The Association of Deployment-Related Life and Family Concerns with Eating Pathology in Active-Duty Service Women

Angelina-Rose Thalheimer, BA,[1,2] Mary Quattlebaum, PhD,[1,2] Katherine Thompson, PhD,[1,2,3] Zoe Sinkford, BA,[1,2] Victoria Thomas, BA,[1,2] Autumn Mains, MSN, RN,[1,2] Stephanie Rioux, BSN, RN,[1,2] Mark Haigney, MD,[1,3] Natasha Schvey, PhD,[7] Marian Tanofsky-Kraff, PhD,[3,7] David Klein, MD, MPH,[4,5,6] & Jason Lavender, PhD[1,2,3]

[1]Military Cardiovascular Outcomes Research (MiCOR) Program, USU, Bethesda, MD, [2]The Metis Foundation, San Antonio, TX, [3]Department of Medicine, USU, Bethesda, MD, [4]Department of Family Medicine, David Grant Medical Center, Travis Air Force Base, CA, [5]Department of Family Medicine, USU, Bethesda, MD, [6]Department of Pediatrics, USU, Bethesda, MD, [7]Department of Medical and Clinical Psychology, USU, Bethesda, MD

The opinions and assertions expressed herein are those of the authors and do not reflect the official policy or position of the Uniformed Services University of the Health Sciences, the U.S. Air Force, or the Department of War.

This research was funded by the Military Women's Health Research Program.

INTRODUCTION

- 34% of active-duty Service women (ADSW) screen positive for an eating disorder, putting them 8.4 times greater risk than their male counterparts (Bodell et al., 2014)
- ADSW who deploy face unique stressors including separation from family and subsequent life and family concerns (Briggs et al., 2019)
- Aversive affective and cognitive states (e.g., anxiety, stress, or concerns about one's family) are robust risk factors of eating pathology (Groesz et al., 2012)
- Although deployments have been found to be related to greater eating pathology in military-affiliated youth and spouses (Neyland et al., 2020; Ray et al., 2025), no study has examined this association in ADSW
- The current study examined associations between deployment-related life and family concerns and eating pathology in ADSW

METHODS/MATERIALS

Participants

- ADSW ages 18-35 were recruited through the Military Health System Data Repository to complete a one-time online survey
- N=672 participants reported at least one deployment

Measures

- <u>Deployment-Related Life & Family Concerns</u>: Deployment Risk and Resilience Inventory-2 (DRRI-2) Section L: Life and Family Concerns (Vogt et al., 2013)
 - 15 items were summed to compute a total score
 - Sample item: "During deployment, I was concerned about the safety and well-being of my spouse or significant other"
- <u>Eating Pathology</u>: Eating Pathology Symptoms Inventory (EPSI; Forbush et al., 2013)
 - 5 severity scales assessing behaviors during the past month: binge eating, purging, dietary restriction, excessive exercise, and maladaptive muscle-building
 - Sample item (binge eating): "I ate a very large amount of food in a short period of time (e.g., within 2 hours)"

Statistical Analyses

- A series of multiple linear regression models (adjusted for relevant significant covariates) examined the associations of deployment-related life and family concerns with eating disorder behaviors

