MHSRS-26-6010

XTEMP: Rapid Contactless Core Temperature Monitoring for Heat-Risk Prevention in Operational Environments

Senay Tewolde, PhD[1], Antoniette Vickio[1], Katharine Long, PhD[2], Souleymane Día[1], Cody Butler, Lt Col, USAF, BSC[3], Joshua J Van Wyngaarden, Lt Col, USAF, BSC, DPT, PhD[4]

[1] BLINKTHERMAL, INC, Dallas, TX, 75230, [2] TEXAS TECH UNIVERSITY, Lubbock, TX, 79409, [3] AFRL 711 HPW/RHBC, [4] Wright-Patterson AFB, OH 45433, [4] Special Warfare Human Performance Squadron, JBSA Lackland – Chapman Annex, TX 78236

01 Problem

Exertional heat illness remains one of the most persistent and preventable threats to military readiness. In 2024 the U.S. Armed Forces recorded crude incidence rates of 36.4 heat-stroke and 183.9 heat-exhaustion cases per 100,000 service members [1], underscoring the continued operational burden of heat-related illness.

Core body temperature is the most direct available physiological indicator of thermal strain and is often among the earliest vital signs to reflect a developing heat injury. However, current methods — including rectal and esophageal probes — are invasive, logistically constrained, and impractical for routine surveillance during military training or operational activity. This leaves a critical gap between the need for early heat-risk detection and the tools available in the field.

A contactless, deployable core-temperature monitoring method, resilient to operational and environmental variability, could close this gap by enabling earlier recognition of rising thermal strain and supporting objective decisions around risk, recovery, and readiness.

02 How XTemp Works

XTEMP is a contactless platform that monitors thermal activity — core body temperature — from the back of the eye. A dual optical and long-wave infrared camera captures the face; 68-point facial-landmark detection isolates the ocular surface and extracts pixel-wise temperature from roughly 3,900 pixels of the exposed eye surface

XTEMP leverages that extracted surface temperature to evaluate the posterior temperature of the eye using inverse heat transfer and a gradient-based optimisation method. The physical model of the eye is grouped into five regions — sclera/choroid/iris, cornea, aqueous humour, lens and vitreous humour — and two boundary conditions: the exposed and the closed surfaces of the eye. The vascularised regions are treated as a perfused porous medium, while the exposed surface carries convective, radiative and tear-evaporative boundary conditions.

Core temperature is then recovered by inverse solution: a trial value is proposed, a forward solve of the heat equation predicts the resulting surface field, and gradient-based optimisation minimises the fit residual against the measured image. The reported value is the temperature at the posterior boundary of the eye — an interior state variable of a heat-transfer model, not a surface reading. No contact, no consumable, no per-subject calibration, under one second.

