Portable Brain Injury Detection to Support Triage and Evacuation for Suspected TBI in Contested Combat Environments

Alexander Chan, PhD[1]; Robert Tiller[1]; Christian Wight, PhD[1]; Forough Khandan[1]; Scott Kirkland[1]; Stuart Crozier, E/Prof[1,2]

1 EMVision Medical Devices, Sydney, Australia

2 University of Queensland, Brisbane, Australia

Introduction

Traumatic brain injury (TBI), particularly blast-related injury, remains a signature wound of modern conflict. In prolonged field care and large-scale combat operations (LSCO), overtriage strains evacuation assets and may divert limited resources from warfighters who require urgent intervention.

Between 2001 and 2018, 68% of Service members with isolated mild TBI were evacuated by rotary wing (Dengler et al., 2022), with more than half returning to duty.

Capability Description

We describe the EMVision First Responder Brain Scanner (Figure 1), a portable structural brain injury detection device that identifies the presence and location of intracranial bleeding. The prototype system is lightweight (< 25 lb), compact (17 in × 11 in × 14 in), and produces actionable results in < 5 minutes.

Figure 1
The EMVision First Responder Brain Scanner and accessories.

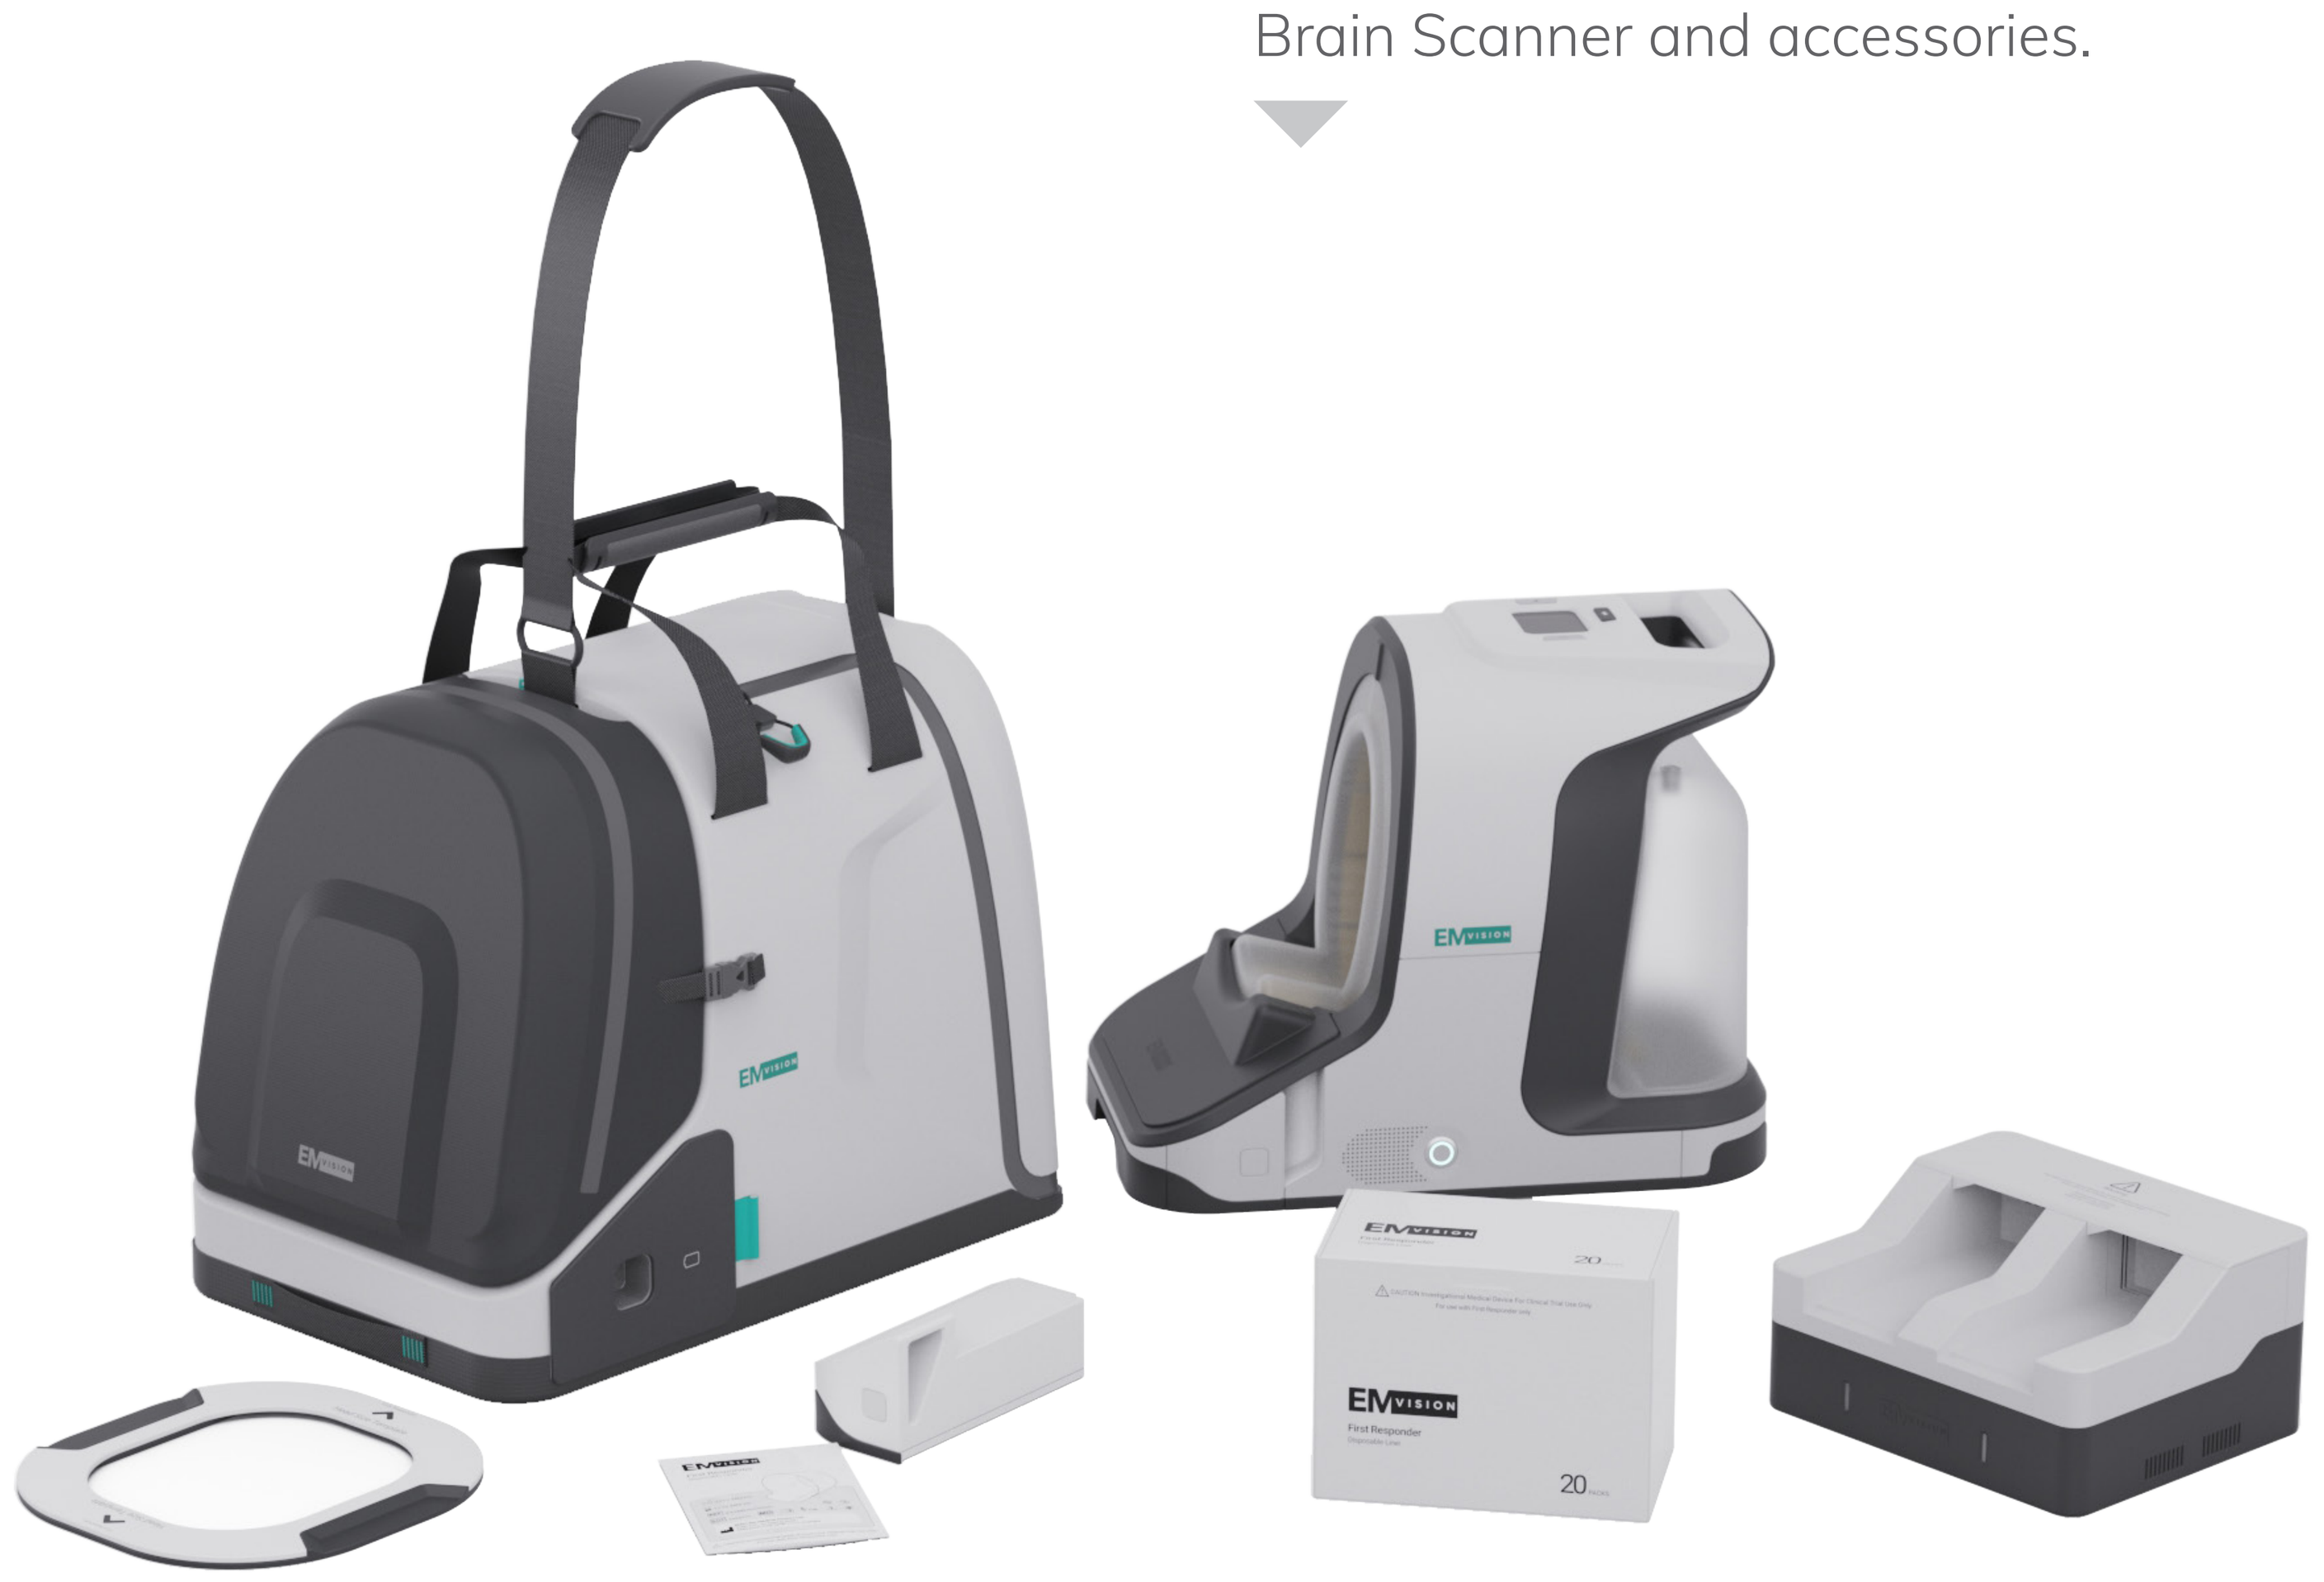

Technical Approach

The device uses low-power, non-ionizing radiofrequency signals to measure cranial dielectric profiles associated with hemorrhage. It can operate on battery or mains power, enabling use in mobile and fixed settings.

Results

Usability testing has been completed in remote locations serviced by fixed-wing aircraft and in emergency ambulance response environments (Figure 2), demonstrating feasibility in prehospital settings.