DEMOGRAPHICS

Age

M_{age} = 28.35 years

Ethnicity

16.1% Latina or Hispanic

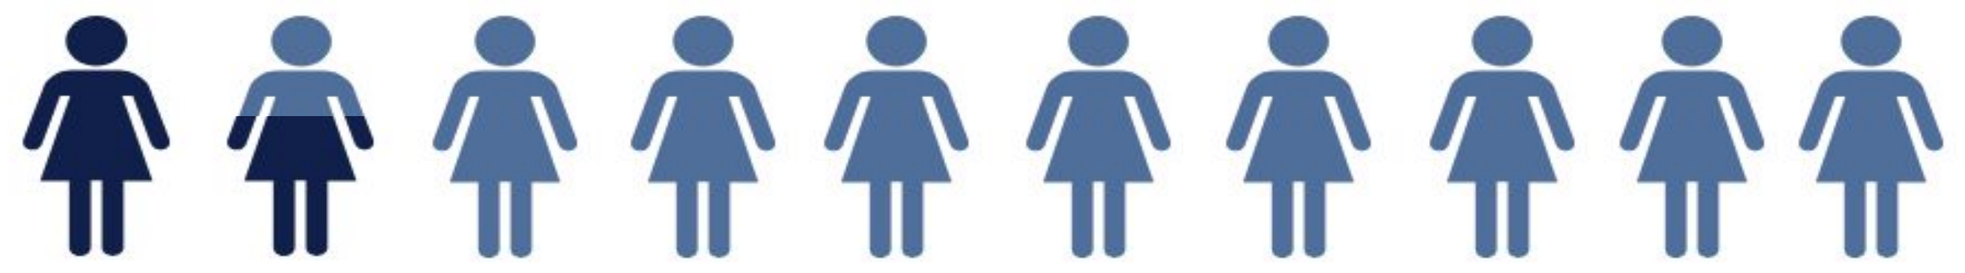

Race

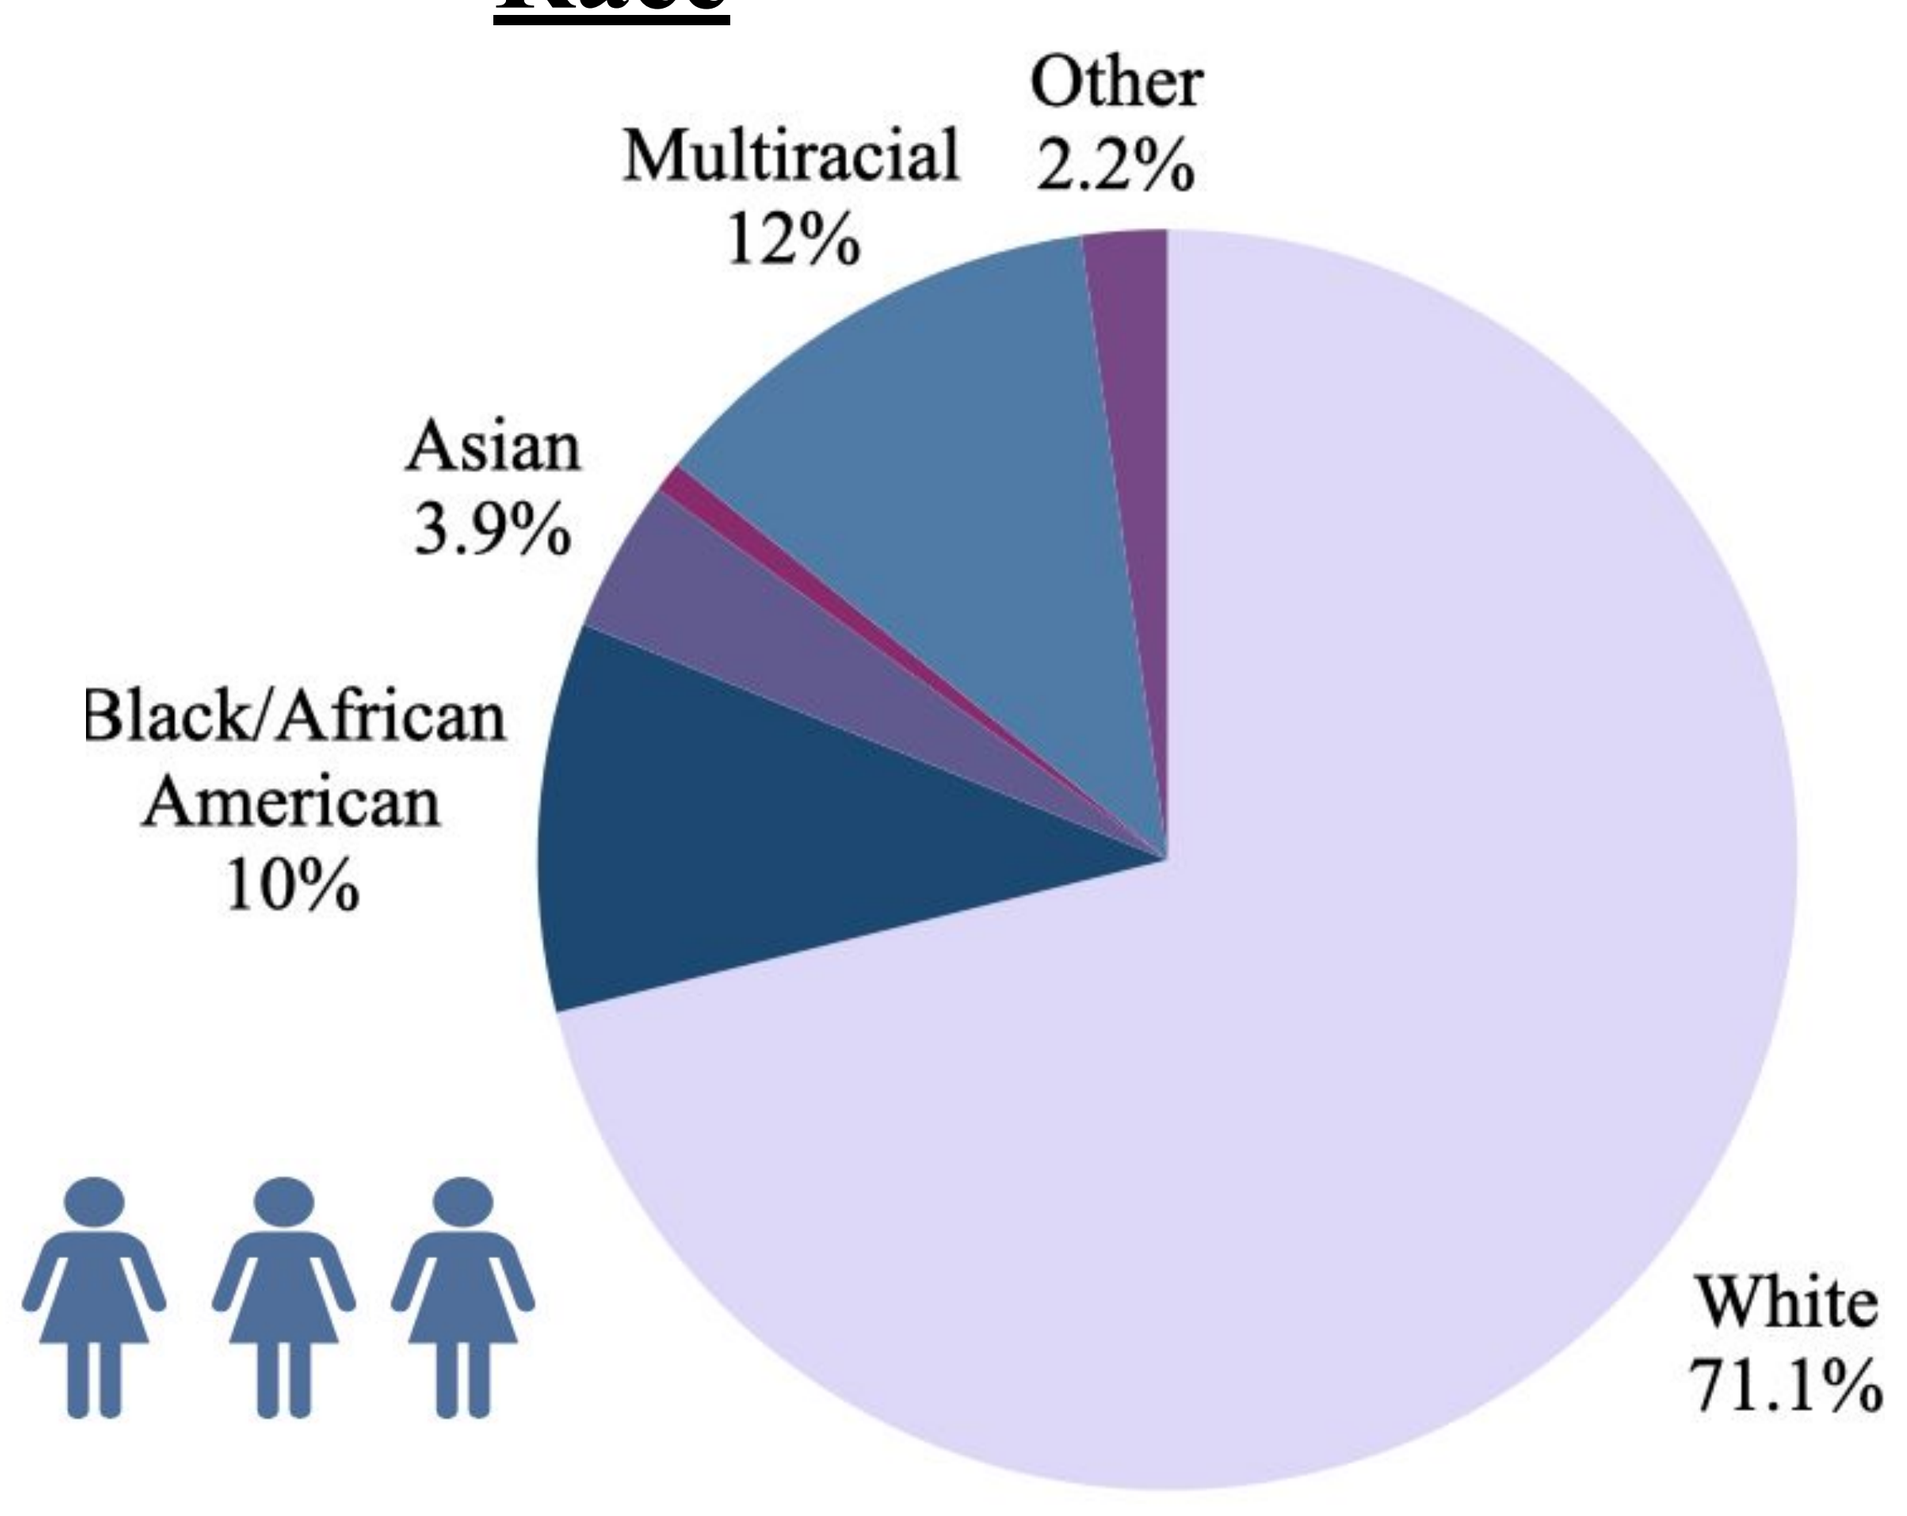
Other
2.2%
Multiracial
12%
Asian
3.9%
Black/African
American
10%
White
71.1%