Acquire - < 1 second

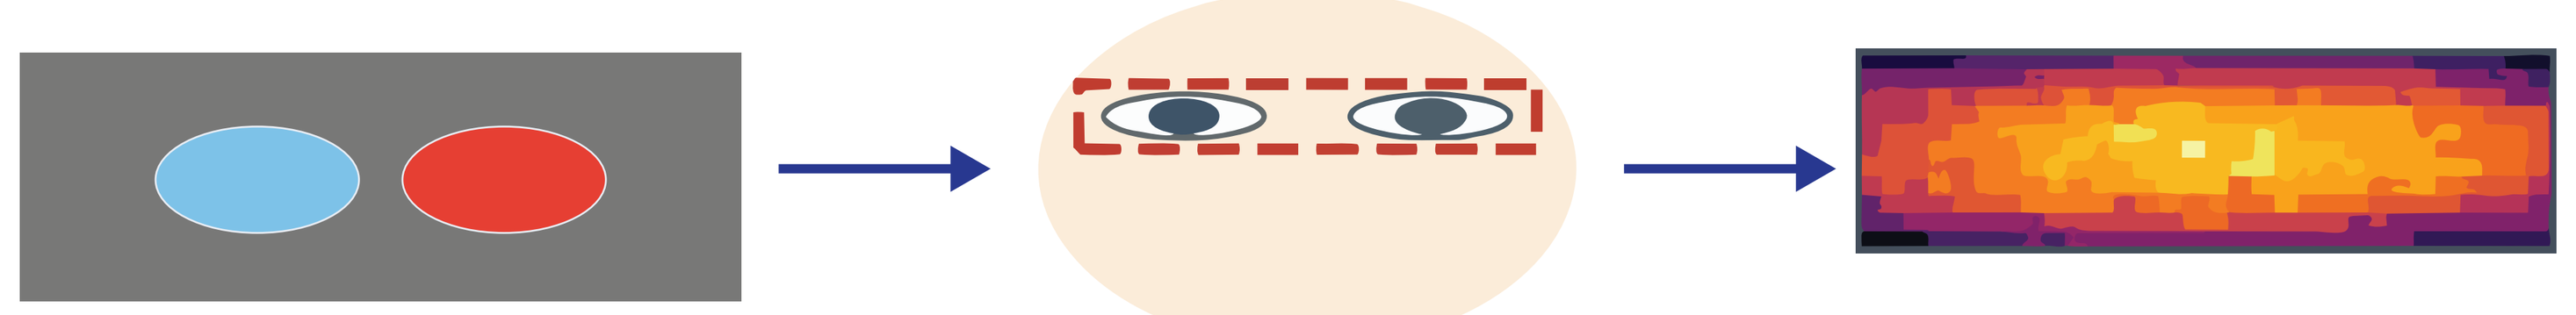

Optical camera + IR Thermal Imaging *non-contact

68-point facial landmark detection

≈39,000 ocular paths identified with temperature

Model

