NEUROFIT

MHSRS SESSION

Data to Decision: Optimizing Cognitive Performance

Exploratory biomarker convergence · US cohort, Jan 2023 – Jul 2026
10,174 users · 747 with pre-check-in PPG
Signal-quality-3 readings · user-balanced estimates

Toward Hardware-Free Autonomic Readiness Monitoring: Daily Somatic Self-Report and Smartphone PPG in 10,174 Users

A one-minute daily somatic read-out, paired with smartphone-camera physiology.

Andrew Hogue, BS[1] · Lisa Kang, MSPH[2]

[1] NEUROFIT, Miami, Florida

r = 0.14
Heart rate vs reported stress, pre-check-in

r = -0.10
HRV-RMSSD vs reported stress, pre-check-in

6.5×
RMSSD link is stronger before than after check-in

α = 0.81
NF8 internal consistency

r = 0.70
NF8 convergence with the PHQ-9

THE GAP

Neither self-report nor wearables alone gives a daily readiness signal.

- **Established instruments** (PHQ-9, GAD-7, DASS-21, PCL-5) recall 1–4 weeks — too coarse for a deployment or training cycle.
- **Wearables stream physiology** but lack validated linkage to subjective state.
- **Added hardware is a barrier** to fielding at scale.
- **Our pairing:** the **NF8** daily somatic scale plus phone-camera photoplethysmography — no extra device.

WHY THE ESTIMATOR DECIDES THE ANSWER

Readings are wildly unequal across users, so pooling them weights the loudest loggers.

The median user contributes **one** pre-check-in reading while the most active decile contributes **59% of all pairs**. A correlation computed over readings therefore weights users by how much they log.

We report **user-balanced (person-mean)** correlations — every user contributes exactly one observation — and cross-check with a mixed-effects model carrying a user random intercept. Only signal-quality-3 readings are used.

Not a cosmetic decision: it reverses two of the three markers (right).