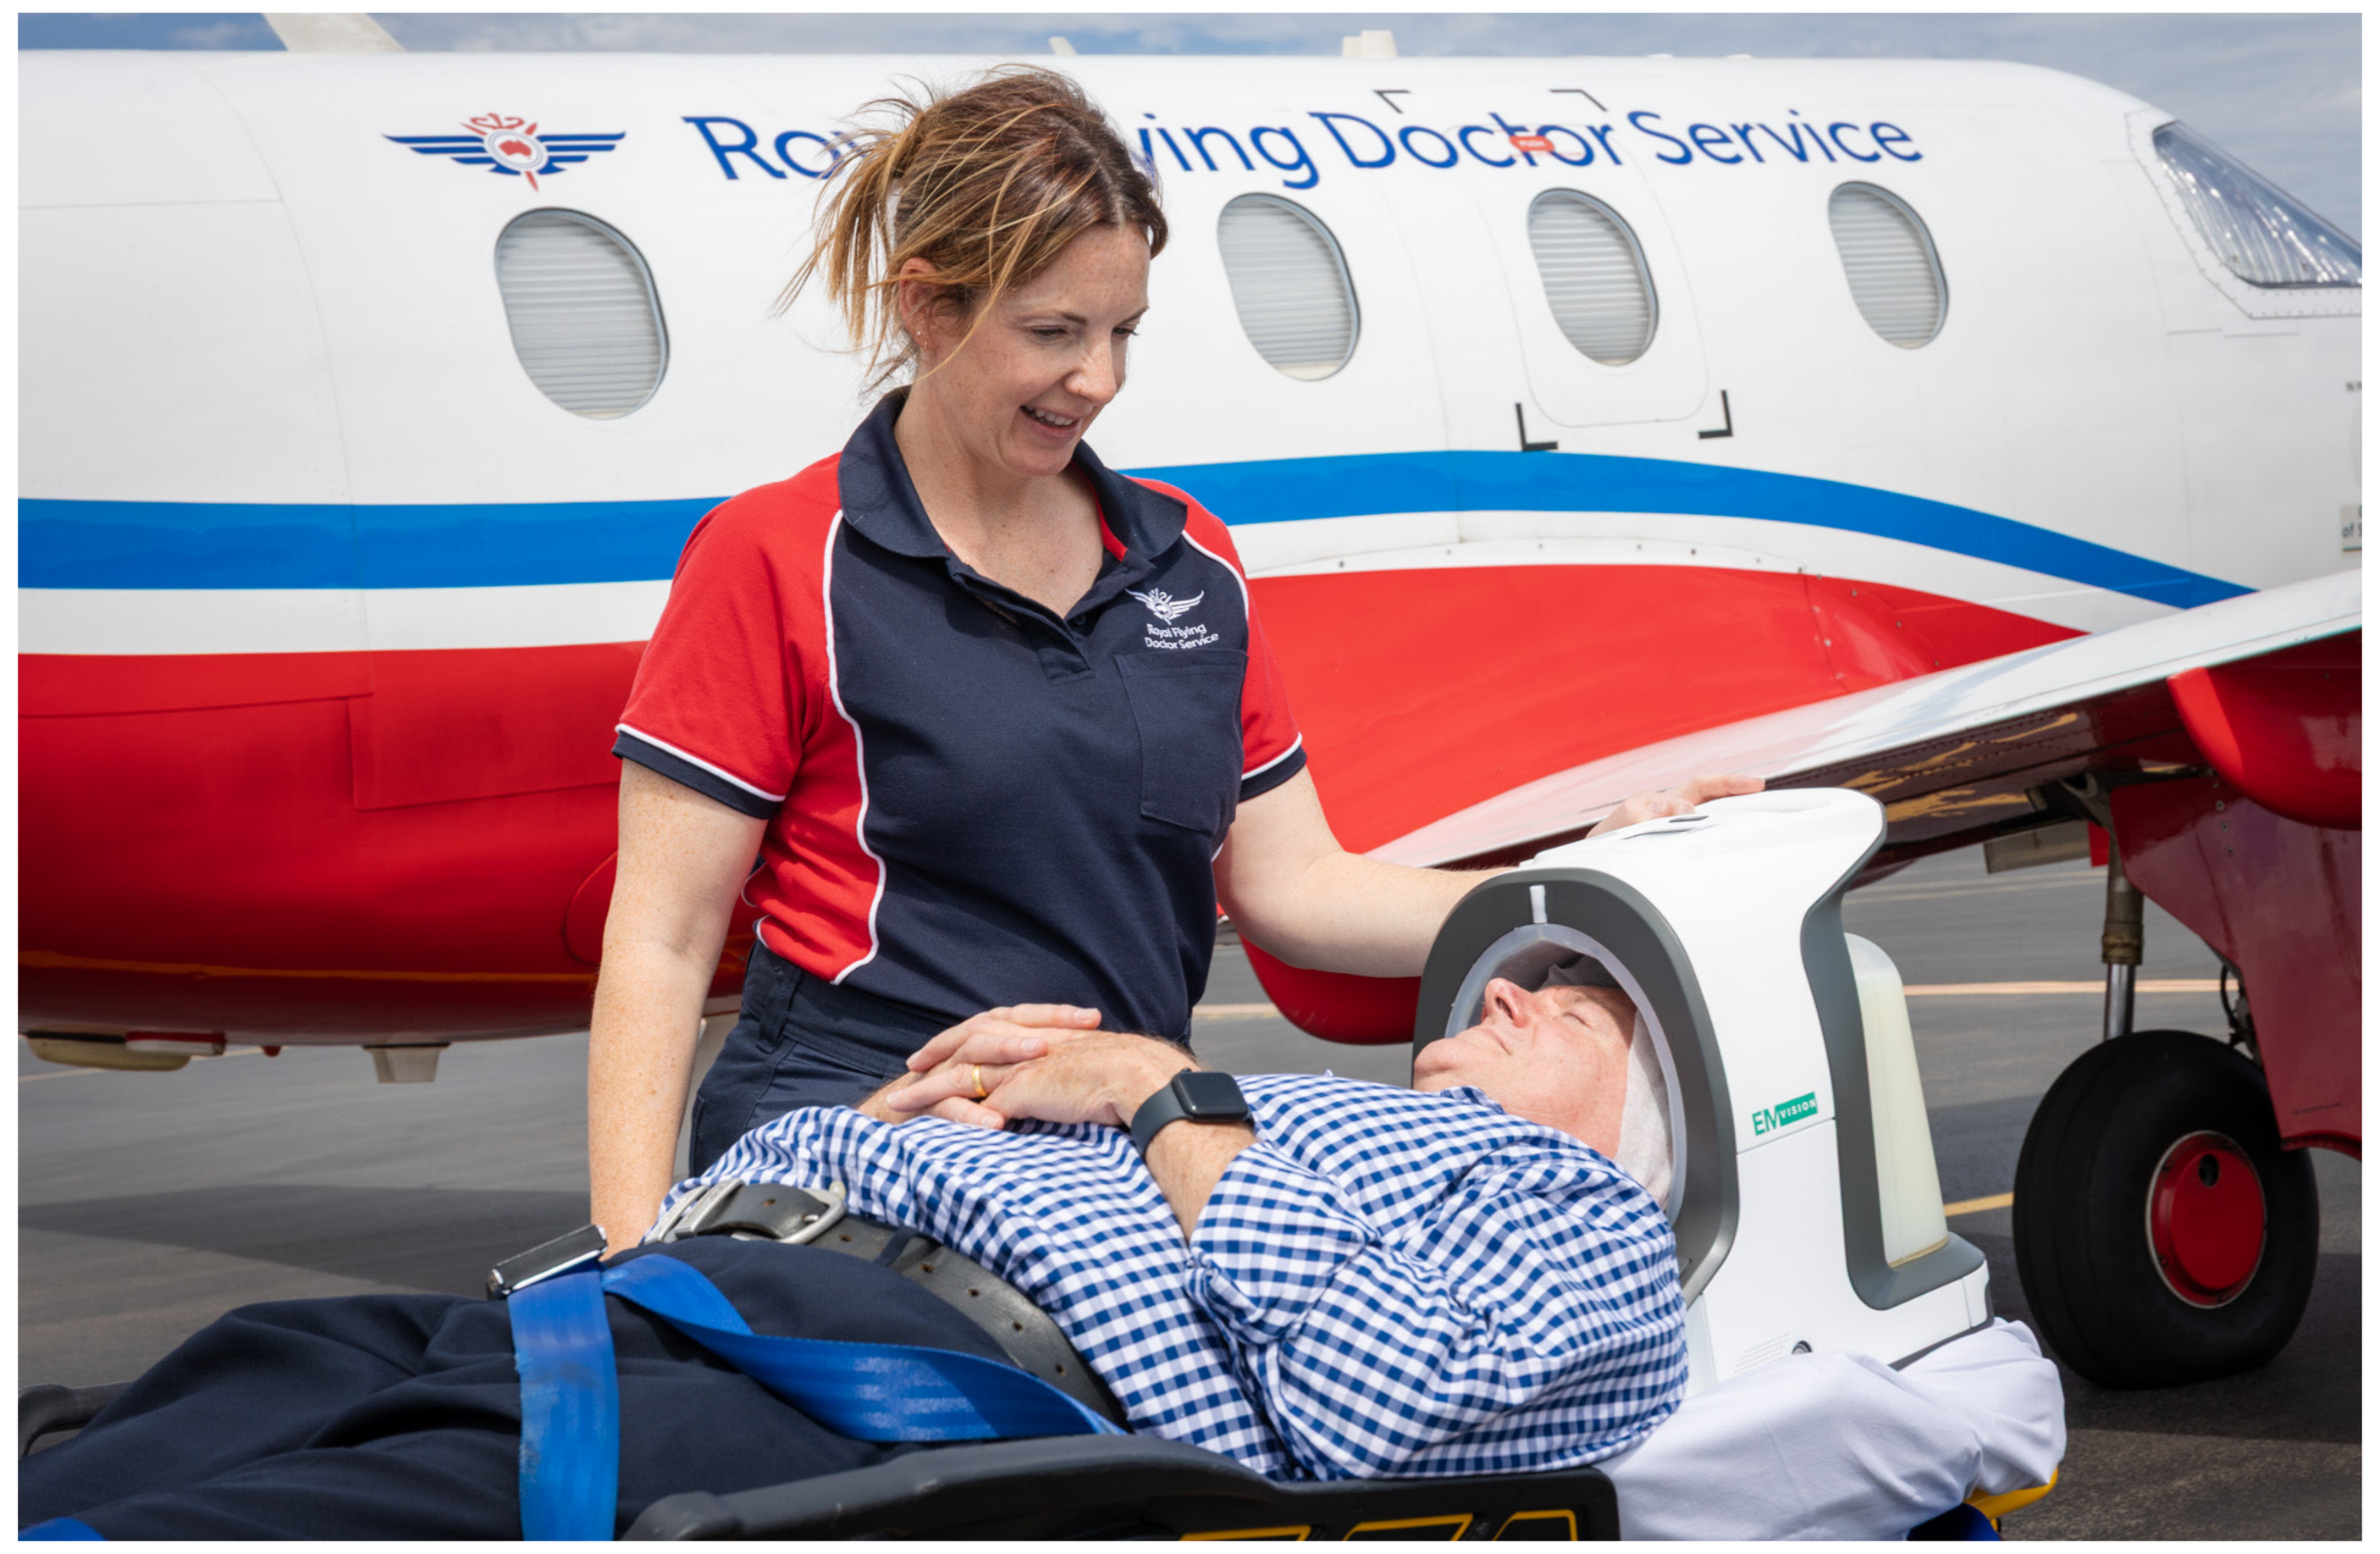

Figure 2
Usability testing has concluded in aeromedical retrievals conducted by the Royal Flying Doctor's Service (RFDS) (above) and in the a mobile stroke unit.

Impact to the Warfighter

In LSCO, routine long-range evacuation for suspected TBI is unsustainable. Higher accuracy, rapid structural brain injury detection may improve prioritization of scarce evacuation resources and, when evacuation is delayed or unavailable, support earlier initiation of appropriate interventions and efficient decision-making.

Applicability to Medical Roles of Care

This device is well suited to Role 2 settings, where advanced neuroimaging is typically unavailable. Algorithms under development are expected to exceed 90% sensitivity and specificity, in line with prior generations of the technology.

The scan procedure aligns with the MACE-2 assessment pathway, supporting integration into existing workflows while enhancing triage and transfer decision-making. The brain scanner does not require specialised training to operate or interpret and there are no risks to repeated exposure.

Developmental Status

The device is in advanced prototype development, has completed in-field workflow implementation trials, and is undergoing continued usability evaluation to support further development. Engineering efforts are underway to further reduce system weight for a production equivalent device before progressing to validation trials.

References

Dengler, B. A., Agimi, Y., Stout, K., Caudle, K. L., Curley, K. C., Sanjakdar, S., Rone, M., Dacanay, B., Fruendt, J. C., Phillips, J. B., & Meyer, A.-C. L. (2022). Epidemiology, patterns of care and outcomes of traumatic brain injury in deployed military settings: Implications for future military operations. Journal of Trauma and Acute Care Surgery, 93(2), 220–228.

Disclaimer

AC, RT, FK, CW, SK, and SC are employees of EMVision Medical Devices and own shares in EMVision Medical Devices.