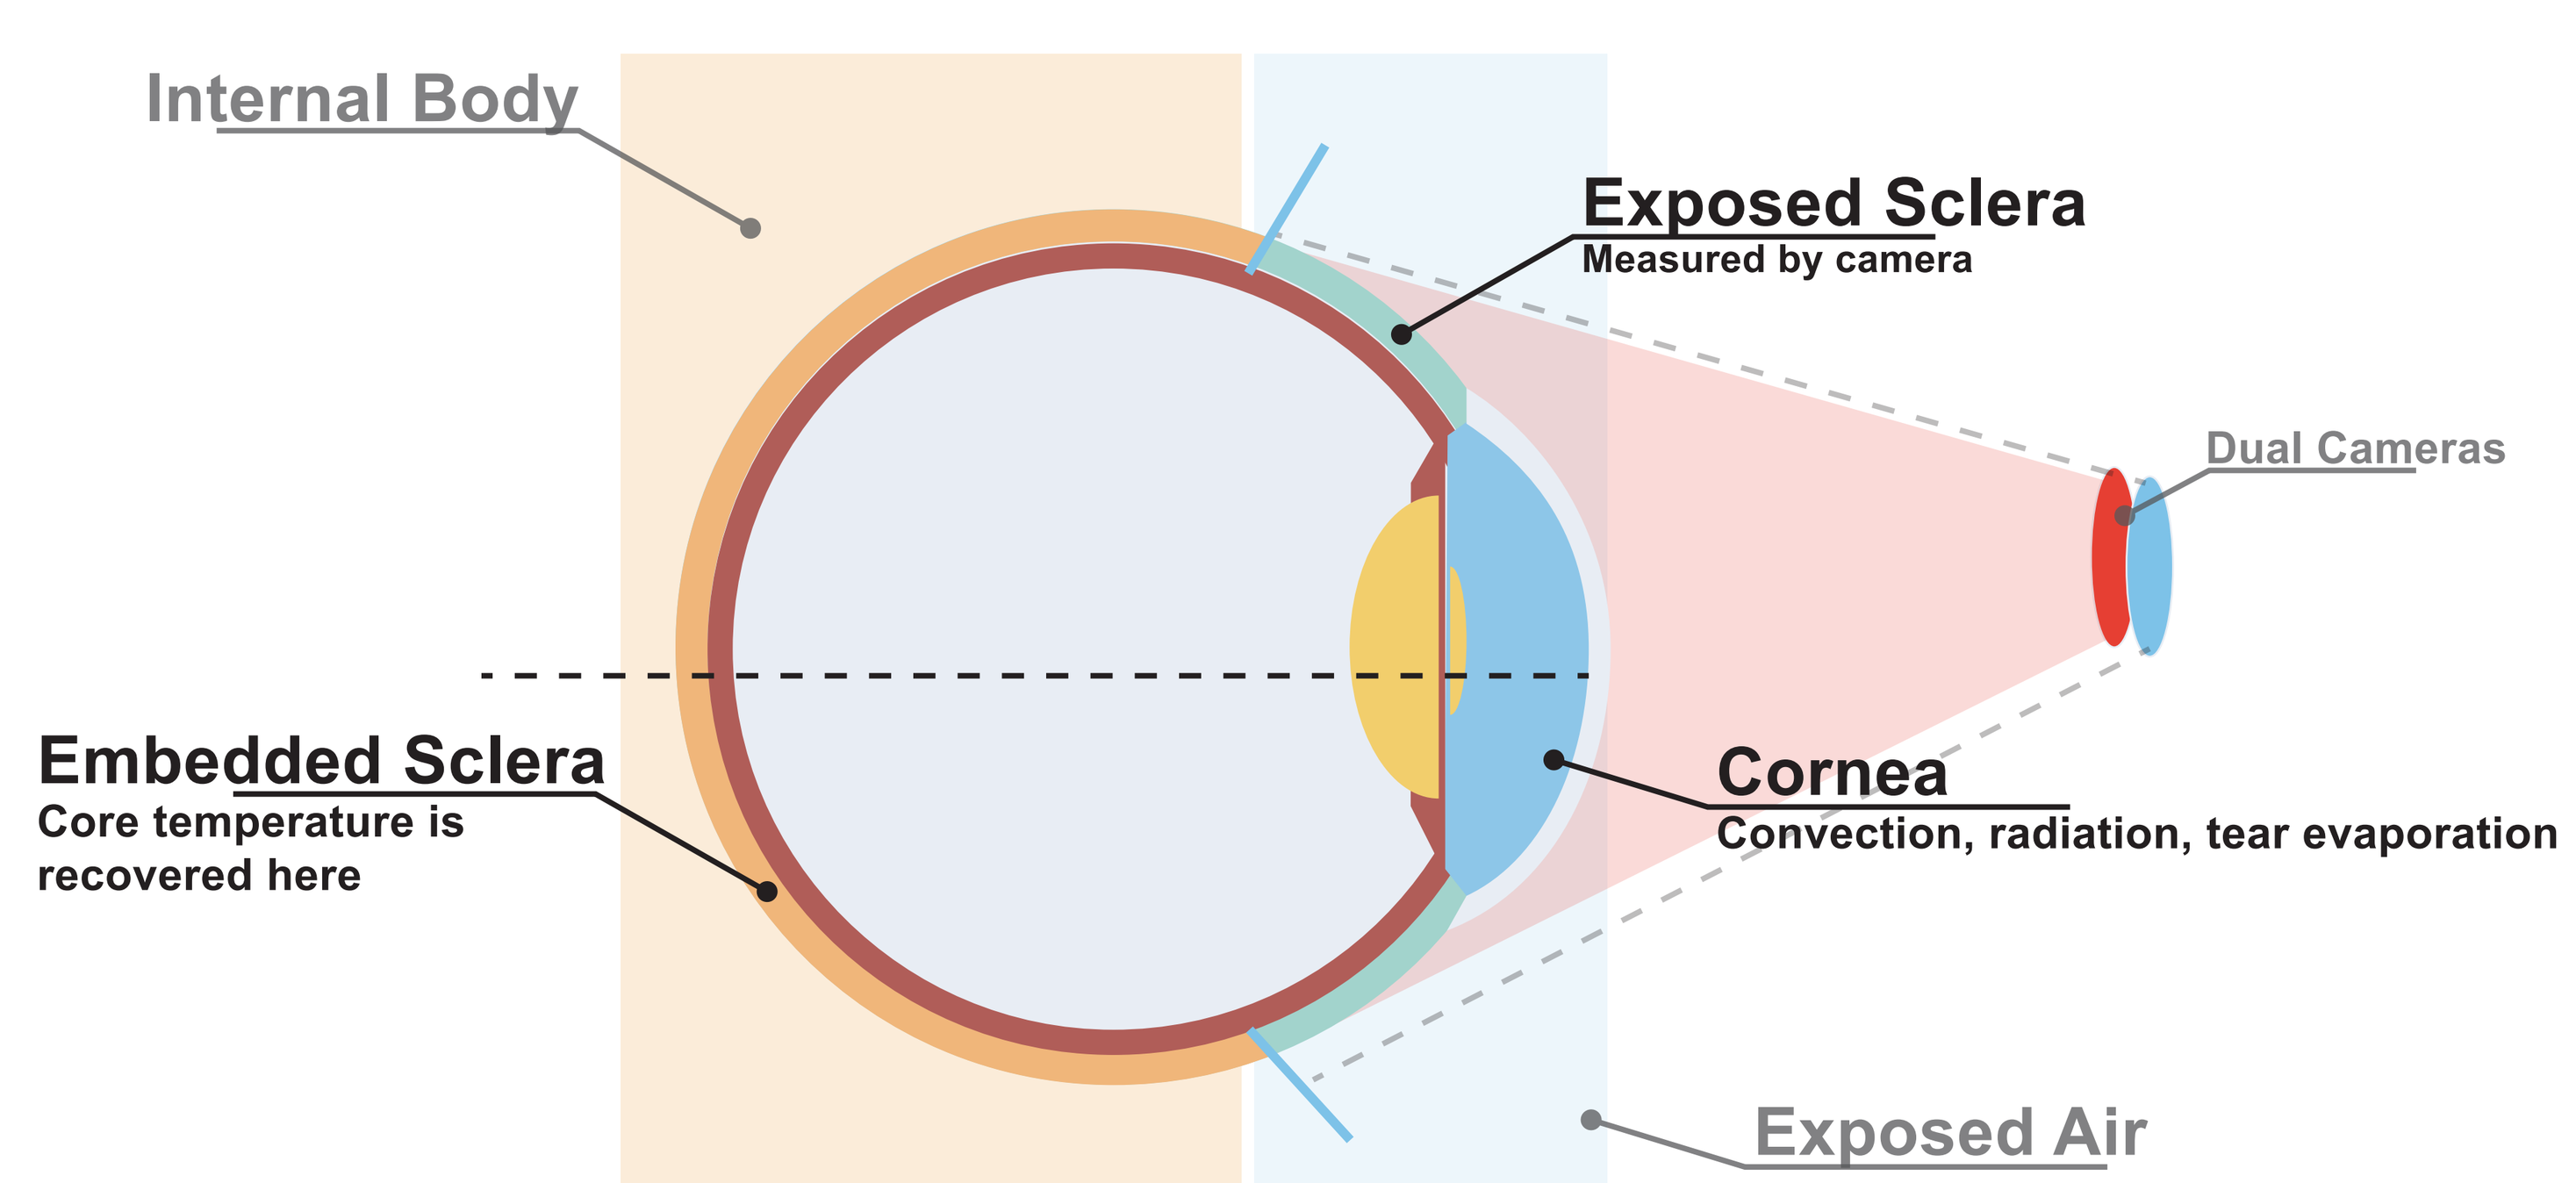
Internal Body
Exposed Sclera
Measured by camera
Dual Cameras
Embedded Sclera
Core temperature is recovered here
Cornea
Convection, radiation, tear evaporation
Exposed Air

