Validation of a Sensor-Embedded Sock Against Gold-Standard Measures of Landing Biomechanics

Jordan Coker, MS[1]; Isabel Erickson, MS[1]; Natalie Zesch[1]

[1] Aptima, Inc., Woburn, MA

PROBLEM INTRODUCTION

FIELD ASSESSMENT REQUIRES VALID WEARABLE MEASURES

Parachute landing falls expose Service Members to substantial lower-extremity loading and may contribute to ankle and other musculoskeletal injuries. Laboratory systems can characterize landing forces, joint motion, plantar loading, and muscle activation, but their size and complexity limit use during airborne training and field operations.

AERO-PI was developed to determine whether a wearable sensor system could capture biomechanical measures relevant to landing assessment outside a traditional laboratory environment.

The AERO-PI concept is intended to support objective assessment of landing performance, evaluation of injury-mitigation approaches, and future development of injury-risk analytics.

CHALLENGES & APPROACH

TRANSLATING LABORATORY MEASURES INTO THE FIELD

Gold-standard biomechanical equipment provides high-quality data but generally requires specialized facilities, trained personnel, and controlled testing conditions. A field-capable system must estimate similar measures while remaining **wearable, minimally intrusive, and compatible with operational movement.**

The AERO-PI prototype combines inertial measurement, electromyography, strain, and plantar force sensing within a sensor-embedded sock. Each sensing modality was intended to represent a laboratory measure associated with landing biomechanics, including landing-event timing, impact loading, ankle movement, plantar pressure distribution, and muscle activation. The central research question was not simply whether the sensors produced data, but whether their outputs corresponded with synchronized gold-standard measurements collected during controlled human movement tasks.

The project's intended metrics include ankle kinematics, impact force, pressure distribution, muscle contraction, ankle stiffness, and landing-form characteristics

METHOD

SYNCHRONIZED WEARABLE AND LABORATORY DATA COLLECTION

Nineteen adults participated in an Institutional Review Board-approved validation study conducted in Auburn University's biomechanical engineering laboratory. Participants completed static calibration, walking, range-of-motion, balance, jump-apart, jump-together, and box-jump tasks while wearing the AERO-PI prototype.

Wearable outputs were collected concurrently with laboratory reference systems, including force platforms, motion capture, Delsys electromyography, and plantar pressure equipment. The Auburn laboratory includes Vicon motion capture, embedded AMTI force platforms, Delsys Trigno sensors, and plantar pressure measurement systems.

Analyses compared the systems across:

- Landing-event timing
- Relative impact-loading trends
- Muscle-activation timing and patterns
- Ankle-motion estimates
- Symmetry and landing-form measures
- Plantar loading characteristics
- Sensor reliability and signal quality

