REBOA-Assisted CPR with Multimodal Monitoring Improves Systemic and Cerebral Perfusion and Neurologic Recovery in Cardiac Arrest

Masih Rafi, MD[1], Nishanth Chamarthi, BS[1], Miranda N Cepkinian, BS[1], Renee E Lotfy, BS[1], Catherine Vu, BS[1], Ryan Farazmand[1], Abdul Manan Mohtasebzada, BS[1], Ryan Hatfield, BS[1], Brian Hyunmin Chung, BS[1], Alexander Liu, BS[1], Afsheen Bazrafkan, BS[1], Seyed Sepehr Khatami, MD[1], Anshiqbal Singh, BS[1], Raul Tabares, BS[1], Katelyn Mai, BS[1], Pedram Borhani Khabaz, MD[1], Zoe E Flyer, DO[8], Thinh Phan, PhD[6], Bernard Choi, PhD[4,5,6], Yigit S Guner, MD, MS[5,8], Yama Akbari, MD, PhD[1-3,6,7]

[1] Department of Neurology, [2] Department of Anatomy and Neurobiology, [3] Department of Neurological Surgery, [4] Department of Biomedical Engineering, [5] Department of Surgery, [6] Beckman Laser Institute and Medical Clinic, [7] Reeve-Irvine Research Center, University of California, Irvine, CA, [8] Children's Hospital of Orange County, Orange, CA

INTRODUCTION

- Cardiac arrest (**CA**) is a common outcome of combat injuries and prolonged field care. Survival, recovery, and neurological outcome are determined by the quality of coronary and cerebral perfusion during cardiopulmonary resuscitation (**CPR**).[1,2,4]
- Our experiments utilize the resuscitative endovascular balloon occlusion of the aorta (**REBOA**) as a mechanical vasopressor, replacing the need for additional epinephrine and improving resuscitation. The efficacy of this mechanism has not yet been studied in a CA model with multimodal neuro-monitoring (**MMM**) to assess cerebral perfusion in real time.
- In order to optimize systemic and cerebral perfusion during CPR, a translational model of REBOA+MMM was developed in a rat model of asphyxial CA (**ACA**).

METHODOLOGY

Fasted adult male Wistar rats underwent an induced 7 minute ACA with reduced epinephrine.