$$\nabla(K_{eff}\nabla T_1) - Q_{eff} = 0 \qquad \nabla(K_i\nabla T_1) = 0$$

Solve

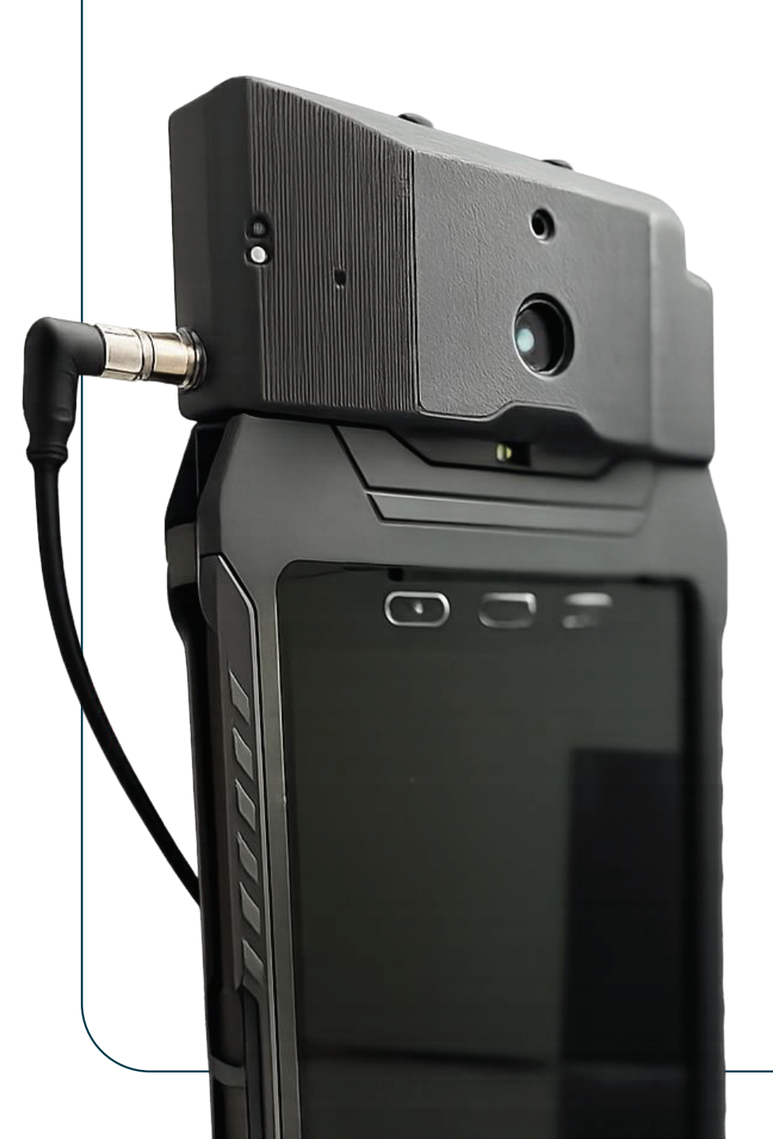

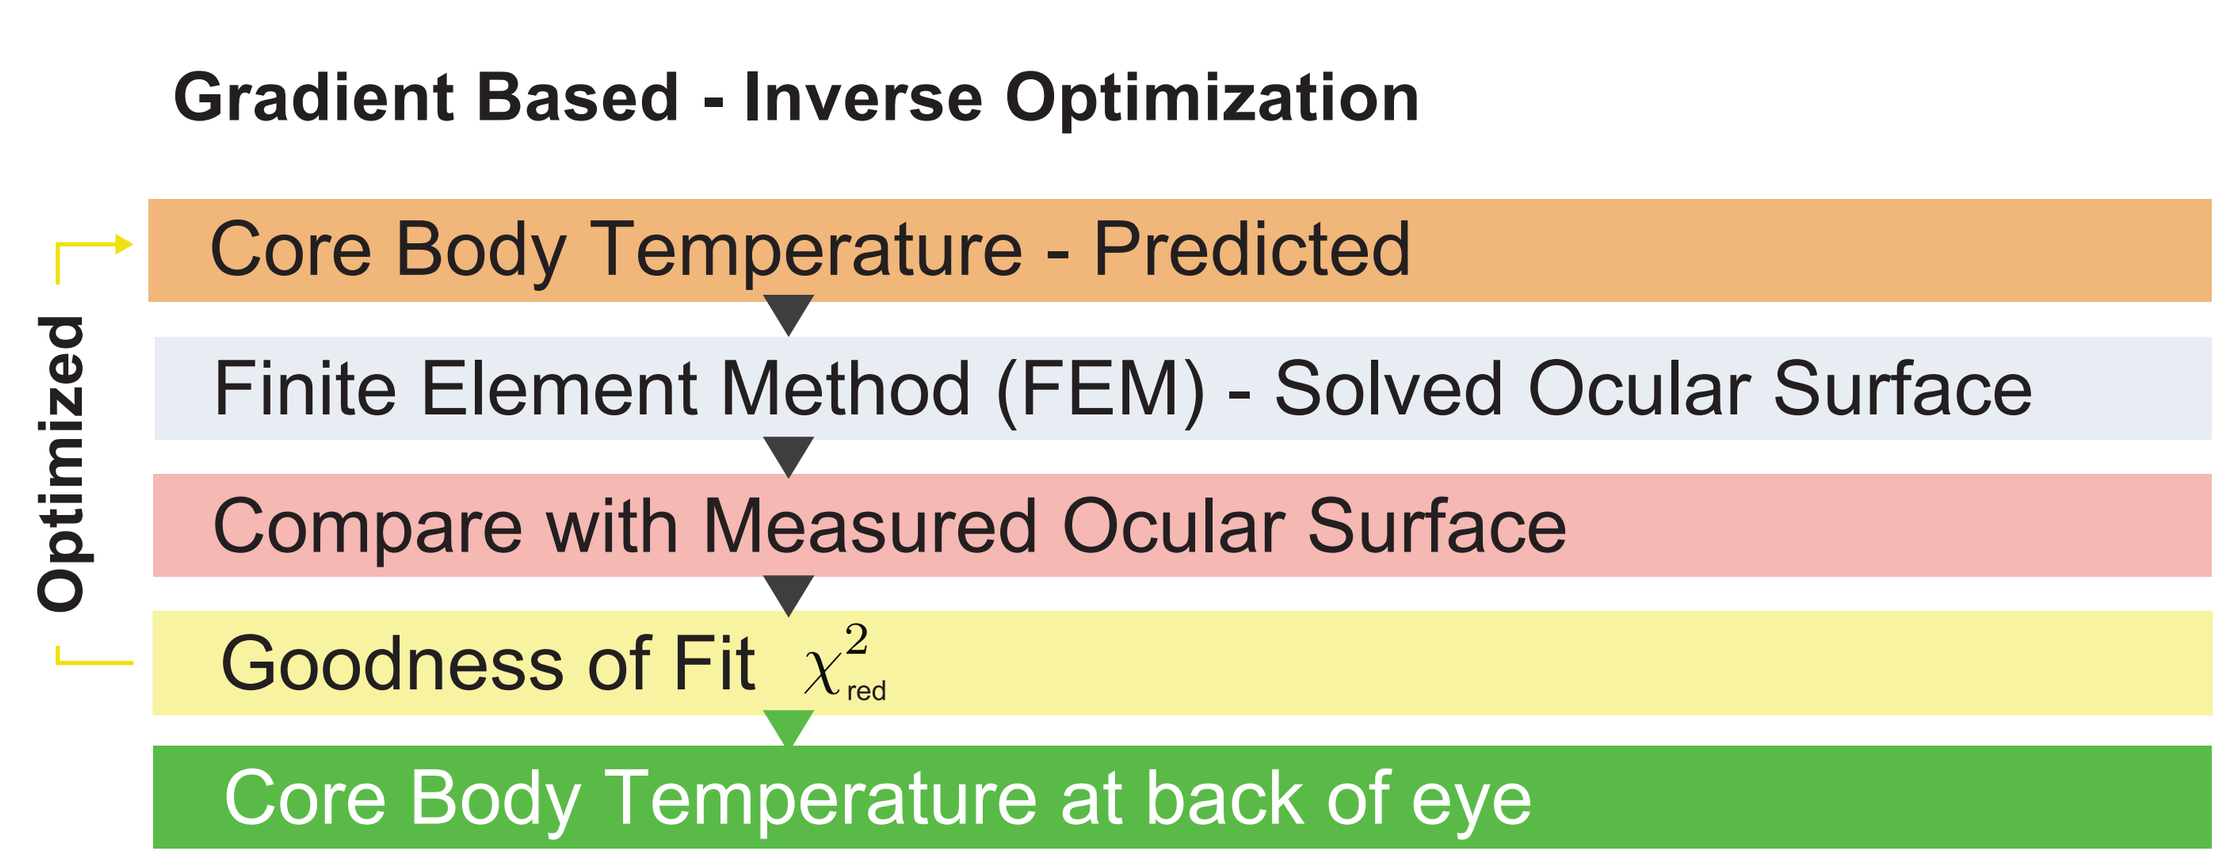
Gradient Based - Inverse Optimization
Optimized
Core Body Temperature - Predicted
Finite Element Method (FEM) - Solved Ocular Surface
Compare with Measured Ocular Surface
Goodness of Fit χ^2_{red}
Core Body Temperature at back of eye

