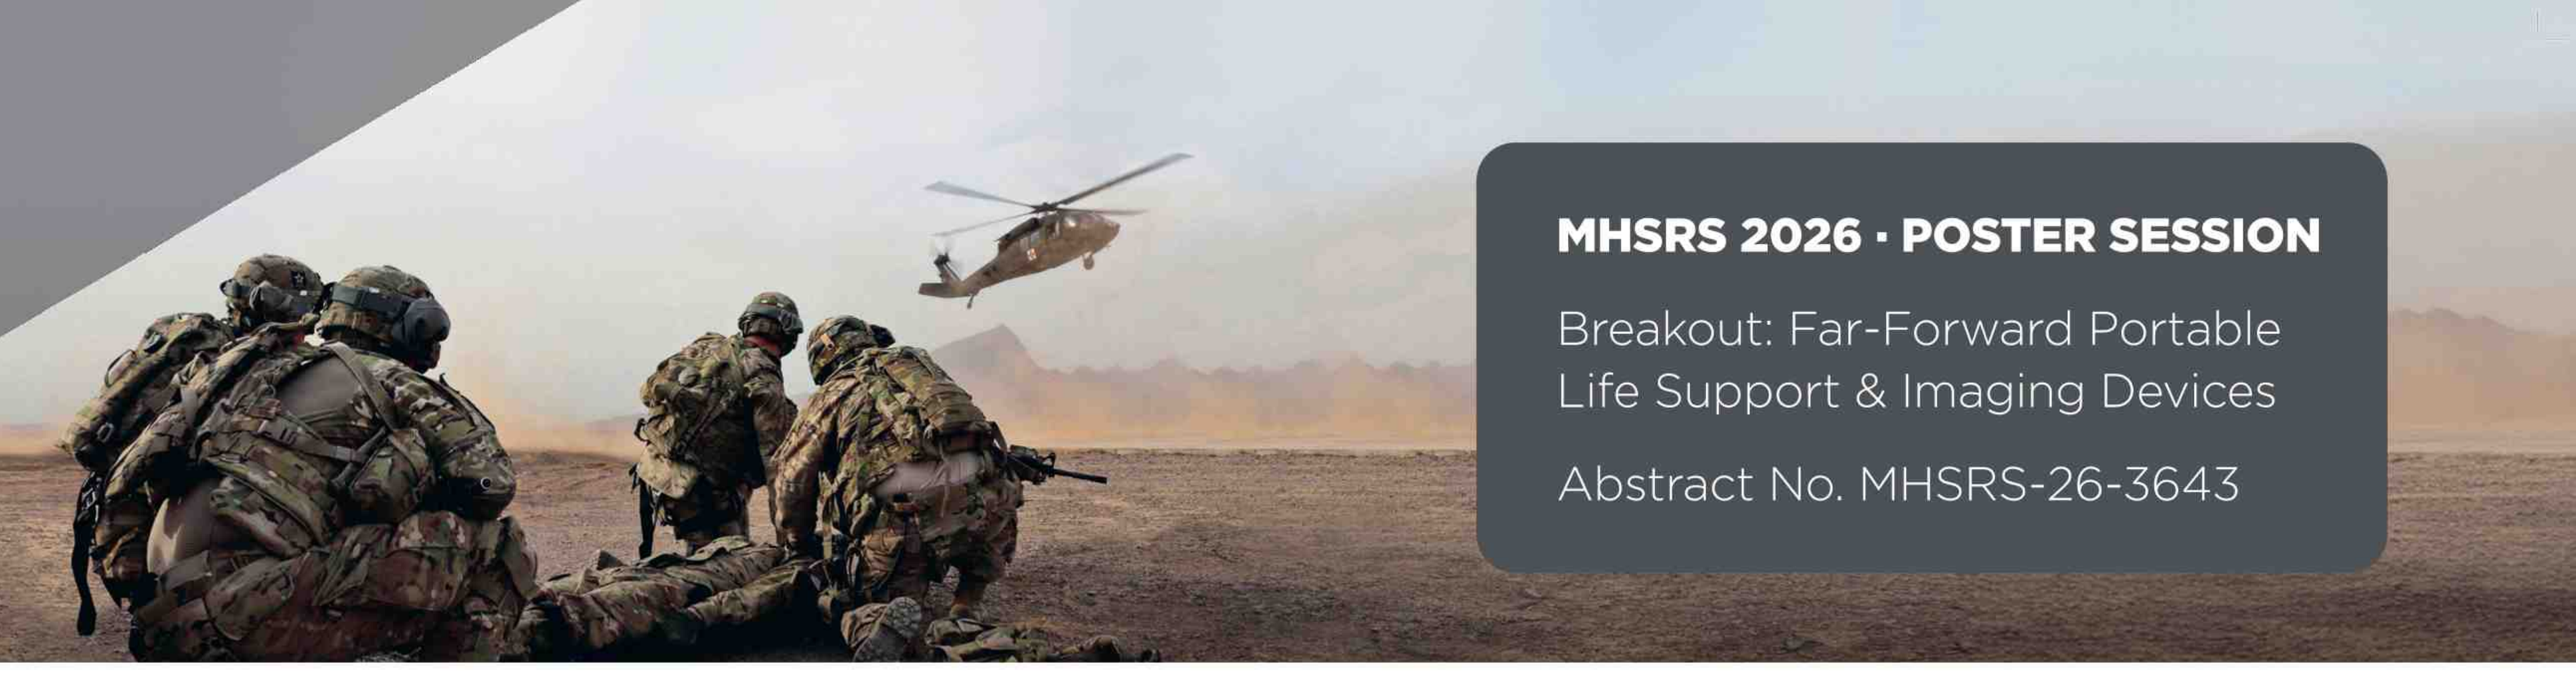

MHSRS 2026 · POSTER SESSION

Breakout: Far-Forward Portable Life Support & Imaging Devices

Abstract No. MHSRS-26-3643

LO 1 — LEARNING OBJECTIVE 1: LIMITATIONS
Describe the operational limitations of conventional oxygen delivery systems during prolonged field care and delayed evacuation.

LO 2 — LEARNING OBJECTIVE 2: CHARACTERISTICS
Analyze the characteristics of chemical oxygen generation as a power-independent oxygen source for austere medical environments.

LO 3 — LEARNING OBJECTIVE 3: INTEGRATION
Discuss integration of portable oxygen generation into point-of-injury and Role 1 operations to support casualty stabilization during delayed evacuation.