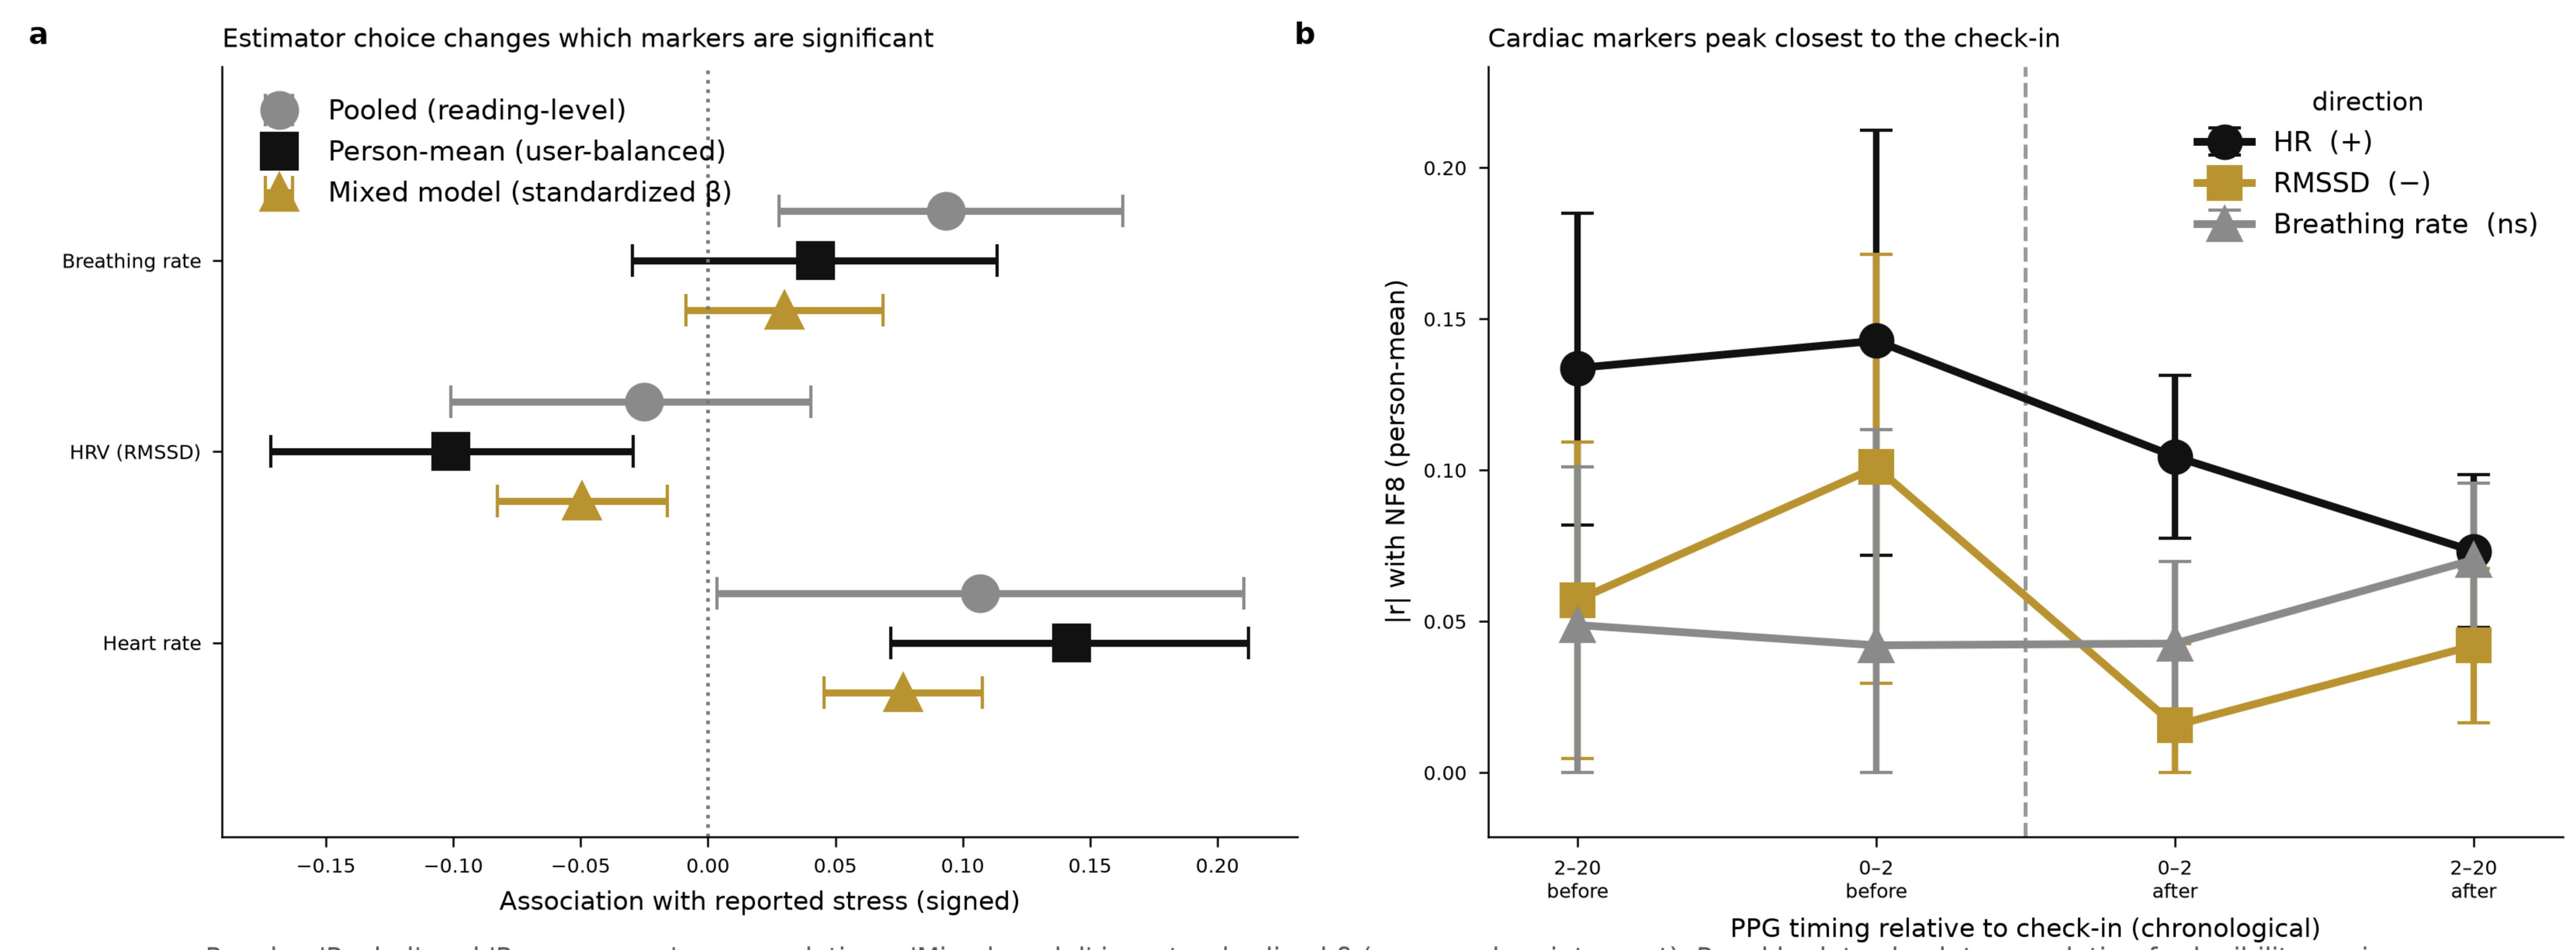
a
Estimator choice changes which markers are significant
Pooled (reading-level)
Person-mean (user-balanced)
Mixed model (standardized β)
Breathing rate
HRV (RMSSD)
Heart rate
−0.15
−0.10
−0.05
0.00
0.05
0.10
0.15
0.20
Association with reported stress (signed)
b
Cardiac markers peak closest to the check-in
direction
HR (+)
RMSSD (−)
Breathing rate (ns)
|r| with NF8 (person-mean)
0.00
0.05
0.10
0.15
0.20
2–20 before
0–2 before
0–2 after
2–20 after
PPG timing relative to check-in (chronological)