03 Clinical Validation

Conducted under Development and Validation of Rapid Contactless Core Body Temperature Measurement Capability, approved by the Texas Tech University Institutional Review Board (**IRB #2024-840**).
Participants completed two randomised environmental-chamber visits, one warm and one hot, separated by a washout interval. Each visit included a 60-minute intermittent treadmill protocol: a resting baseline, 20 minutes of jogging at 5–8 mph, a 2-minute transition to don the weighted vest, and 38 minutes of weighted-vest walking with a 20–40 lb load. The present analysis reports the hot-condition data (6 male, 2 female).

XTEMP was compared with rectal temperature to evaluate contactless core-temperature tracking over time. Agreement was assessed using correlation, regression, Bland–Altman analysis, error metrics, intraclass correlation and Lin's concordance correlation. The Heat Strain Index was calculated separately from XTEMP and from rectal temperature to compare heat-strain tracking between methods.

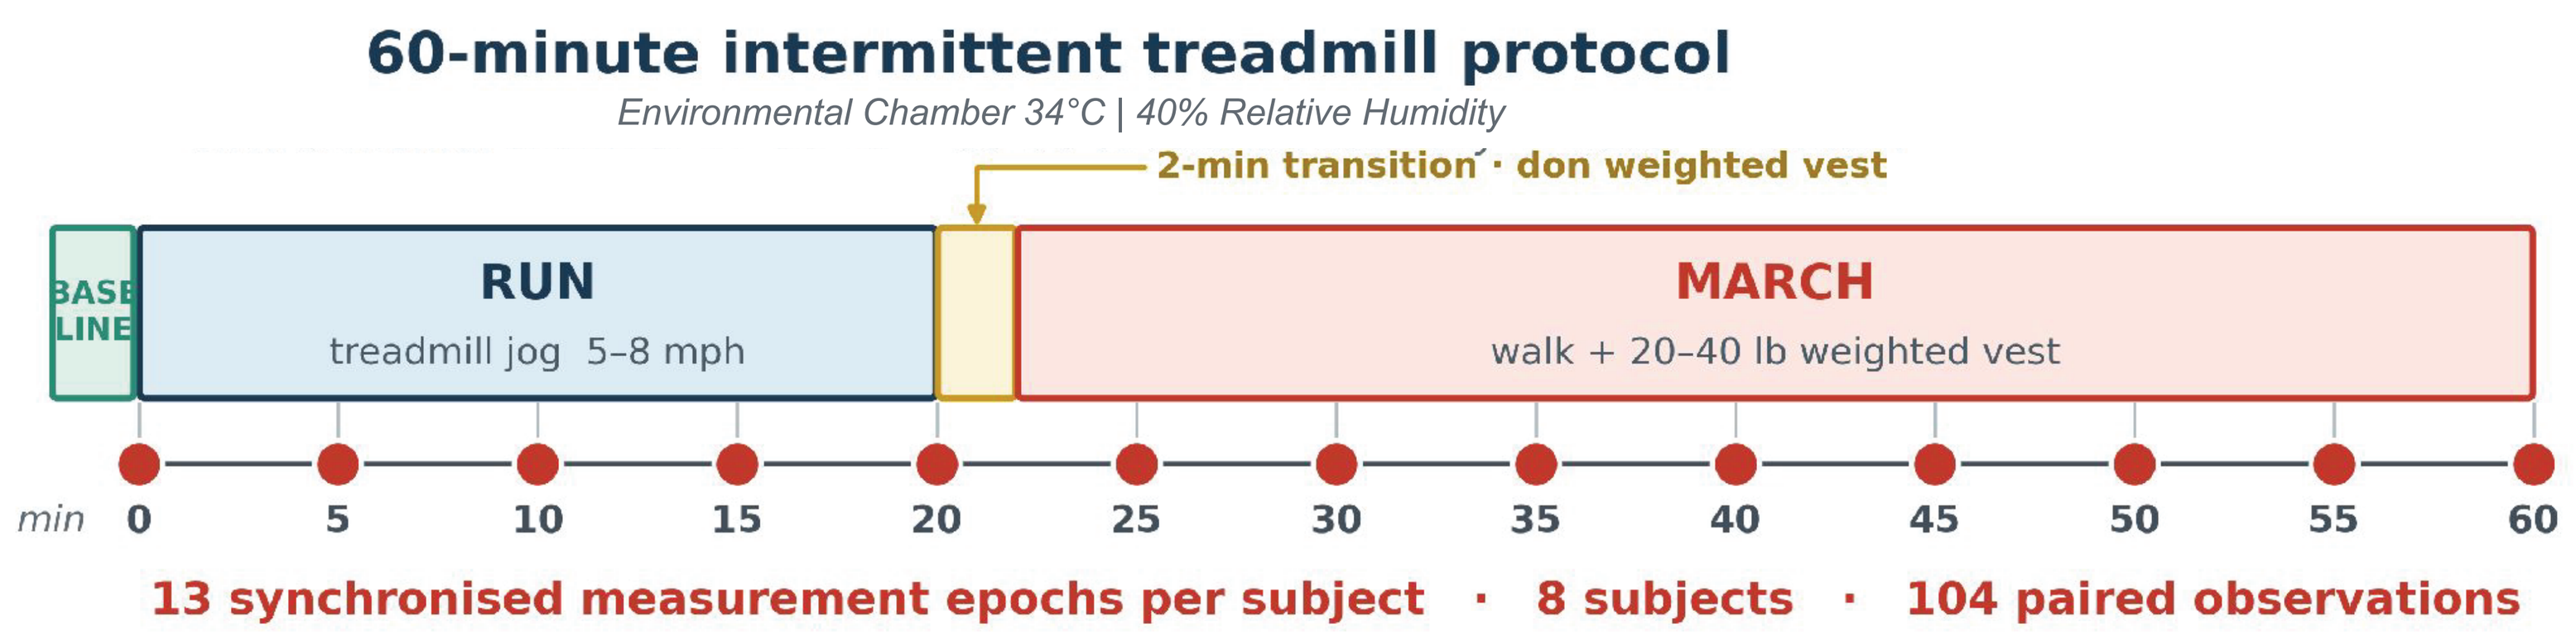
60-minute intermittent treadmill protocol
Environmental Chamber 34°C | 40% Relative Humidity
2-min transition · don weighted vest
BASE LINE
RUN
treadmill jog 5–8 mph
MARCH
walk + 20–40 lb weighted vest
min 0 5 10 15 20 25 30 35 40 45 50 55 60
13 synchronised measurement epochs per subject · 8 subjects · 104 paired observations

Instruments
XTEMP Camera | Rectal Probe (YSI probe, BIOPAC) | Hear Rate - Polar H10 chest strap