Branch

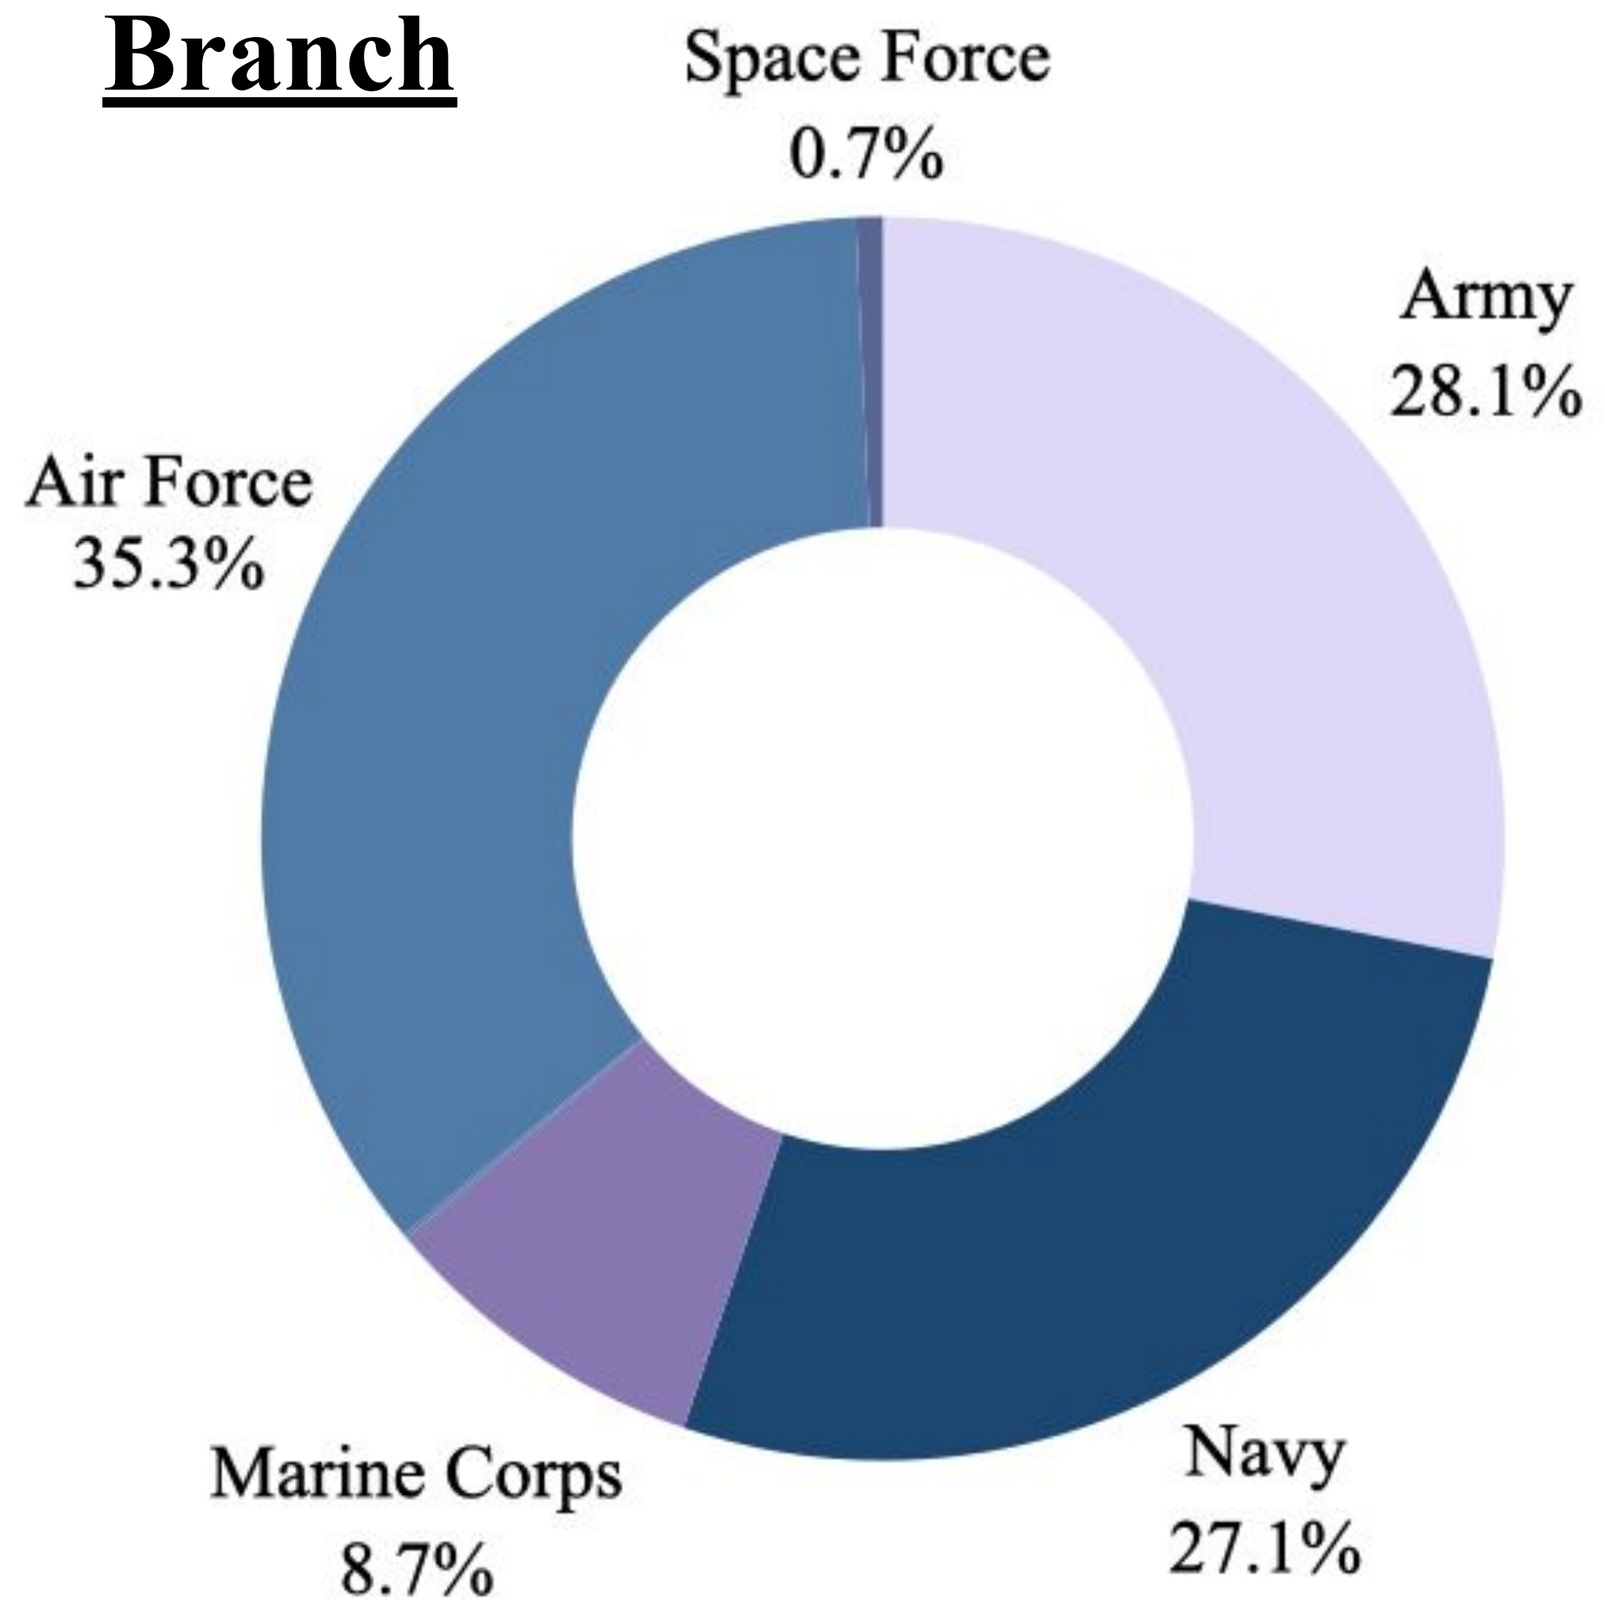
Space Force
0.7%
Army
28.1%
Air Force
35.3%
Marine Corps
8.7%
Navy
27.1%