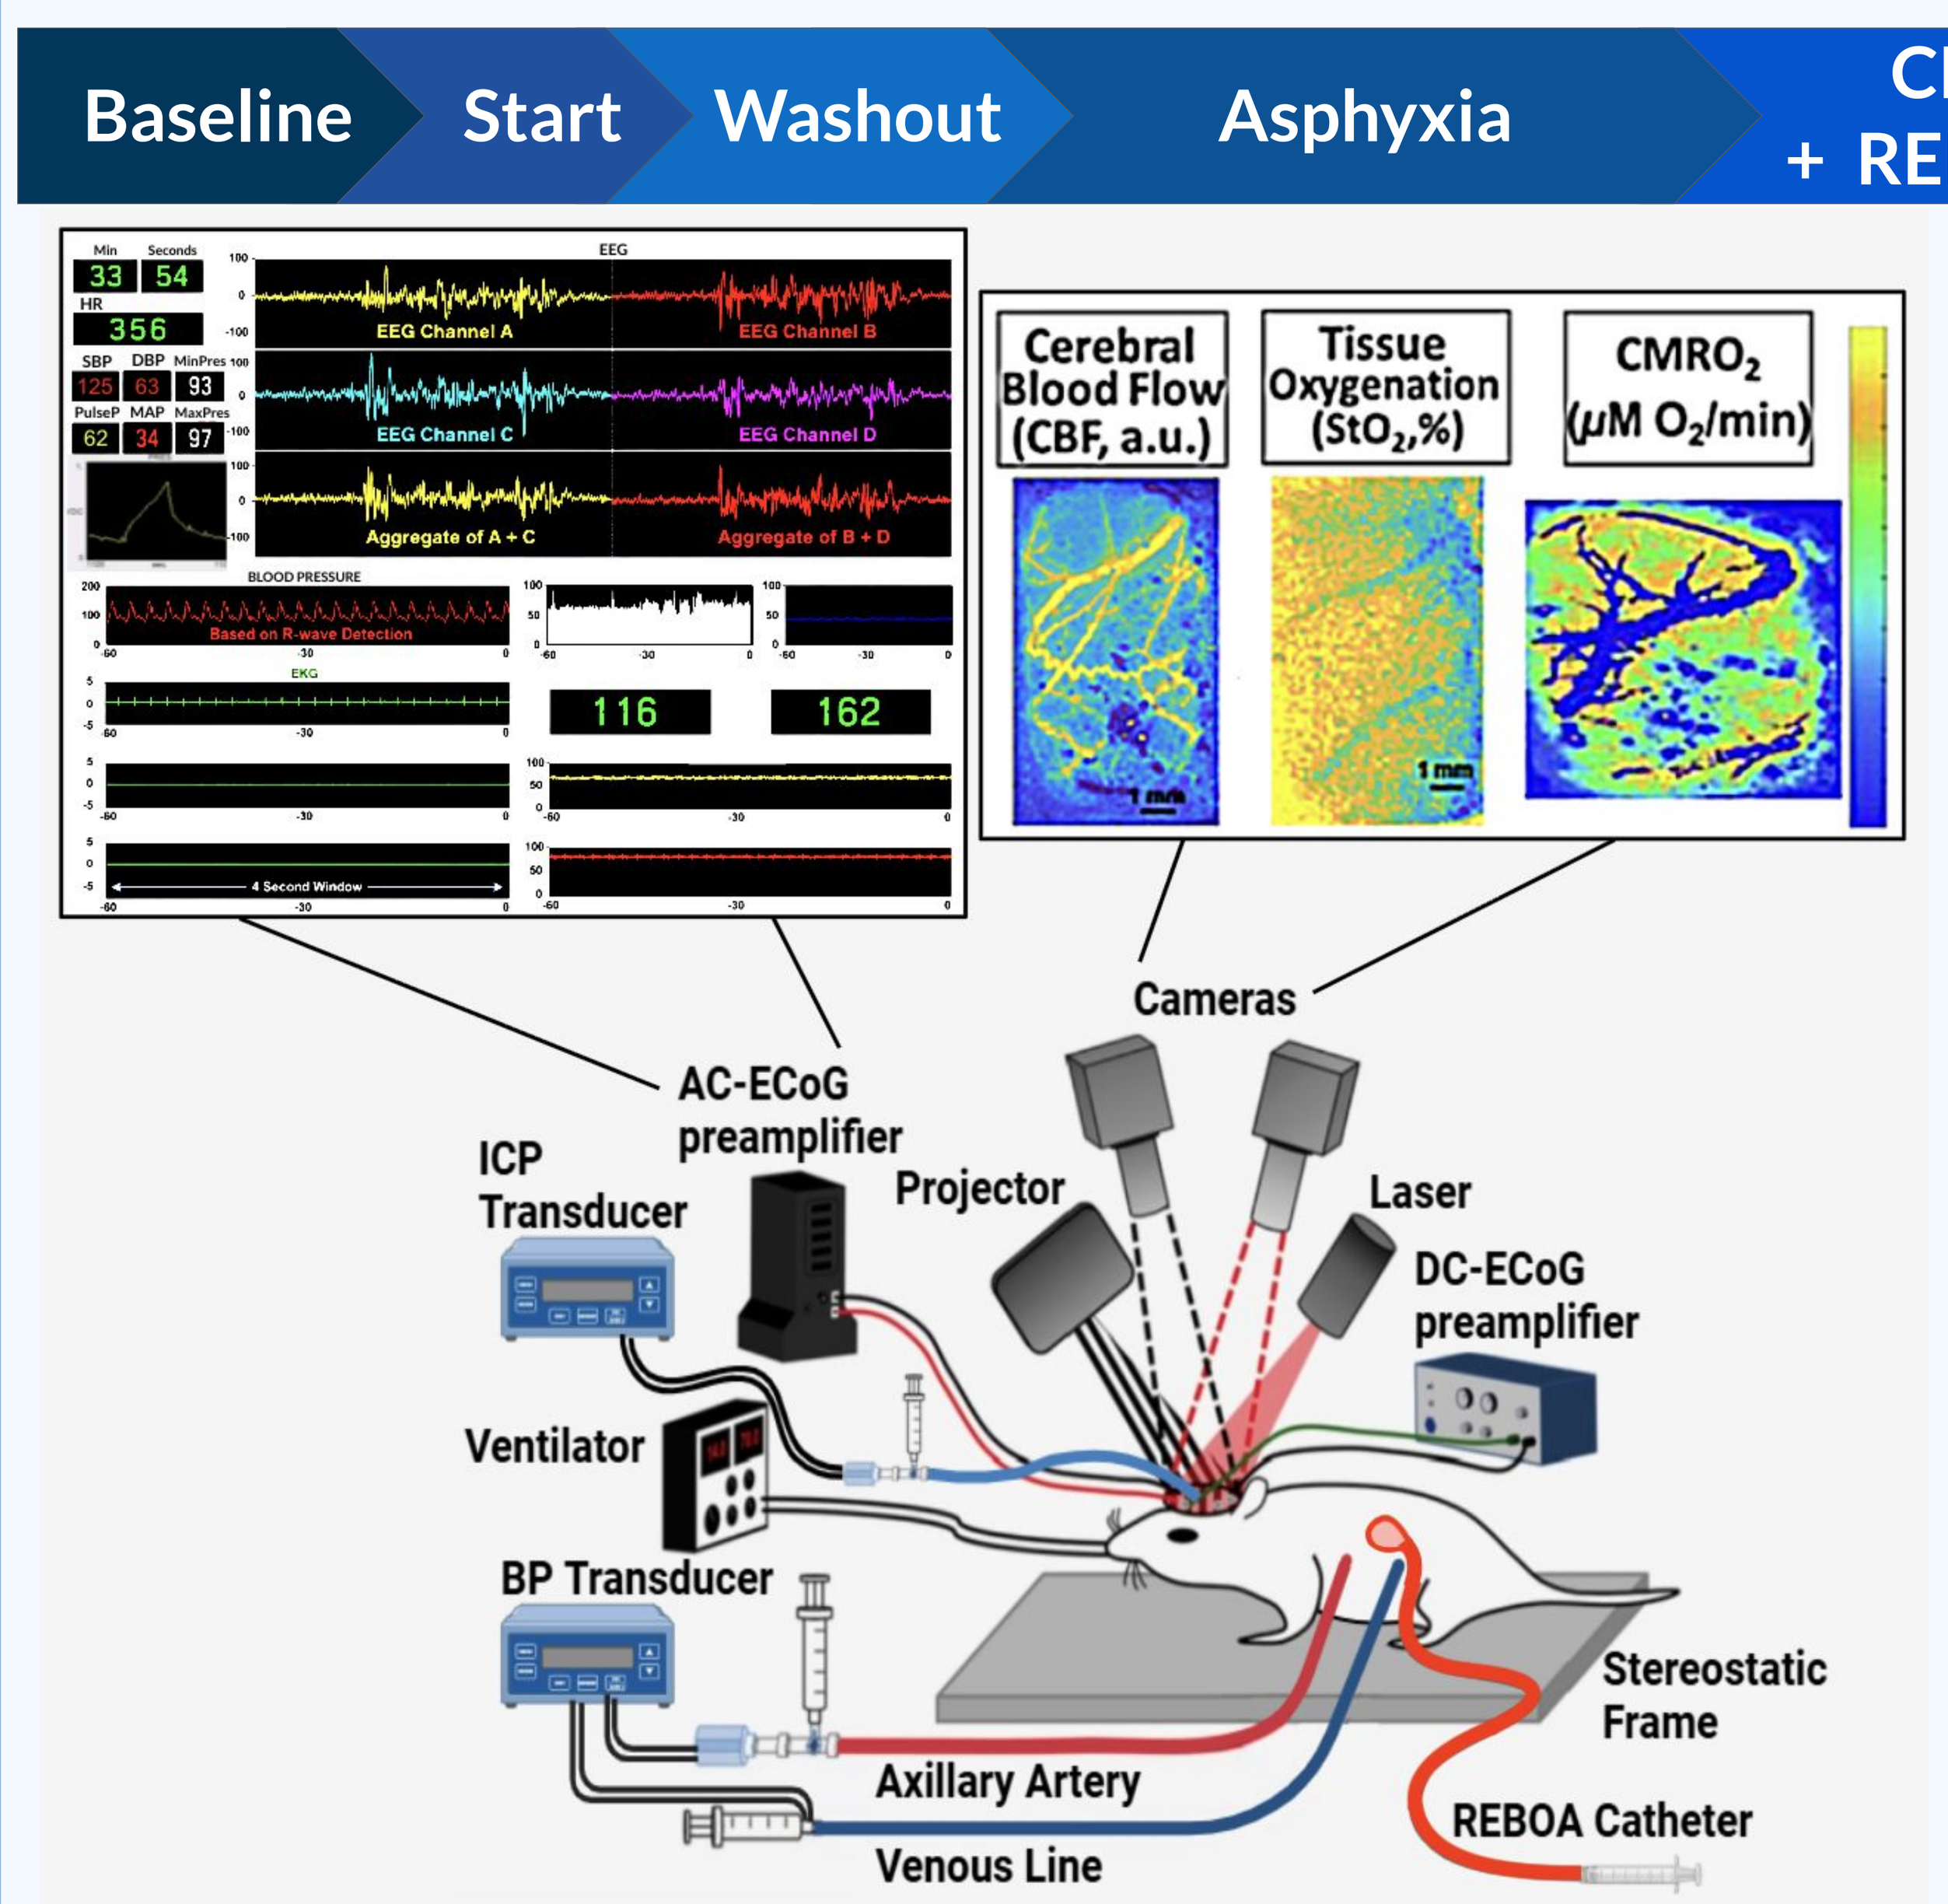
Baseline
Start
Washout
Asphyxia
CPR + REBOA
ROSC
Recovery
EEG Channel A
EEG Channel B
EEG Channel C
EEG Channel D
Aggregate of A + C
Aggregate of B + D
BLOOD PRESSURE
Based on R-wave Detection
EKG
116
162
4 Second Window
Cerebral Blood Flow (CBF, a.u.)
Tissue Oxygenation (StO_2,%)
CMRO$_2$ (μM O_2/min)
1 mm
Cameras
AC-ECoG preamplifier
ICP Transducer
Projector
Laser
DC-ECoG preamplifier
Ventilator
BP Transducer
Stereostatic Frame
Axillary Artery
Venous Line
REBOA Catheter