04 Results

Thermal and Cardiovascular Responses

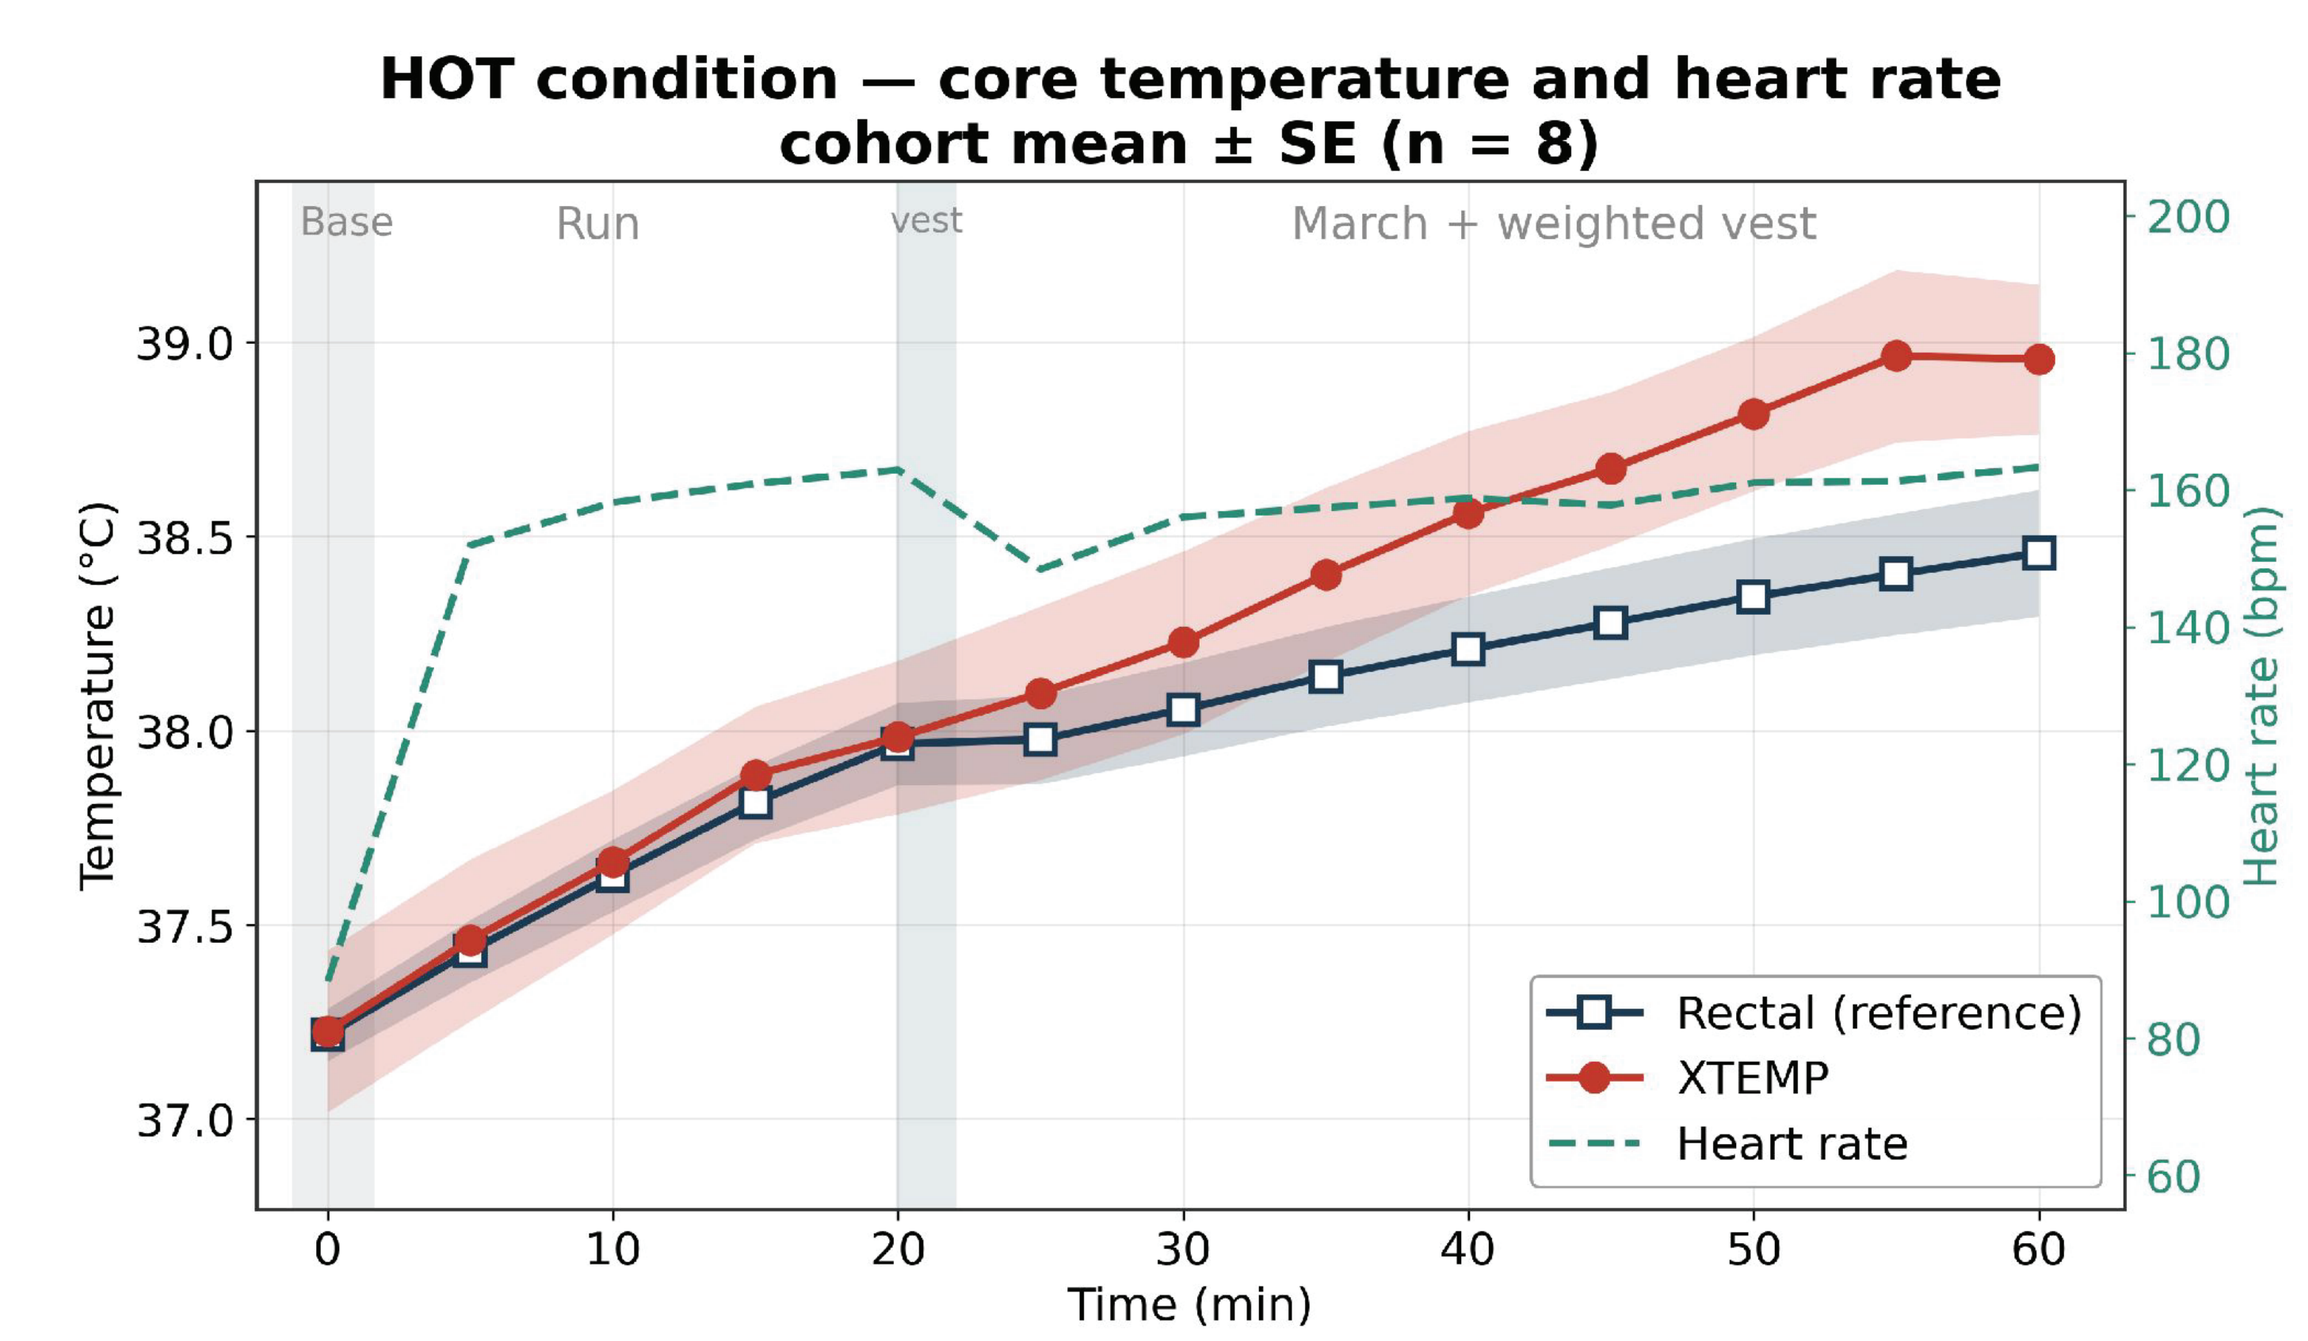
HOT condition — core temperature and heart rate
cohort mean ± SE (n = 8)
Base
Run
vest
March + weighted vest
Temperature (°C)
Heart rate (bpm)
Time (min)
Rectal (reference)
XTEMP
Heart rate