Military Rank

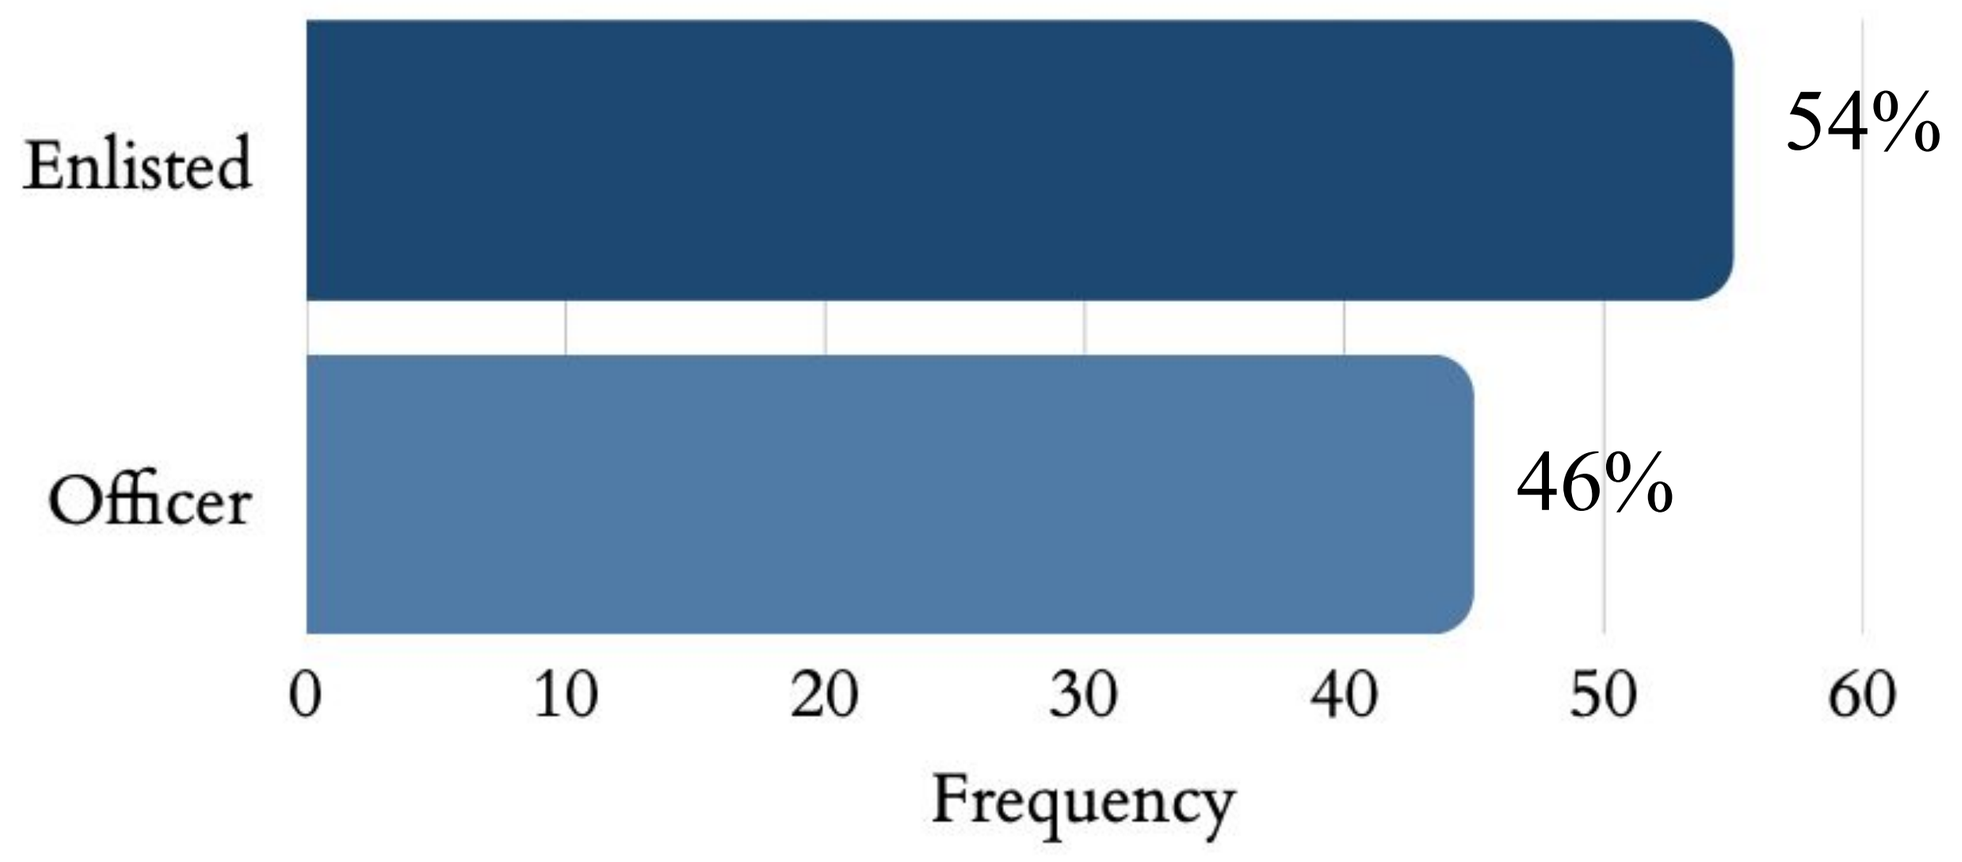
Enlisted
54%
Officer
46%
0
10
20
30
40
50
60
Frequency

RESULTS

Higher deployment-related life and family concerns were associated with more severe binge eating,[1] purging,[2] and dietary restriction,[3] but not excessive exercise (*p*=.17) or maladaptive muscle-building behaviors (*p*=.40)