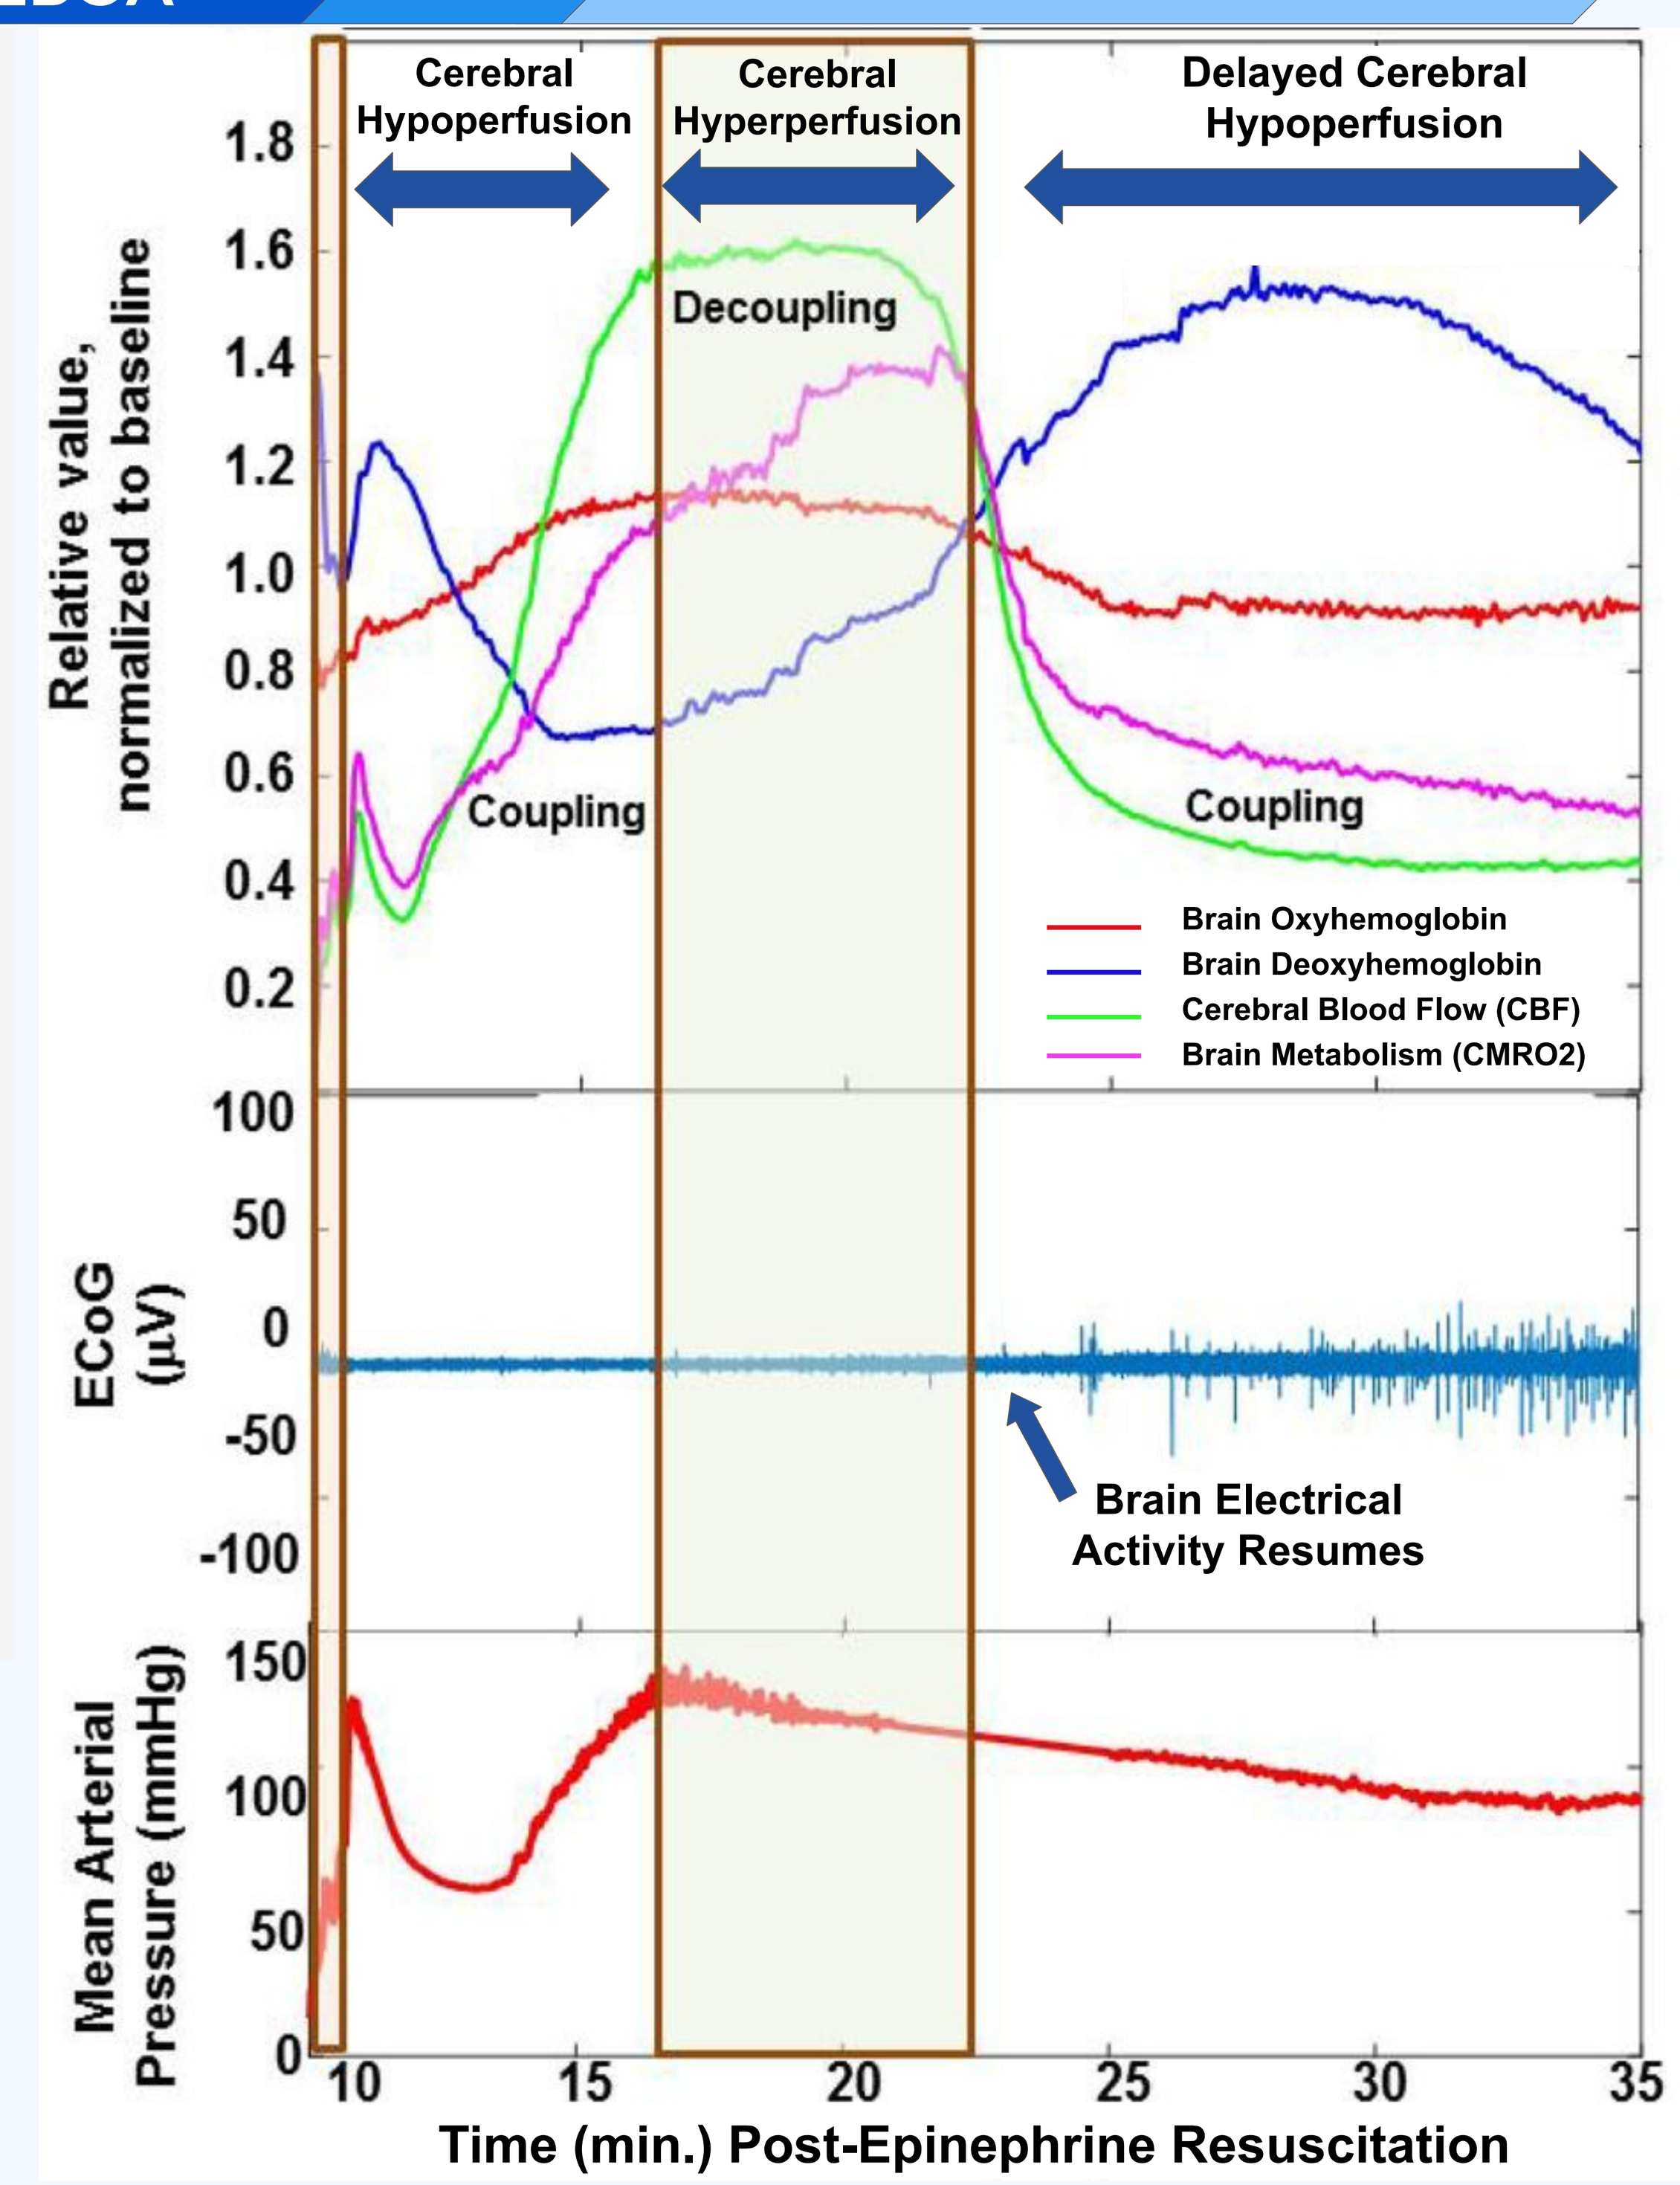
Cerebral Hypoperfusion
Cerebral Hyperperfusion
Delayed Cerebral Hypoperfusion
Decoupling
Coupling
Coupling
Brain Oxyhemoglobin
Brain Deoxyhemoglobin
Cerebral Blood Flow (CBF)
Brain Metabolism (CMRO2)
Relative value, normalized to baseline
ECoG (μV)
Brain Electrical Activity Resumes
Mean Arterial Pressure (mmHg)
Time (min.) Post-Epinephrine Resuscitation