The protocol produced progressive, uncompensable heat strain: rectal temperature rose 1.24 ± 0.48 °C (37.22 → 38.46 °C; peak 39.31 °C) and heart rate reached 169 ± 26 beats·min ¹, remaining elevated throughout the march. No participant reached a stopping criterion.

XTEMP reproduced the rectal trajectory with no detectable lag. Bias remained within ±0.04 °C from baseline through the run, the two methods moved in the same direction across 80.2 % of epoch transitions, and all 8 of 8 participants tracked individually at r > 0.70 (within-subject r = 0.935). Separation emerged only after the transition to load carriage, growing monotonically to +0.56 °C by 55 minutes — a systematic, phase-locked divergence consistent with compartment-specific thermal kinetics under sustained thermal and circulatory load, not a loss of signal.

Bland - Altman Agreement

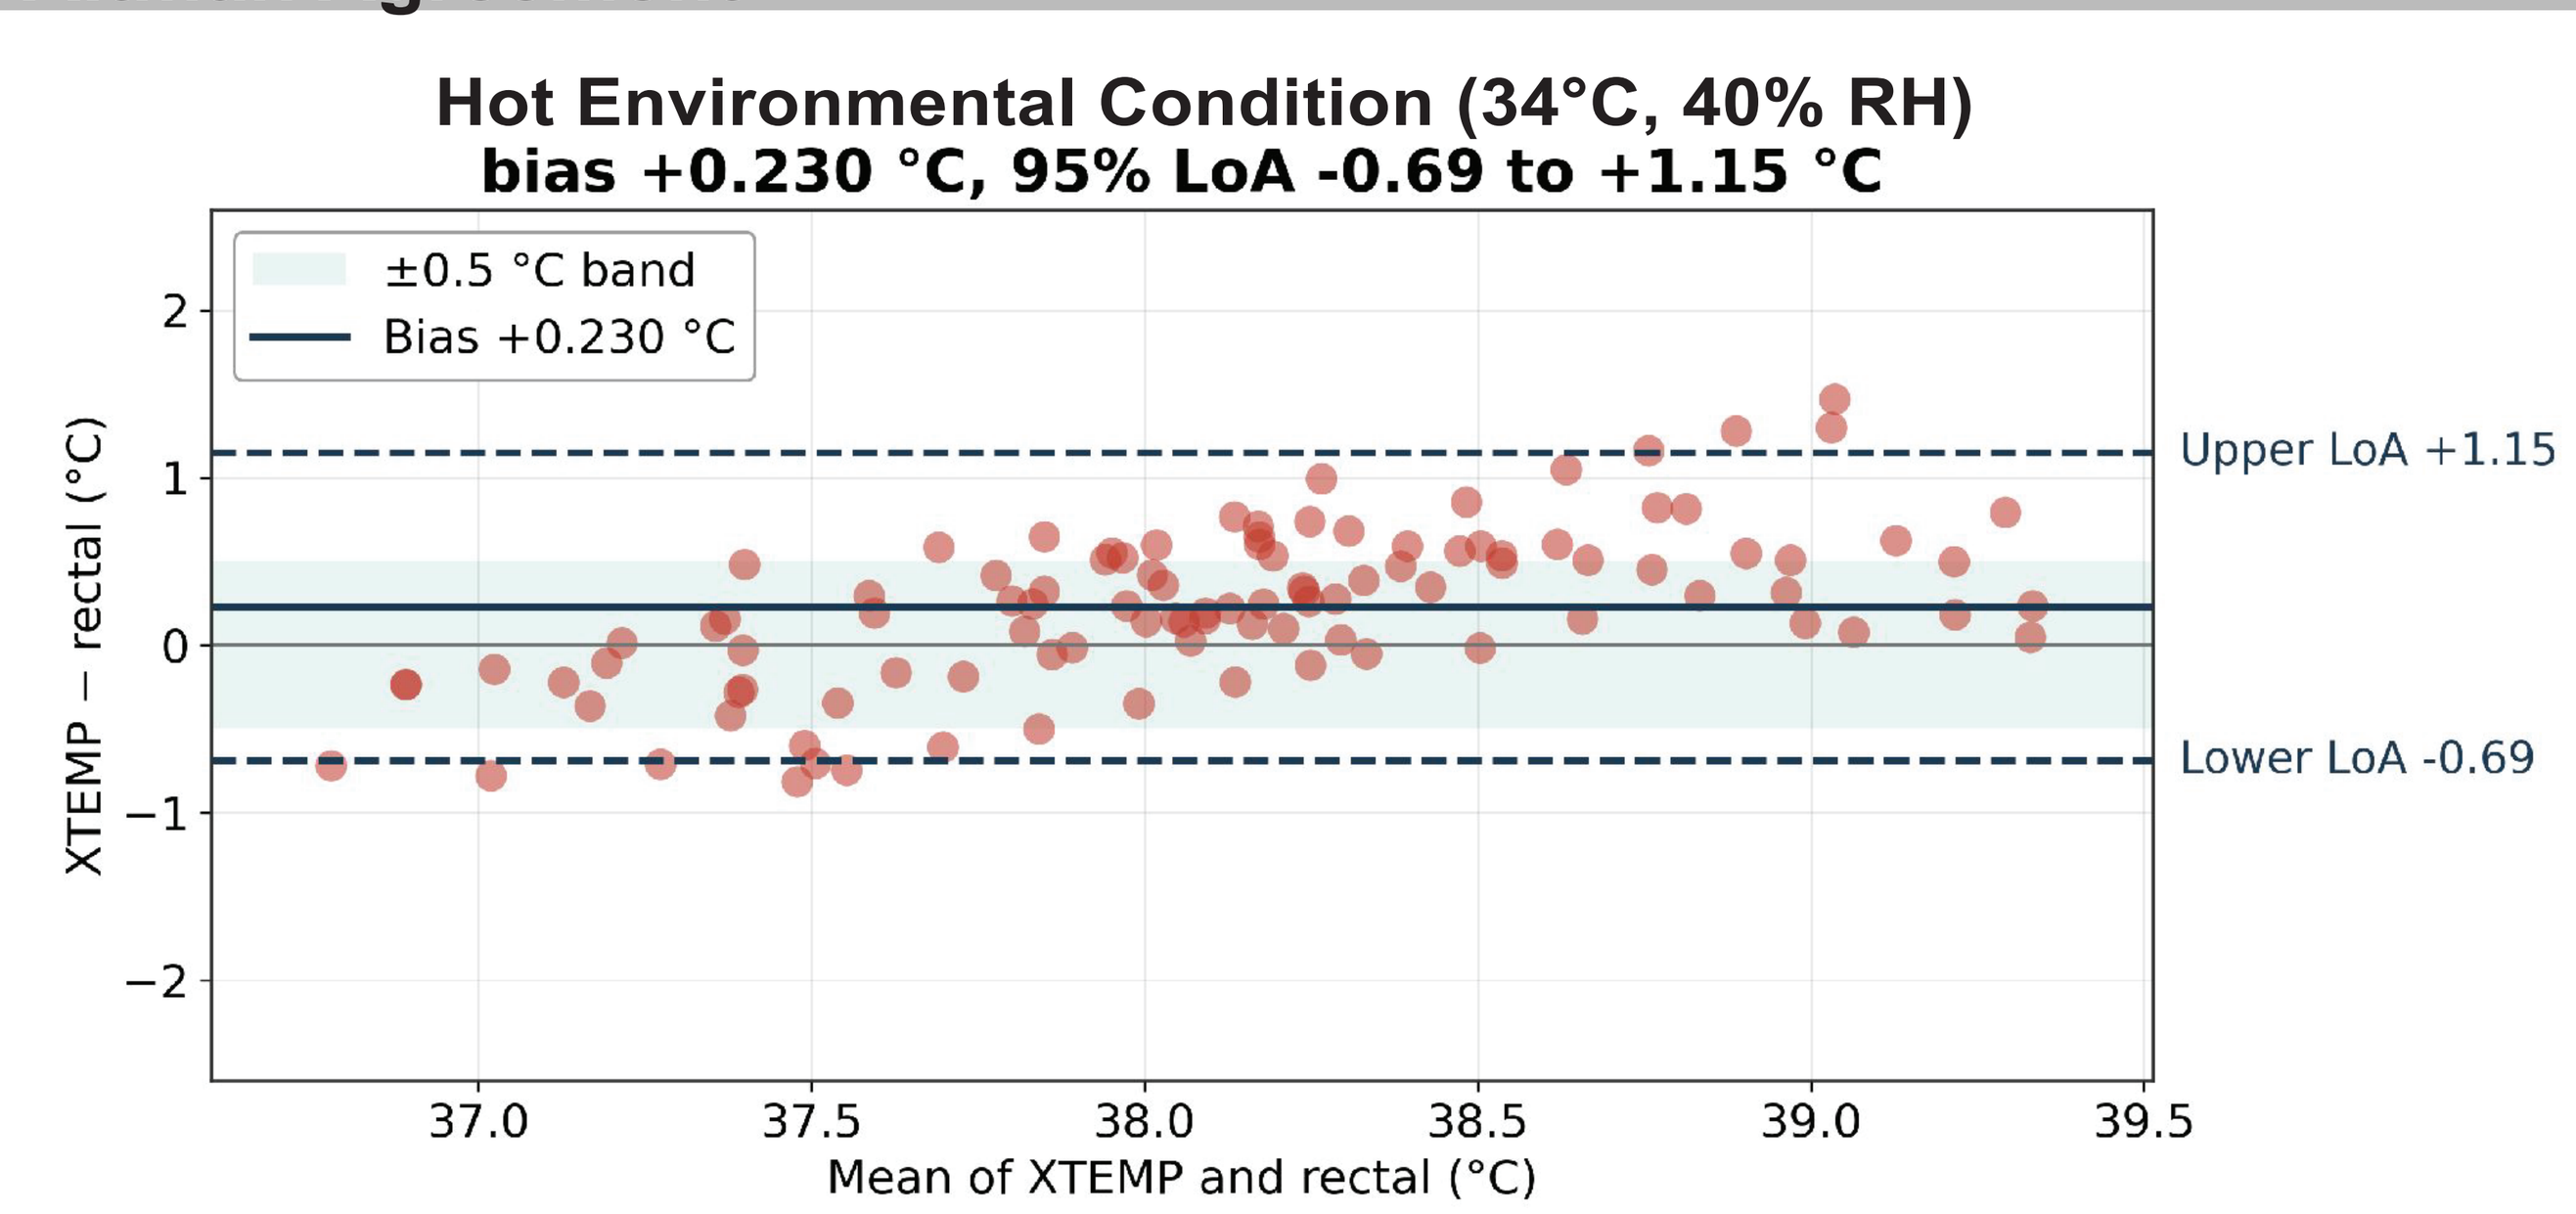
Hot Environmental Condition (34°C, 40% RH)
bias +0.230 °C, 95% LoA -0.69 to +1.15 °C
±0.5 °C band
Bias +0.230 °C
Upper LoA +1.15
Lower LoA -0.69
XTEMP − rectal (°C)
Mean of XTEMP and rectal (°C)

Across all epochs XTEMP was strongly associated with rectal temperature (r = 0.818, ρ = 0.823, p < 0.001) but differ by a characterisable offset and scale – mean bias +0.230 °C (95 % CI +0.139 to +0.322), 95 % limits of agreement -0.69 to +1.15 °C, RMSE 0.521 °C, ICC(2,1) = 0.697, and 60.6 % of readings within ±0.5 °C – rather than varying independently.

04 Results

Metric	Value	Interpretation
Pearson r · Spearman ρ	0.818 · 0.823	Strong linear association
Within-subject r	0.935	Individual tracking preserved
Bias (95 % CI), °C	+0.230 (+0.139, +0.322)	Significant positive offset
95 % limits of agreement, °C	-0.69 to +1.15	Exceeds ±0.5 °C equivalence margin
ICC(2,1) · Lin's CCC	0.697 · 0.695	Moderate absolute agreement
OLS slope (SE)	1.29 (0.09)	Gain error; slope > 1, p < 0.001
Difference–mean slope, °C·°C ¹	0.49 (p < 0.001)	Proportional, not fixed, bias

Per-participant bias spanned -0.53 to +0.75 °C while within-participant correlation remained 0.878–0.990, indicating displacement of the individual regression intercept rather than loss of signal. The SD of the difference was invariant across epochs (0.35–0.50 °C) while bias rose from +0.01 °C at baseline and +0.04 °C through the run to +0.35 °C under load carriage — a load-dependent divergence in scale between compartments, not independent variation. A gain term of known form is invertible.