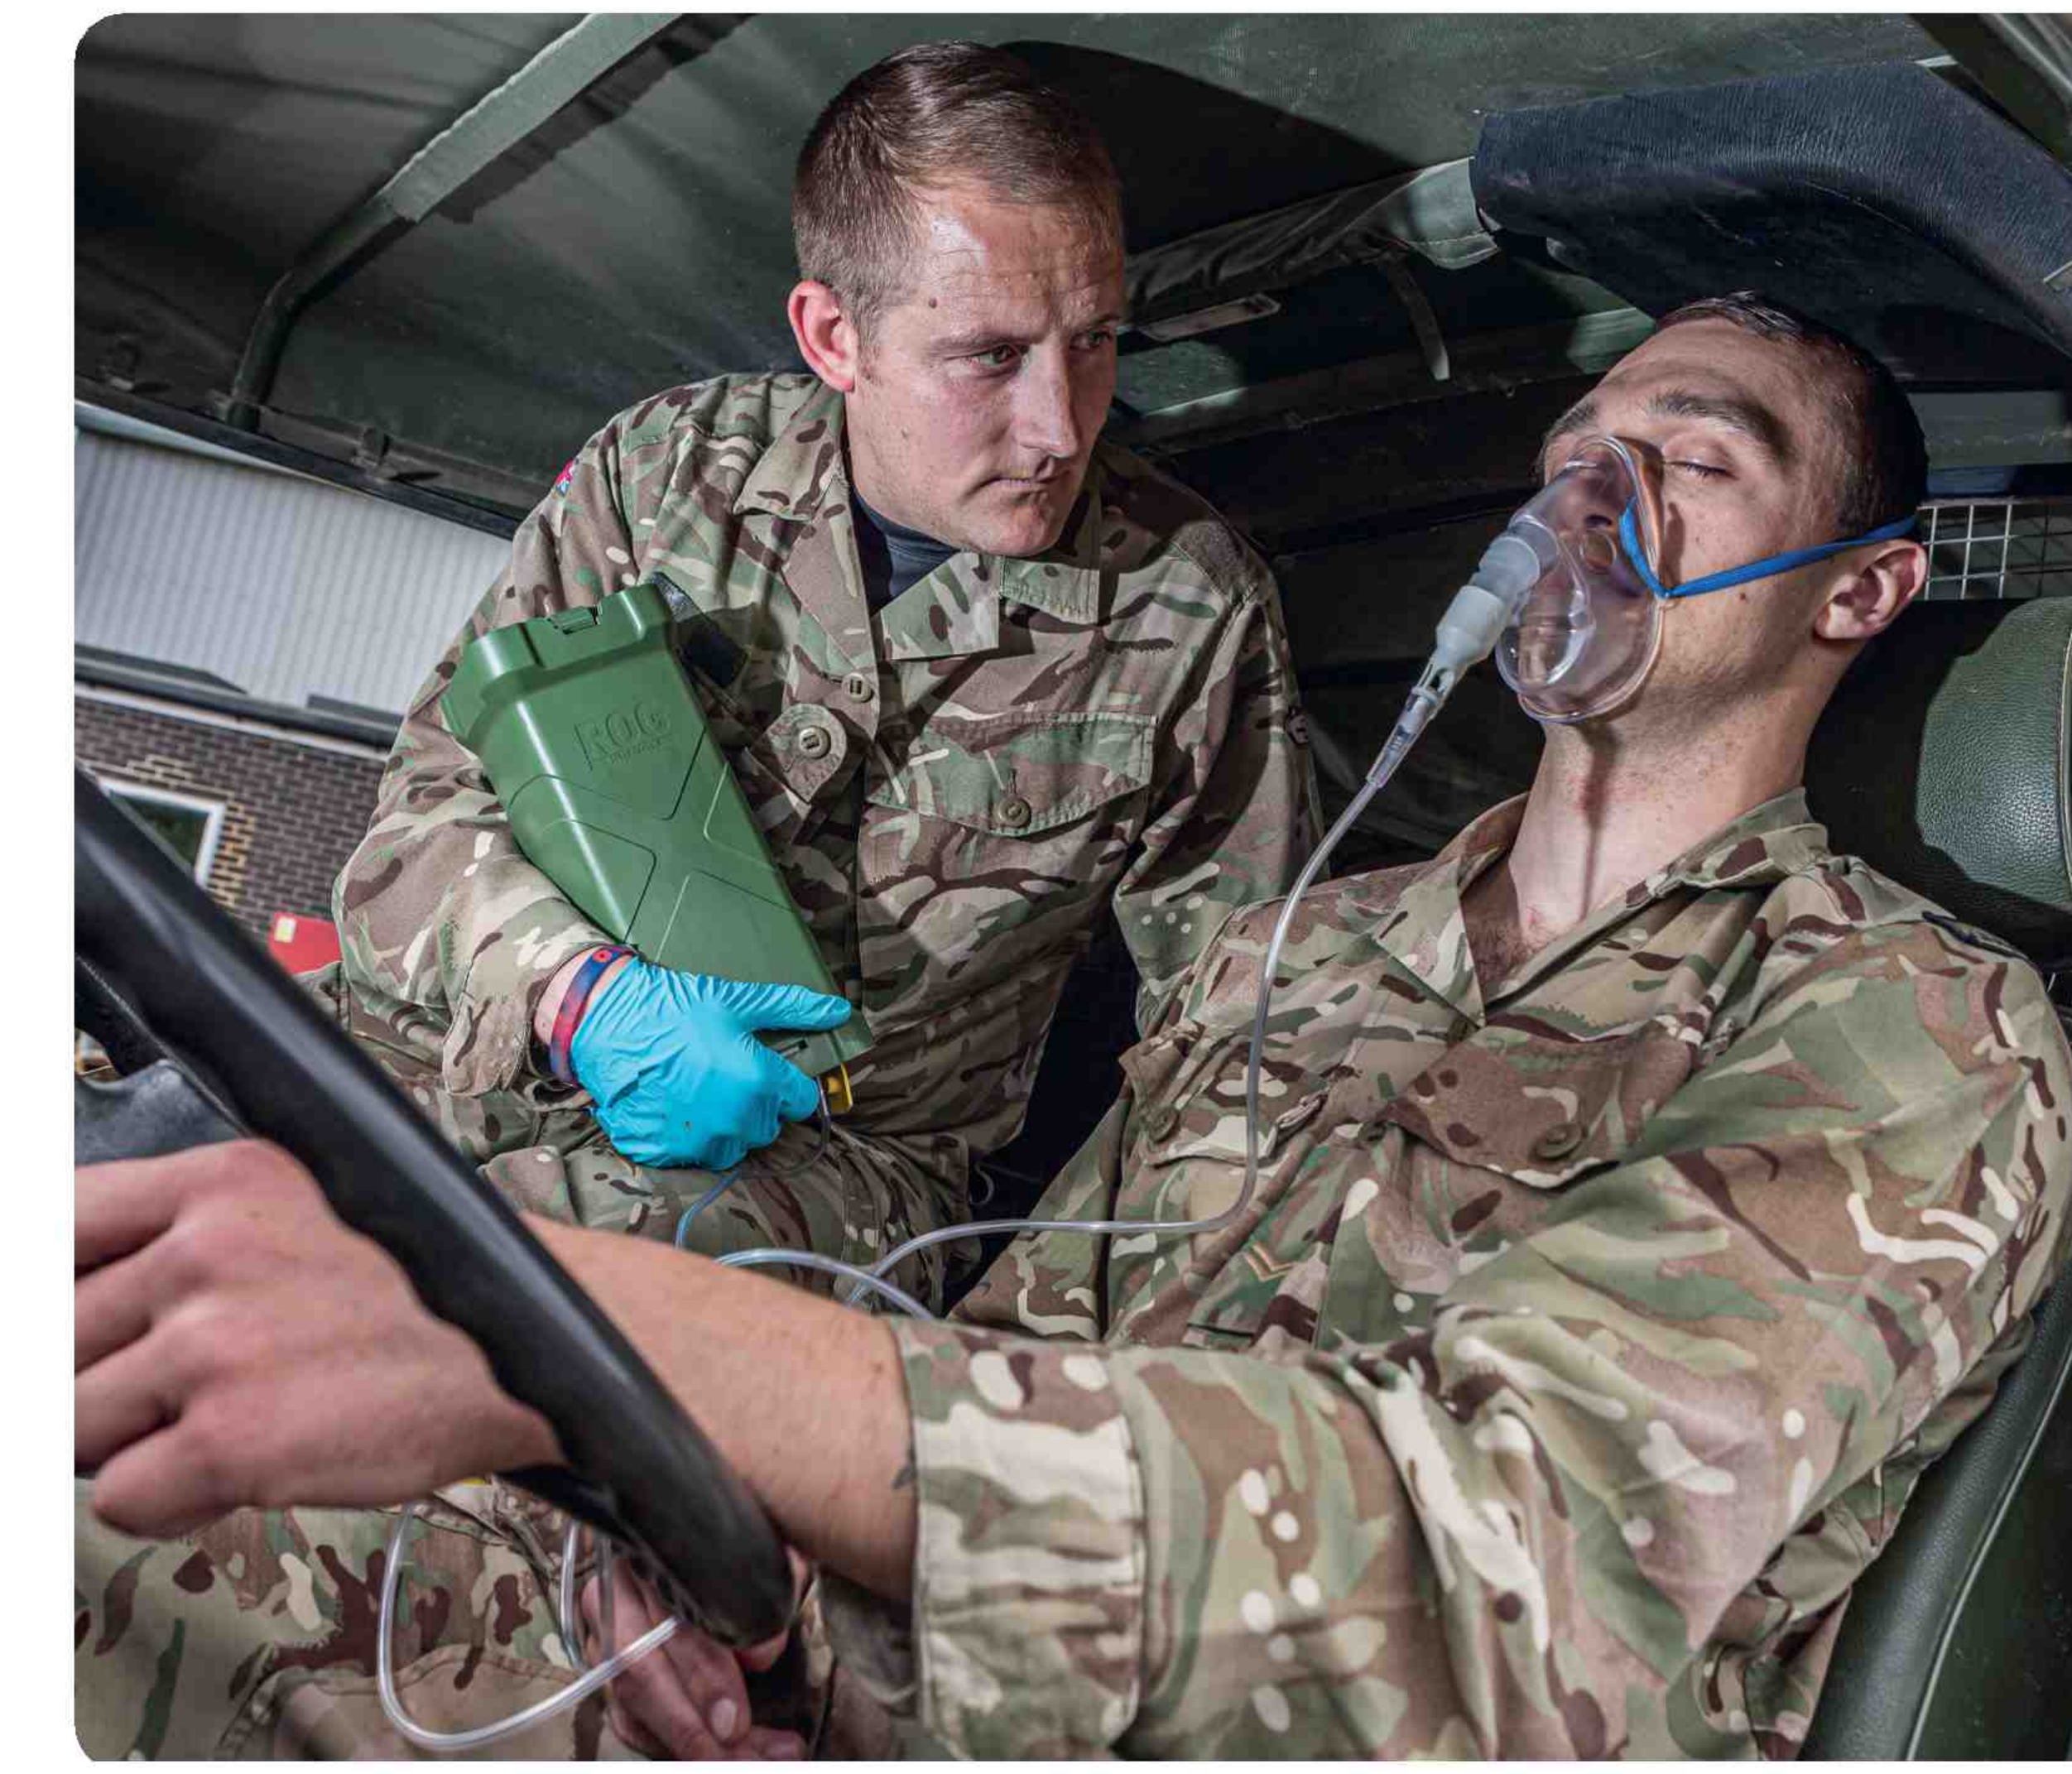

WHEN EVACUATION IS DELAYED, HOW DOES OXYGEN REACH THE CASUALTY?

A portable, power-independent oxygen generation capability was evaluated for austere environments where compressed gas and powered concentrators are impractical — supporting oxygen delivery at the point of injury and during prolonged prehospital care.

Reaction is exothermic

$$2NaClO_{3(s)} \rightarrow 3O_{2(g)} + 2NaCl_{2(s)} + \Delta$$

SUMMARY OF BENCH & OPERATIONAL TEST RESULTS

≥96%	≥6 L/min	≥15 min	≥90 L	5.4 lb	0 w
oxygen purity	average flow	delivery duration	delivery duration	2.45 kg, handheld	external power

1 INTRODUCTION (LO 1)

23% of U.S. combat fatalities were potentially survivable

87% of those deaths occurred before reaching a medical treatment facility

- Hypoxia contributes to preventable trauma mortality — particularly in traumatic brain injury, airway compromise, burns, and thoracic injury.
- Compressed cylinders: fragmentation hazard, weight, and refill logistics limit employment at the point of wounding (LO1).
- Powered concentrators: dependent on batteries or generators; purity and flow degrade with altitude, heat, dust, and humidity (LO1).
- Army Futures Command and the Medical CDID identify far-forward oxygen delivery as a critical capability gap.
- Future operations are expected to extend evacuation timelines and reliance on prolonged prehospital care.

CLINICAL CONTEXT — TCCC GUIDELINES (MAY 2026)

Updated TCCC guidelines raised the prehospital SpO_2 target from >90% to ≥92%. Sustaining this target through delayed evacuation requires oxygen sources that can be employed forward of Role 1.

2 CAPABILITY DESCRIPTION (LO 2)

Sealed chemical oxygen generation produces medical-grade oxygen through a self-contained, non pressurized reaction. The process requires no external power; passive thermal moderation conditions the gas to ambient breathing temperature before delivery. The system is single-use and maintenance-free in storage.