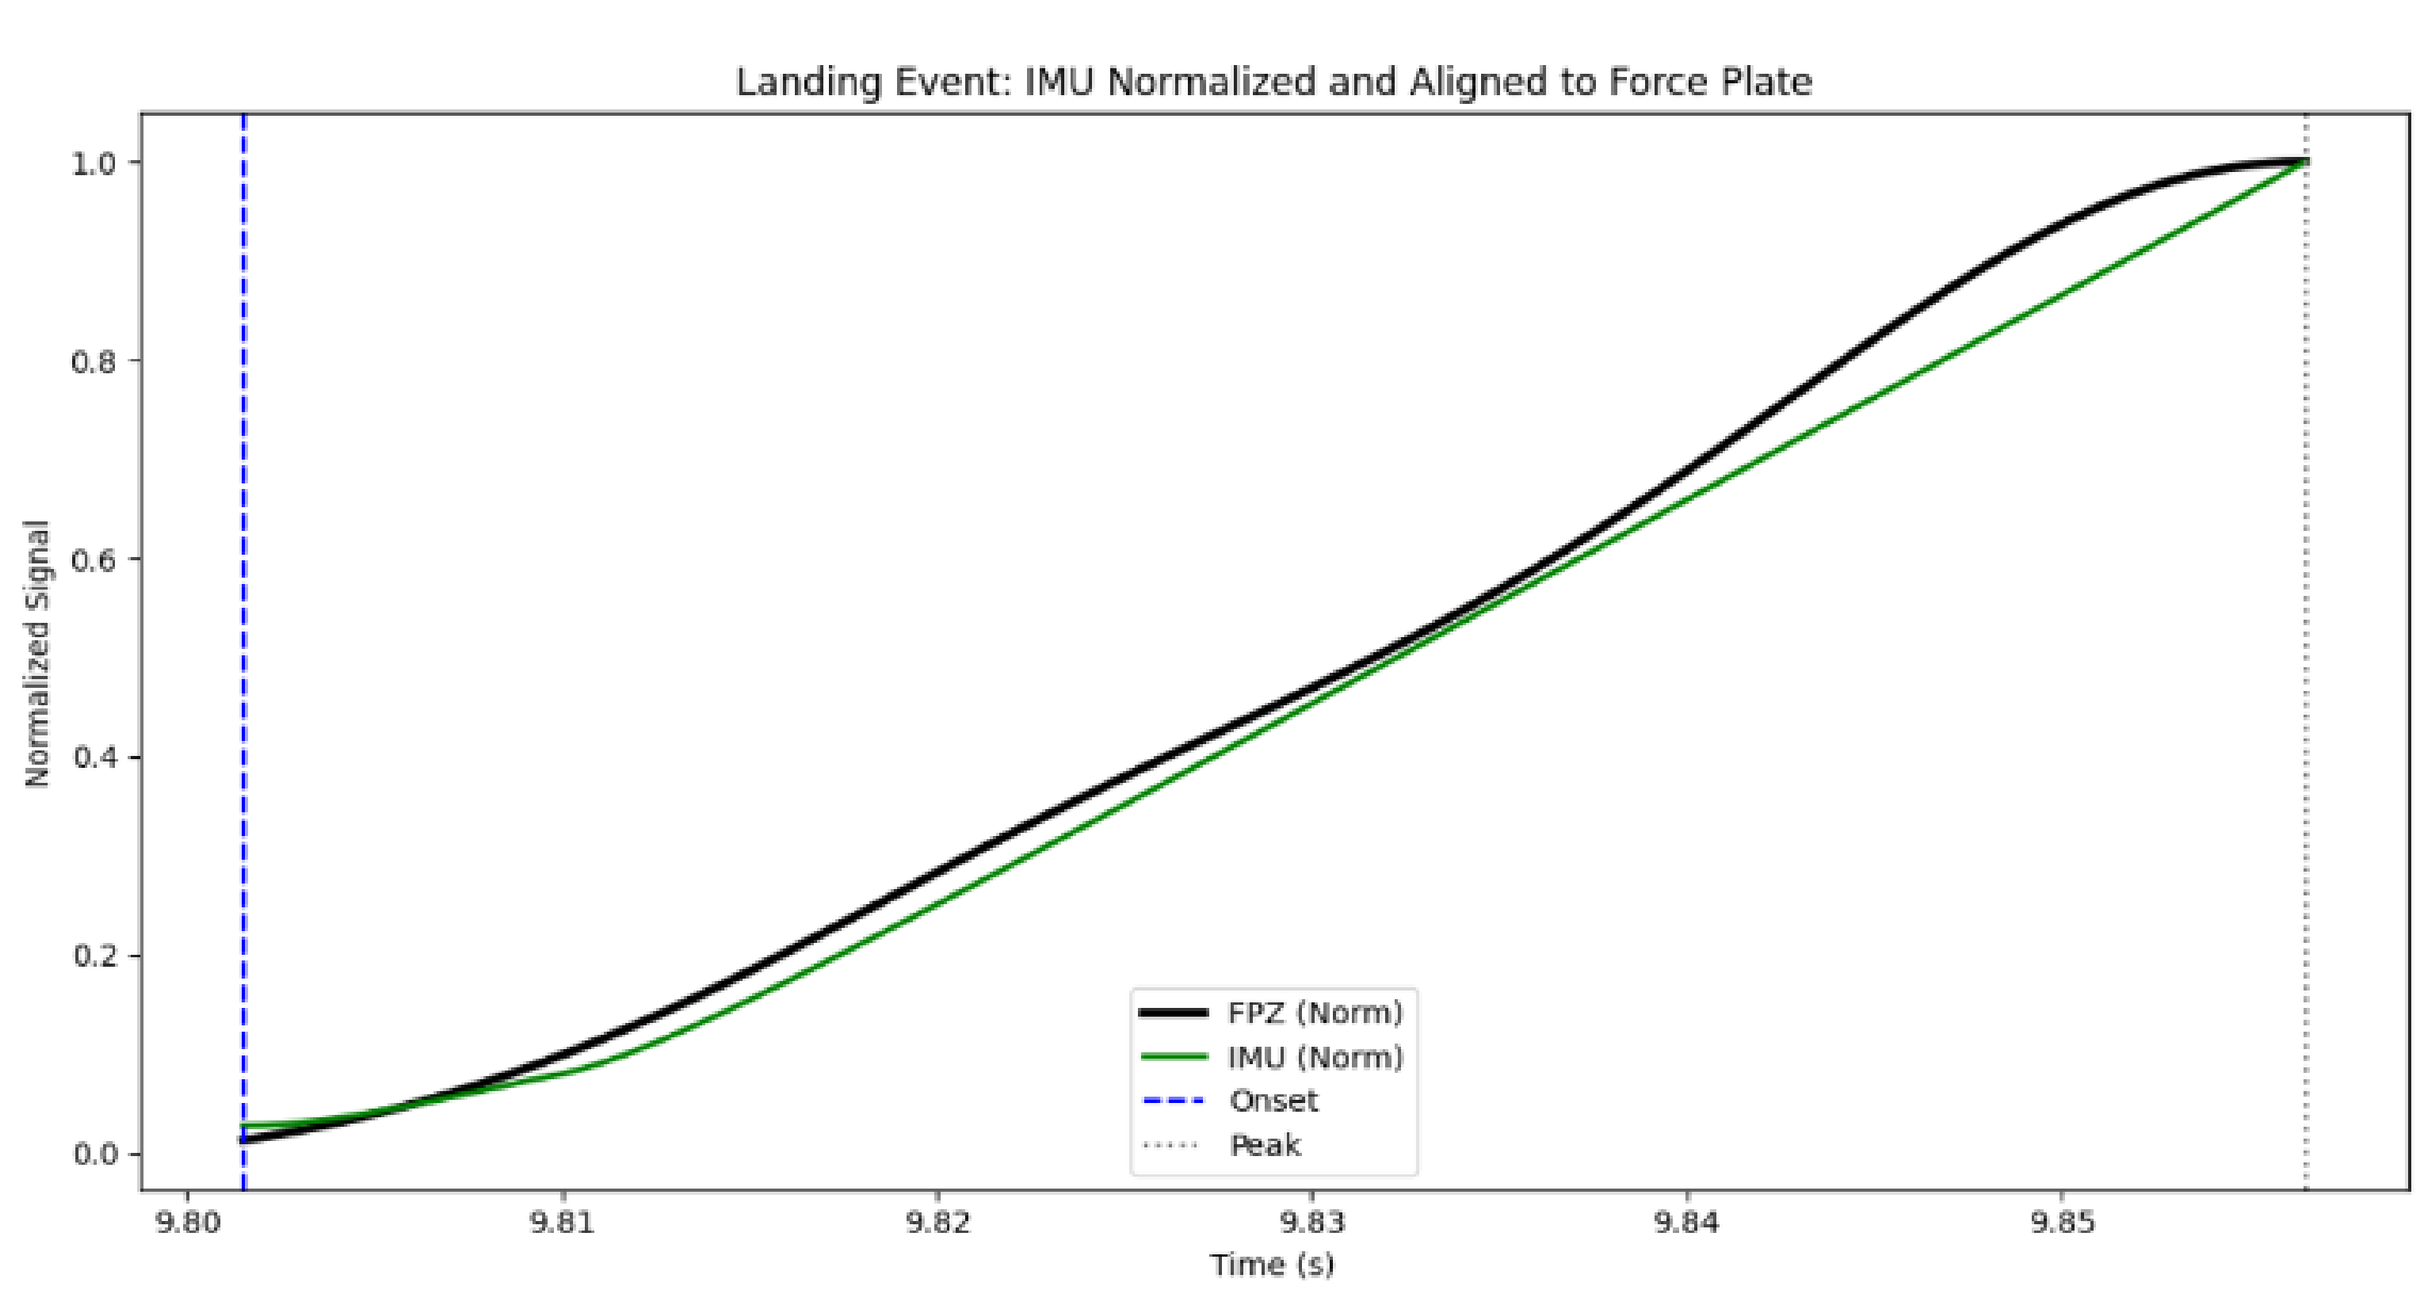
Landing Event: IMU Normalized and Aligned to Force Plate
Normalized Signal
Time (s)
FPZ (Norm)
IMU (Norm)
Onset
Peak
0.0
0.2
0.4
0.6
0.8
1.0
9.80
9.81
9.82
9.83
9.84
9.85