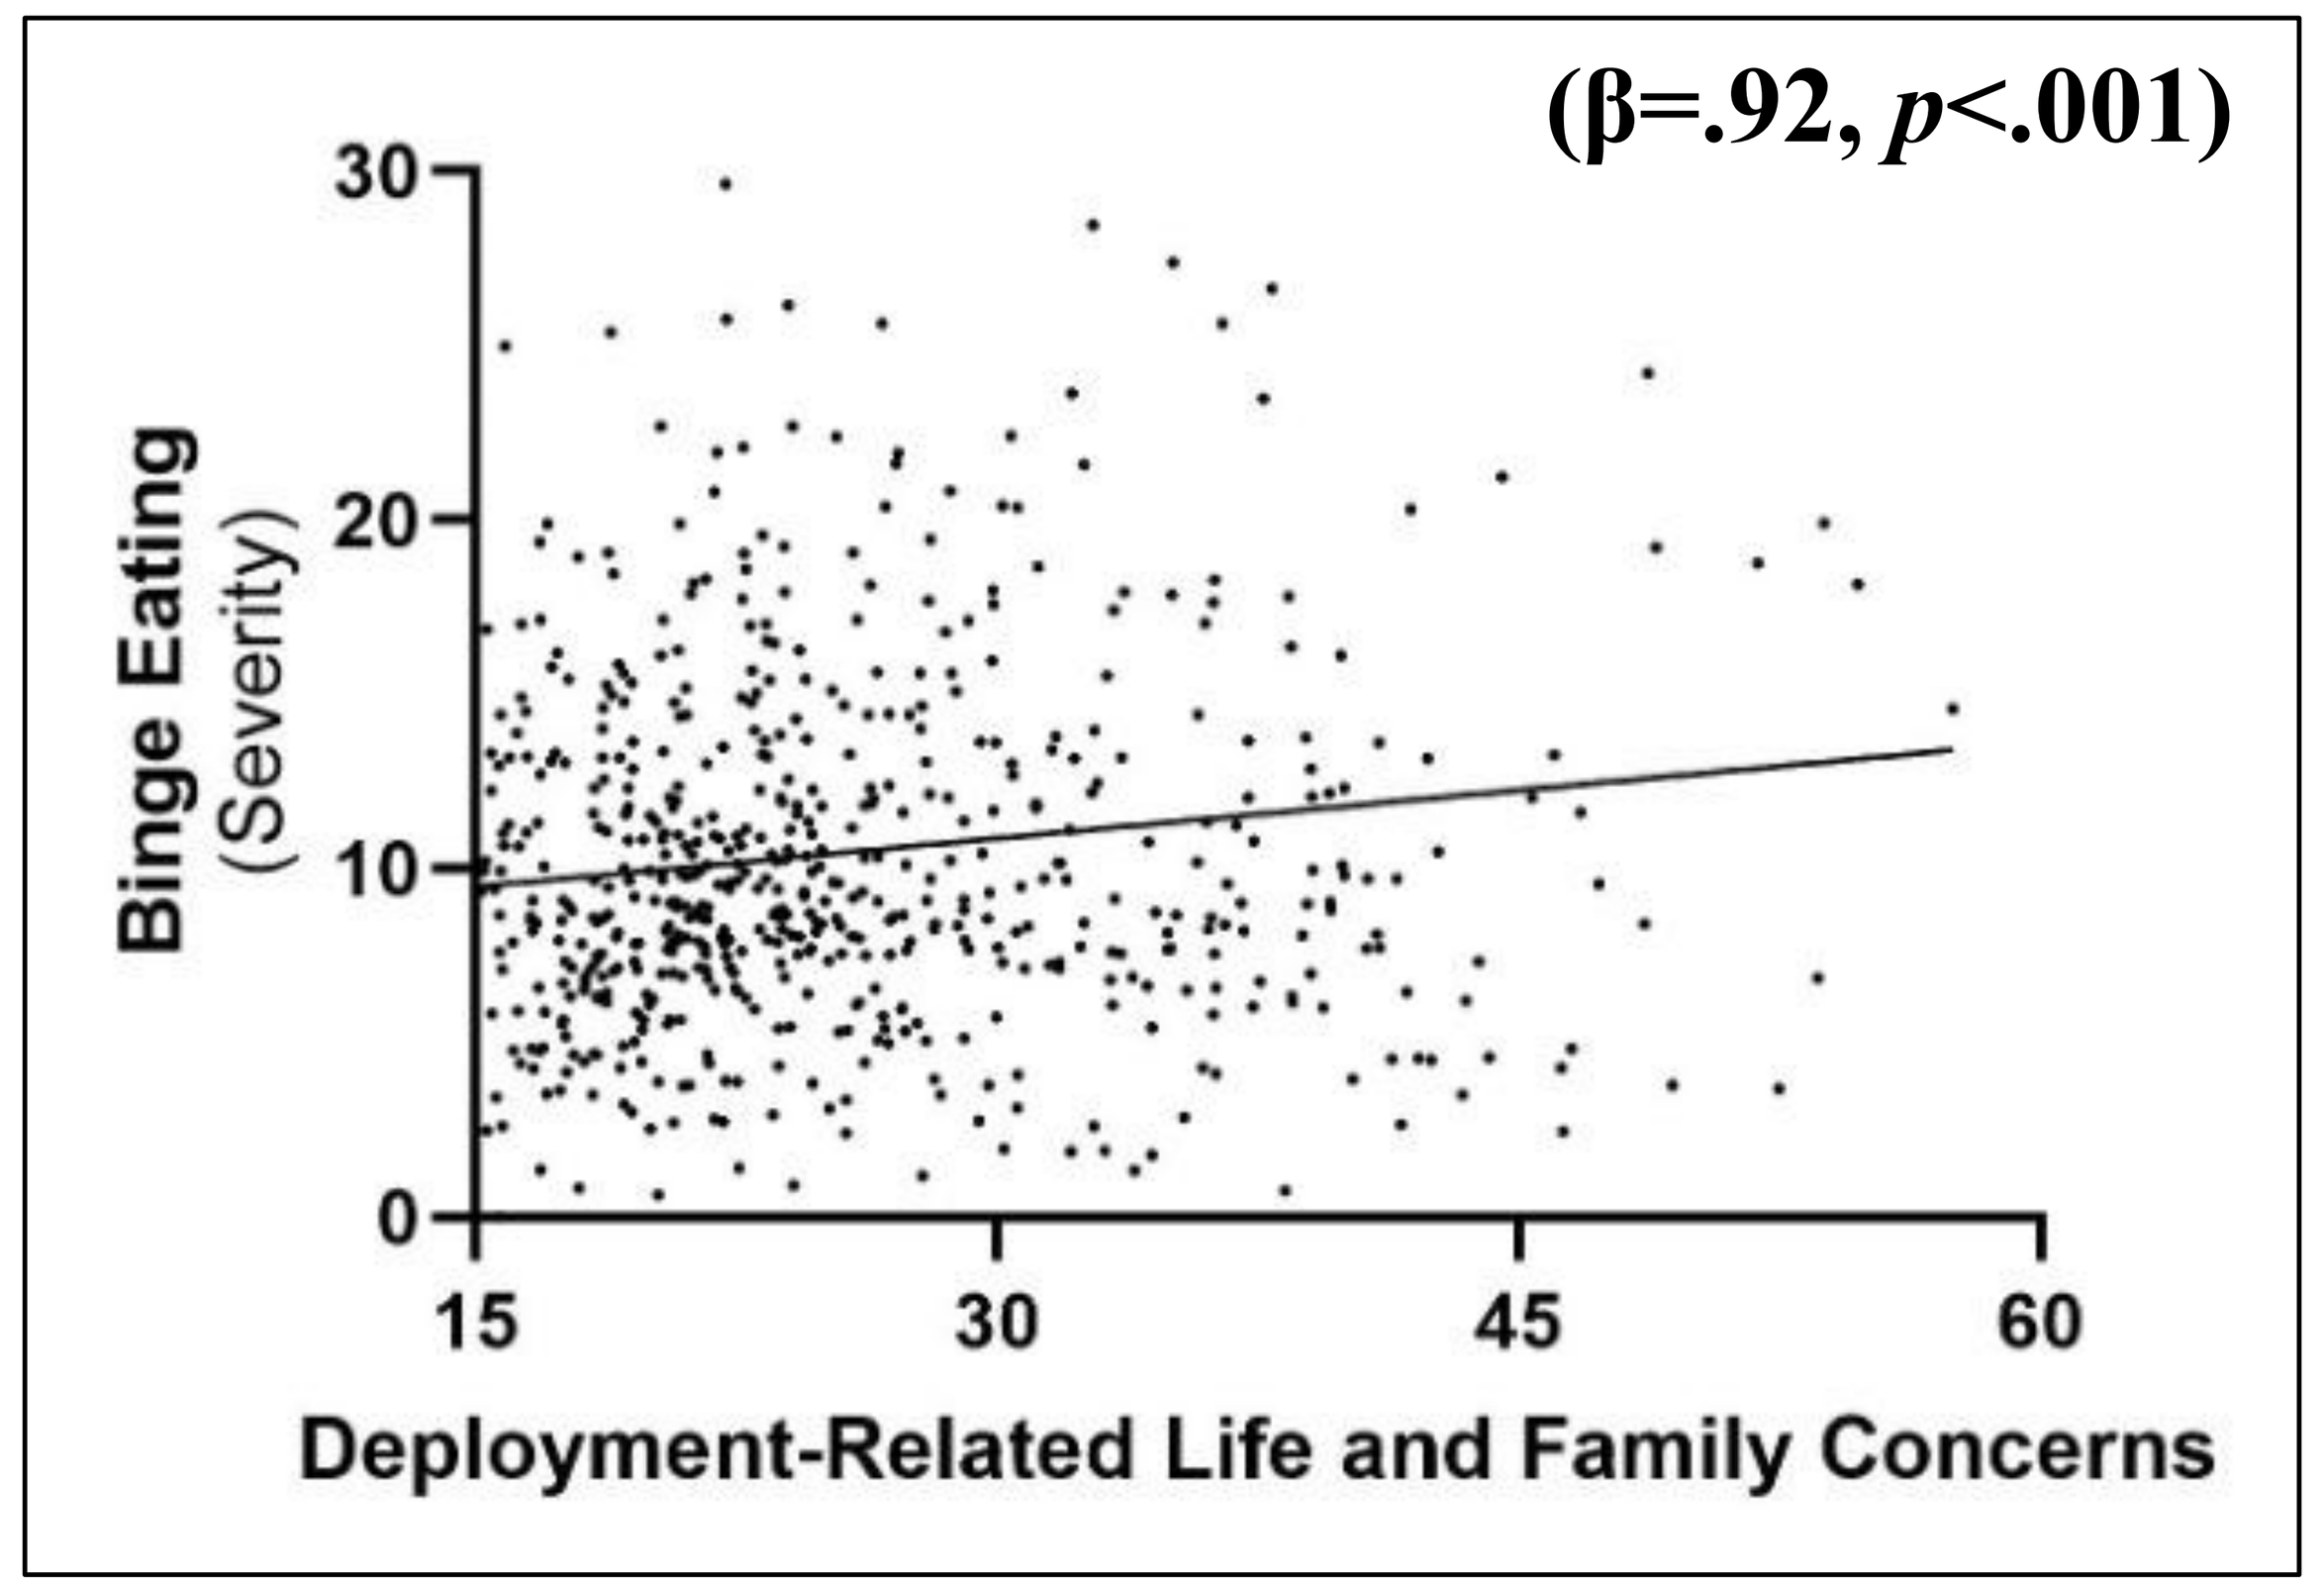
(β=.92, p<.001)
Binge Eating
(Severity)
0
10
20
30
15
30
45
60
Deployment-Related Life and Family Concerns