Figure 1: Rat model with real time multimodal monitoring.

- REBOA was inflated at CPR onset for the experimental group and as a fail safe 3 minutes into CPR for Sham rats. Deflation of REBOA occurred 30 seconds after return of spontaneous circulation (**ROSC**).
- Real time MMM (Figure 1) includes:[3]
 - Laser speckle imaging (**LSI**) for cerebral blood flow (**CBF**) and spatial frequency domain imaging (**SFDI**), which were combined to calculate cerebral metabolic rate of oxygen consumption (**CMRO2**).
 - Mean arterial pressure (**MAP**) and intracranial pressure (**ICP**) for calculation of cerebral perfusion pressure.
 - Quantitative electrocorticography (**ECoG**) to calculate burst suppression ratio (**BSR**).
- Neurological assessment included neurological deficit score (**NDS**) and open field testing (**OFT**).

RESULTS

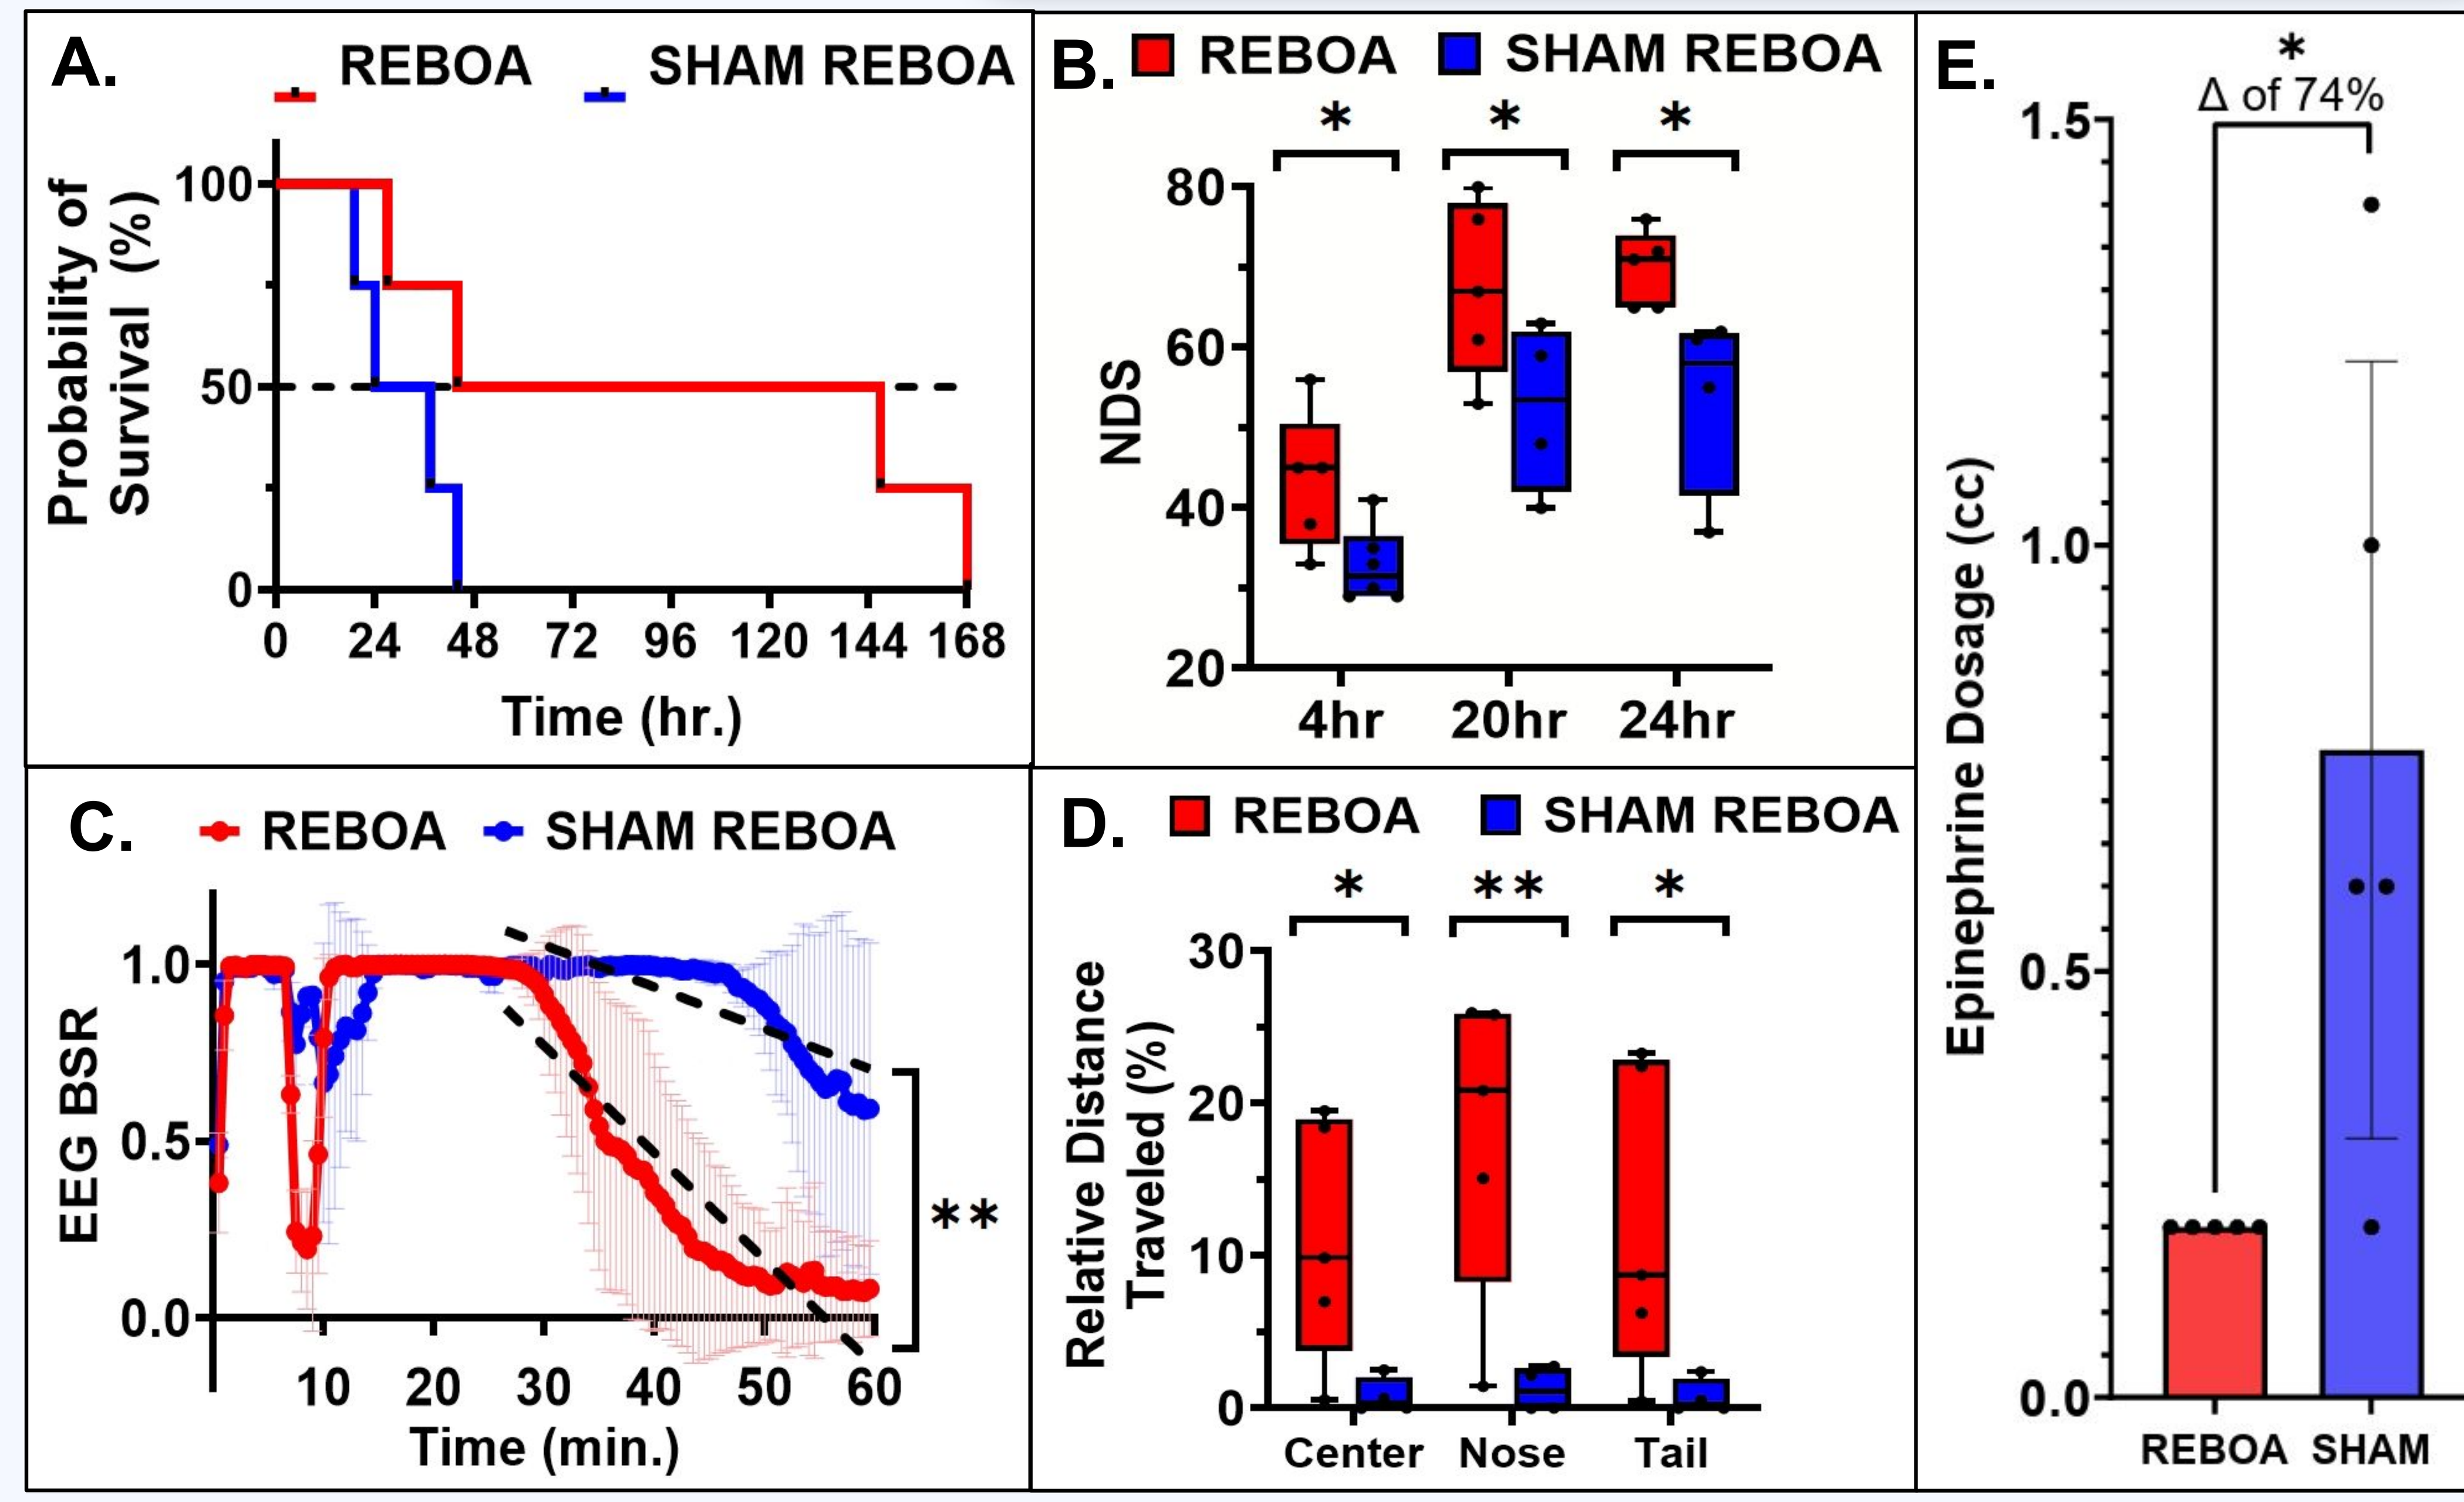
A.
REBOA
SHAM REBOA
Probability of Survival (%)
Time (hr.)
B.
REBOA
SHAM REBOA
NDS
4hr
20hr
24hr
C.
REBOA
SHAM REBOA
EEG BSR
Time (min.)
D.
REBOA
SHAM REBOA
Relative Distance Traveled (%)
Center
Nose
Tail
E.
Δ of 74%
Epinephrine Dosage (cc)
REBOA
SHAM