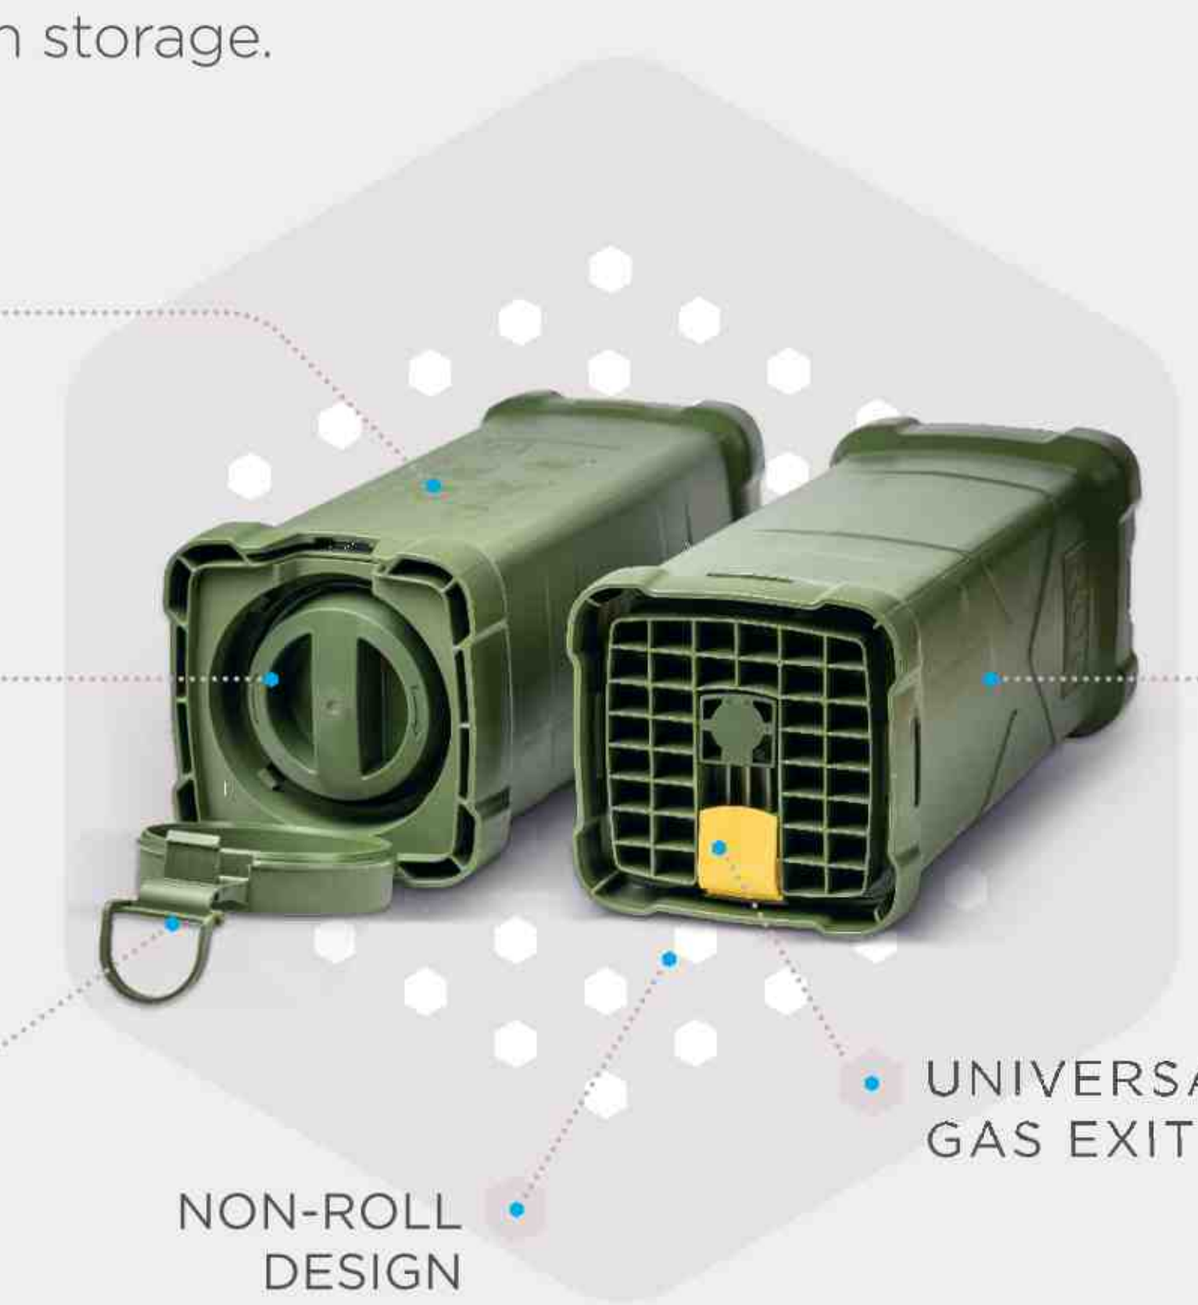
PICTOGRAM INSTRUCTIONS FOR USE
SIMPLE TWIST ACTUATOR DIAL
TAMPER EVIDENT COVER LATCH
NON-ROLL DESIGN
RUGGED CASING
UNIVERSAL, FLEXIBLE GAS EXIT PORT

Activation: open lid → turn dial 2–3 turns → oxygen flows. No valves, regulators, or power sequencing. Standard outlet (BS EN 13544-2) is compatible with existing masks, tubing, and airway equipment — no workflow changes.