IMU Landing Forces Correlated to Force Plate Measurements after Normalization

AERO-PI

WEARABLE MULTIMODAL LANDING ASSESSMENT

AERO-PI is a sensor-embedded wearable and supporting analytics approach designed to capture lower-extremity biomechanical data in environments where full laboratory systems are impractical. The prototype integrates inertial measurement units, electromyography, strain sensing, and force-sensitive resistors to measure complementary aspects of movement and landing.

The system is intended to transform wearable signals into measures that may support objective performance assessment, injury-mitigation research, and future field-based data collection. The broader architecture connects wearable sensing with data-processing pipelines and user-facing analytics.

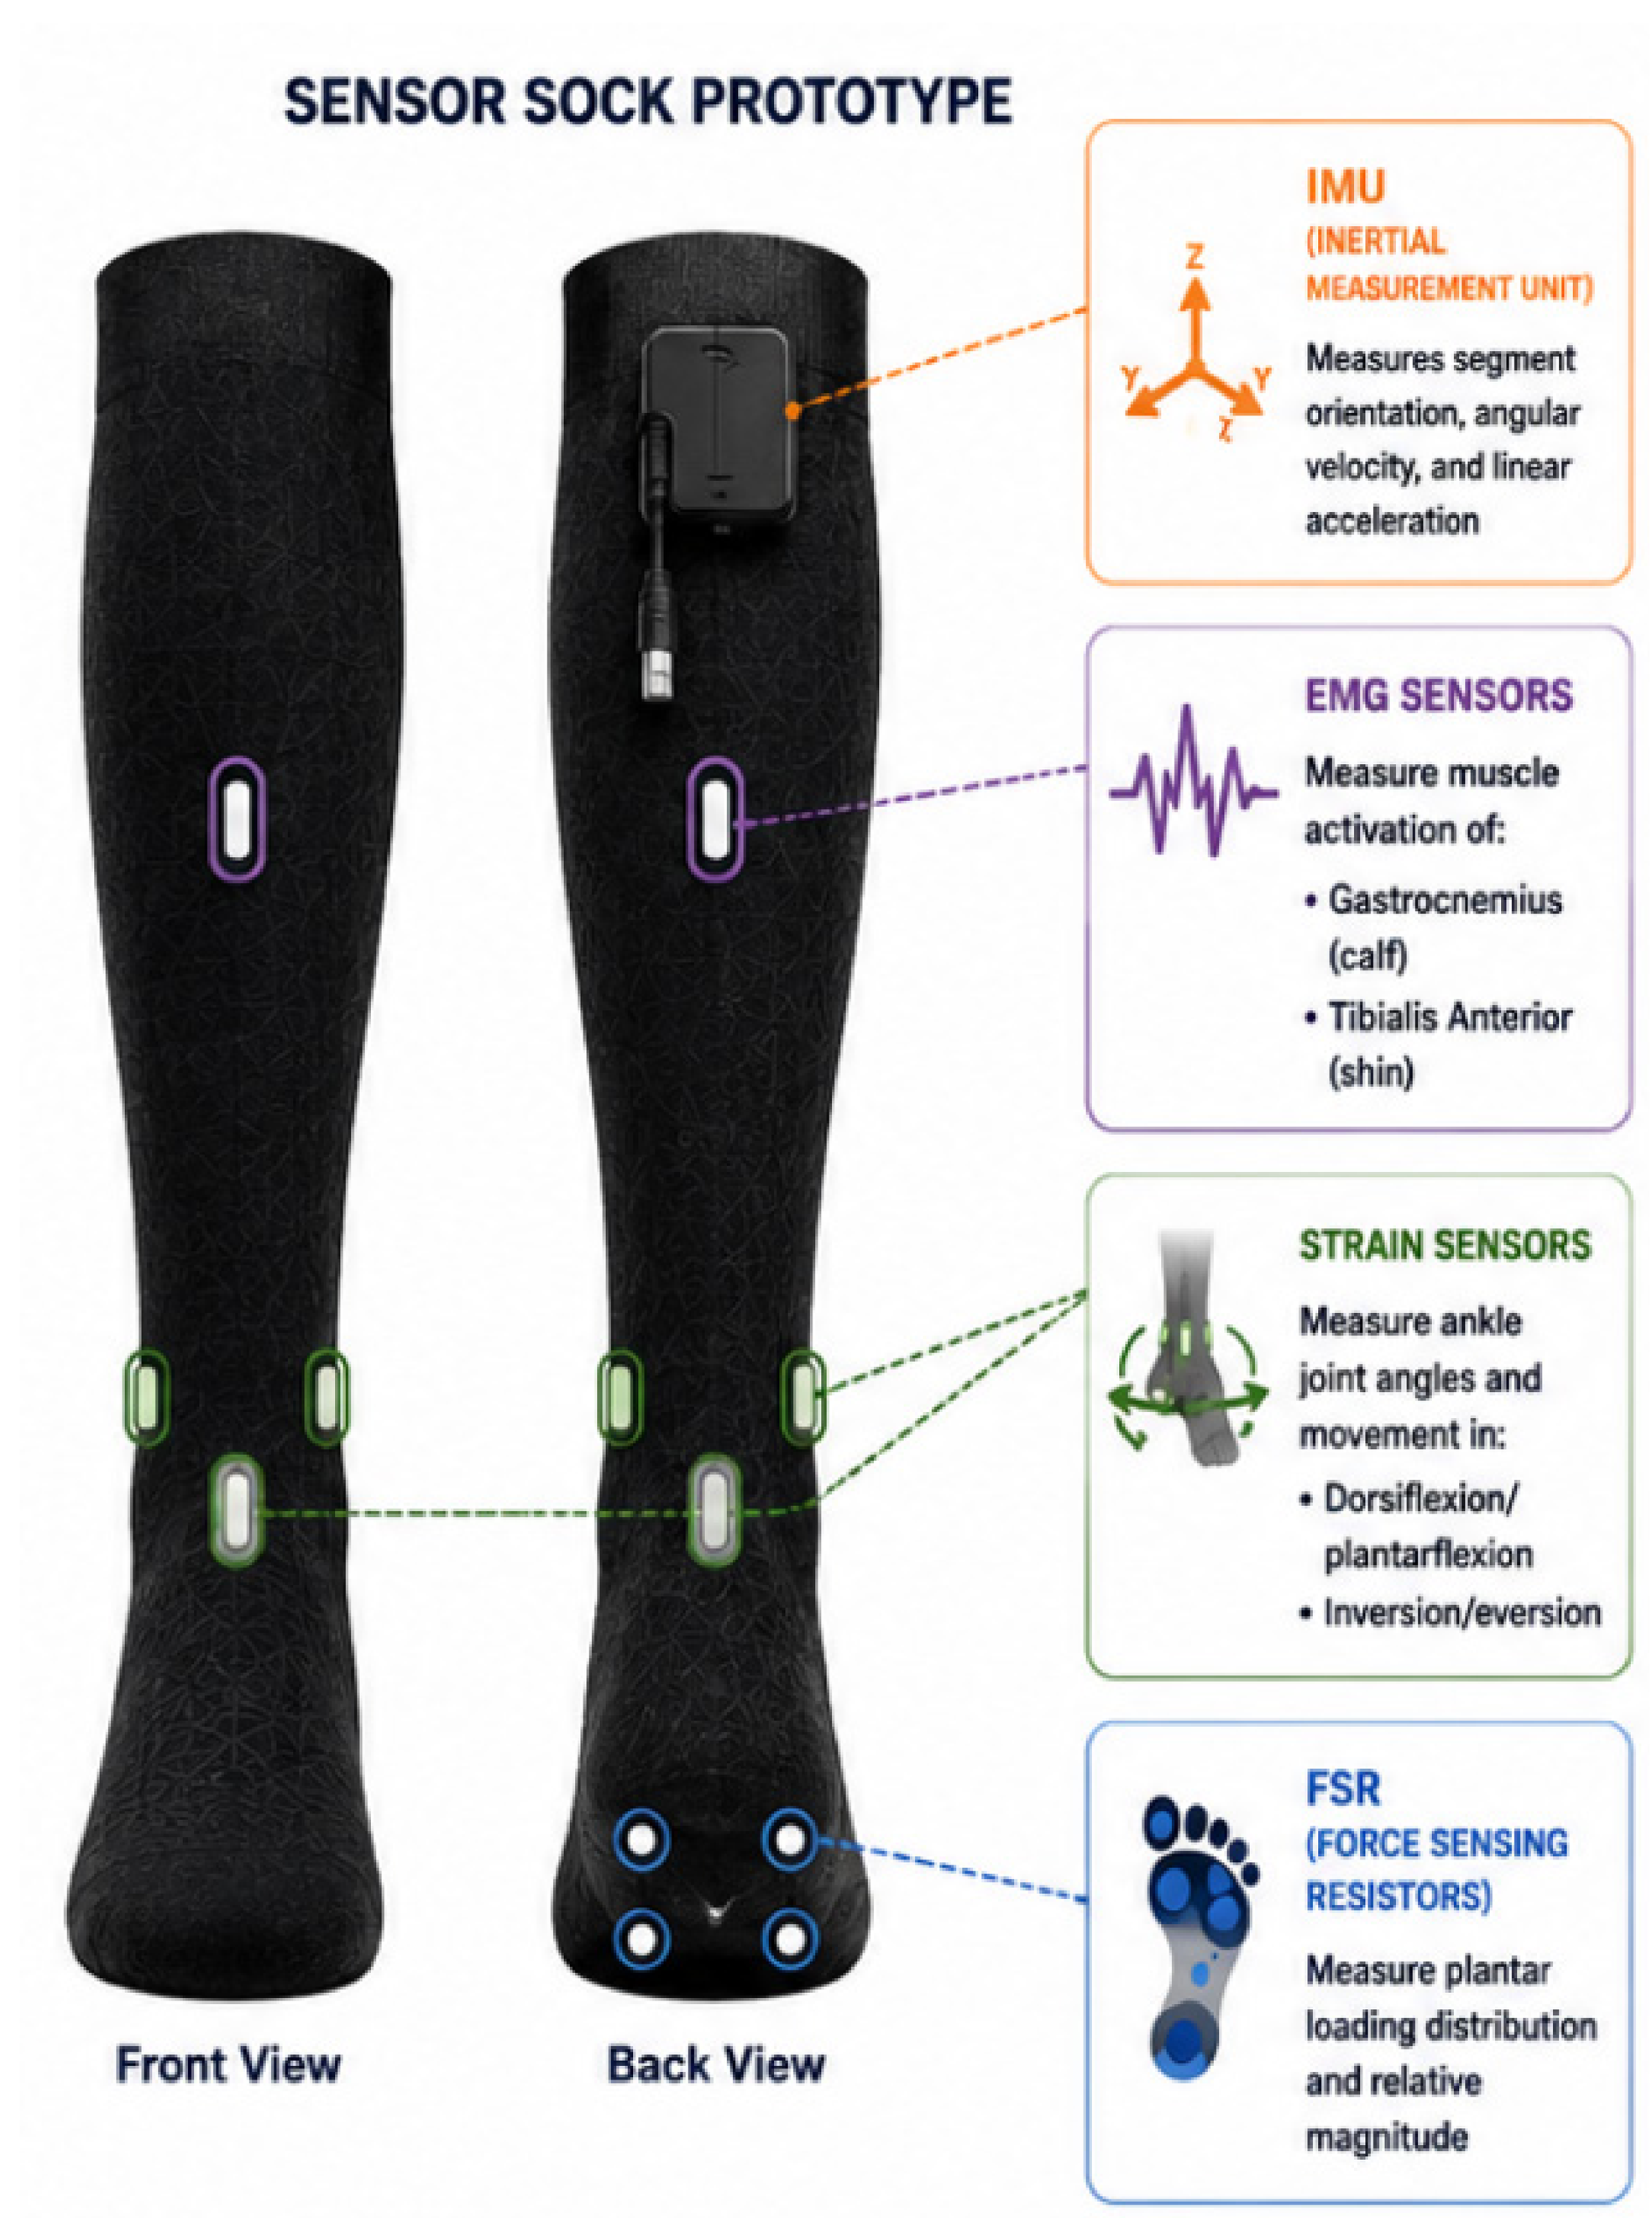
SENSOR SOCK PROTOTYPE
IMU
(INERTIAL MEASUREMENT UNIT)
Measures segment orientation, angular velocity, and linear acceleration
EMG SENSORS
Measure muscle activation of:
• Gastrocnemius (calf)
• Tibialis Anterior (shin)
STRAIN SENSORS
Measure ankle joint angles and movement in:
• Dorsiflexion/ plantarflexion
• Inversion/eversion
FSR
(FORCE SENSING RESISTORS)
Measure plantar loading distribution and relative magnitude
Front View
Back View