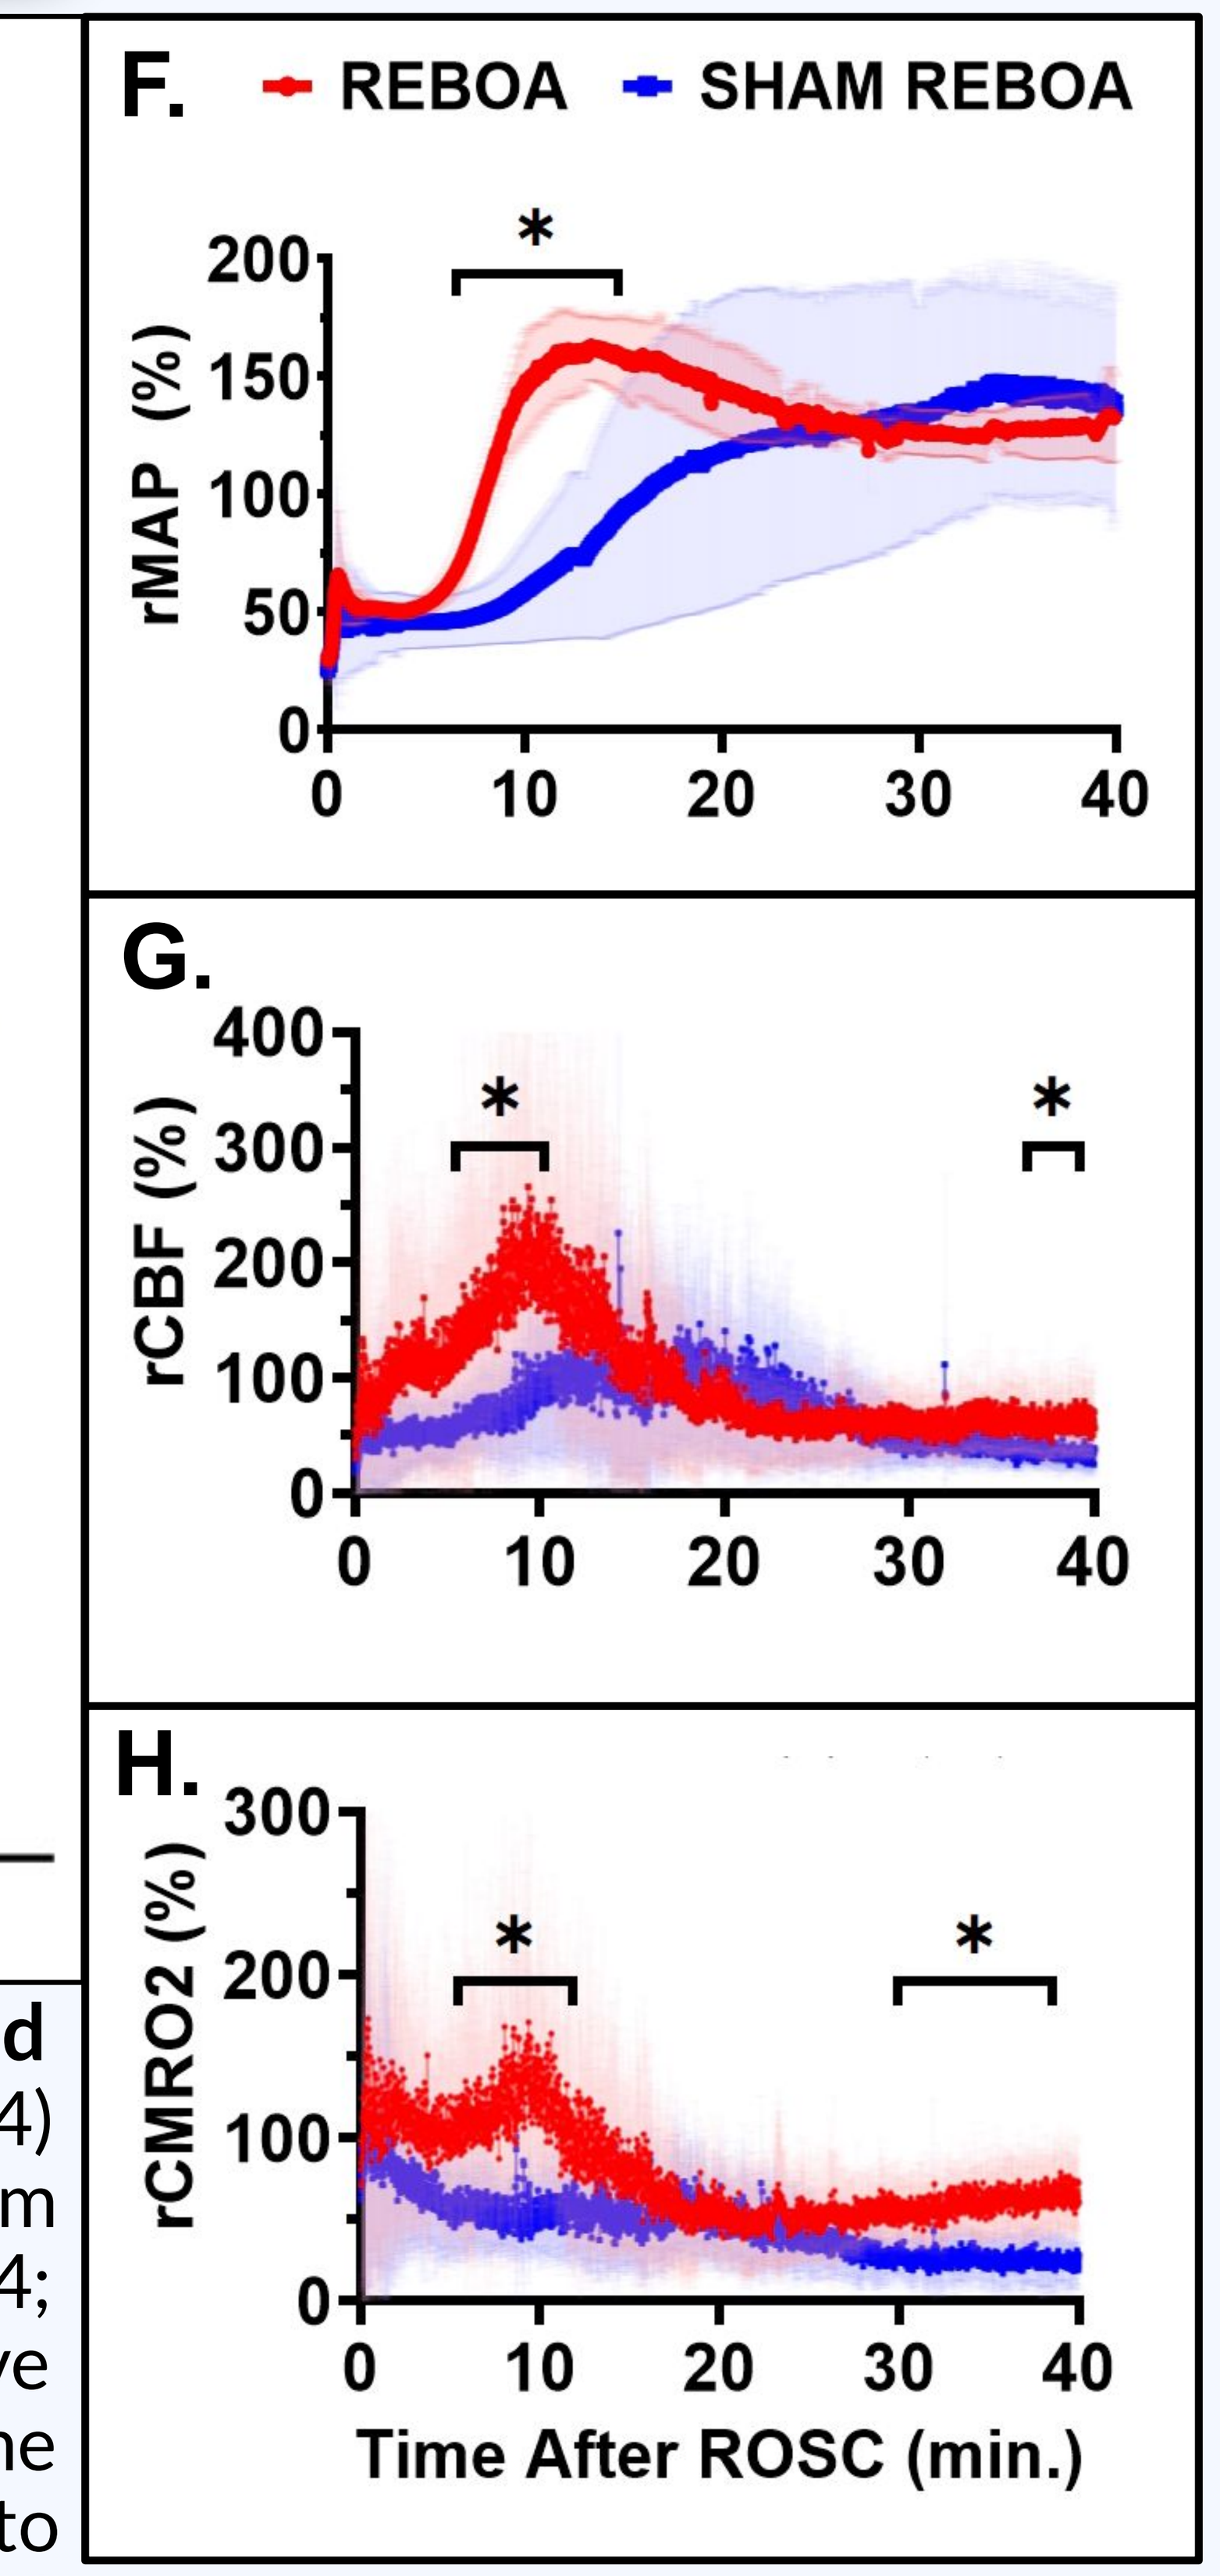
F.
REBOA
SHAM REBOA
rMAP (%)
G.
rCBF (%)
H.
rCMRO2 (%)
Time After ROSC (min.)