CHARACTERISTICS VS. CONVENTIONAL OXYGEN SOURCES (LO1, LO2):

CHARACTERISTIC	CHEM. GEN.	CYLINDER	CONCENTR
Non-pressurized (no blast hazard)	✓	✗	✓
No electrical power required	✓	✓	✗
Purity maintained in extreme	✓	✓	LTD
Man-portable for dismounted use	✓	LTD	✗
Silent / no electro-optical signature	✓	✓	✗
Safe for aircraft & tactical vehicles	✓	LTD	✗

LTD = possible but commonly constrained by hazard, power, or sustainment limitations.

3 METHODS (LO 2)

Bench and operational assessments evaluated oxygen purity, flow stability, activation reliability, environmental durability, and compatibility with standard oxygen delivery equipment. Environmental testing aligned with MIL-STD-810G/H. Ventilator compatibility was assessed in prehospital and Role 1 configurations (Zoll Impact 731, simulated patient settings).

4 RESULTS (LO 2)

Bench and operational assessments evaluated oxygen purity, flow stability, activation reliability, environmental durability, and compatibility with standard oxygen delivery equipment. Environmental testing aligned with MIL-STD-810G/H. Ventilator compatibility was assessed in prehospital and Role 1 configurations (Zoll Impact 731, simulated patient settings).

CHARACTERISTIC	METHOD — PARAMETERS	RESULTS
Vibration	514.8 — 5–500 Hz, 60 min/axis	✓
High temperature	501.5 — 104°F, 20 h	✓
Humidity + heat	507.5 — 95% RH	✓
Low temperature	502.5 — 0.4°F, 20 h	✓
Altitude	500.5 — 10k / 15k / 20k ft	✓
Blown sand	510.6 — 19 m/s, 140°F, 3 h	✓
Blown rain	506.6 — 4 in/h, 40 mph wind	✓
Drop	516.6 — 3 ft operational drop	✓
Mechanical shock	516.7 — 40 g ground / 20 g flight	✓

REGULATORY & TRANSPORT STATUS

FDA 510(k) cleared. Certified for global transport (UN 3356): commercial & military airlift, ground, maritime.

5 ROLES OF CARE (LO 3)

POINT OF INJURY ▶ PROLONGED CASUALTY CARE ▶ ROLE 1 / CASEVAC

Power-independent generation enables oxygen delivery at the point of injury and during prolonged casualty care when evacuation is delayed. It may support stabilization of casualties with respiratory compromise and integrates with existing airway equipment without workflow changes (LO3).

6 IMPACT / SIGNIFICANCE (LO 3)

- May expand the ability of forward medical personnel to maintain oxygenation during delayed evacuation and prolonged field care.
- Compact, shelf-stable design may support autonomous resupply — including unmanned aerial delivery — enabling distributed oxygen availability in contested environments.

7 DEVELOPMENTAL STATUS

- ✓ **Environmental qualification (MIL-STD-810G/H)**
- ✓ **Ventilator compatibility assessment**
- ✓ **Regulatory clearance — FDA 510(k)**
- ✓ **Transport certification (UN 3356)**
- Planned: user evaluations & integration studies

FURTHER INFORMATION

Andrew Maier | Molecular Products Inc., Louisville, CO
amaier@molprod.com

molecular Filtration Group

Disclaimer: The views expressed in this poster are those of the authors and do not represent the official policy or position of the Department of Defense or the U.S. Government. The technology described is presented for discussion of capability development and operational evaluation and does not constitute endorsement or promotion of any specific product.

Reference: Eastridge BJ, et al. Death on the battlefield (2001–2011): implications for the future of combat casualty care. J Trauma Acute Care Surg. 2012.