AERO-PI Sensor Sock Design and Sensor Layout

RESULT

STRONGEST AGREEMENT IN TIMING AND MOTION

The study cohort included 14 male and 5 female participants. Comparisons with laboratory reference systems showed that the prototype's performance varied by sensing modality.

Inertial measurement: IMU-derived landing events aligned well with force-platform event timing and captured relative changes in impact-loading trends across jump conditions. These results support the IMU's use for detecting movement events and comparing relative landing intensity.

Electromyography: Sock-integrated EMG captured muscle-activation timing and general activation patterns that corresponded with Delsys measurements. Agreement was most promising for activation onset, pre-landing activation, and co-contraction-related features rather than absolute signal magnitude.

Strain sensing: Strain-derived estimates showed promise for tracking ankle motion. Performance was stronger for inversion and eversion than for dorsiflexion and plantarflexion.

Plantar force sensing: Force-sensitive resistor outputs were less consistent than the other modalities. The sensors did not reliably distinguish dominant plantar loading regions across all trials, identifying plantar sensing as a priority for hardware and calibration refinement.

Overall, IMU, EMG, and selected strain-derived measures showed the greatest potential for continued development.

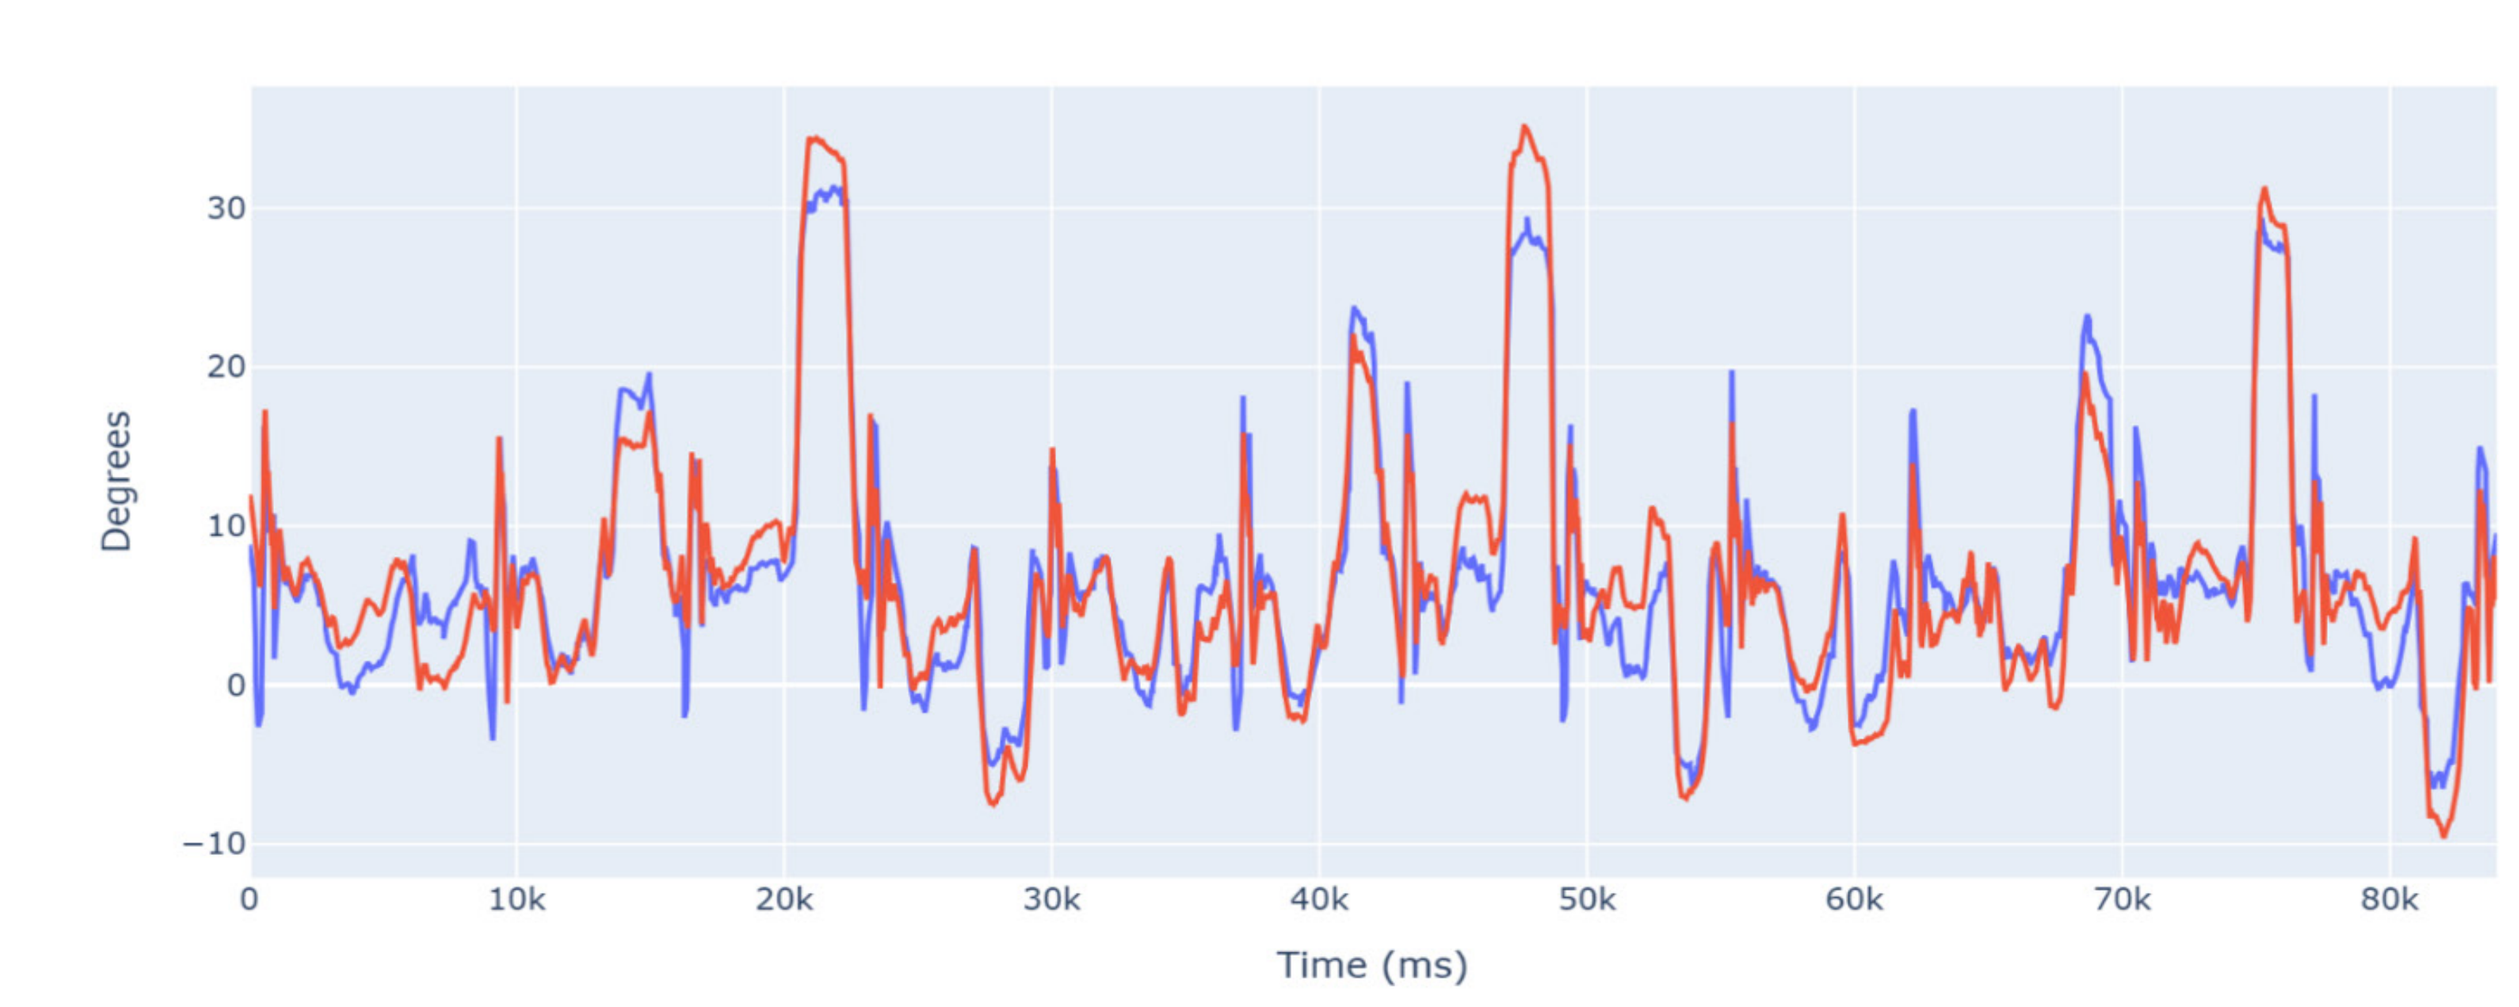
Degrees
Time (ms)
30
20
10
0
−10
0
10k
20k
30k
40k
50k
60k
70k
80k