Figure 2: REBOA improves survival and neurological outcomes after ROSC via enhanced cerebral perfusion. (A) REBOA rats showed higher median survival rate (REBOA n=4 vs. Sham n=4) and (B) improved NDS 4h, 20h, and 24h post-ROSC (REBOA n=5 vs. Sham n=6) compared to Sham rats ($p<0.05$). (C) BSR of REBOA rats displayed faster EEG recovery (REBOA n=6 vs. Sham n=4; $p<0.05$ AUC, slope). (D) OFT showed that relative to pre-CA baseline movement, REBOA rats have significantly greater movement at 24h post-ROSC, indicating better neurobehavioral outcome (REBOA n=5 vs Sham n=4; $p<0.05$). (E) REBOA significantly reduced epinephrine need compared to Sham experiments (REBOA n=5 vs. Sham n=5, $p<0.05$). (F, G, H) REBOA significantly increased relative MAP, CBF, and CMRO2 post-ROSC (rMAP [7.5-15 min]; rCBF [7.5-10.5, 35-40 min]; rCMRO2 [7.5-10.5, 29-40 min]: REBOA n=5 vs. SHAM n=5; $p<0.05$).

DISCUSSION

REBOA+MMM during CPR improved systemic and cerebral perfusion during post-ROSC hyper- and hypoperfusion while also enhancing neurobehavioral and electrophysiological recovery (Figure 2). A commercial device, suitable for simultaneous MMM monitoring of cerebral perfusion, metabolism, and EEG, coupled with AI enabled precision-guided resuscitation to optimize REBOA use, may reduce reliance on vasopressor drugs while improving cardiac and neurological outcomes and prognostication.

REFERENCES

1. Crouzet C, Wilson RH, Bazrafkan A, Farahabadi MH, Lee D, Alcocer J, Tromberg BJ, Choi B, Akbari Y. Cerebral Blood Flow Is Decoupled From Blood Pressure and Linked to EEG Bursting After Resuscitation From Cardiac Arrest. Biomedical Optics Express. 2016;7(11):4660-4673. https://doi.org/10.1364/BOE.7.004660
2. Crouzet C, Wilson RH, Lee D, Bazrafkan A, Tromberg BJ, Akbari Y, Choi B. Dissociation of Cerebral Blood Flow and Femoral Artery Blood Pressure Pulsatility After Cardiac Arrest and Resuscitation in a Rodent Model: Implications for Neurological Recovery. *JAHA*. 2020;9(1). https://doi.org/10.1161/JAHA.119.012691
3. Wilson RH, Crouzet C, Torabzadeh M, Bazrafkan A, Maki N, Alcocer J, Tromberg BJ, Choi B, Akbari Y. Cerebral perfusion and metabolism coupling during a critical time window provides rapid assessment of cardiac arrest severity and prognosis in a preclinical model. *bioRxiv*. 2019. https://doi.org/10.1101/785972
4. S. Han, M. Contreras, A. Bazrafkan, M. Rafi, S. Dara, A. Orujyan, A. Panossian, C. Crouzet, B. Louper, B. Choi, R. Wilson, and Y. Akbari. (2022). Cortical Anoxic Spreading Depolarization During Cardiac Arrest is Associated with Remote Effects on Peripheral Blood Pressure and Postresuscitation Neurological Outcome. Neurocritical care, 37(Suppl 1), 139–154. https://doi.org/10.1007/s12028-022-01530-2

FUNDING

This work was funded by Children's Hospital of Orange County (CHOC) PSF grant award and the Roneet Carmell Memorial Endowment Fund.