[1]Adjusted for BMI, stress, & race

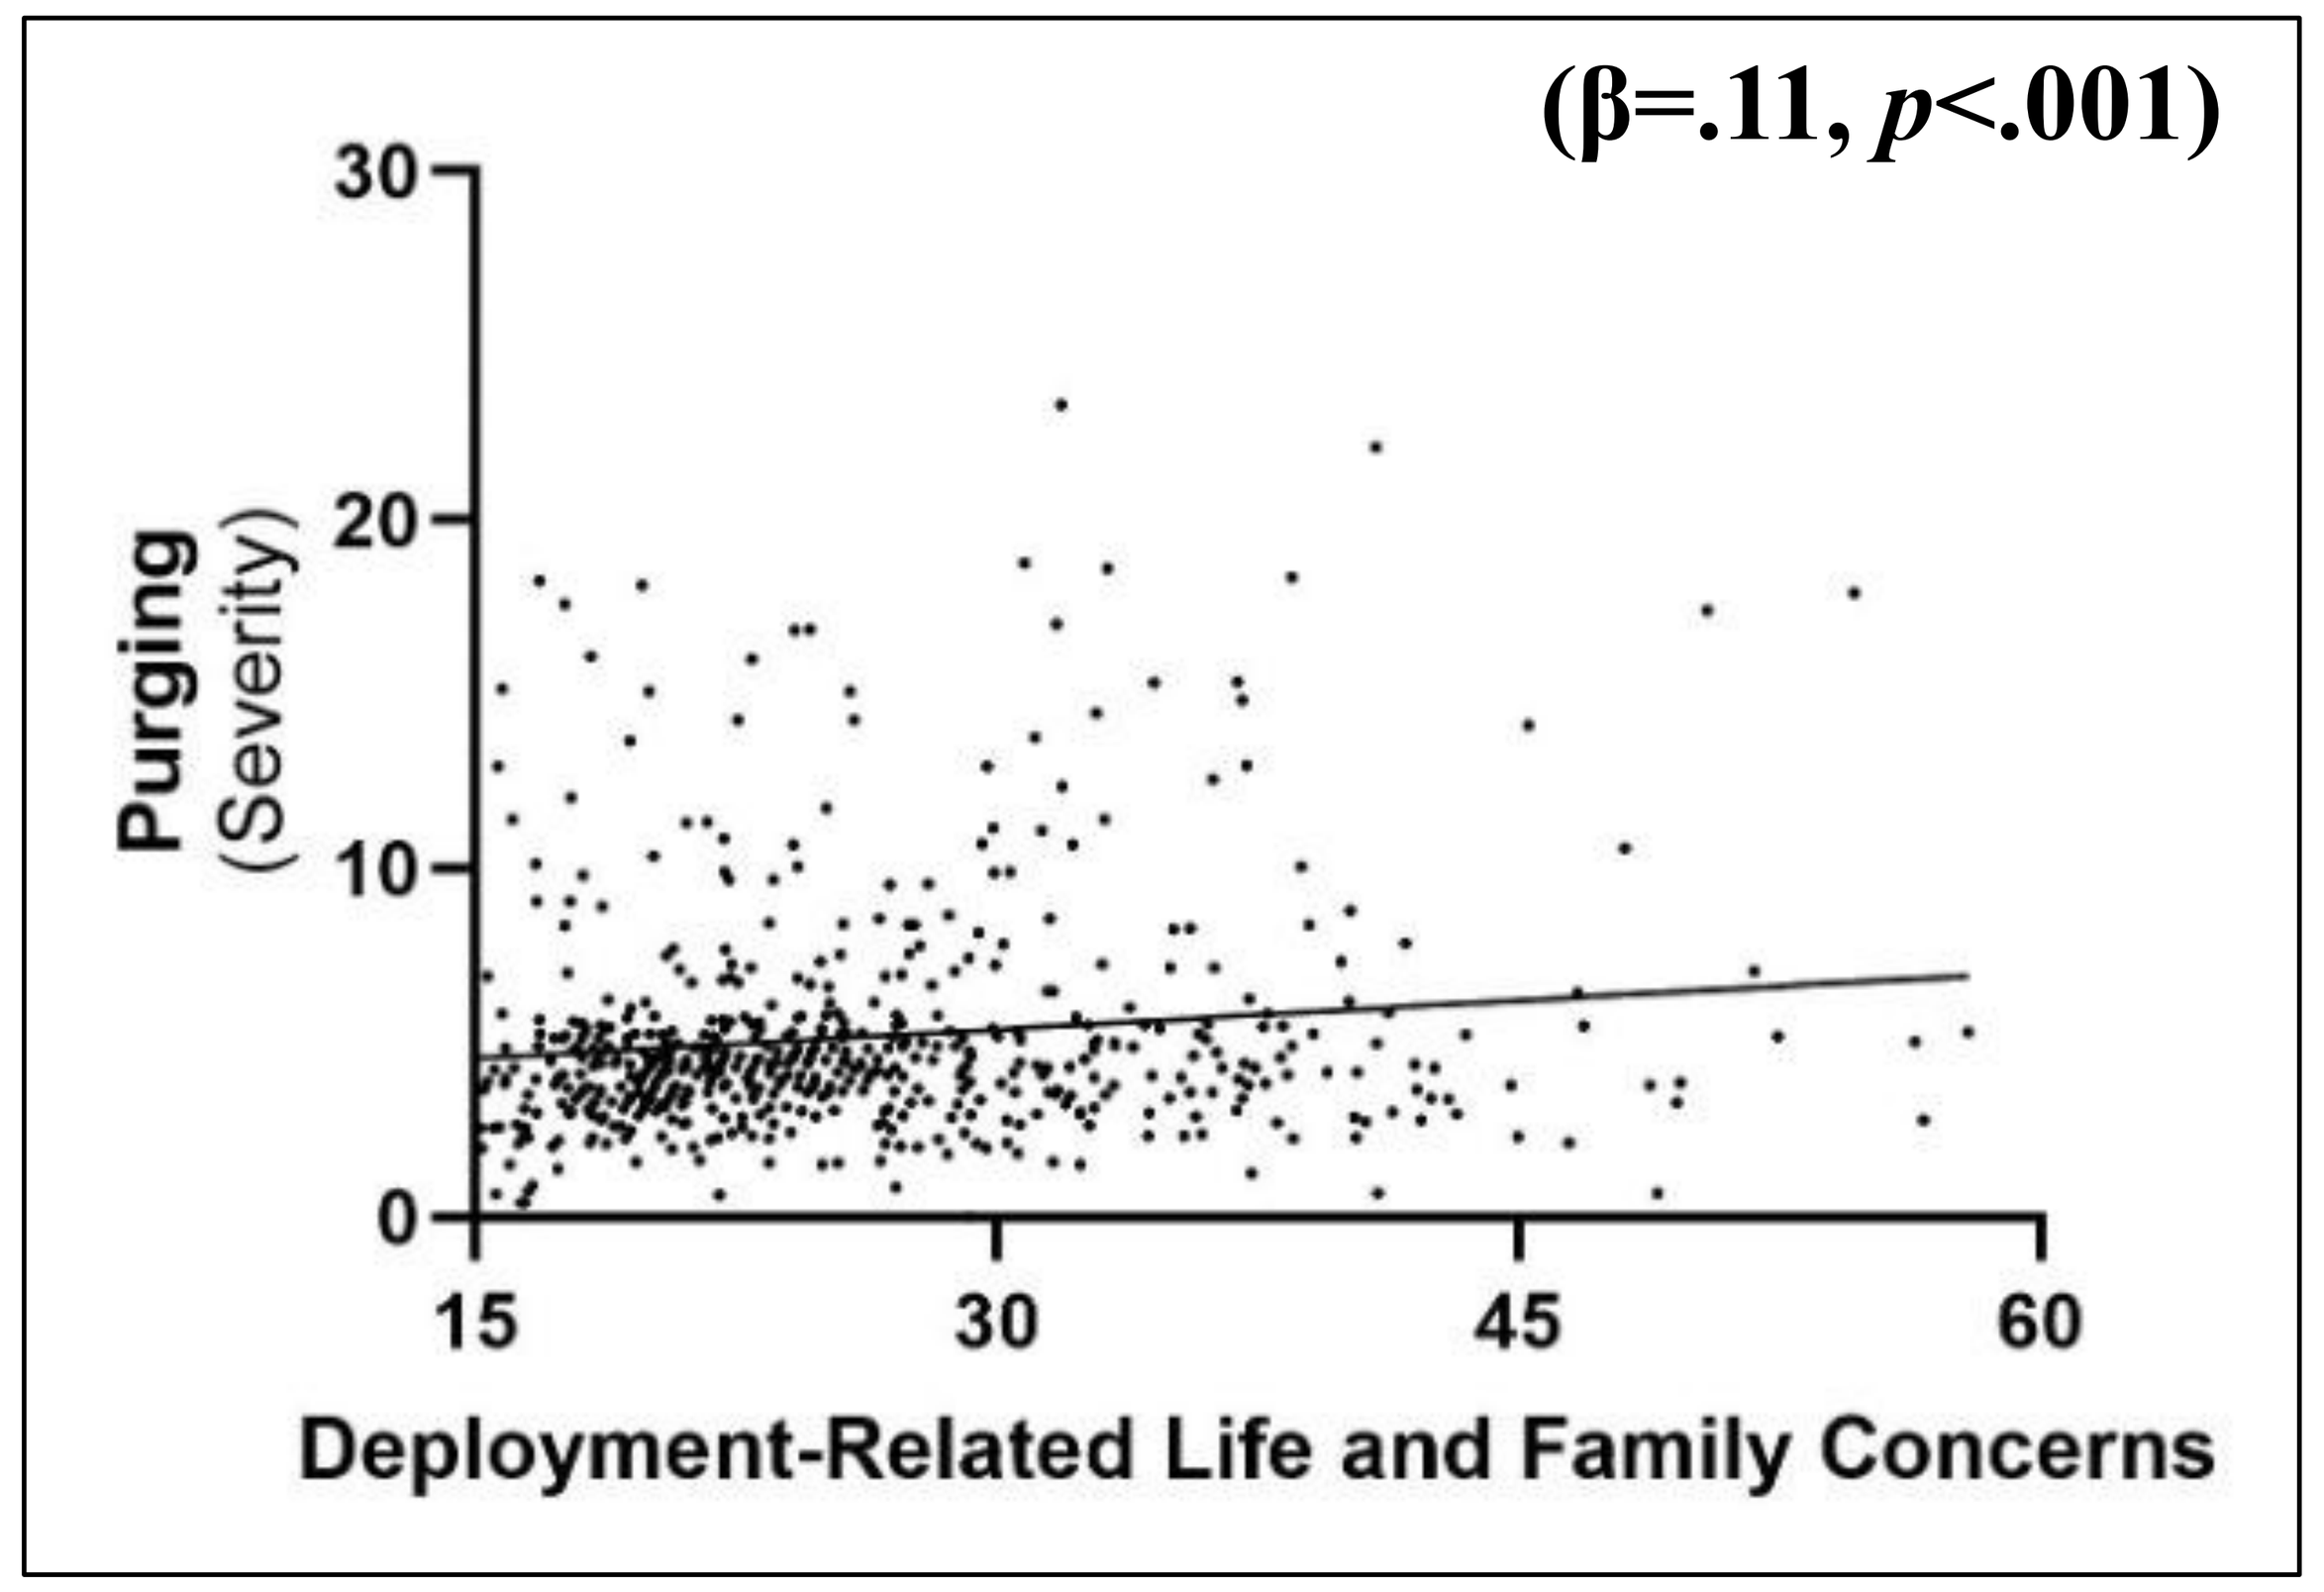
(β=.11, p<.001)
Purging
(Severity)
0
10
20
30
15
30
45
60
Deployment-Related Life and Family Concerns