Sock Flex Sensor vs Gold Standard Motion Capture

REFERENCES & CITATIONS

Knapik JJ, Steelman R, Grier T, et al. Military parachuting injuries, associated events, and injury risk factors. Aviat Space Environ Med. 2011;82(8):797–804.

Knapik JJ, Spiess A, Swedler DI, et al. Systematic review of the parachute ankle brace: Injury risk reduction and cost effectiveness. Am J Prev Med. 2010;38(1 Suppl):S182–S188.

Sun T, Li D, Fan B, Tan T, Shull PB. Real-time ground reaction force and knee extension moment estimation during drop landings via modular LSTM modeling and wearable IMUs. IEEE J Biomed Health Inform. 2023;27(7):3222–3233.

ACKNOWLEDGEMENT

This work was performed under the SBIR Phase II project, AERO-PI: Advanced EXOJUMP to Reduce Occurrences of Parachute Injuries, Contract W911QY-24-C-0024, and was supported in part by preceding Phase I activities under Contract W911QY23P0006. The authors thank Samuel Corner and Brian Huffman of the U.S. Army DEVCOM Soldier Center for their guidance and collaboration. We also acknowledge instructors and personnel across airborne training programs and our collaborators at Auburn University, particularly Michael Zabala, Sierra Eady, and members of the AUBE Lab, for their contributions to data collection, biomechanical analysis, and validation.