Heat Strain Index

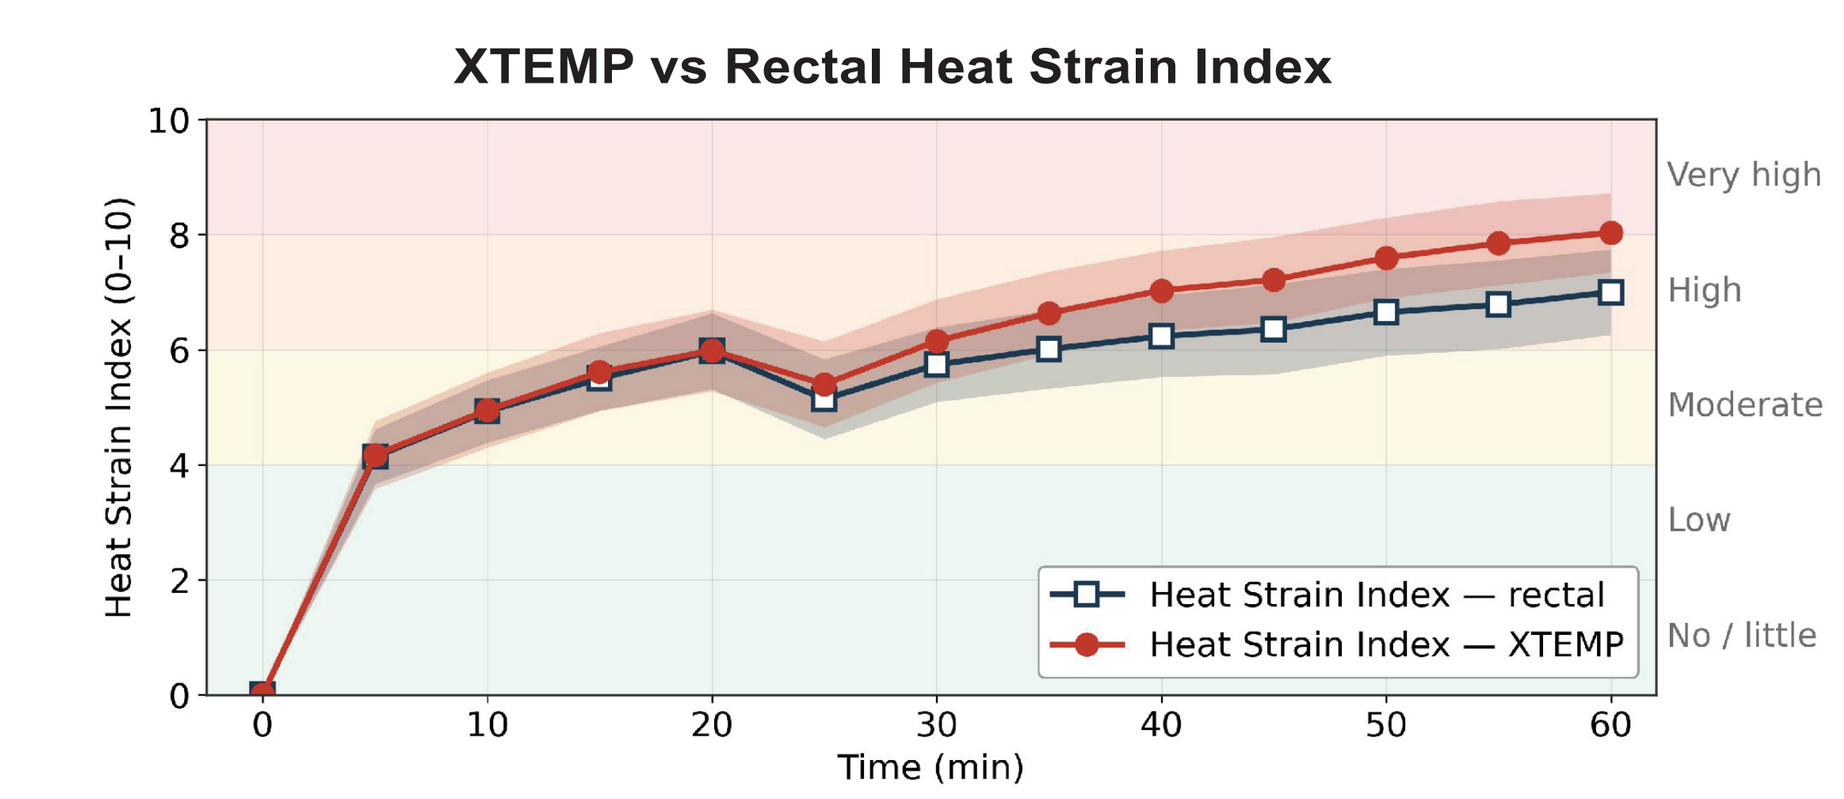
XTEMP vs Rectal Heat Strain Index
Heat Strain Index (0–10)
Very high
High
Moderate
Low
No / little
Heat Strain Index — rectal
Heat Strain Index — XTEMP
Time (min)

Heat strain was quantified with the Heat Strain Index (the Physiological Strain Index of Moran et al. [3]), a 0–10 scale combining core temperature and heart rate against each participant's own resting baseline. It was computed independently from each temperature source using the identical measured heart-rate series, so any difference arises solely from the temperature term.

Agreement between the two indices was substantially better than between the raw temperatures: ICC(2,1) = 0.940 (excellent) against 0.697 for temperature, with r = 0.961 and bias +0.48 index units on a scale whose risk bands are 2 units wide. Because the index references each participant to their own baseline, an individual offset in the temperature estimate largely cancels.

XTEMP identified every participant and every epoch that crossed the high-strain threshold of ≥ 7 — 100 % sensitivity, 87.5 % specificity, zero false negatives — a median 10 minutes ahead of the reference and never later. For a screening instrument this is the correct error profile: it warns early and it does not miss.

100%
Sensitivity to high-strain crossing

10 min
median lead over the reference

0.940
ICC for the Heat Strain Index

05 Impact

What the temperature result means: XTEMP closely tracked the direction and timing of the rectal response; the later separation likely reflects differences in thermal-compartment response under sustained heat, workload and elevated circulatory demand rather than random measurement variability. The rectum is a low-perfusion, high-inertia compartment, while the posterior orbit is perfused through the ophthalmic branch of the internal carotid. During a sustained monotonic rise the two need not agree in magnitude even when they agree in direction and timing — and the divergence here was phase-locked and proportional, with scatter unchanged. A simultaneous esophageal or ingestible reference would separate compartment physiology from device error.

What the heat-strain result enables: The Heat Strain Index is referenced to each participant's own baseline, individual offsets largely cancel — agreement rises from ICC 0.70 for raw temperature to 0.94 for strain, with complete detection of the high-strain crossing and lead time over the reference. Strain, not absolute degrees, is what an operator acts on, and it is already performing. Enrollment continues toward a target of 30 participants across both hot and warm conditions, which will support condition-specific limits of agreement, sex- and body-composition-stratified analysis, and prospective validation of a load-phase correction.

Path to a deployable capability: A measurement requiring only a camera — no contact, no consumable, no calibration, under one second — fits three operational needs: rapid screening and triage support at Role 1 where contact measurement is impractical or unsafe; readiness screening before and during heat-stress training, where risk is individual and environmental flags cannot resolve it; and recovery and return-to-duty monitoring, where the trend matters more than the absolute value.

06 Conclusion

In the first clinical validation of physics-based inverse ocular thermography during exertional heat stress, XTEMP reproduced the core-temperature trajectory with high fidelity and no detectable lag (within-subject r = 0.935; 8 of 8 participants above r = 0.70). Absolute point agreement is the current limitation, but the error is a systematic, phase-locked drift confined to load carriage (+0.35 °C) — a correctable engineering problem, not an irreducible noise floor. Against the endpoint that governs operational decisions, the system already performs.

Next steps Clinical: Complete enrollment to 30 participants across hot and warm conditions; add a second reference standard to resolve compartment lag; and extend beyond temperature toward broader contactless physiological monitoring.

Next steps Product: Continued hardware development in collaboration with Teledyne FLIR, advancing from the current tethered prototype toward embedded integration with tactical end-user devices and ATAK, and ultimately a wearable glasses form factor for hands-free, continuous assessment at the point of need.

REFERENCES
1. Maule AL, et al. MSMR. 2025;32(4).
2. Giersch GEW, et al. Am J Physiol Regul Integr Comp Physiol. 2023;324:R15-19.
3. Moran DS, Shitzer A, Pandolf KB. Am J Physiol. 1998;275:R129-34.
4. Tewolde S, Long KR. US Patent 11,971,311 B2. 2024.

LIMITS OF THE PRESENT EVIDENCE
Single site, single condition, small cohort. Validated for hyperthermia only; performance in hypothermia and in trauma populations is untested. Two participants were excluded a priori for failed baseline acquisition (|XTEMP - rectal| > 0.75 °C at rest).

DISCLAIMER
The views expressed are those of the authors and do not necessarily reflect the official policy or position of the Department of Defense, the Defense Health Agency, or the U.S. Government. Reference to commercial products does not constitute endorsement.