Panel a: 'Pooled' and 'Person-mean' are correlations; 'Mixed model' is a standardized β (user random intercept). Panel b plots absolute correlation for legibility — signs are fixed in the legend (heart rate POSITIVE, RMSSD NEGATIVE, both the expected autonomic direction). A CI spanning zero is drawn with a lower bound of 0; person-mean weights each user equally (n=747 users, 0–2-before window).

Figure 1. (a) The three estimators in the 0–2 min pre-check-in window. (b) Chronological temporal gradient. Panel detail in the figure footnote.

Reading time vs check-in	N (users)	Heart rate r	HRV-RMSSD r	Breathing rate r
2–20 min before	1,392	+0.134	-0.057	+0.049
0–2 min before	**747**	**+0.143**	**-0.101**	**+0.042**
0–2 min after	5,198	+0.104	-0.016	+0.043
2–20 min after	5,930	+0.073	-0.042	+0.070

User-balanced (person-mean) correlations with the NF8; bold row = primary window. Heart rate is positive and HRV-RMSSD negative throughout — the expected joint autonomic signature of sympathetic arousal with parasympathetic withdrawal. Breathing rate shows no pre/post gradient (1.0×) against 1.4× for heart rate and 6.5× for RMSSD.

WHAT USER-BALANCING REVERSES

Two of three markers change verdict.

- **HRV-RMSSD gains significance:** pooled r = -0.03 (interval crosses zero) becomes -0.10 [-0.17, -0.03], and the mixed model agrees.
- **Breathing rate loses it:** pooled +0.09 becomes +0.04 [-0.03, 0.11].
- **Mechanism:** the most active decile carries an RMSSD correlation of the *opposite sign* to the other 90%, so pooling cancelled a real association.
- **Robustness:** the RMSSD estimate survives deleting the 20 heaviest loggers.

EACH USER AS THEIR OWN CONTROL

Within-person concordance agrees, and one clean negative.

Among users with ≥3 pre-check-in readings, a user's own day-to-day NF8 change tracked their own heart rate (r = +0.217 [0.13, 0.31], N = 361) and RMSSD (r = -0.118 [-0.21, -0.03]); breathing rate was null (r = +0.014).

This design is immune to the clustering problem by construction and agrees with the user-balanced cross-sectional result.

A negative result worth stating: in a matched day-7 subsample (n = 1,759) the NF8 fell substantially (d = +0.76) with *no* corresponding biomarker movement and no within-person coupling between a user's NF8 change and their own biomarker change. Over a week, self-reported change and resting physiology are dissociated.

WHAT THIS DOES AND DOES NOT SHOW

A validated daily self-report scale; the biomarker linkage is exploratory and small.

The NF8 has established convergent validity (PHQ-9 r = 0.70, GAD-7 0.55, DASS-21 Stress 0.52; α = 0.81), and pre-check-in cardiac markers move with it in the physiologically expected directions.

These correlations are small, cross-sectional and observational. The temporal gradient is suggestive construct-validity evidence, not proof of causal ordering. RMSSD from a short mobile-camera recording is the noisiest of the three metrics by construction, and breathing rate showed no association once users were weighted equally. **Neither cognitive performance nor readiness was measured in this dataset, and no participant was identified as military** — operational relevance motivates the planned military validation study; it is not a finding of this work.

PPG correlations are user-balanced (person-mean) over signal-quality-3 readings; US-based cohort. Python primary with independent R re-implementation. Companion preprints in preparation (Hogue & Kang, 2026).

MHSRS 2026