[2]Adjusted for BMI, stress, marital status, & rank

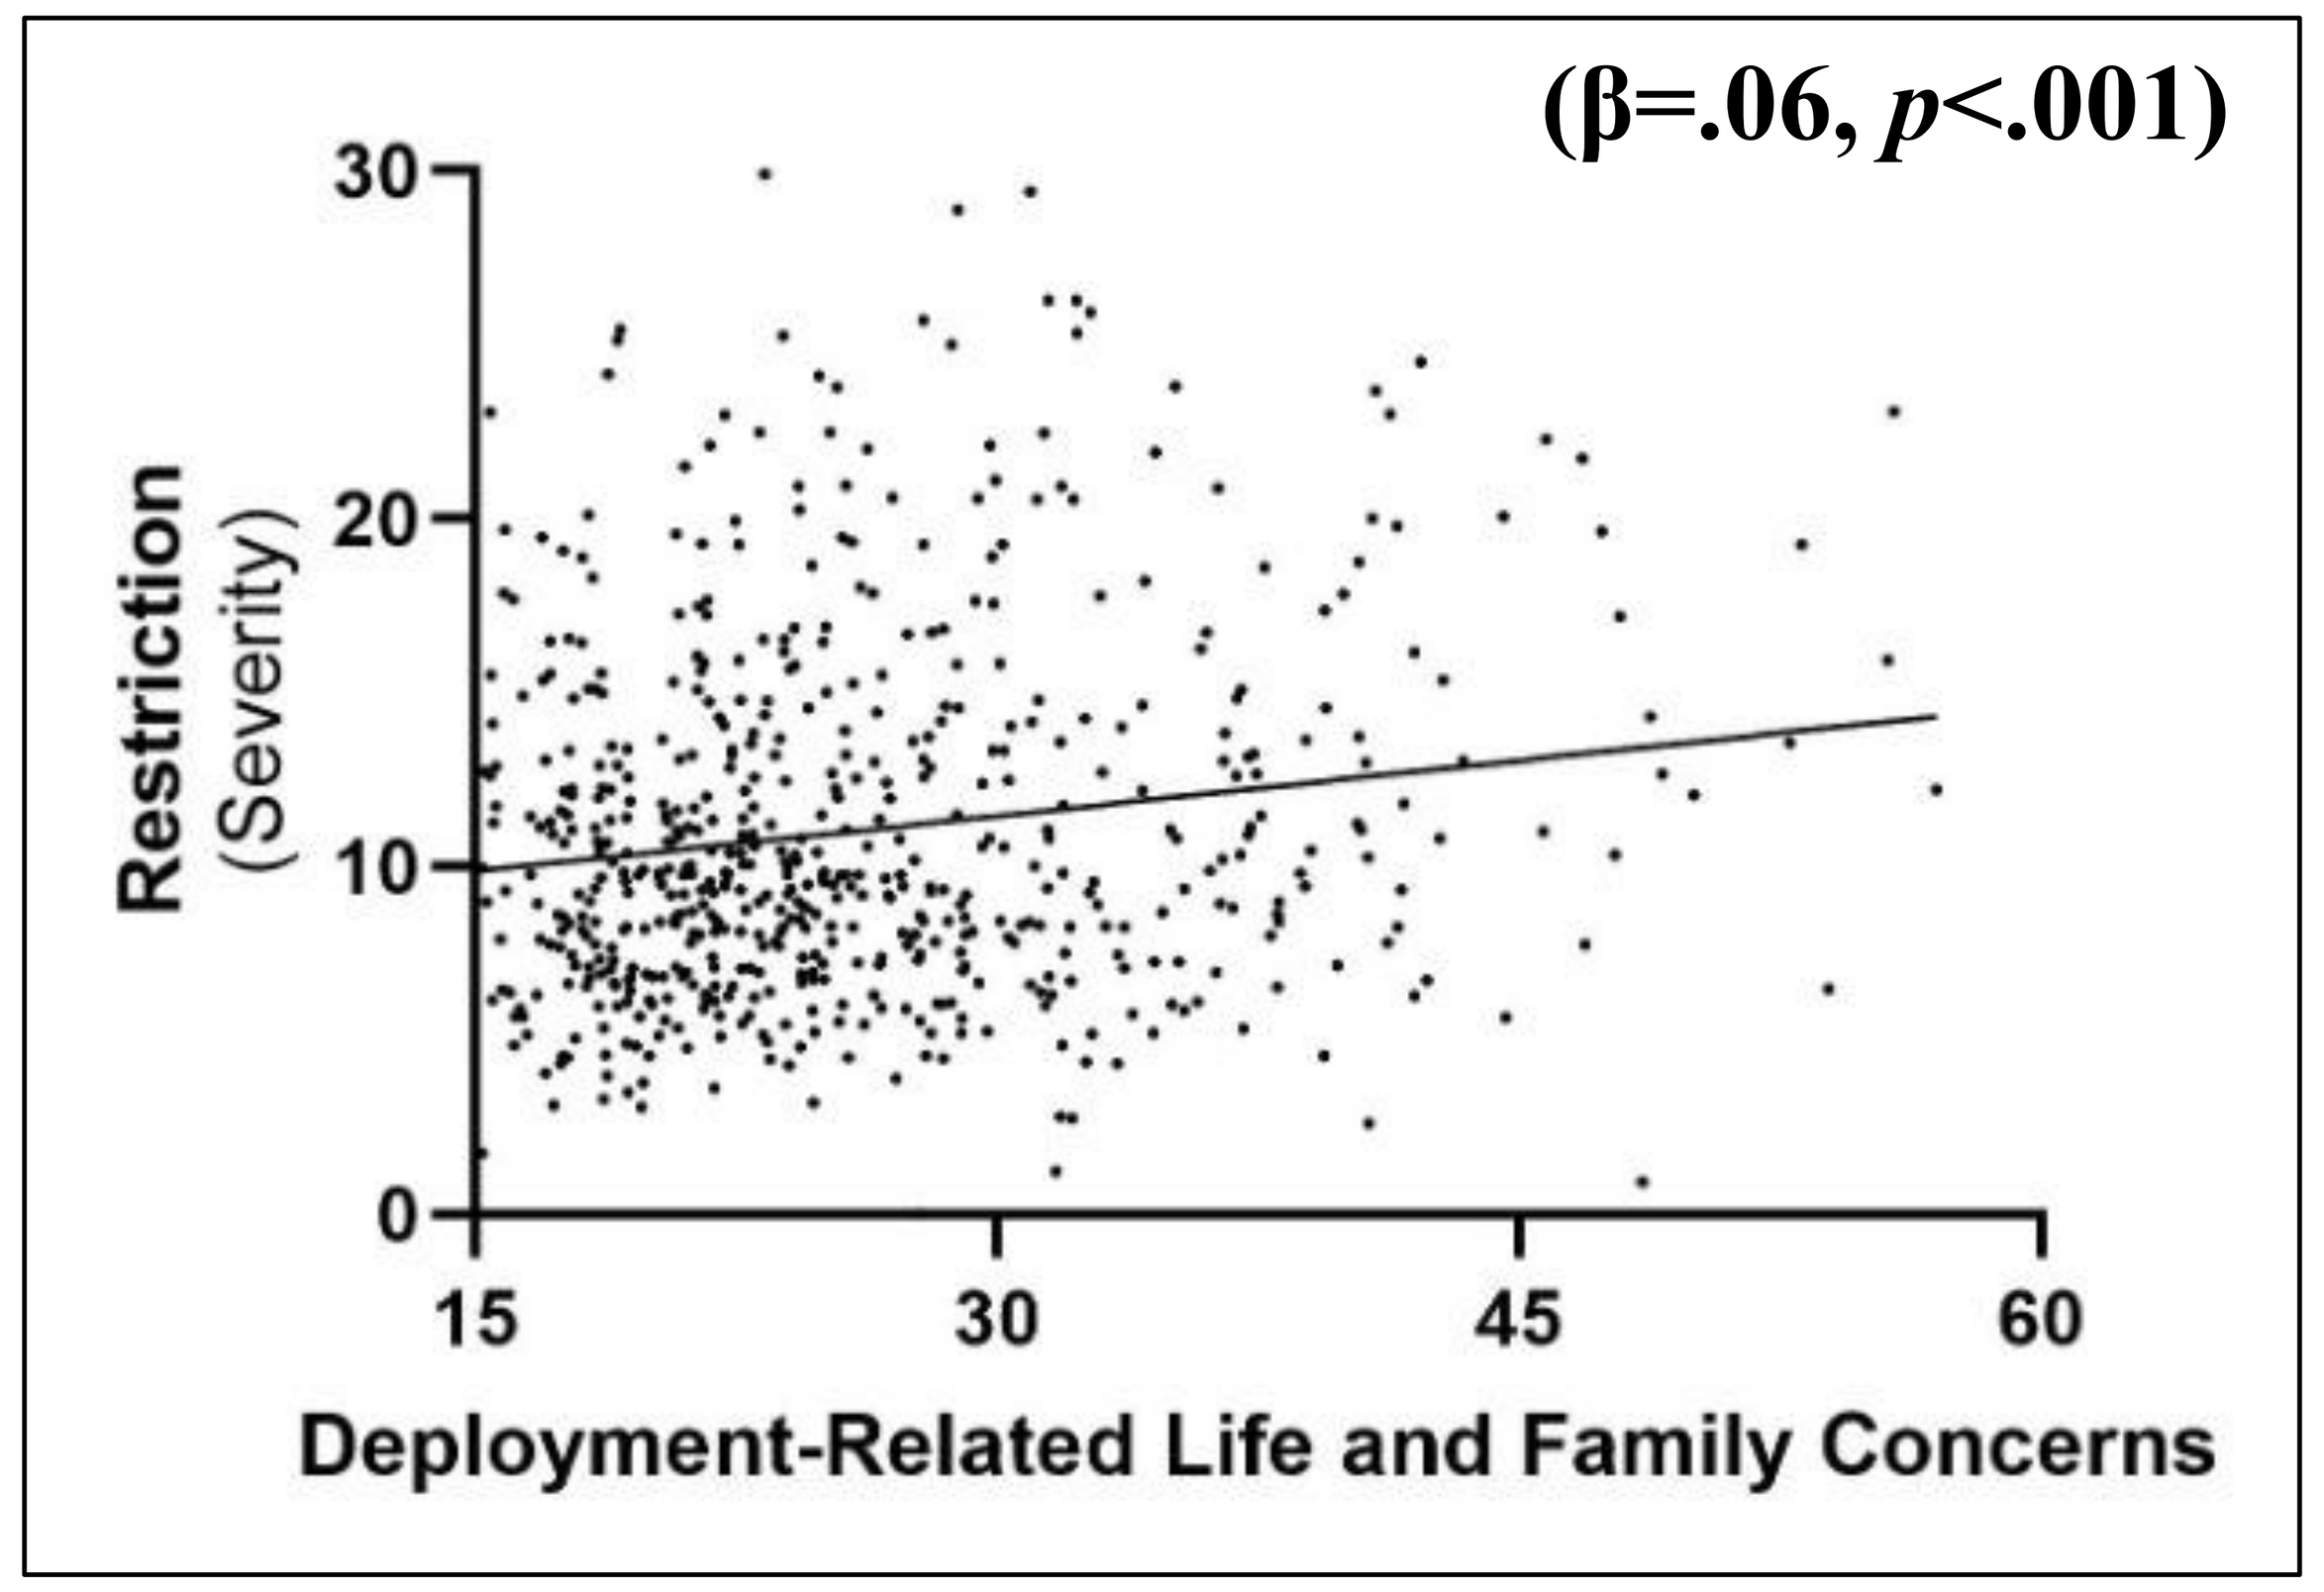
(β=.06, p<.001)
Restriction
(Severity)
0
10
20
30
15
30
45
60
Deployment-Related Life and Family Concerns

[3]Adjusted for BMI, stress, marital status, & rank

CONCLUSIONS

- Although cross-sectional, findings are consistent with theoretical models that posit binge eating, restriction, and purging may function as maladaptive coping strategies in response to aversive internal states and functioning (Freeman & Gil, 2004; Sassaroli et al., 2015)
- Eating pathology can compromise ADSW health, exacerbating healthcare costs and reducing military readiness (Himmerich et al., 2025; Mariotti, 2015)
 - Associated with fatigue, cognitive impairment, musculoskeletal injuries, cardiometabolic dysfunction, co-occurring mental health concerns
- Teaching behavioral strategies (e.g., stress coping) pre-deployment to mitigate impacts of life and family concerns during deployment may reduce eating pathology risk among ADSW (Jimenez-Padilla et al., 2025)

<u>Author Contact</u>: angelinarose.thalheimer.ctr@usuhs.edu